In vitro assays to predict ADC hematological toxicity: Contribution of antibody, linker, and payload

Yongzhao Huang,[1] Emer Clarke,[1] Graham A. E. Garnett,[2] Manuel Lasalle,[2] Jodi Wong,[2] Ambroise Wu,[2] Araba Sagoe-Wagner,[2] Elena Barbosa,[2] Khushi Lodaya,[2] Catalina Suarez,[2] Stuart D. Barnscher,[2] Jamie R. Rich,[2] Raffaele Colombo[2]

1. Discovery Life Sciences, Seattle, WA, USA; 2. Zymeworks Inc., Vancouver, BC, Canada

Abstract #5482

Introduction

Antibody-drug conjugates (ADCs) are one of the fastest-growing therapeutic modalities, with 12 FDA-approved ADCs and more than 280 different ADCs in clinical development. Despite their success, significant hurdles remain. Notably, translating preclinical findings to the clinic remains challenging. Hematological toxicities are commonly associated with many ADCs and may arise from the direct killing of hematopoietic cells by the ADC itself or indirectly from payload released elsewhere in the body.

Development of *in vitro* assays capable of predicting clinical findings could improve ADC development and guide the selection of optimal linkers and payloads.

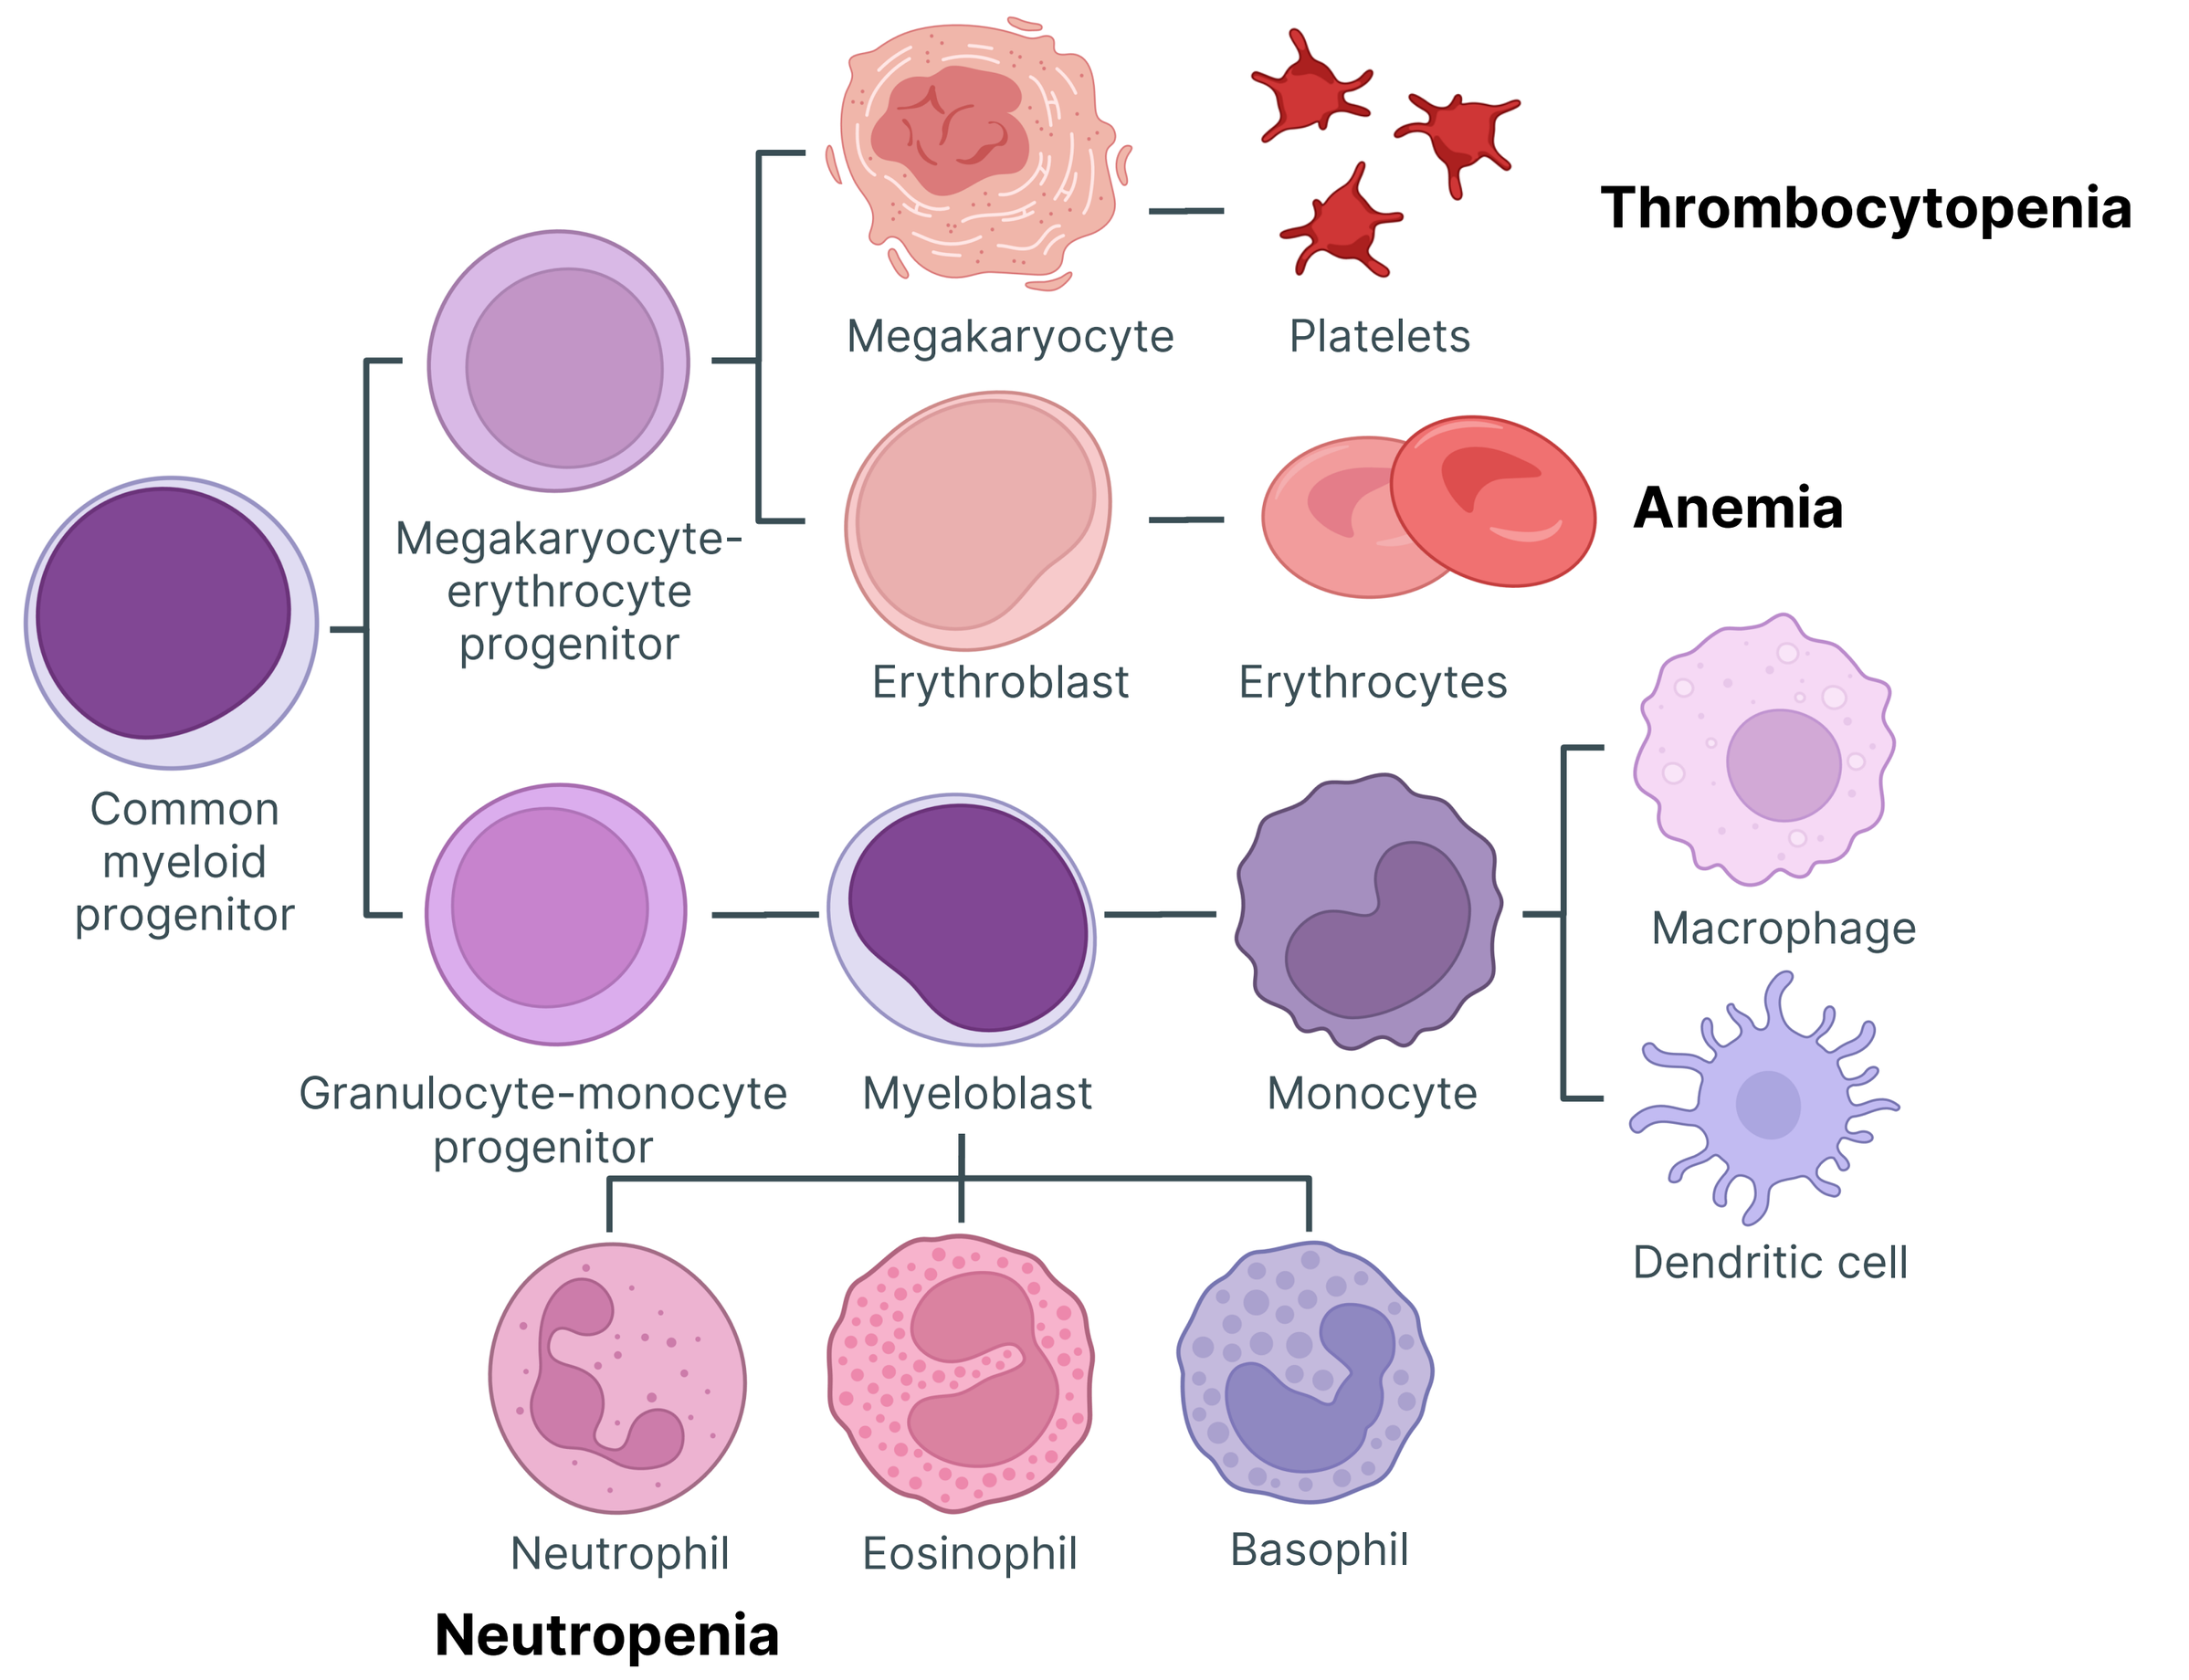
Thrombocytopenia
Megakaryocyte
Platelets
Megakaryocyte-erythrocyte progenitor
Erythroblast
Erythrocytes
Anemia
Common myeloid progenitor
Macrophage
Granulocyte-monocyte progenitor
Myeloblast
Monocyte
Dendritic cell
Neutrophil
Eosinophil
Basophil
Neutropenia

