Mechanistic QSP modeling and translational strategy for determining an FIH dose for ZW171, a bispecific 2+1 T-cell engager molecule targeting mesothelin and CD3

Abstract number: 1062
SITC 2024

Nicole Afacan[1*], Carter L. Johnson[2*], Georgi I. Kapitanov[2], Saheli Sarkar[2], Kurt Stahl[1], Patricia Zwierzchowski[1], Chayne Piscitelli[1], Thomas Spreter Von Kreudenstein[1], Paul A. Moore[1], Nina Weisser[1], Shang-Chiung Chen[1]

[1] Zymeworks Inc., Vancouver, BC, Canada; [2] Certara, Concord, MA, USA; * These authors contributed equally

Introduction

ZW171 is a clinical-stage humanized bispecific T-cell engager (TCE) molecule that can simultaneously bind to mesothelin (MSLN) on tumor cells and to CD3ε expressed on T-cells. ZW171 is comprised of a 2+1 antibody format that incorporates two single chain variable fragments (scFvs) that bind to the extracellular domain (ECD) of MSLN, and one fragment antigen binding domain (Fab) that binds to CD3ε. Binding of the molecule to CD3 on T-cells and MSLN on tumor cells results in the formation of a trimolecular complex (CD3ε-ZW171-MSLN), or a tetramolecular complex (CD3ε-ZW171-MSLN-MSLN) due to ZW171's ability to bind to two MSLN molecules. This mechanism of action mimics the standard immune synapse, triggering T cell activation and killing of MSLN+ tumor cells.

In contrast to traditional MABEL approach solely relying on the most sensitive *in vitro* assays[1,2], a Quantitative System Pharmacology (QSP) model was developed for ZW171 using *in vitro* data, pharmacokinetics (PK) data from cynomolgus monkey, and literature data (e.g., CD3 receptors per T cells, number of T cells in central and peripheral compartments, and clinical PK data of MSLN-targeting TCE) to facilitate the selection of ZW171 starting dose for phase 1 clinical study.

Method

The human QSP model was developed by a stepwise approach:

(1) An *in vitro* binding model (including cis-avidity of ZW171 (i.e. accumulated binding strength of ZW171 to two MSLN molecules) and the ability of ZW171 to crosslink between MSLN+ tumor cells and T-cells).

(2) A two compartment (central and peripheral) *in vivo* cynomolgus monkey model.

(3) A three compartment (central, peripheral and tumor) *in vivo* human model.

The *in vitro* model was used to capture cytotoxicity data[3] and establish criteria for starting and efficacious doses in the clinic. The *in vivo* cynomolgus monkey model was developed to capture PK data and include drug binding to targets (MSLN and CD3) *in vivo*. Human PK parameters of ZW171 were allometrically scaled from those of cynomolgus monkey[4,5], and the *in vivo* human model was constructed to determine the ZW171 doses at which the criteria outlined in the in vitro models were met.

