Design and evaluation of mRNA translation inhibitors for use as antibody-drug conjugate payloads

Mark E. Petersen, Jodi Wong, Samir Das, Vidhi Khanna, Kurt Stahl, Micheal G. Brant, Truman Hirkala-Schaefer, Renee Duan, Jesse Leblanc, Victoria K. Harman-McKenna, Manuel Lasalle, Graham A. E. Garnett, Rehan Higgins, Catalina Suarez, Kaylee J. Wu, Linglan Fu, Germanna Righetto, Devika Sim, Adele Chan, Allysha Bissessur, Ambroise Wu, Araba Sagoe-Wagner, Luying Yang, Andrea Hernandez Rojas, Dunja Urosev, Sam Lawn, Vincent Fung, Stuart D. Barnscher, Raffaele Colombo, Jamie R. Rich

Zymeworks Inc., Vancouver, BC, Canada

Abstract #2062

Introduction

Eukaryotic initiation factor 4A (eIF4A) mediates recruitment of ribosomes to mRNA and catalyzes the unwinding of mRNA secondary structures, thereby facilitating ribosome scanning and translation initiation.[1] Inhibitors of eIF4A (e.g., silvestrol, zotatifin, and other rocaglates) block translation of select mRNAs that contain a high degree of secondary structure (e.g. mRNAs encoding MYC, KRAS, and CDK4/6, among others), resulting in inhibition of cell proliferation and induction of apoptosis.[2] Despite limited efficacy observed for zotatifin in a phase I clinical trial,[3] antibody conjugation of a novel eIF4A payload offers the possibility of enhanced exposure and improved tolerability, which could lead to more robust responses. Furthermore, eIF4Ai antibody-drug conjugates (ADCs) would constitute a novel mechanism with the potential to address payload-specific resistance mechanisms that have been reported following progression on ADCs with current payloads.

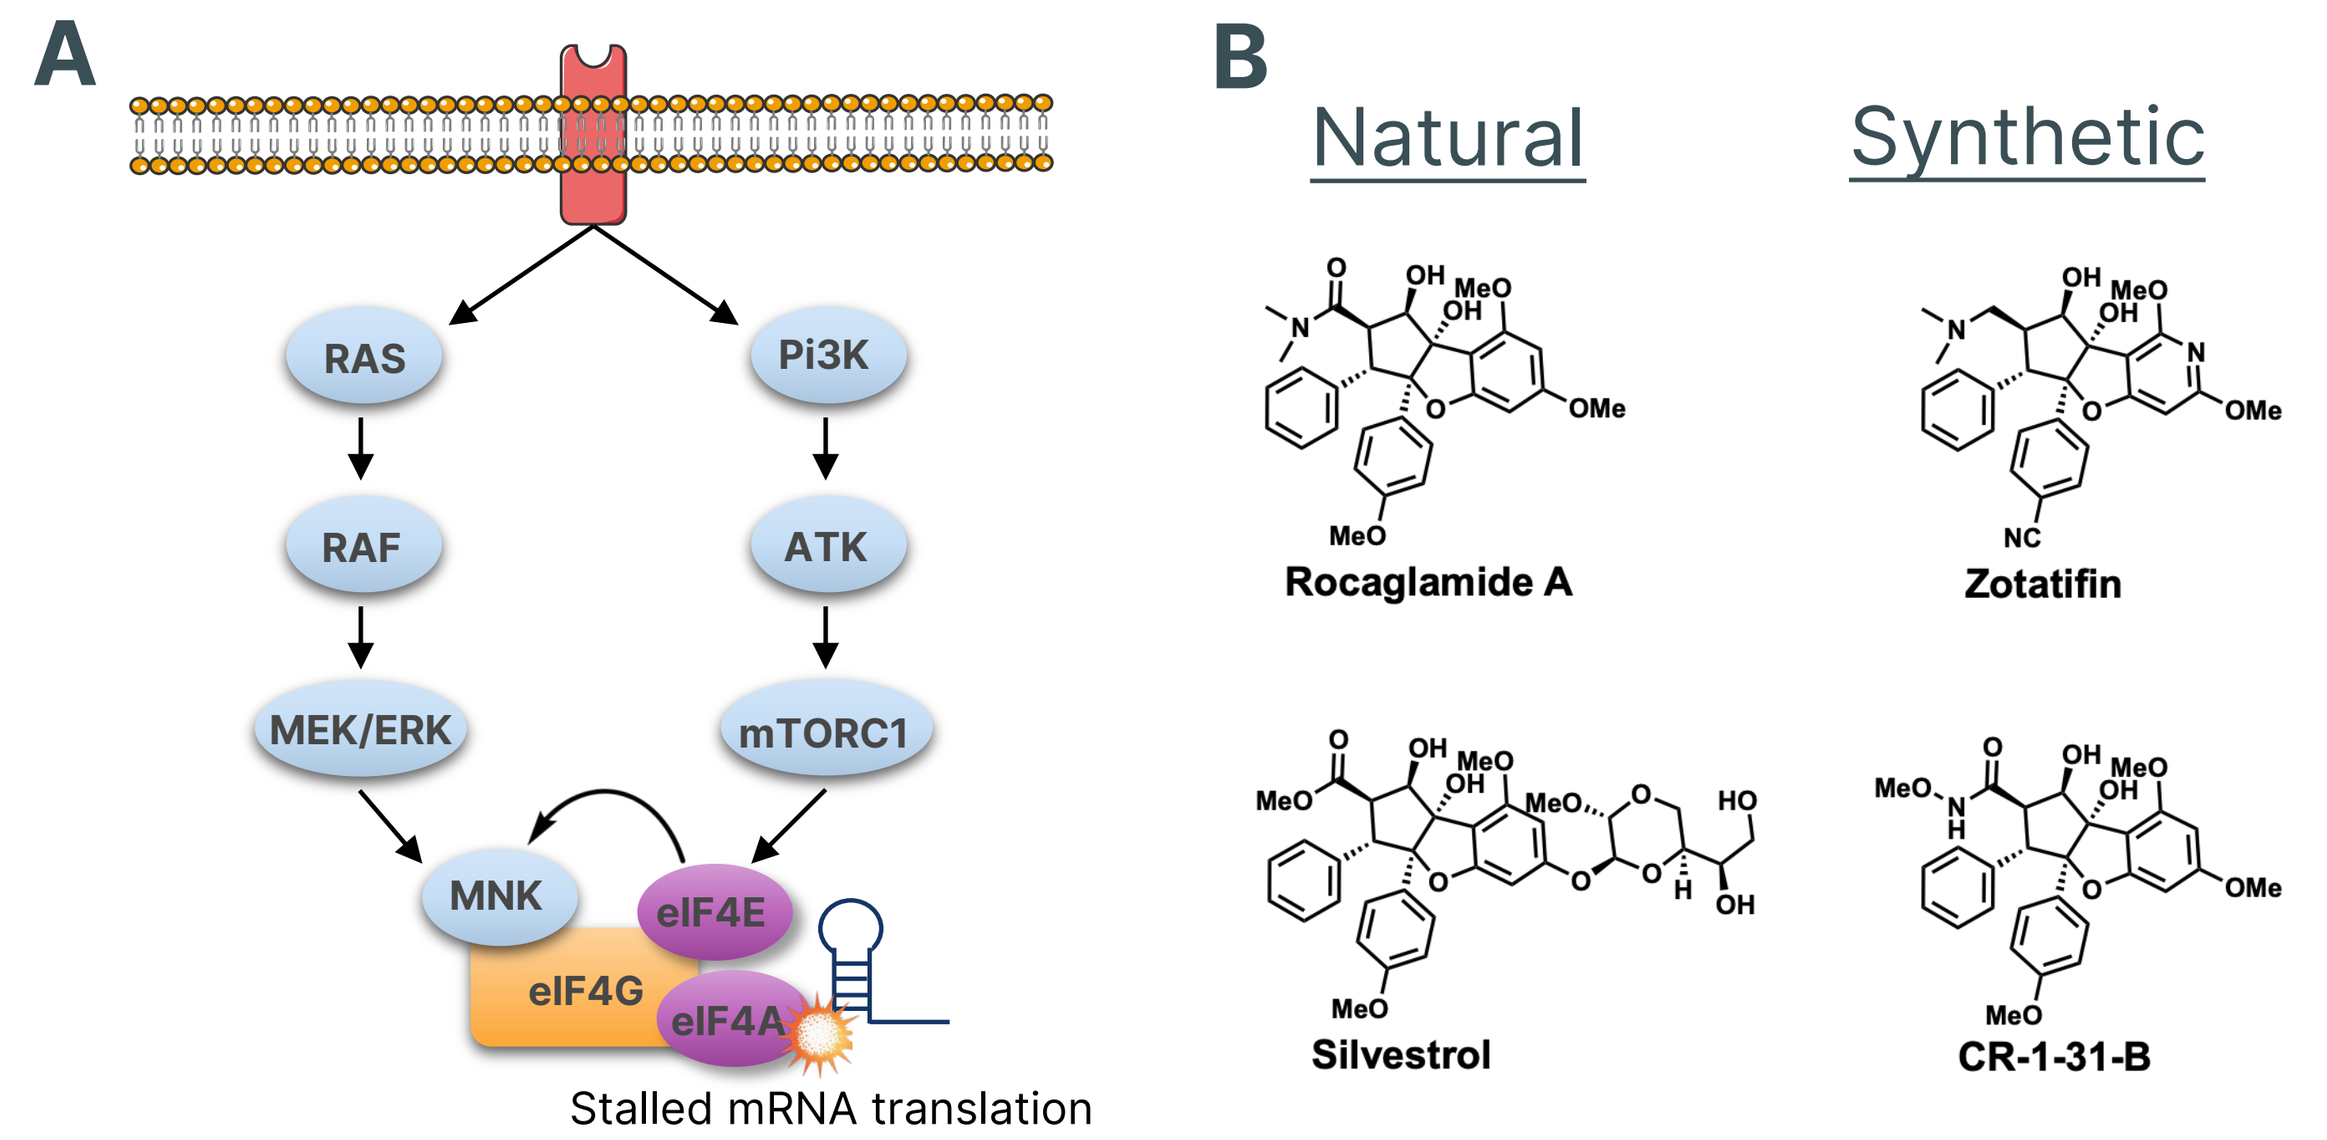
A
RAS
Pi3K
RAF
ATK
MEK/ERK
mTORC1
MNK
eIF4E
eIF4G
eIF4A
Stalled mRNA translation
B
Natural
Synthetic
Rocaglamide A
Zotatifin
Silvestrol
CR-1-31-B

Figure 1: **A.** The eIF4F complex is essential for cap dependent mRNA translation and consists of three components: the scaffolding protein eIF4G, the mRNA 5′ cap binding protein eIF4E, and the RNA helicase eIF4A which mediates the unwinding of secondary structures. **B.** The natural products Rocaglamide and Silvestrol as well as the synthetic analogs Zotatifin and CR-1-31-B bind to and form a ternary complex with eIF4A and mRNA. Transcripts that contain a high degree of secondary structure are more sensitive to downregulation by eIF4A inhibitors.