Figure 1. Hierarchy of hematopoietic cells during normal differentiation of bone marrow-derived hematopoietic stem/progenitor cells into lineage-specific cells and their associated hematological toxicities.

Materials

Different ADC payloads (exatecan, DXd, SN-38, ZD06519, DM1, DM4, MMAE, MMAF) were tested to understand the intrinsic free payload contribution to off-target toxicities.

Different ADCs were generated by conjugating targeted and non-targeted antibodies to a common hemiasterlin microtubule inhibitor, while varying the protease-cleavable linkers to evaluate linker contribution to on-target and off-target cytotoxicity *in vitro*.

Finally, two clinically validated ADCs with the same drug-linker were tested to assess the contribution of the antibody in ADC off-target toxicities.

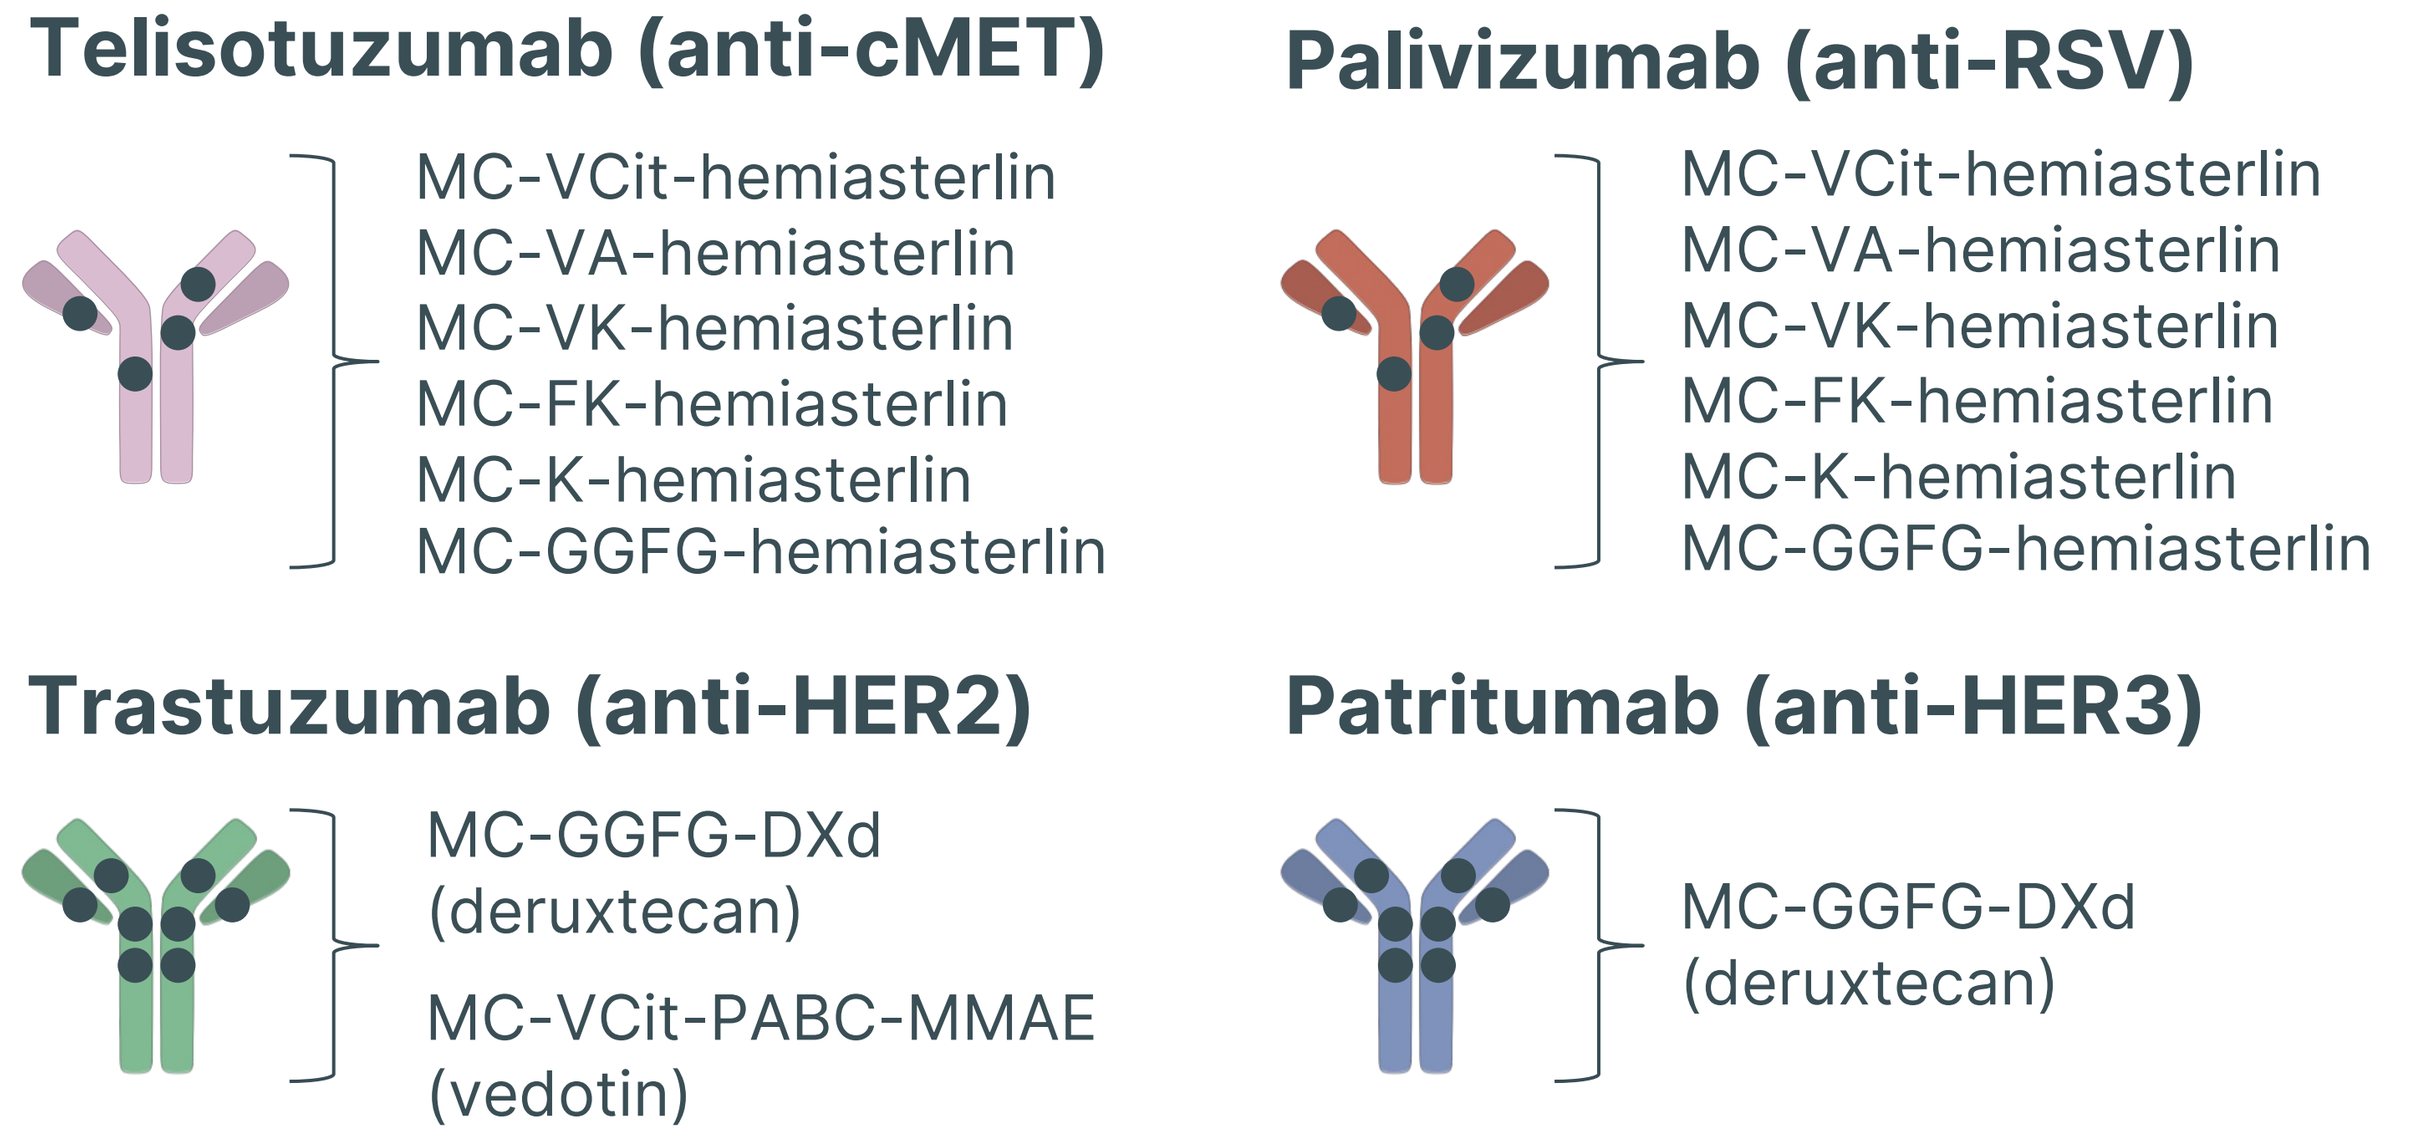
Telisotuzumab (anti-cMET)
MC-VCit-hemiasterlin
MC-VA-hemiasterlin
MC-VK-hemiasterlin
MC-FK-hemiasterlin
MC-K-hemiasterlin
MC-GGFG-hemiasterlin
Palivizumab (anti-RSV)
MC-VCit-hemiasterlin
MC-VA-hemiasterlin
MC-VK-hemiasterlin
MC-FK-hemiasterlin
MC-K-hemiasterlin
MC-GGFG-hemiasterlin
Trastuzumab (anti-HER2)
MC-GGFG-DXd (deruxtecan)
MC-VCit-PABC-MMAE (vedotin)
Patritumab (anti-HER3)
MC-GGFG-DXd (deruxtecan)

Figure 2. Schematic representation of ADCs included in the colony formation, cleavability, cytotoxicity, and/or stability *in vitro* assays. Telisotuzumab and palivizumab ADCs were conjugated as DAR=4 (±0.2), whereas trastuzumab and patritumab ADCs were conjugated as DAR=7.8 and 7.7, respectively.