Result

Model Diagram

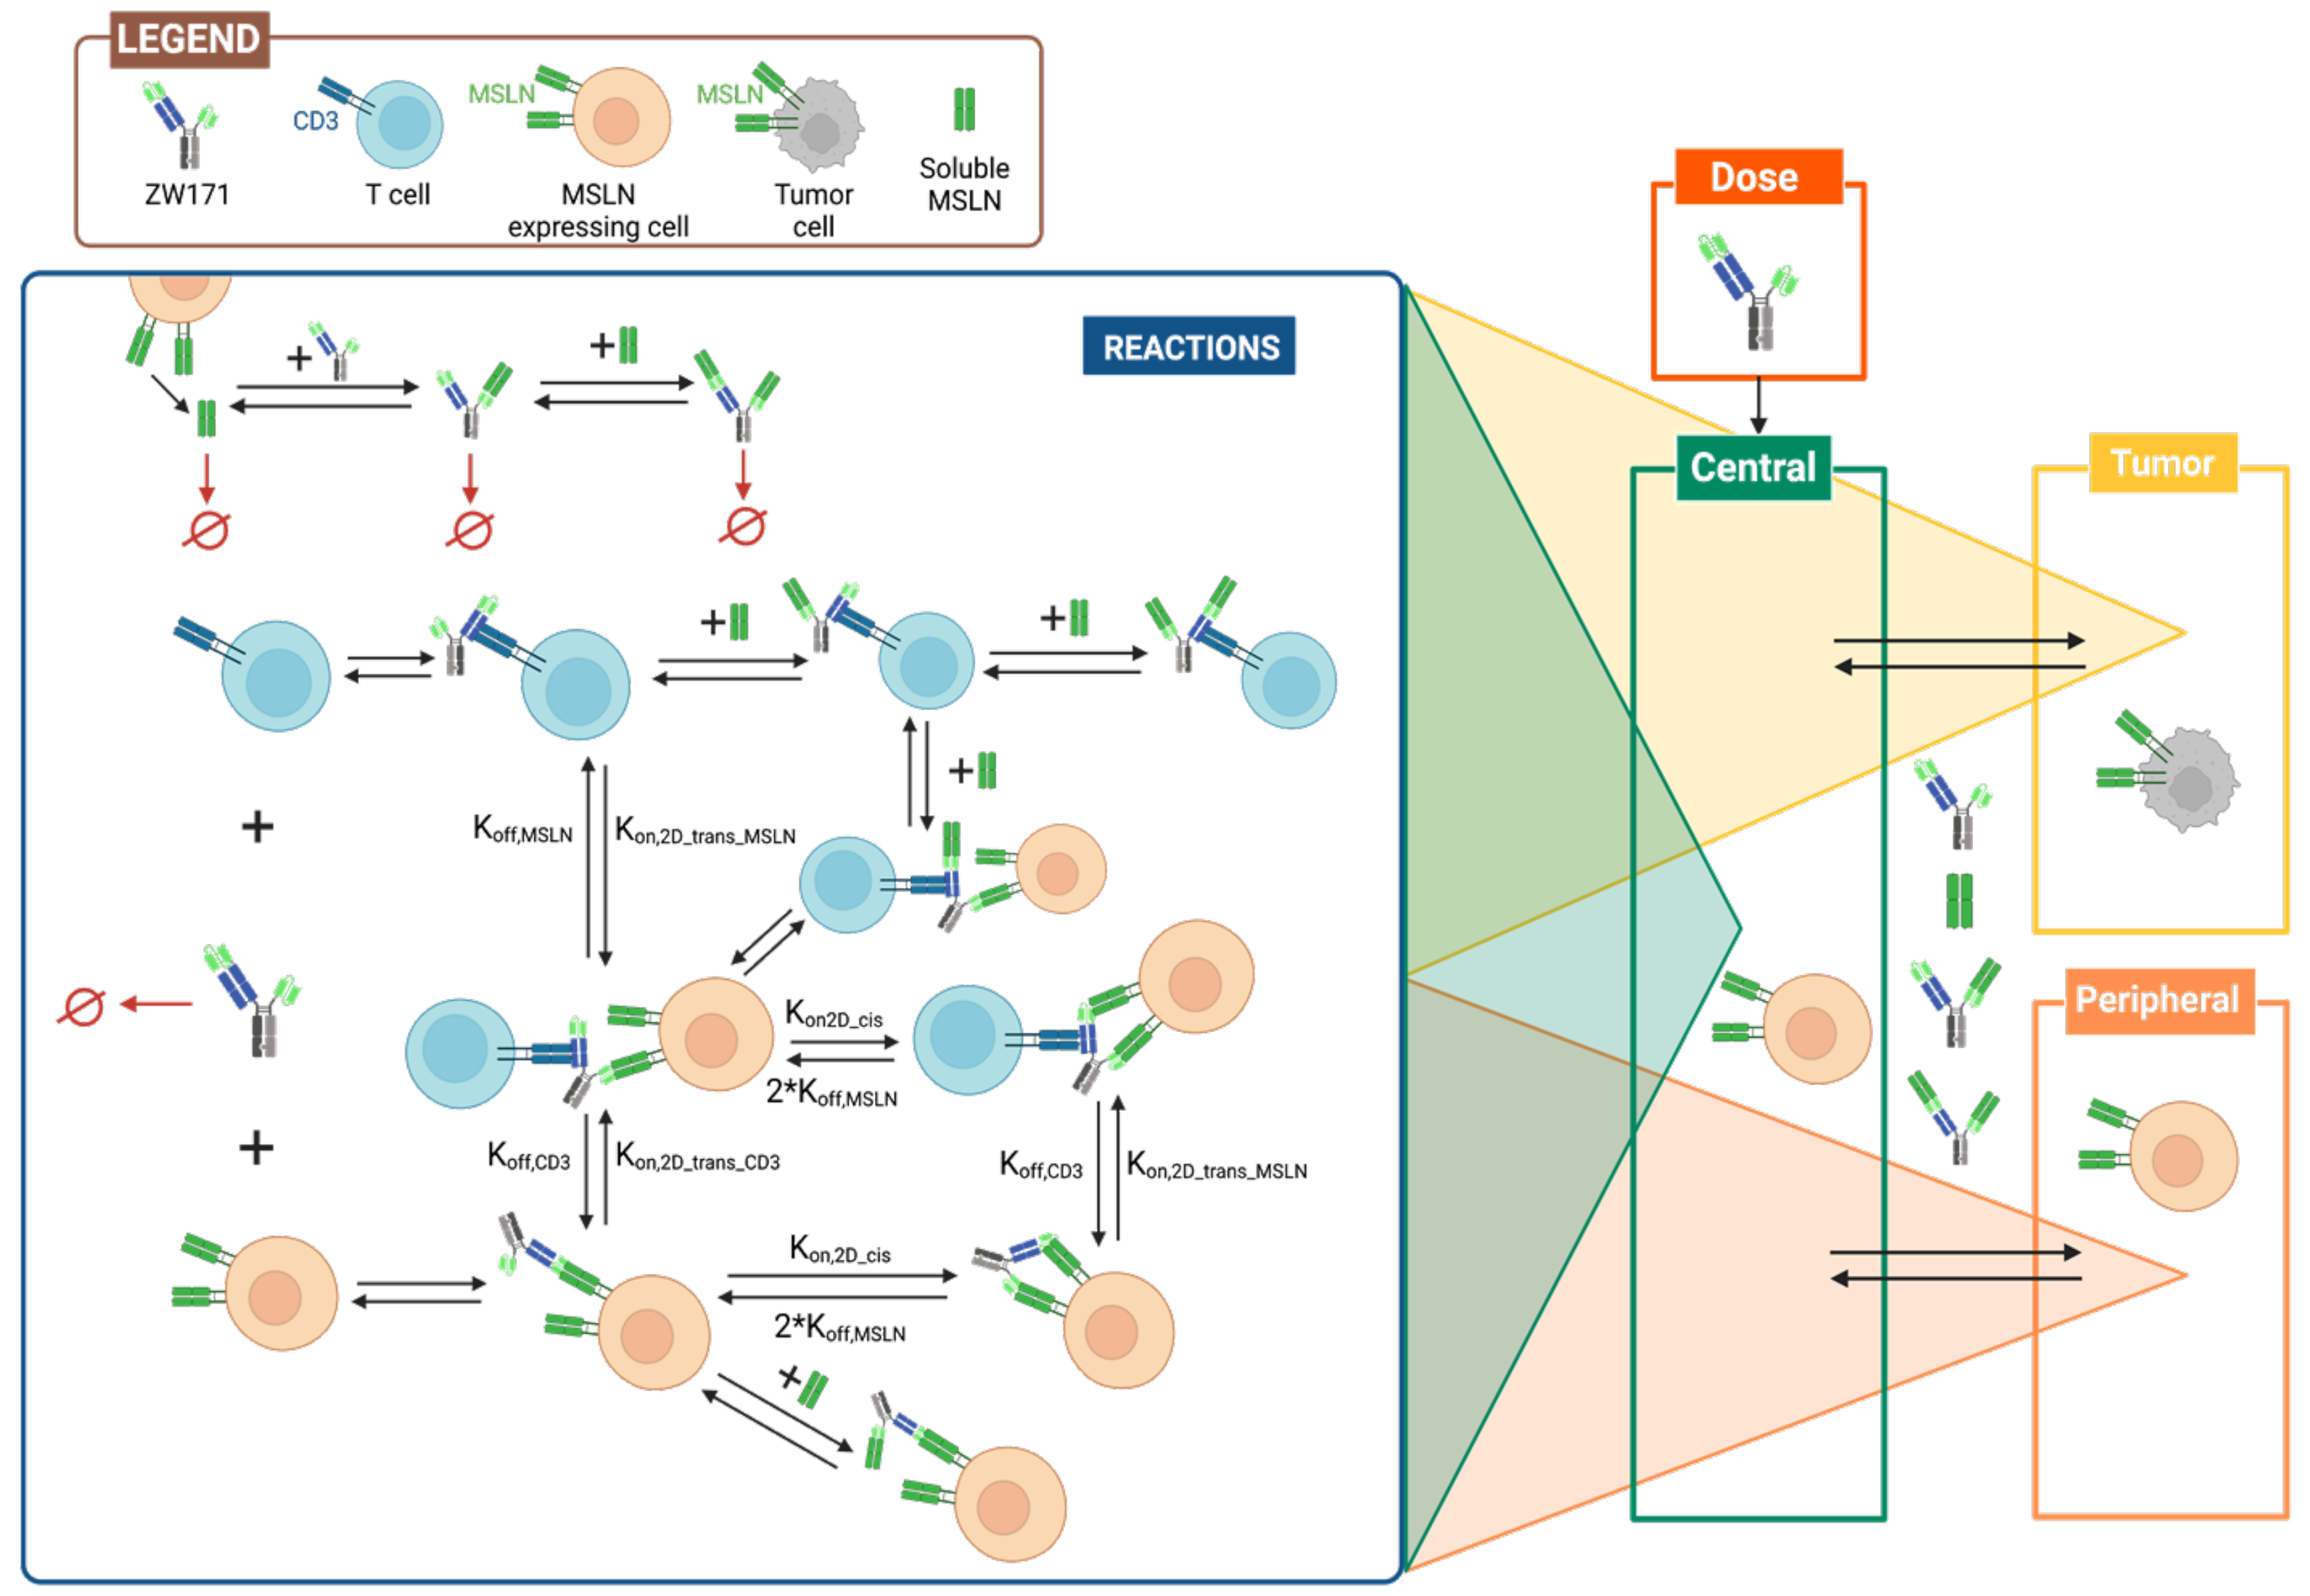
LEGEND
ZW171
CD3
T cell
MSLN
MSLN expressing cell
Tumor cell
Soluble MSLN
REACTIONS
Dose
Central
Tumor
Peripheral