Structural diversification identifies payloads with broad range of potency, efficacy, and tolerability

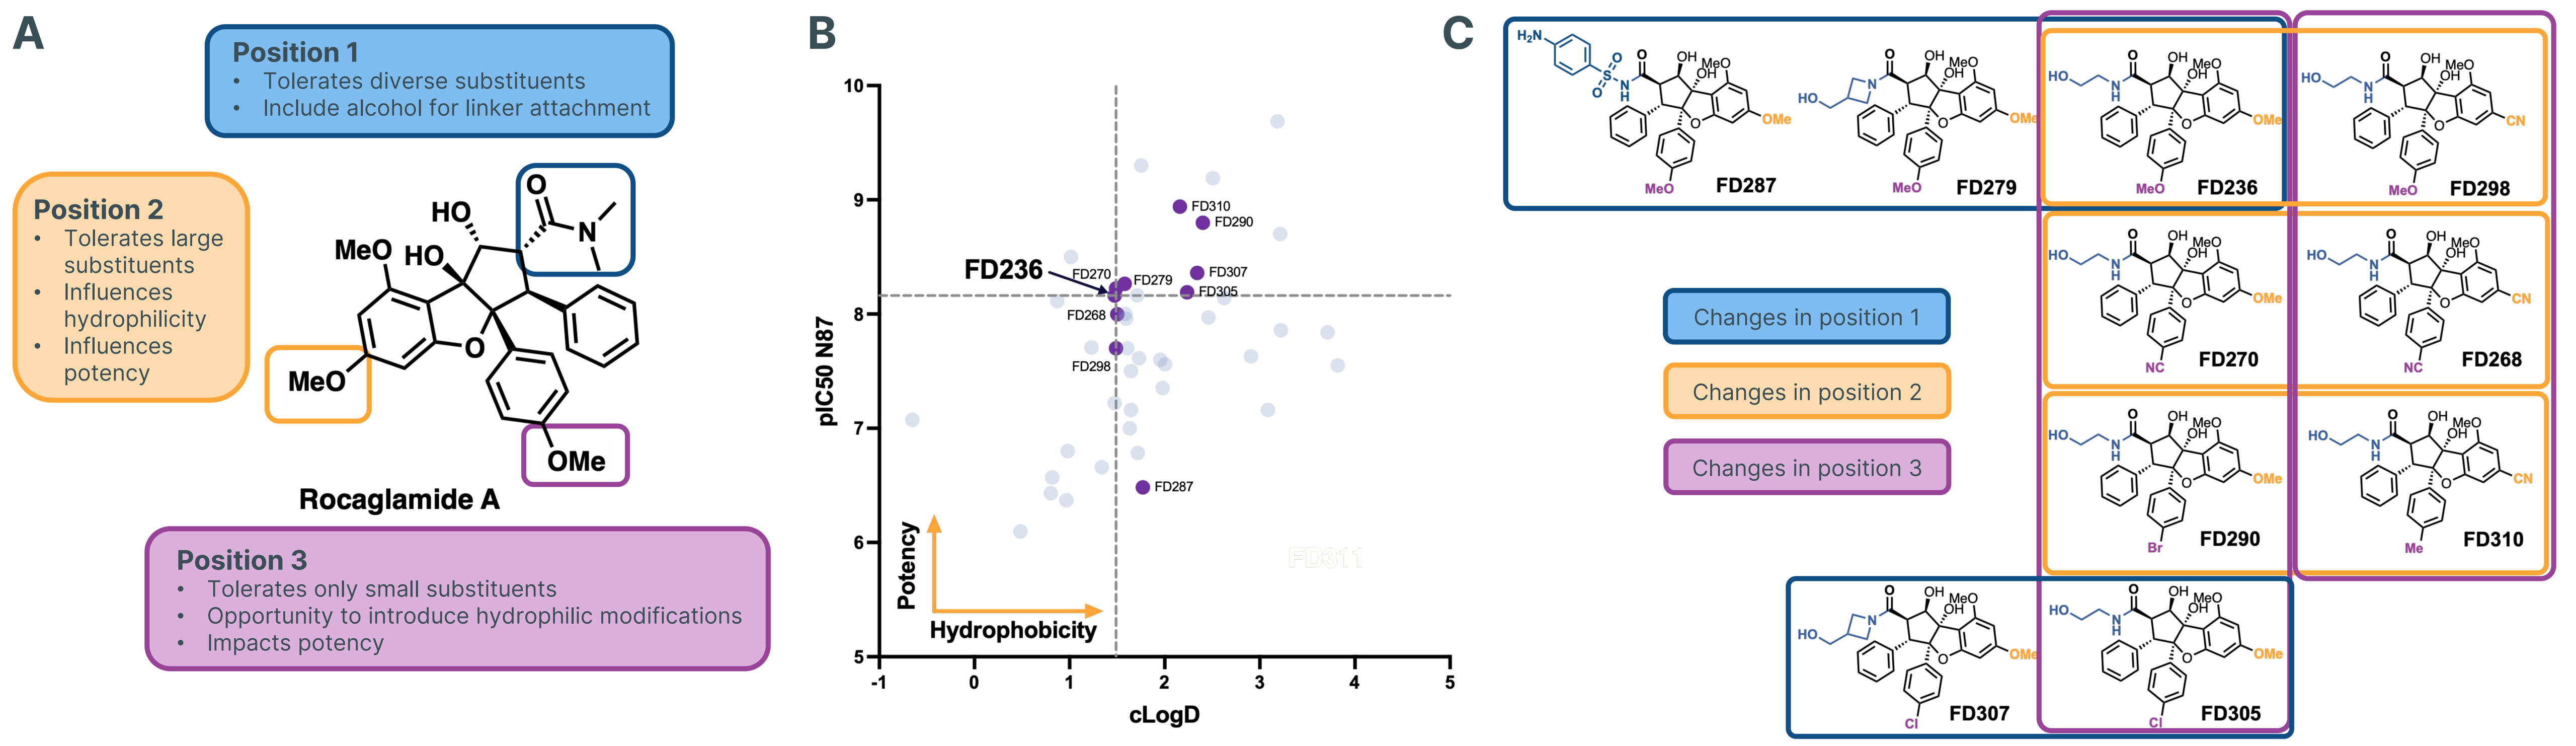
A
Position 1
Tolerates diverse substituents
Include alcohol for linker attachment
Position 2
Tolerates large substituents
Influences hydrophilicity
Influences potency
Position 3
Tolerates only small substituents
Opportunity to introduce hydrophilic modifications
Impacts potency
Rocaglamide A
B
FD236
pIC50 N87
cLogD
Potency
Hydrophobicity
C
Changes in position 1
Changes in position 2
Changes in position 3
FD287
FD279
FD236
FD298
FD270
FD268
FD290
FD310
FD307
FD305