Human colony formation cell (CFC) assays can predict certain clinical toxicities observed with ADCs

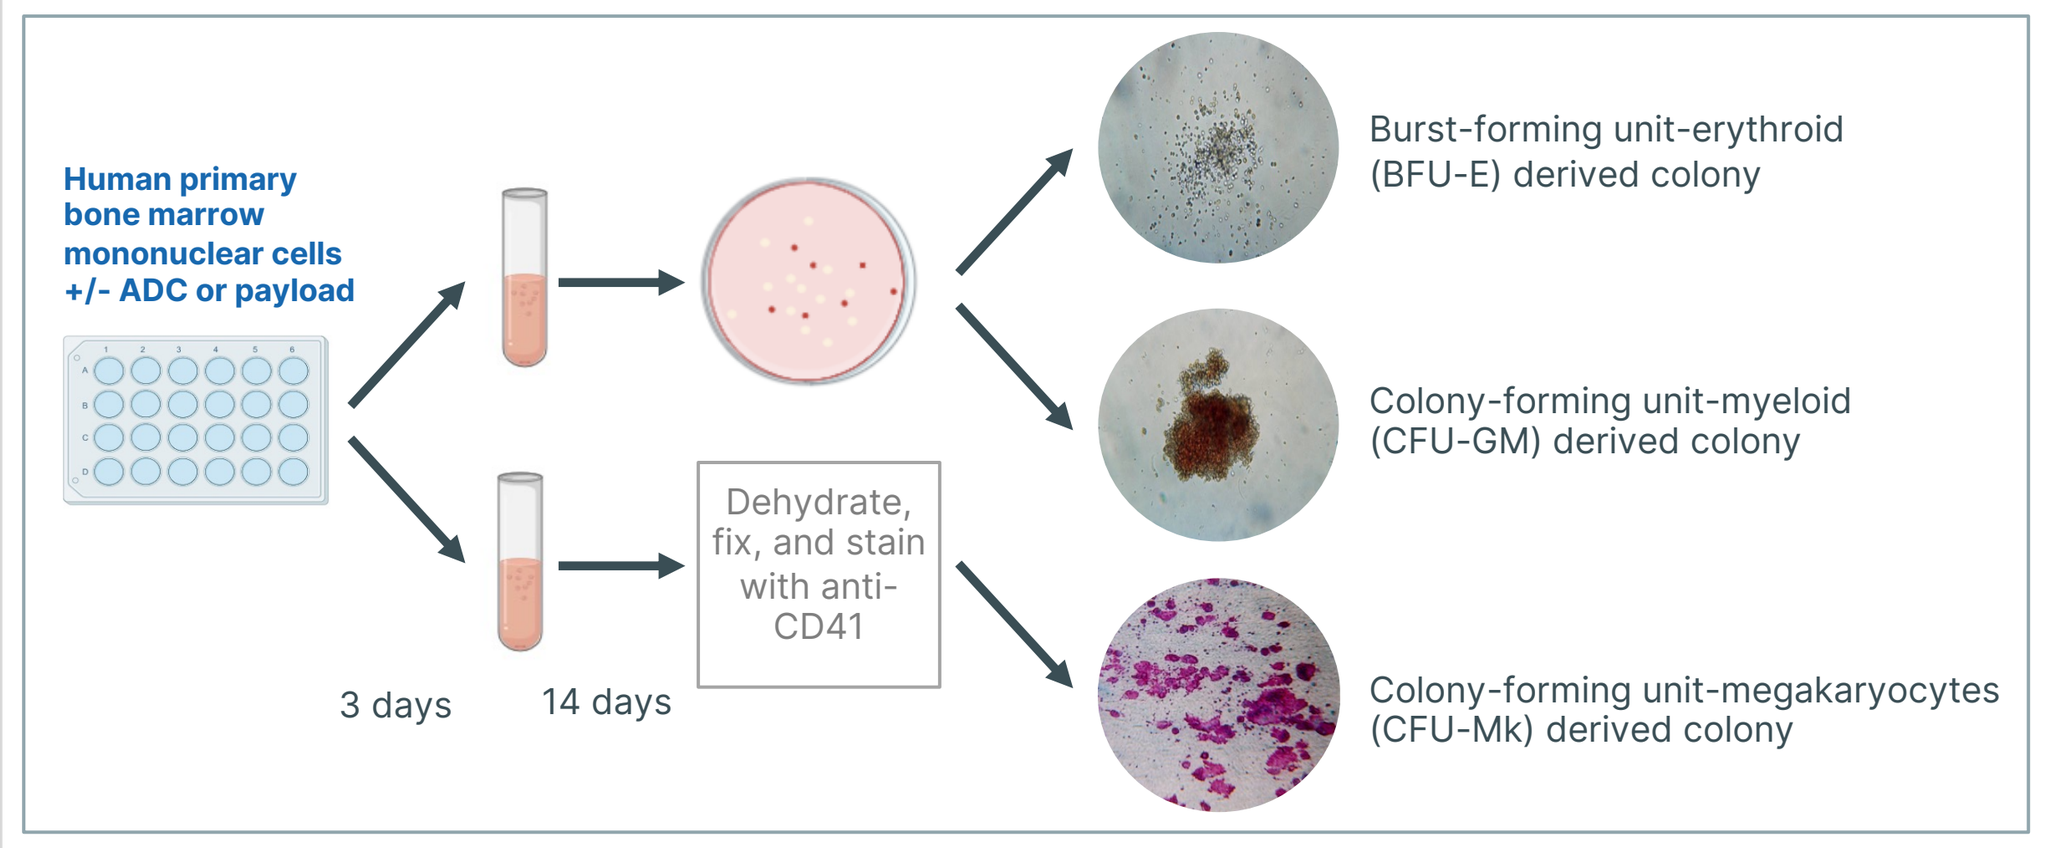
Human primary bone marrow mononuclear cells +/- ADC or payload
3 days
14 days
Dehydrate, fix, and stain with anti-CD41
Burst-forming unit-erythroid (BFU-E) derived colony
Colony-forming unit-myeloid (CFU-GM) derived colony
Colony-forming unit-megakaryocytes (CFU-Mk) derived colony

Figure 3. Schematic representation of CFC assays workflow.

Healthy human CD34+ hematopoietic stem cells isolated from bone marrow were seeded with various cytokines to induce differentiation into erythroid (BFU-E), myeloid (CFU-GM), or megakaryocyte (CFU-Mk) progenitors, and treated with various concentration of ADC, payload, or formulation buffer only (solvent control). After incubation, colonies were counted, and the percent of colony inhibition was determined by comparing colony numbers in the treatment conditions to those in the solvent controls.

Payloads – ADC payloads have different intrinsic cytotoxicity on human erythroid, myeloid, and megakaryocyte progenitors

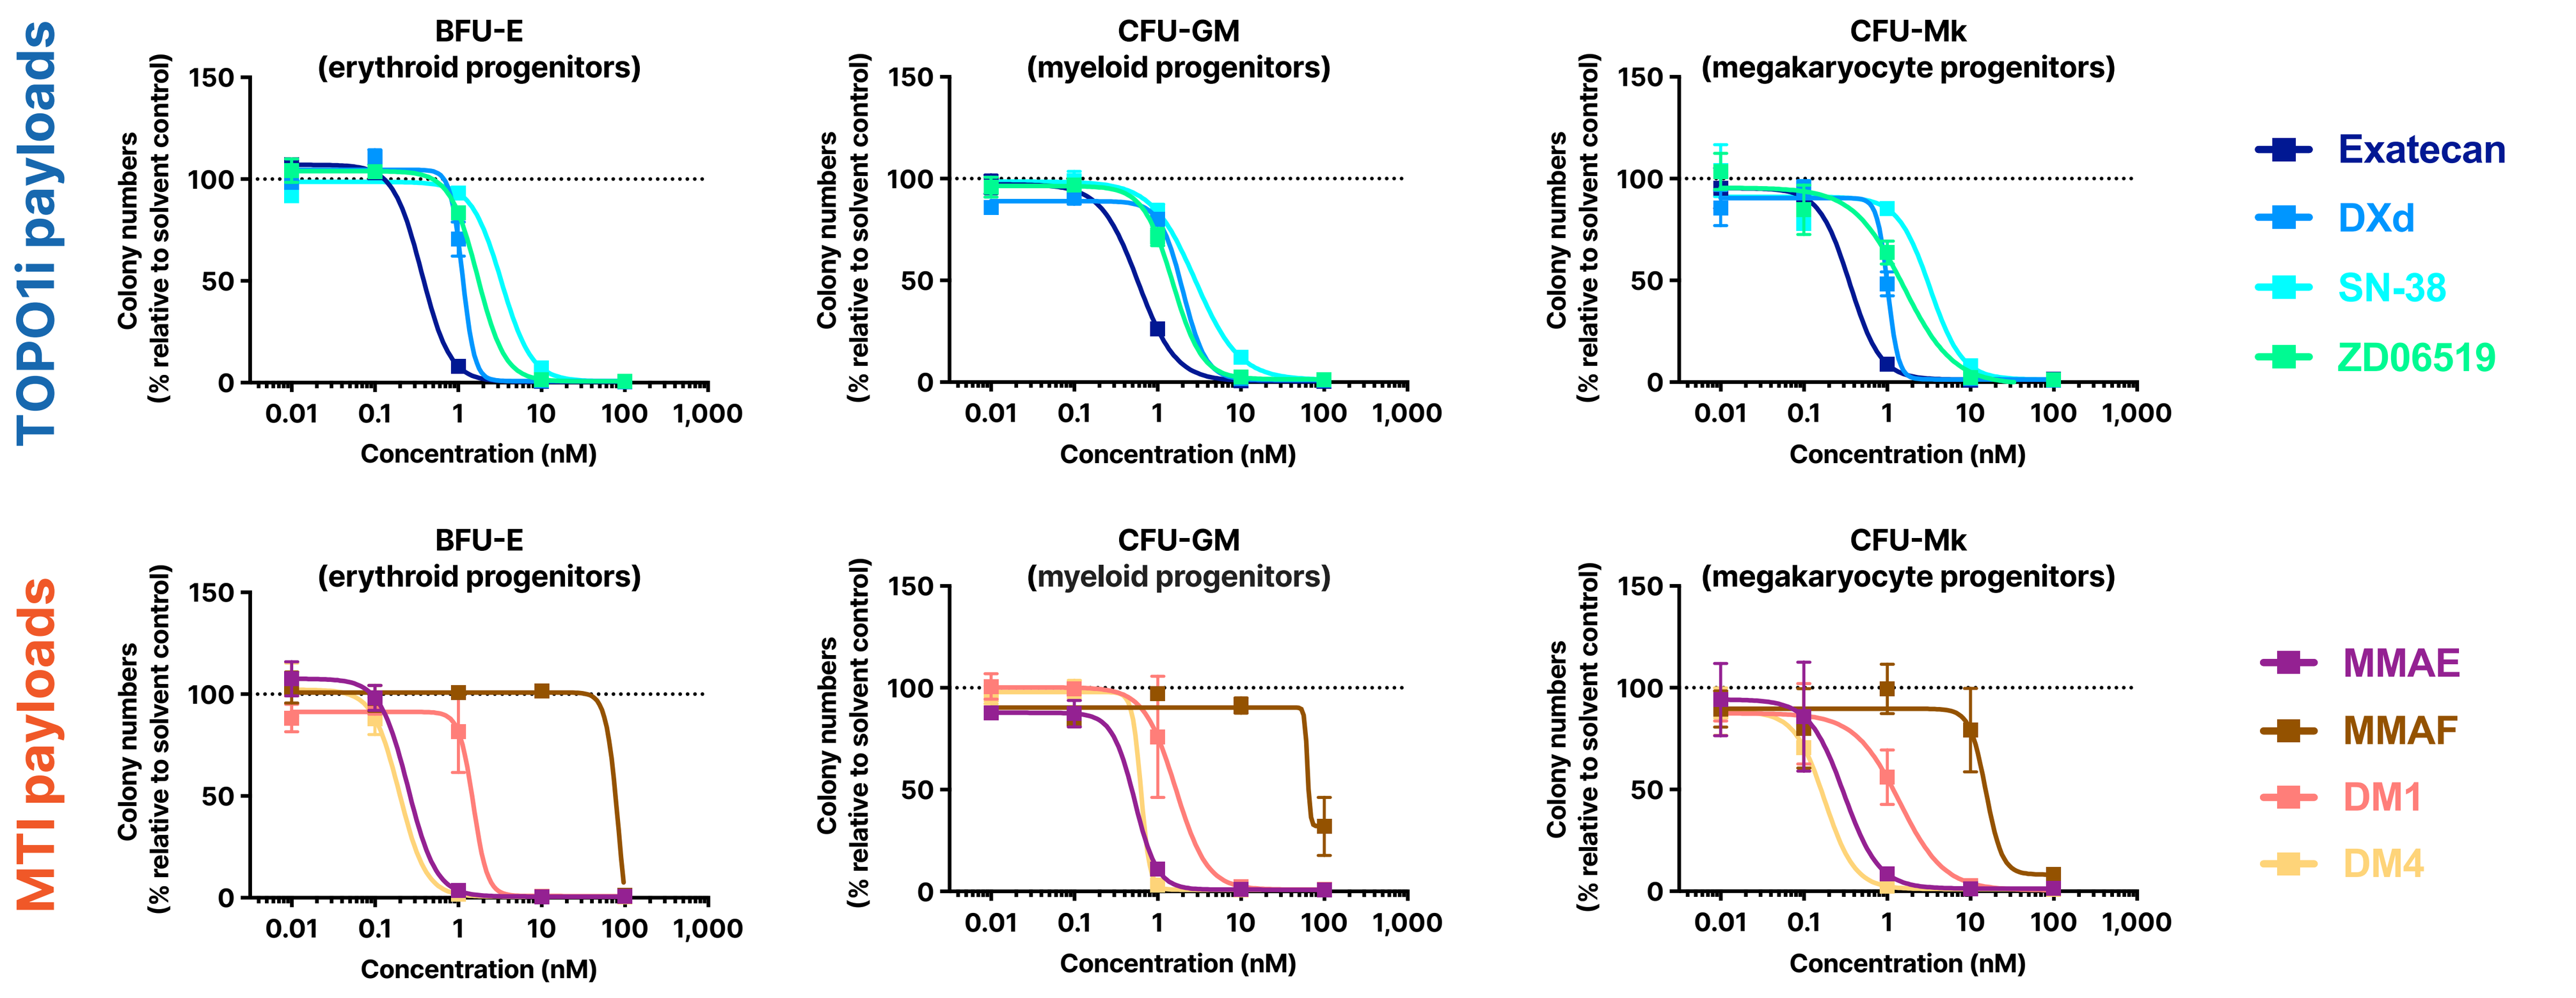
TOPO1i payloads
MTI payloads
BFU-E (erythroid progenitors)
CFU-GM (myeloid progenitors)
CFU-Mk (megakaryocyte progenitors)
Colony numbers (% relative to solvent control)
Concentration (nM)
Exatecan
DXd
SN-38
ZD06519
MMAE
MMAF
DM1
DM4

Figure 4. *In vitro* cytotoxicity of free topoisomerase 1 inhibitor (TOPO1i) and microtubule inhibitor (MTI) payloads in the CFC assays (performed as depicted in Fig. 3). Exatecan showed higher cytotoxicity than other TOPO1i payloads in the CFC assays. Similarly, for MTI payloads, DM4 and MMAE showed the highest cytotoxicity, followed by DM1 and MMAF.