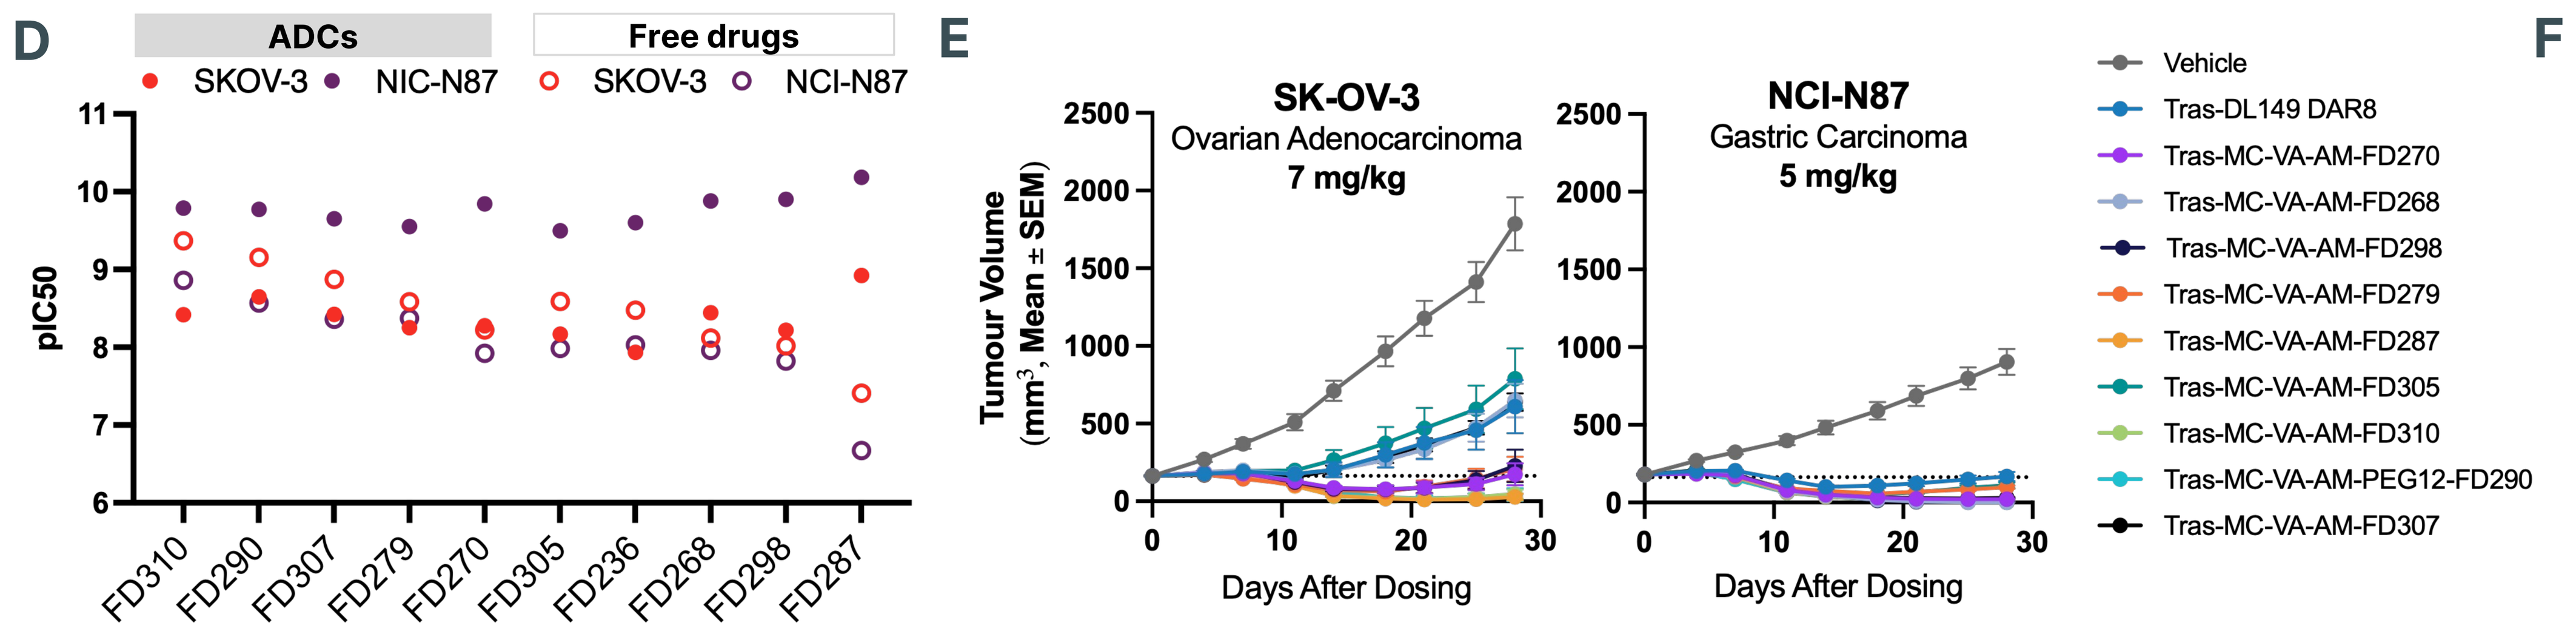
D
ADCs
Free drugs
SKOV-3
NIC-N87
SKOV-3
NCI-N87
pIC50
E
SK-OV-3
Ovarian Adenocarcinoma
7 mg/kg
NCI-N87
Gastric Carcinoma
5 mg/kg
Tumour Volume (mm³, Mean ± SEM)
Days After Dosing
Vehicle
Tras-DL149 DAR8
Tras-MC-VA-AM-FD270
Tras-MC-VA-AM-FD268
Tras-MC-VA-AM-FD298
Tras-MC-VA-AM-FD279
Tras-MC-VA-AM-FD287
Tras-MC-VA-AM-FD305
Tras-MC-VA-AM-FD310
Tras-MC-VA-AM-PEG12-FD290
Tras-MC-VA-AM-FD307

F

Tolerability of trastuzumab ADCs in non-tumor bearing BALB/c mice		
Payload	Dose, mg/kg	Tolerated
FD236	15	✓
FD236	30	✓
FD236	50	X
FD310	15	X
FD310	30	X
FD270	15	✓
FD270	30	X
FD279	15	✓
FD279	30	✓
FD298	15	✓
FD298	30	X
FD307	15	✓
FD307	30	✓

Figure 3. (A) Identified structure activity of relationships of rocaglamides with specific focus on three positions. **(B)** Over 40 linkable eIF4Ai that span a range of potency and hydrophilicity were synthesized. **(C)** Selection of 10 payloads that represent structural modification at each of the three positions and span a range of potency. Colored boxes represent changes at a specific position within a molecular series. **(D)** pIC50 of trastuzumab-ADCs compared to free drugs in 3D spheroids of NCI-N87 and SK-OV-3 cells. **(E)** In vivo activity of trastuzumab-eIF4A ADCs at 7 mg/kg and 5 mg/kg in SK-OV-3 and NCI-N87 cell line derived xenografts. **(F)** Tolerability of select ADCs in non-tumor bearing BALB/c mice. ADCs carrying payloads FD236, 279, and 307 were tolerated up to 30 mg/kg. ADCs carrying payloads FD298, and 270 were tolerated up 15 mg/kg and an ADC with FD310 was not tolerated at either dose.

Lead drug-linkers show broad activity, but toxicology findings indicate a narrow preclinical therapeutic index