Linkers – Comparison of non-targeted ADCs with the same antibody and payload but different linkers highlights the potential contribution of linker to off-target toxicities

Antibody-drug conjugate	Cytotoxicity EC_{50} (nM): BFU-E (erythroid progenitors)	Cytotoxicity EC_{50} (nM): CFU-GM (myeloid progenitors)	Cytotoxicity EC_{50} (nM): CFU-Mk (megakaryocyte progenitors)
Palivizumab-MC-VCit-hemiasterlin	6.7	6.4	0.8
Palivizumab-MC-VA-hemiasterlin	3.0	5.0	<0.8*
Palivizumab-MC-K-hemiasterlin	57.5	56.9	21.0
Palivizumab-MC-VK-hemiasterlin	4.7	4.3	2.0
Palivizumab-MC-FK-hemiasterlin	9.1	9.6	12.1
Hemiasterlin payload (unconjugated)	7.5	8.5	1.7

Table 1. Cytotoxicity (EC_{50}, reported as nM) of different ADCs in CFC assays (performed as depicted in Fig. 3). *incomplete curve.

Antibodies – Comparison of ADCs with the same payload and linker but different antibodies highlight the potential contribution of the antibody to off-target toxicities

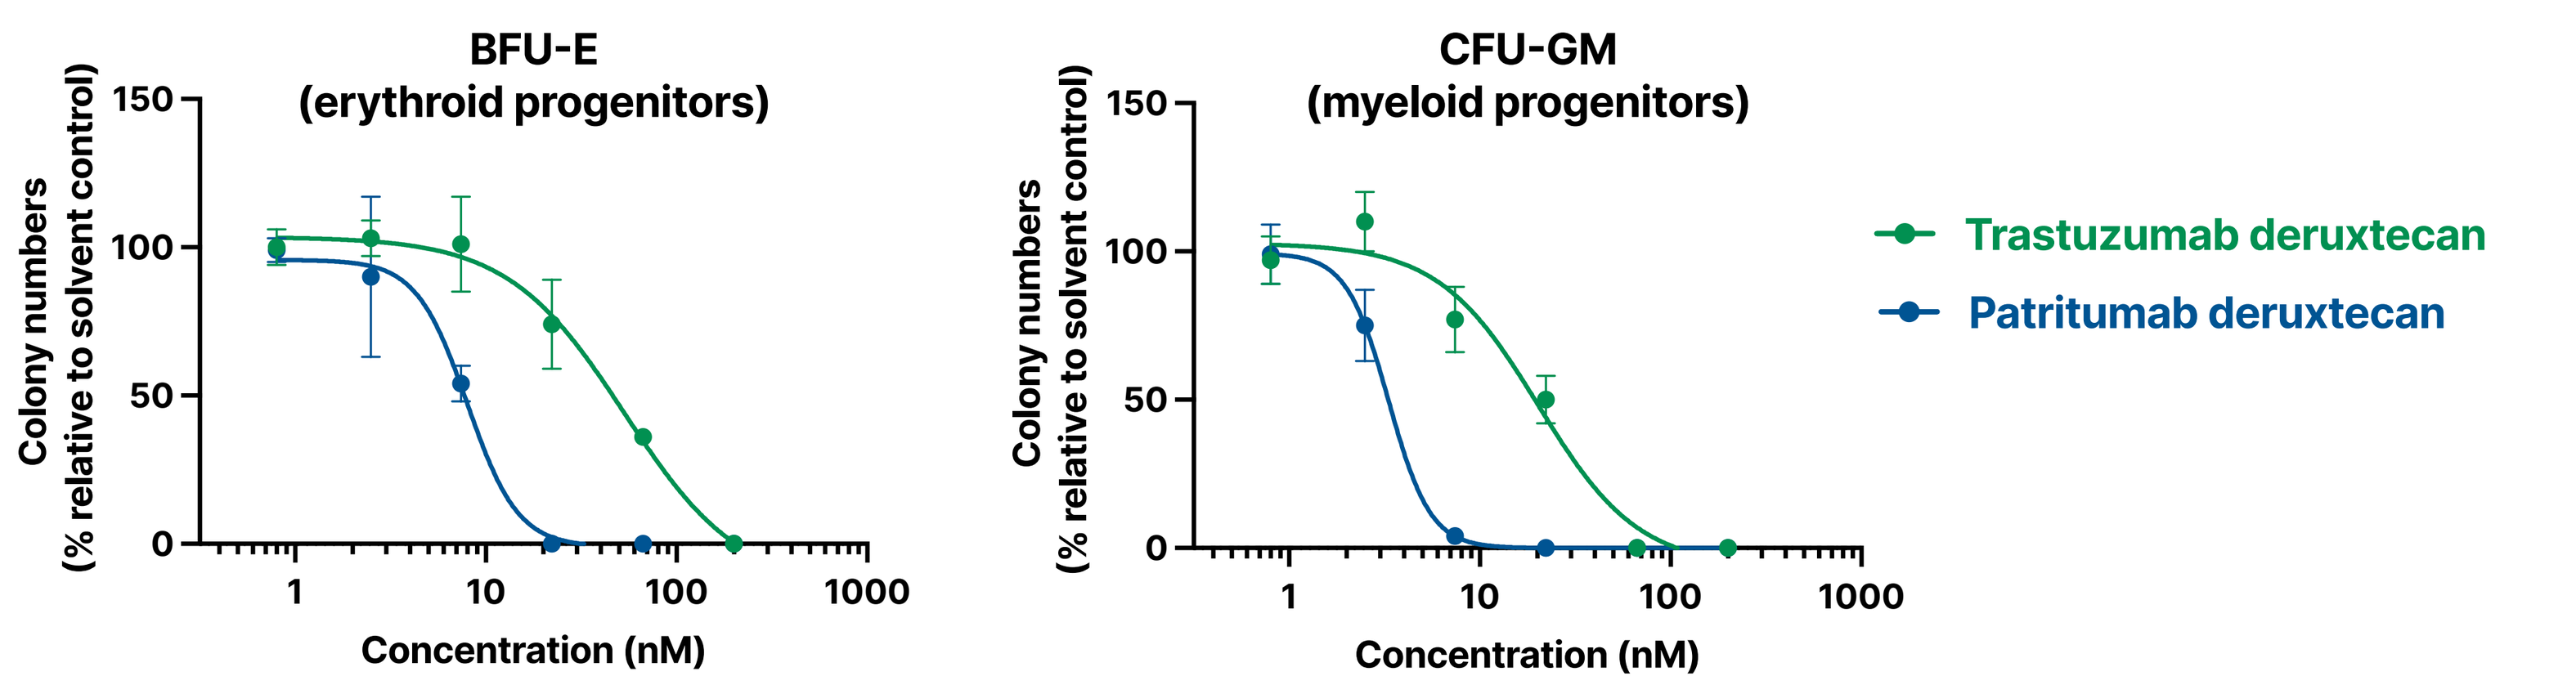
BFU-E (erythroid progenitors)
CFU-GM (myeloid progenitors)
Colony numbers (% relative to solvent control)
Concentration (nM)
Trastuzumab deruxtecan
Patritumab deruxtecan

Naked mAbs comparison

Hydrophobic interaction chromatography (HIC) retention time:

- Trastuzumab = 9.7 min
- Patritumab = 10.2 min

Isoelectric point (pI):

- Trastuzumab = 8.9
- Patritumab = 8.6

Figure 5. Patritumab deruxtecan (HER3-DXd) exhibited greater cytotoxicity in the CFC assay (performed as depicted in Fig. 3) compared to trastuzumab deruxtecan (T-DXd), in alignment with the higher incidence of hematological toxicities observed in patients treated with HER3-DXd versus T-DXd in clinical settings (Ref. 1, 2).

In vitro linker cleavage and cytotoxicity assays reveal different payload release kinetics

Different linkers released payloads at different rates in a lysosomal assay

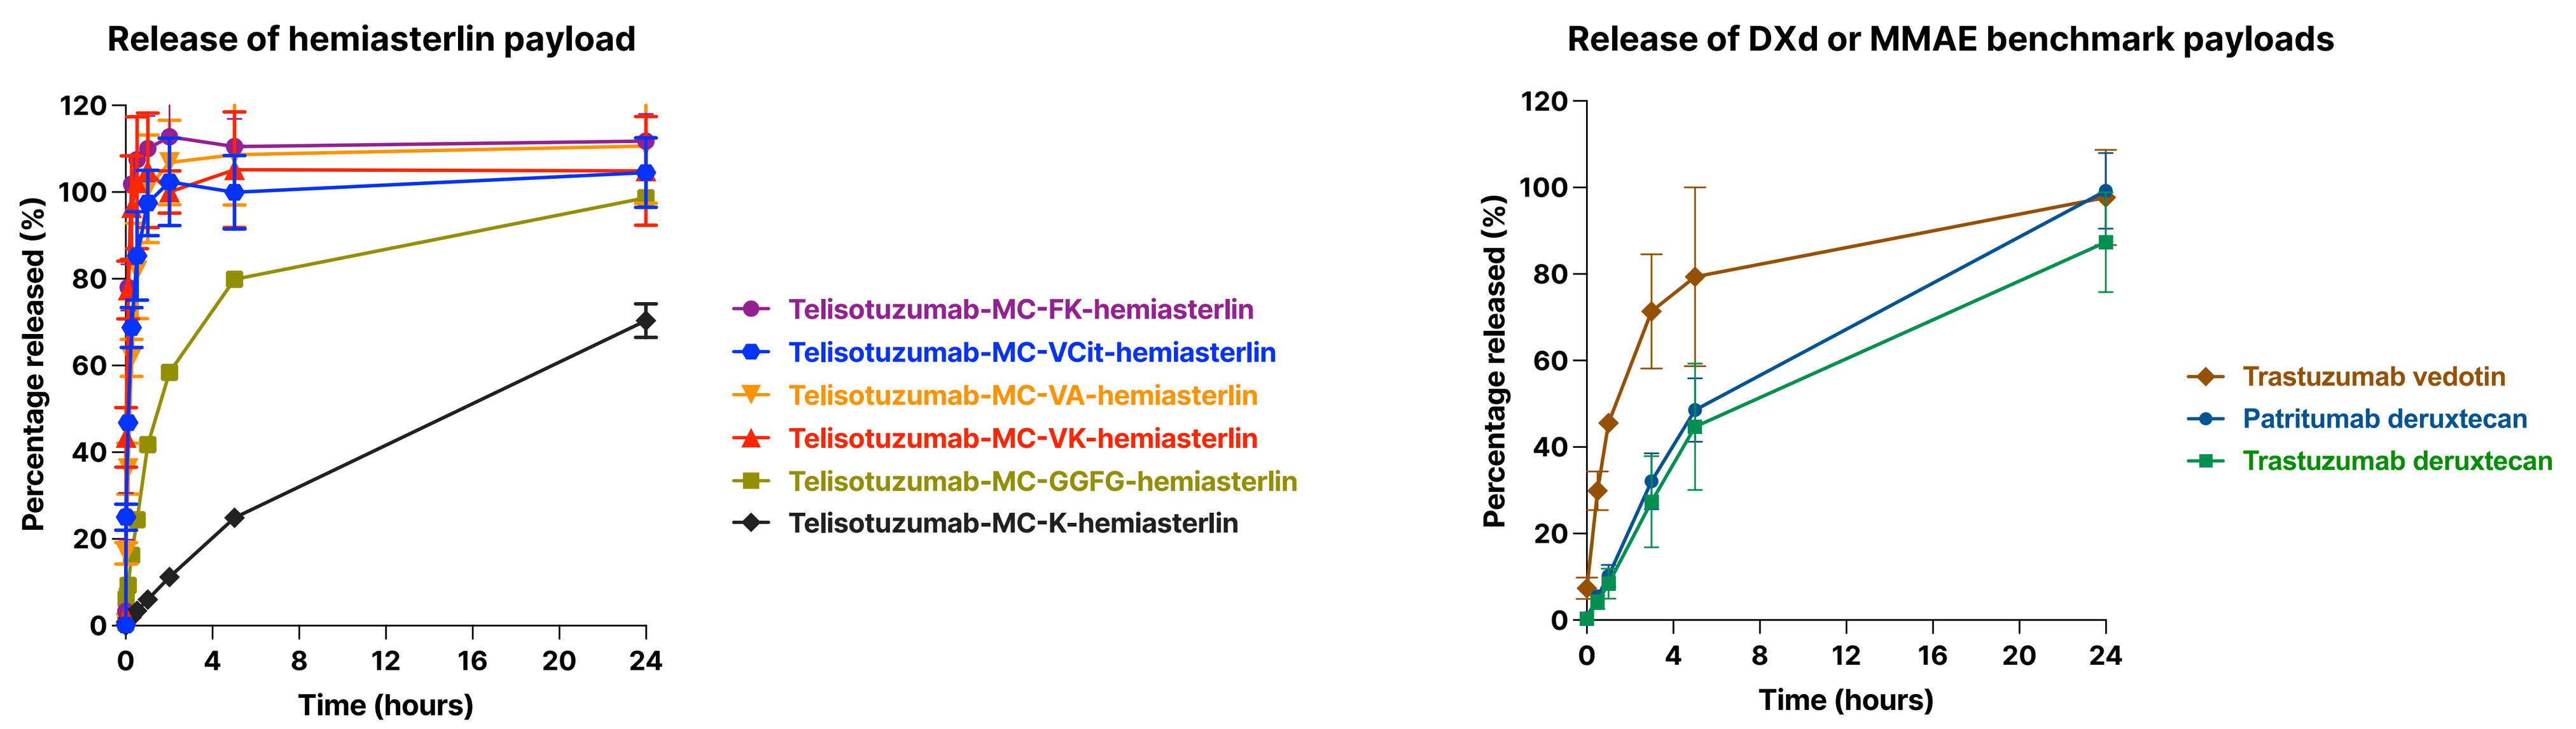
Release of hemiasterlin payload
Percentage released (%)
Time (hours)
Telisotuzumab-MC-FK-hemiasterlin
Telisotuzumab-MC-VCit-hemiasterlin
Telisotuzumab-MC-VA-hemiasterlin
Telisotuzumab-MC-VK-hemiasterlin
Telisotuzumab-MC-GGFG-hemiasterlin
Telisotuzumab-MC-K-hemiasterlin
Release of DXd or MMAE benchmark payloads
Trastuzumab vedotin
Patritumab deruxtecan
Trastuzumab deruxtecan

Figure 6. Released payload from ADCs was measured by spiking human lysosomal extract with the corresponding test article (1-3 µM). Released payload was measured using LC-MS. Notable disconnection from linker cleavage rate in a biochemical assays and *in vitro* cytotoxicity were also reported by P. L. Salomon et al. (Ref. 3).