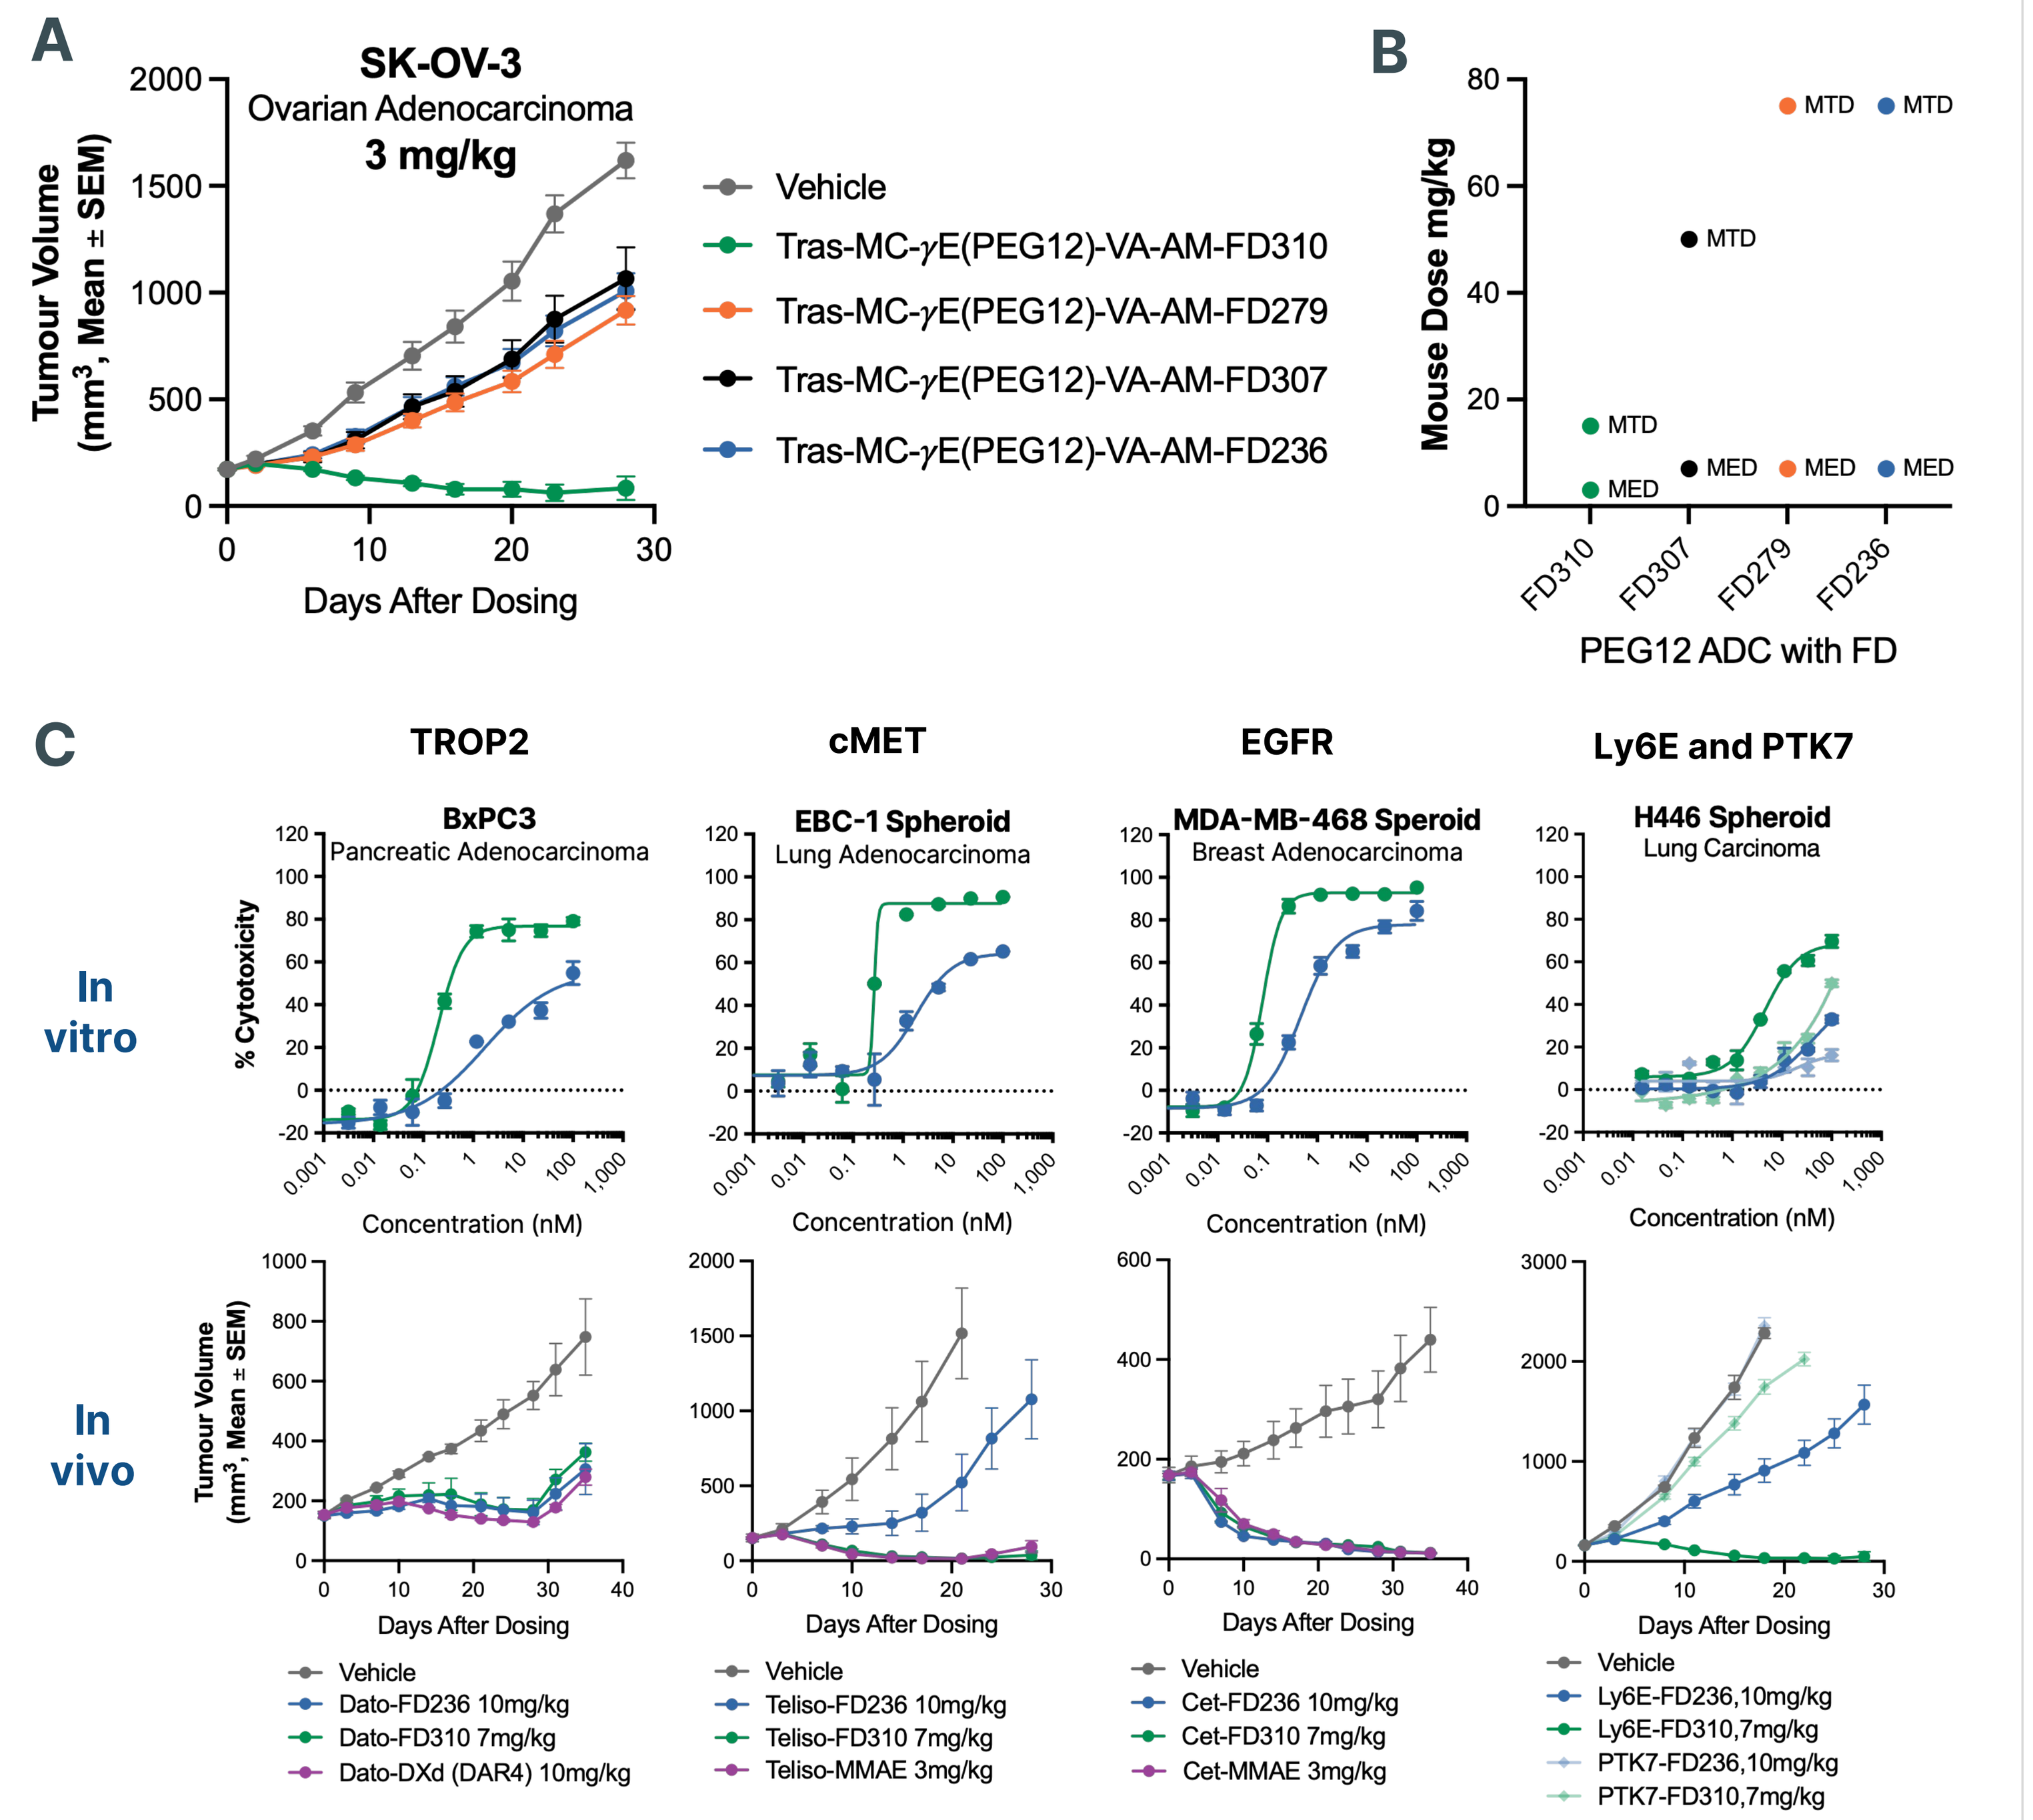
A
SK-OV-3
Ovarian Adenocarcinoma
3 mg/kg
Tumour Volume (mm³, Mean ± SEM)
Days After Dosing
Vehicle
Tras-MC-γE(PEG12)-VA-AM-FD310
Tras-MC-γE(PEG12)-VA-AM-FD279
Tras-MC-γE(PEG12)-VA-AM-FD307
Tras-MC-γE(PEG12)-VA-AM-FD236
B
Mouse Dose mg/kg
MTD
MED
PEG12 ADC with FD
C
TROP2
cMET
EGFR
Ly6E and PTK7
In vitro
In vivo
BxPC3
Pancreatic Adenocarcinoma
EBC-1 Spheroid
Lung Adenocarcinoma
MDA-MB-468 Speroid
Breast Adenocarcinoma
H446 Spheroid
Lung Carcinoma
% Cytotoxicity
Concentration (nM)
Days After Dosing
Vehicle
Dato-FD236 10mg/kg
Dato-FD310 7mg/kg
Dato-DXd (DAR4) 10mg/kg
Teliso-FD236 10mg/kg
Teliso-FD310 7mg/kg
Teliso-MMAE 3mg/kg
Cet-FD236 10mg/kg
Cet-FD310 7mg/kg
Cet-MMAE 3mg/kg
Ly6E-FD236,10mg/kg
Ly6E-FD310,7mg/kg
PTK7-FD236,10mg/kg
PTK7-FD310,7mg/kg

Figure 5: (A) Trastuzumab targeted ADCs carrying 4 lead payloads were evaluated with PEG12 containing linkers in a SK-OV-3 cell line derived xenograft. **(B)** ADCs with the FD310 payload demonstrated tumor regression at dose of 3 mg/kg however were burdened with significant toxicity at doses above 15 mg/kg. ADCs carrying either the FD236 or 279 payloads had the largest murine therapeutic index with tumor regression observed at 7 mg/kg and tolerability up to 75 mg/kg. **(C)** Breadth of activity of ADCs targeting TROP2 (datopotamab), cMET (telisotuzumab), EGFR (cetuximab), Ly6E, and PTK7 was evaluated using the most potent payload (FD310) and best tolerated payload (FD236).