In vitro evaluation of cMet and palivizumab hemiasterlin ADCs with different linkers highlights target-dependent cytotoxicity and instability of certain linkers

EBC-1 = cMet high
HCC827 = cMet moderate
HT-29 = cMet moderate
BT-20 = cMet low

Assay	Measurement	cMET hemiasterlin ADCs: VCit	VA	VK	FK	K	GGFG	Palivizumab hemiasterlin ADCs: VCit	VA	VK	FK	K	GGFG
EBC-1 cytotoxicity	EC_{50} (nM)	0.03	0.02	0.02	0.02	0.02	0.02	>60	>60	10	4.0	>60	16
HT-29 cytotoxicity	EC_{50} (nM)	0.20	0.16	0.13	0.10	0.10	0.20	>60	>60	60	7.9	>60	20
HCC827 cytotoxicity	EC_{50} (nM)	0.29	0.43	0.40	0.51	0.70	0.58	>60	>60	11	3.2	>60	>30
BT-20 cytotoxicity	EC_{50} (nM)	0.14	0.36	1.00	0.97	1.79	1.43	>60	>60	16	4.0	>60	>30
ADC stability in cultured growth medium	Payload released (% of theoretical) after 4 days in cultured growth medium	0.1%	0.3%	12.3%	24.5%	0.9%	2.2%	0.1%	0.3%	9.6%	26.0%	0.8%	2.9%

Table 2. Comparison of *in vitro* cytotoxicity and media stability of targeted vs non-targeted ADCs. For cytotoxicity assay, cancer cells were treated with a serial dilution of test sample and incubated under standard culturing conditions for 4 days. Following treatment, cell viability was quantified using an ATP quantification luminescent reagent. For ADC stability in cultured growth medium, growth media (RPMI 1640 supplemented with 10% fetal bovine serum) was collected after culturing EBC-1 cancer cells for 4 days under standard culture conditions followed by centrifugation to remove cells and debris. ADCs were incubated in cultured growth medium at 1.4 µM for 4 days and payload release (% of theoretical) was measured by mass spectrometry.

Conclusions

- It is important to be able to predict clinical toxicity profile, which largely depends on payload class and key ADC attributes.
- The poor translation of preclinical findings to predict ADC clinical toxicities has led to the empirical testing of ADC designs in patients.
- CFC assays using primary bone marrow cells can be used to evaluate toxicities of both ADCs and their payloads on blood progenitor cell lineages.
- Comparison of ADC clinical toxicities and *in vitro* CFC assay results indicates that the CFC assay can effectively recapitulate specific clinical observations, making it a valuable screening tool for ADCs.
- The prediction of clinical toxicity profiles for ADCs remains a complex challenge due to the intricate ADC disposition in humans.

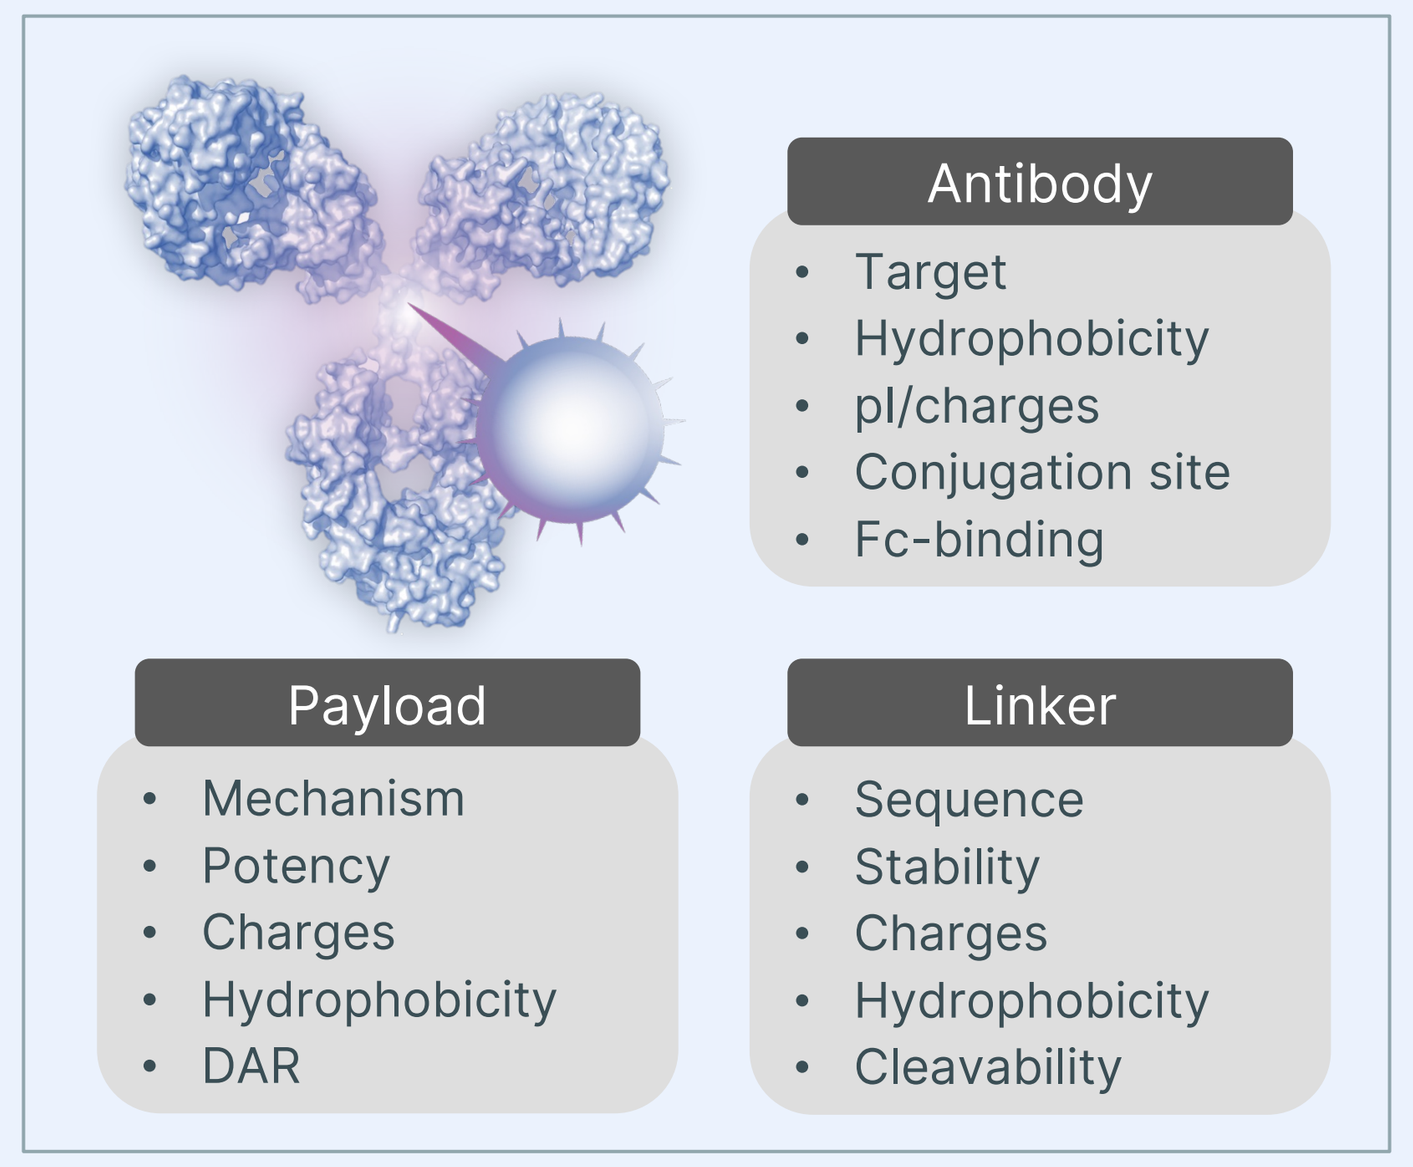
Antibody
Target
Hydrophobicity
pI/charges
Conjugation site
Fc-binding
Payload
Mechanism
Potency
Charges
Hydrophobicity
DAR
Linker
Sequence
Stability
Charges
Hydrophobicity
Cleavability

Figure 7. Schematic representation of ADC key molecular attributes influencing target and off-target toxicities.

References

1. I. E. Krop et al. *J. Clin. Oncol.* **2023**, *41*, 5550-5560
2. S. Modi et al. *N. Engl. J. Med.* **2020**, *382*, 610-621
3. P. L. Salomon et al. *Pharmaceutics* **2019**, *16*, 4817-4825

AACR Annual Meeting 2025