Table 1: Toxicology study in Sprague-Dawley Rats

Finding	Tras-MC-γE(PEG12)-VA-AM-FD279 (DAR8)		Tras-MC-γE(PEG12)-VA-AM-FD279 (DAR4)	
	20 mg/kg	40 mg/kg	40 mg/kg	80 mg/kg
Organ Weights* **(Day 22)**	↓ testes – **absolute**, BW, **BrW** ↓ epididymis – **absolute**, BW, **BrW** ↓ thymus – absolute, BW, BrW	↓ testes – **absolute, BW, BrW** ↓ epididymis – **absolute, BW, BrW** ↓ thymus – absolute, BW, BrW	↓ testes – absolute, BW, BrW ↓ epididymis – absolute, BW, **BrW** ↓ thymus – absolute, BW, BrW	↓ testes – **absolute, BW, BrW** ↓ epididymis – **absolute, BW, BrW** ↓ thymus – absolute, BW, BrW
Clinical Chemistry (Day 22)*	↑ GLO, K ↓ ALB, A/G, CHOL, TG	↑ ALT, AST, GLO, K ↓ ALB, A/G, TG	↓ ALB, A/G, CHOL	↑ ALT, AST, GLO, K ↓ ALB, A/G
Hematology (Day 22)*	↑ WBC, NEUT	↑ PLT, WBC, NEUT, MONO, EOS ↓ RBC, HGB, HCT, RETIC, LYM, EOS	↑ WBC, NEUT, LYM, MONO	↑ PLT, WBC, NEUT, LYM, MONO ↓ RBC, HGB, HCT
Histopathology (Day 22)*				
Kidney	glomerulonephritis, hyaline casts, and/or tubular basophilia		glomerulonephritis, hyaline casts, and/or tubular basophilia	
Eye	mixed cell infiltration/inflammation (ciliary body, cornea & iris), corneal mineralization (20 mg/kg), corneal ulceration & hyperplasia (40 mg/kg)		mixed cell infiltration/inflammation (ciliary body, cornea & iris), corneal hyperplasia, corneal mineralization (80 mg/kg)	
Degeneration/atrophy/necrosis	testes	testes, mammary gland	testes	testes, mammary gland
Mixed or mononuclear cell infiltration/inflammation	lung, brain (choroid plexus), mandibular lymph node, pituitary	lacrimal gland, lung^, testes, mandibular lymph node, pituitary, mammary gland	lung, brain (choroid plexus), pituitary	lung^, brain (choroid plexus), testes, salivary gland, mandibular lymph node, mammary gland
Increased sinusoidal cellularity	--	--	liver	liver

*bold = significantly different from control, BW: relative to body weight, BrW: relative to brain weight, A/G: albumin/globulin ratio, K: potassium, ALB: albumin, LDH: lactate dehydrogenase, ALP: alkaline phosphatase, PHOS: phosphorous, ALT: alanine aminotransferase, TG: triglycerides, AST: aspartate aminotransferase, CHOL: cholesterol, GLO: globulin, BASO: basophils, NEUT: neutrophils, EOS: eosinophils, PLT: platelets, HCT: hematocrit, RBC: red blood cells, HGB: hemoglobin, RETIC: reticulocytes, LYM: lymphocytes, WBC: white blood cells, MONO: monocytes

Table 1: Tolerability of DAR4 and DAR8 trastuzumab-FD279 ADCs were evaluated in Sprague-Dawley Rats. No mortality was observed; however, significant findings highlight that these ADCs were poorly tolerated at all doses.

Proof of concept HER2 targeted ADCs demonstrate in vivo anti-tumor activity

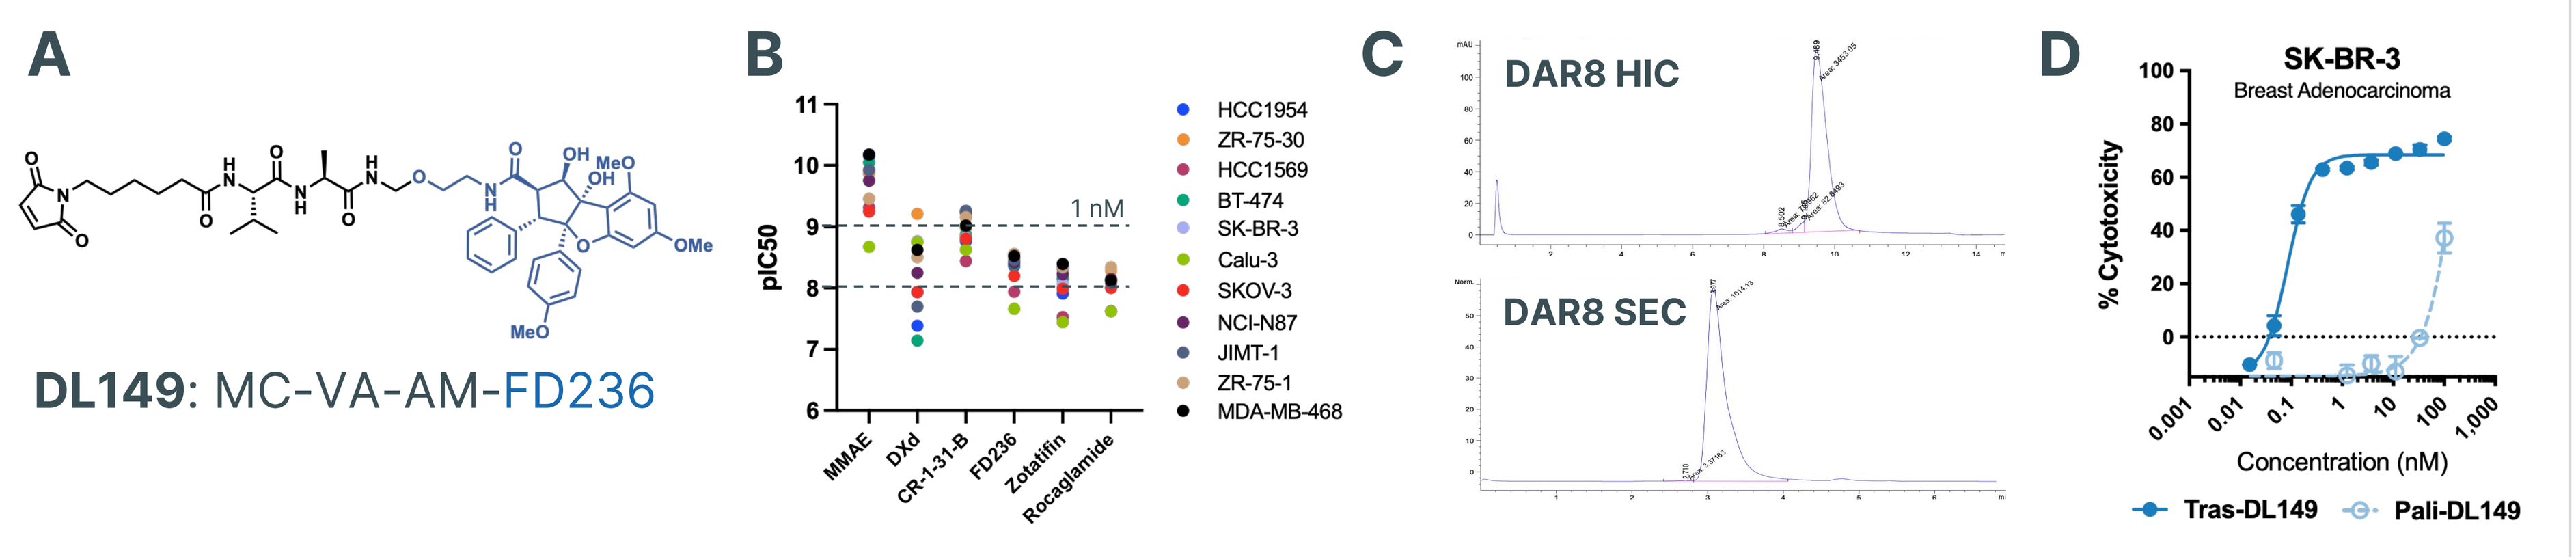
A
DL149: MC-VA-AM-FD236
B
pIC50
1 nM
HCC1954
ZR-75-30
HCC1569
BT-474
SK-BR-3
Calu-3
SKOV-3
NCI-N87
JIMT-1
ZR-75-1
MDA-MB-468
MMAE
DXd
CR-1-31-B
FD236
Zotatifin
Rocaglamide
C
DAR8 HIC
DAR8 SEC
D
SK-BR-3
Breast Adenocarcinoma
% Cytotoxicity
Concentration (nM)
Tras-DL149
Pali-DL149

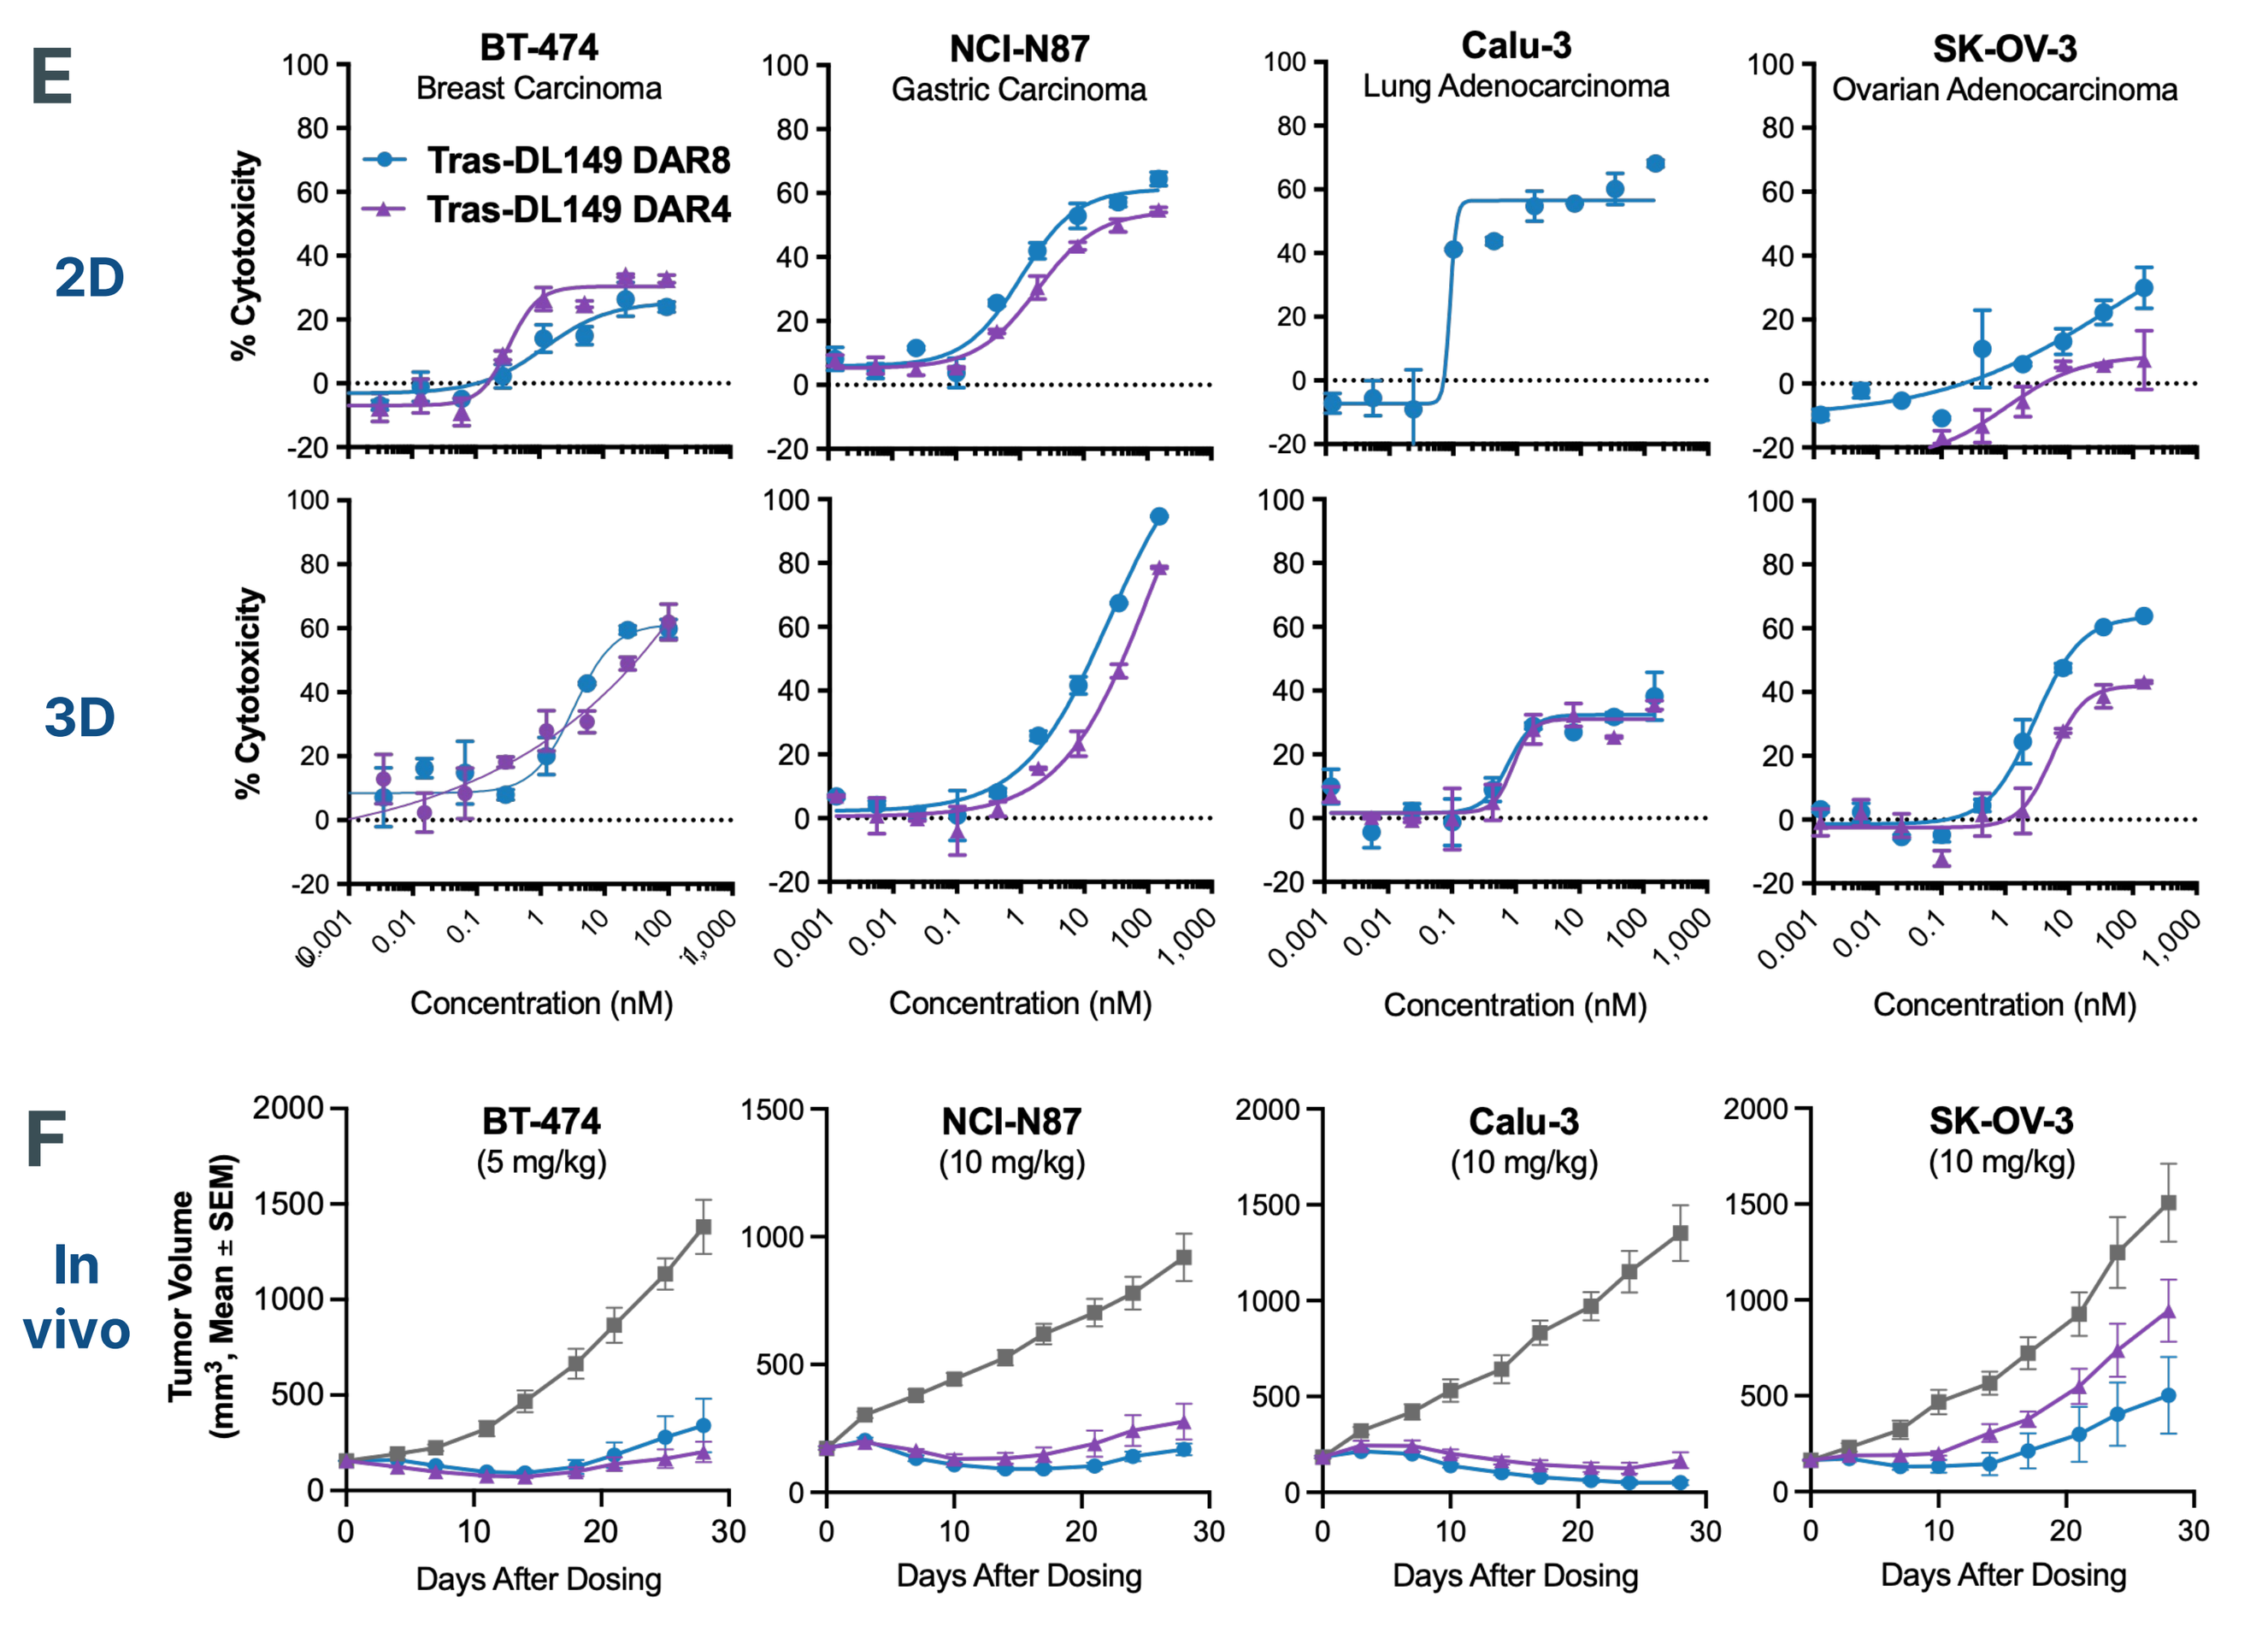
E
2D
3D
BT-474
Breast Carcinoma
NCI-N87
Gastric Carcinoma
Calu-3
Lung Adenocarcinoma
SK-OV-3
Ovarian Adenocarcinoma
Tras-DL149 DAR8
Tras-DL149 DAR4
% Cytotoxicity
Concentration (nM)
F
In vivo
BT-474
(5 mg/kg)
NCI-N87
(10 mg/kg)
Calu-3
(10 mg/kg)
SK-OV-3
(10 mg/kg)
Tumor Volume (mm³, Mean ± SEM)
Days After Dosing

Figure 2. (A) Structure of a proof-of-concept drug-linker that incorporates an eIF4Ai (FD236) that is released after enzymatic cleavage of the Val-Ala dipeptide and self-immolation of the resulting hemi-aminal (AM). **(B)** The FD236 payload demonstrates similar in vitro potency to known eIF4A inhibitors and commonly used ADC payloads in a tumor cell line panel. **(C)** Full conjugation of interchain disulfides to a DAR of 8 was confirmed by hydrophobic interaction chromatography (HIC) without aggregation as determined by size exclusion chromatography (SEC). **(D)** Trastuzumab-DL149 ADC shows potent in vitro activity in HER2 expressing SK-BR-3 cells while a non targeted ADC is inactive. **(E)** DAR8 and DAR4 ADCs are active in a range of HER2 expressing cell lines both in 2D monolayer and 3D spheroid assays. **(F)** Strong in vitro to in vivo activity correlation of trastuzumab targeted eIF4A ADCs. Robust efficacy is observed for both DAR4 and DAR8 ADCs in a BT-474 CDX model at 5 mg/kg and in NCI-N87, Calu-3, and SK-OV-3 at 10 mg/kg.

Increased linker hydrophilicity improves efficacy and tolerability

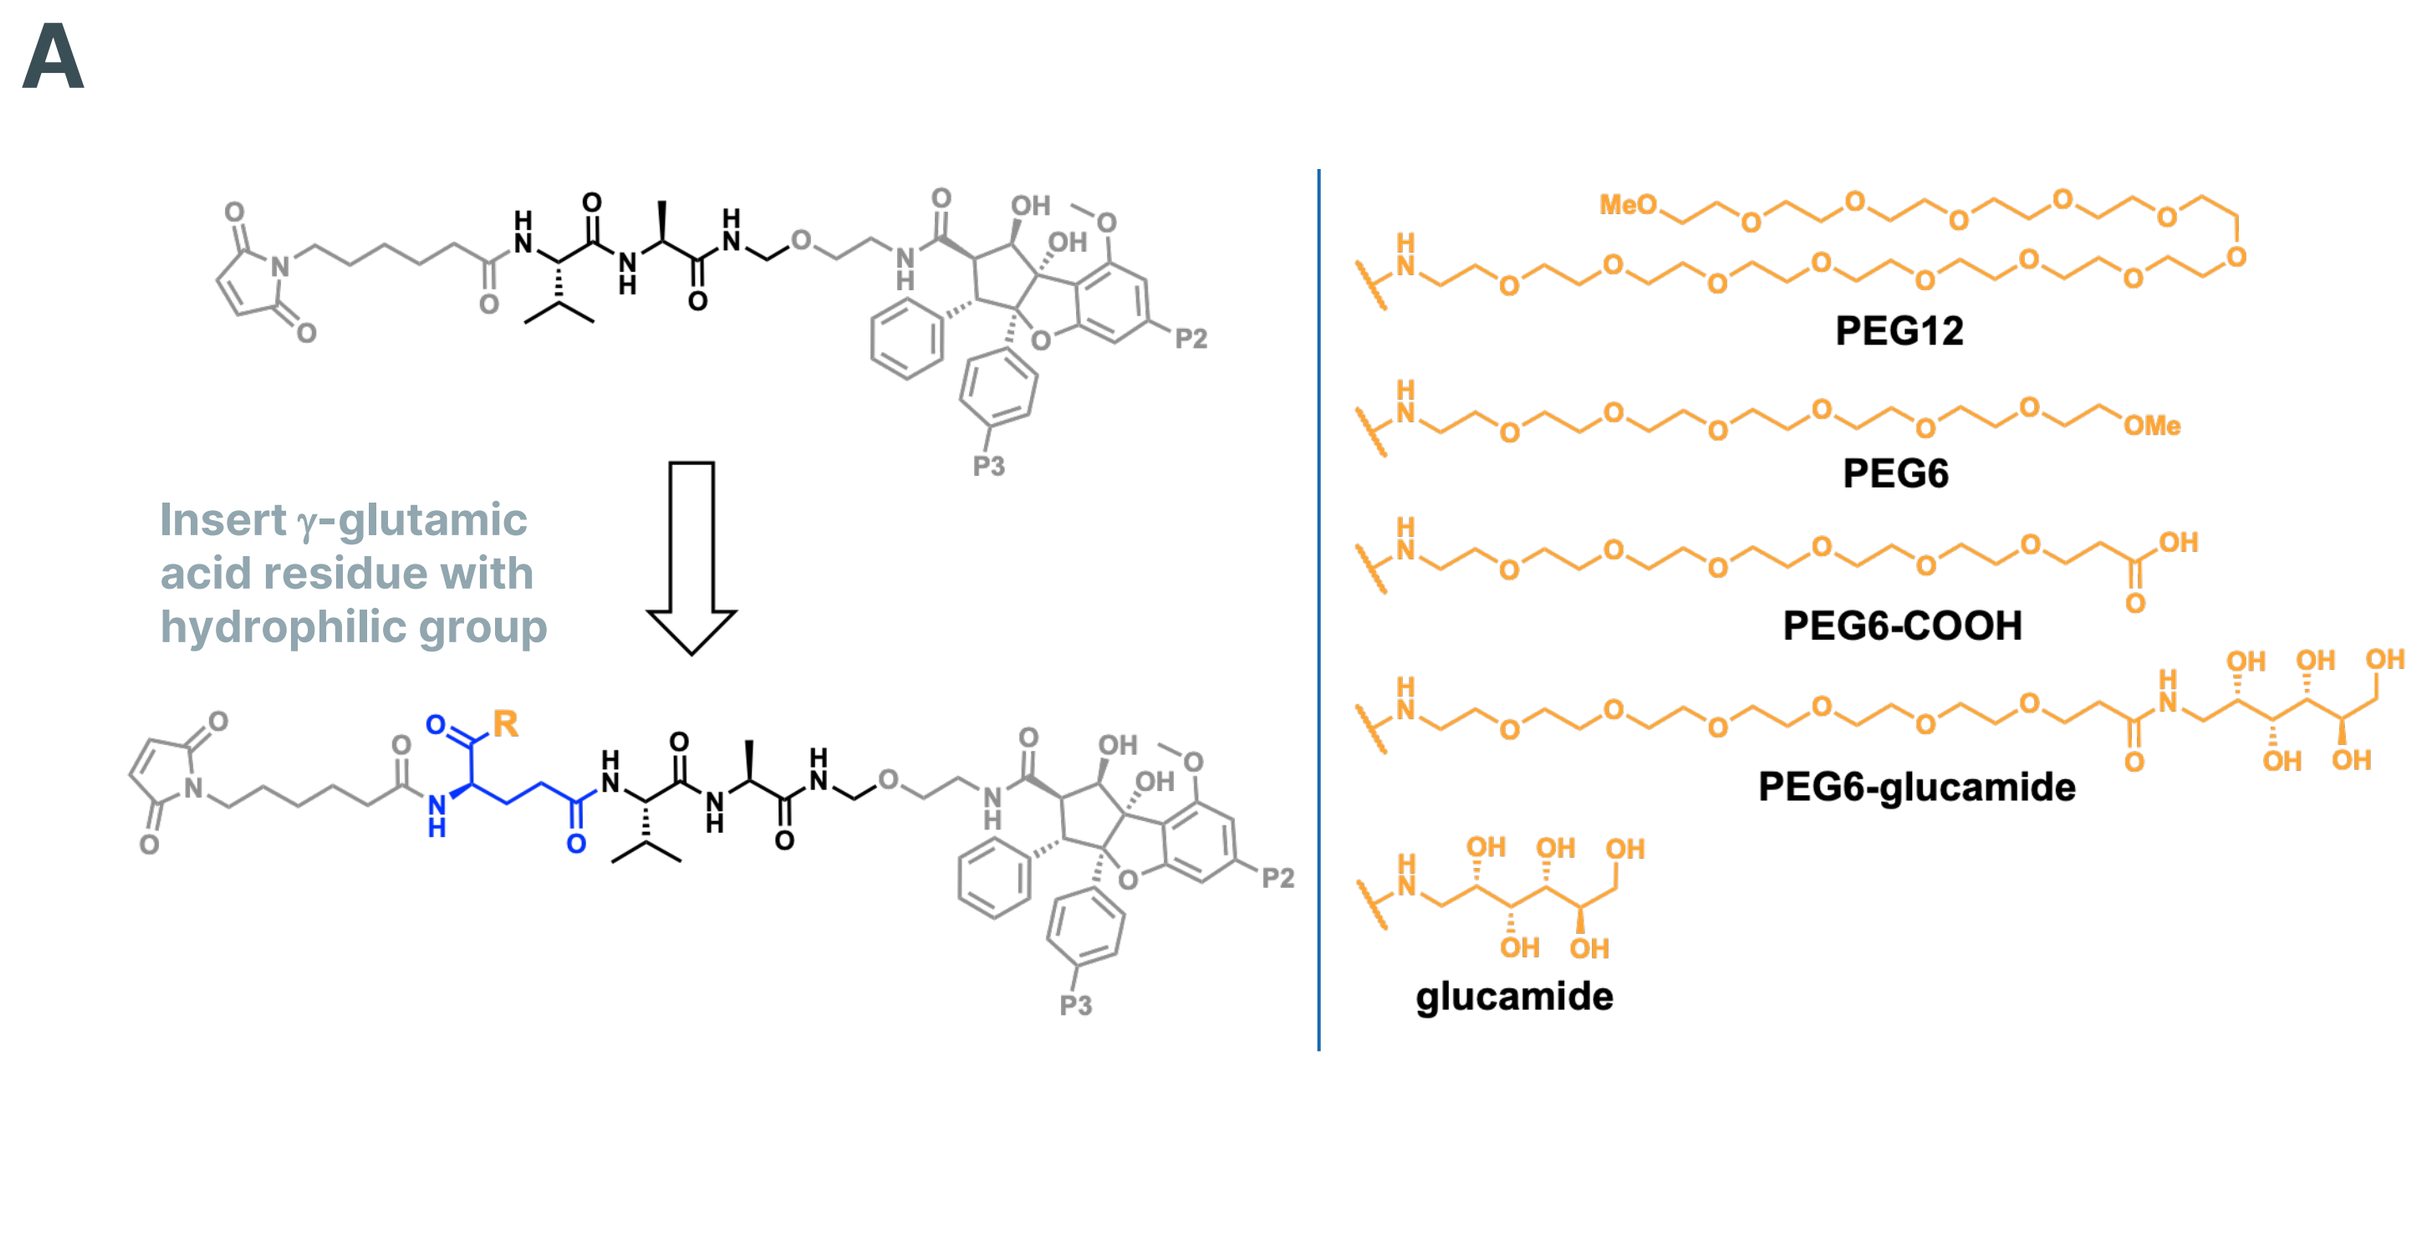
A
Insert γ-glutamic acid residue with hydrophilic group
PEG12
PEG6
PEG6-COOH
PEG6-glucamide
glucamide

B

Tolerability of trastuzumab ADCs in non-tumor bearing BALB/c mice			
Linker	Dose, mg/kg	BW Mean %chg, Day 1-7	Clinical Signs, Day 7
Buffer	-	3.5	Normal
FD236 ADCs			
γEVA	15	-16.3	Lethargy
γE(PEG6)VA	15	-13.2	Lethargy
γE(PEG12)VA	15	-3.6	Normal
γE(glucamide)VA	15	-12.5	Lethargy
γE(PEG6-COOH)VA	15	-7.9	Normal
γE(PEG6-glucamide)VA	15	-12.0	Normal
FD310 ADCs			
γEVA	50	-1.3	Normal
γE(PEG6)VA	50	-2.5	Normal
γE(PEG12)VA	50	4.0	Normal
γE(glucamide)VA	50	-1.8	Normal
γE(PEG6-COOH)VA	50	2.3	Normal

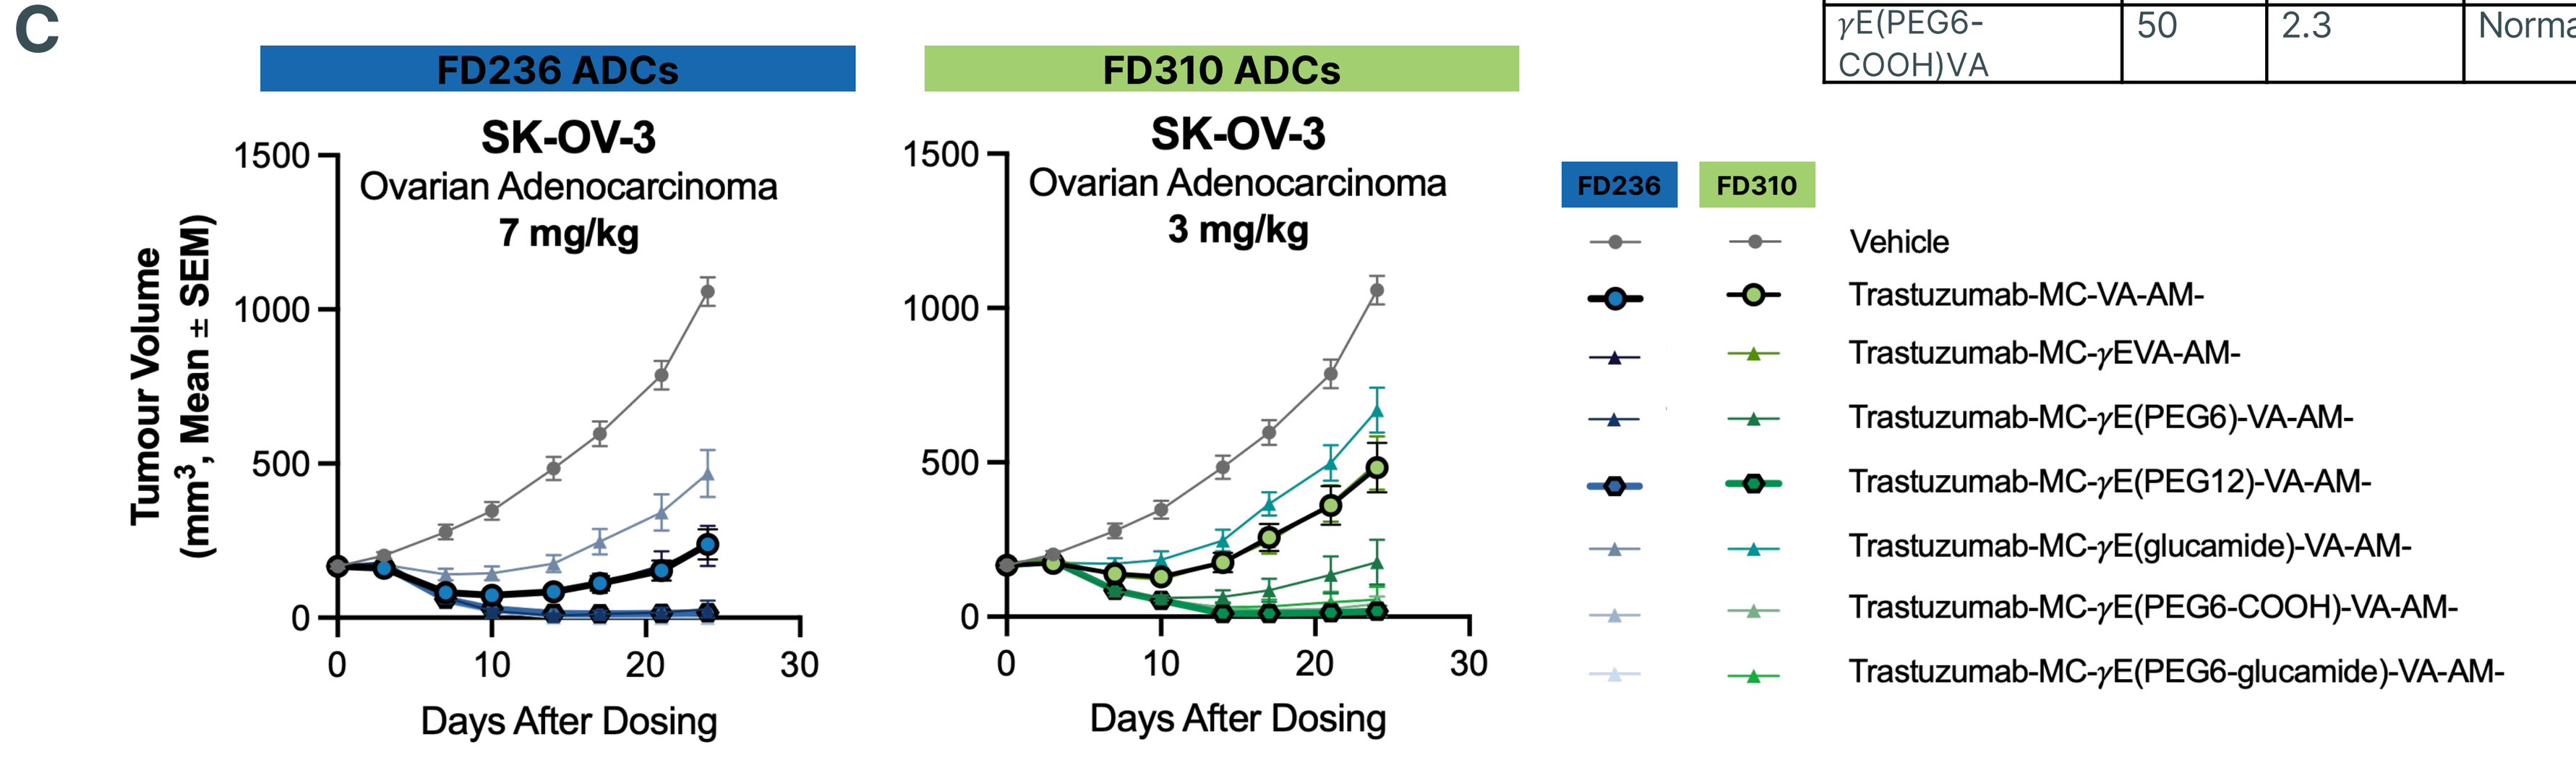
C
FD236 ADCs
FD310 ADCs
SK-OV-3
Ovarian Adenocarcinoma
7 mg/kg
3 mg/kg
Tumour Volume (mm³, Mean ± SEM)
Days After Dosing
FD236
FD310
Vehicle
Trastuzumab-MC-VA-AM-
Trastuzumab-MC-γEVA-AM-
Trastuzumab-MC-γE(PEG6)-VA-AM-
Trastuzumab-MC-γE(PEG12)-VA-AM-
Trastuzumab-MC-γE(glucamide)-VA-AM-
Trastuzumab-MC-γE(PEG6-COOH)-VA-AM-
Trastuzumab-MC-γE(PEG6-glucamide)-VA-AM-

Figure 4. (A) Strategy for incorporating hydrophilicity enhancing groups utilized a glutamic acid residue as a branch point for the addition of PEG chains and glucamides. **(B)** Trastuzumab ADCs with branched drug-linkers incorporating either FD236 or FD310 improved the tolerability in non-tumor bearing BALB/c mice, with the PEG12 containing linkers demonstrating the largest impact. **(C)** Incorporation of a hydrophilic group improved the in vivo efficacy of trastuzumab-FD236 and trastuzumab-FD310 ADCs in a HER2 expressing SK-OV-3 cell line derived xenograft. The original VA and optimized PEG12 containing linkers are highlighted with black borders.

Conclusion

Over 40 unique and linkable rocaglamide analogs were evaluated as free drugs leading to selection of 10 compounds for ADC generation. The successful incorporation of a PEG12 containing hydrophilic linker produced ADCs with improved efficacy and tolerability. Promising in vivo efficacy was demonstrated across multiple tumor associated targets, however the observed toxicity level in both mice and rats suggests a potentially lower than anticipated preclinical therapeutic window.

References:
(1) Xue C, Gu X, Li G, Bao Z and Li L. Expression and Functional Roles of Eukaryotic Initiation Factor 4A Family Proteins in Human Cancers. *Front. Cell Dev. Biol.* **2021**, *9*, 711965. (2) Pelletier J, Graff J, Ruggero, D, Sonenberg, N. Targeting the eIF4F Translation Initiation Complex: A Critical Nexus for Cancer Development. *Cancer Res.* **2015**, *75(2)*, 250-263. (3) Rosen E, Sharma M, Caswell-Jin JL, Spira AI, Fulgar GA, Densel M, Rana N, Sperry S, Warner D, Meric-Bernstram F. Phase 1/2 dose expansion study evaluating first-in-class eIF4A inhibitor zotatifin in patients with ER+ metastatic breast cancer. *J Clin Oncol*, **2023**, *41*, 1080.

AACR Annual Meeting 2026