In Vitro Model

- The *in vitro* model describes the observed receptor occupancy of CD3 on T cells and MSLN on OVCAR3 (human ovarian cancer cell) reasonably well (Fig. 1).
- The model recapitulates the observed *in vitro* cytotoxicity of MSLN-expressing OVCAR3 cancer cells in co-culture with PBMC (Fig. 2).

Figure 1. Model-predicted on-cell binding for CD3 and MSLN binding

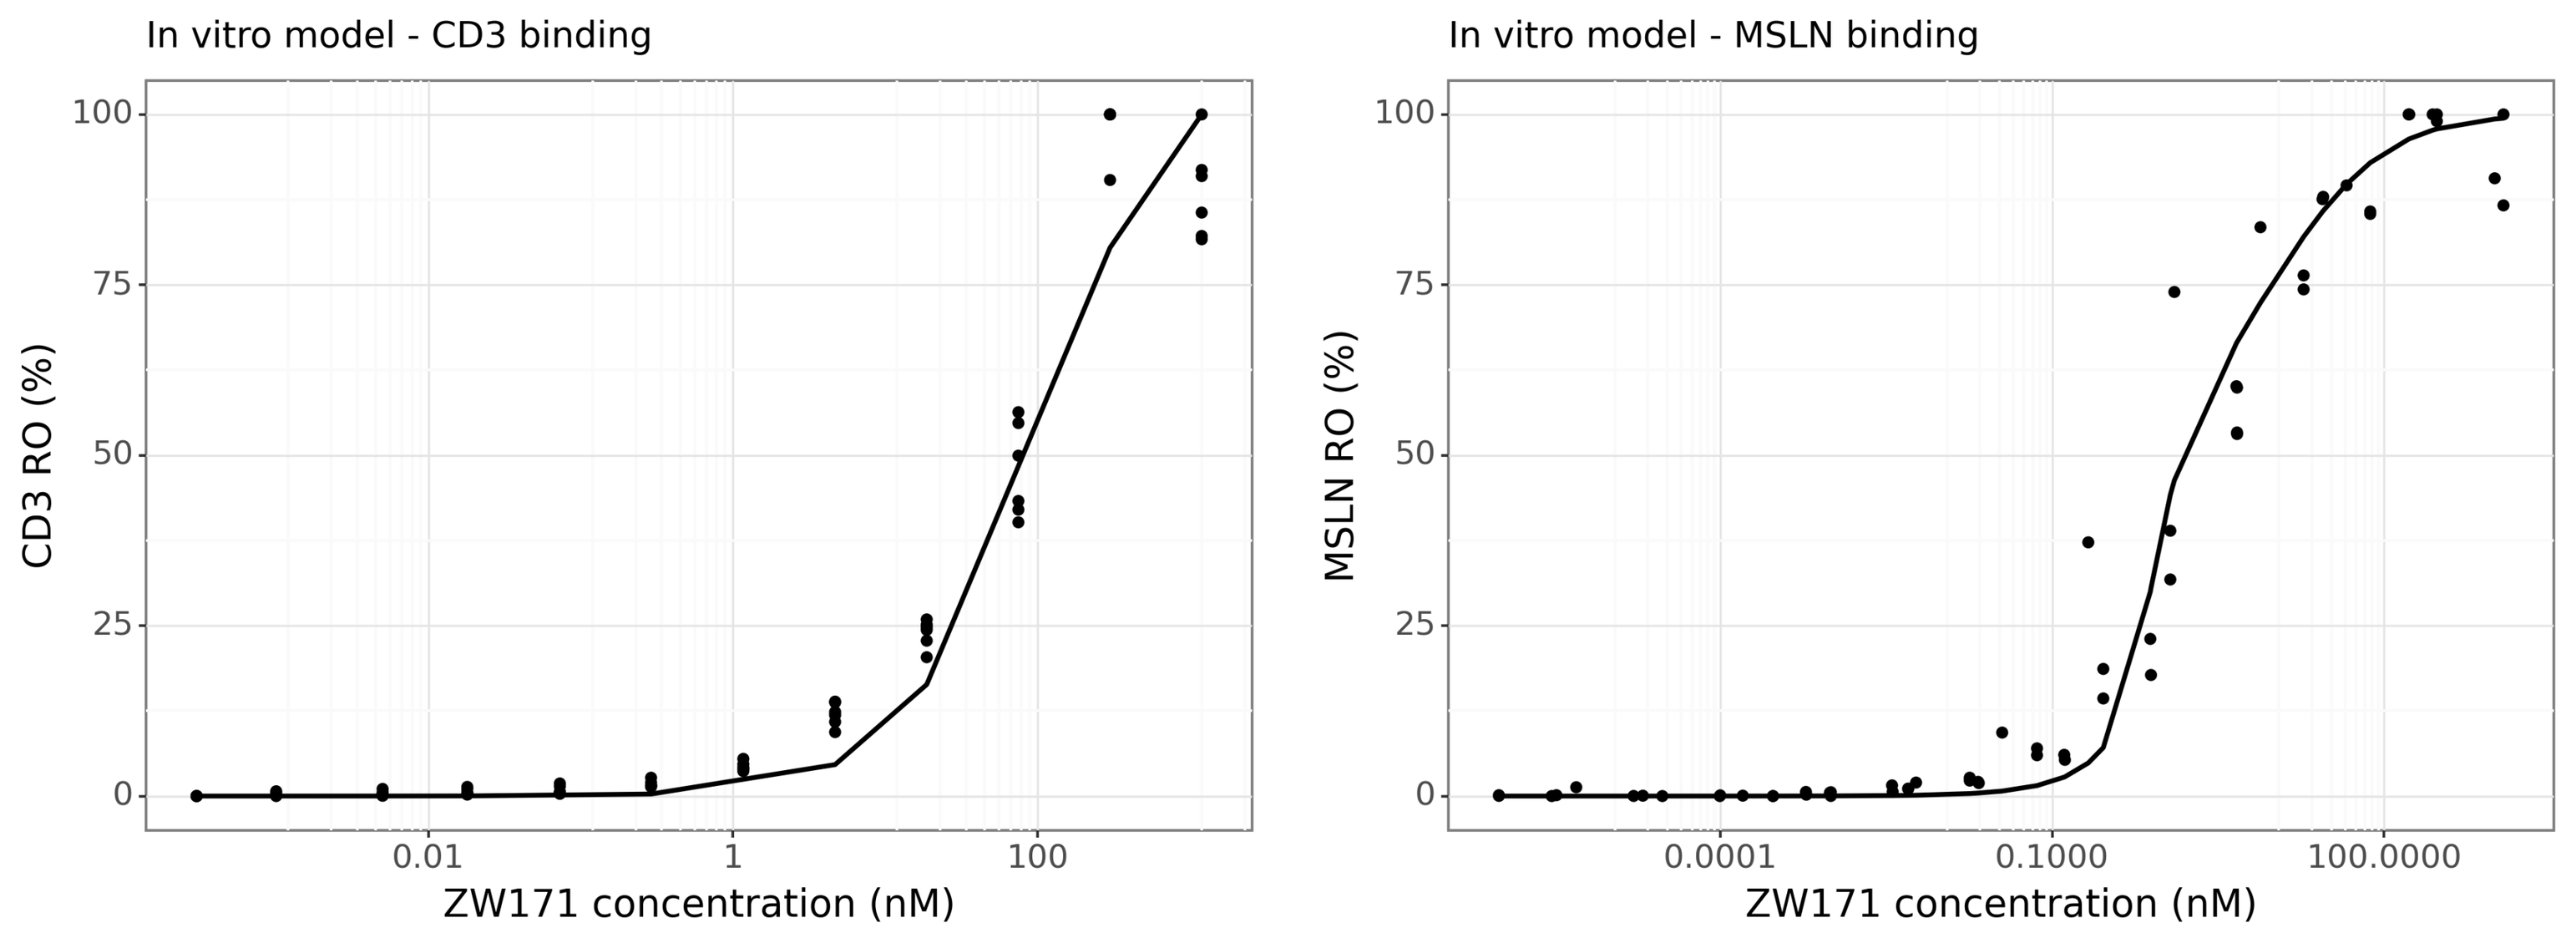
In vitro model - CD3 binding
CD3 RO (%)
ZW171 concentration (nM)
In vitro model - MSLN binding
MSLN RO (%)
ZW171 concentration (nM)

Figure 2. Model predictions and observed *in vitro* cytotoxicity data

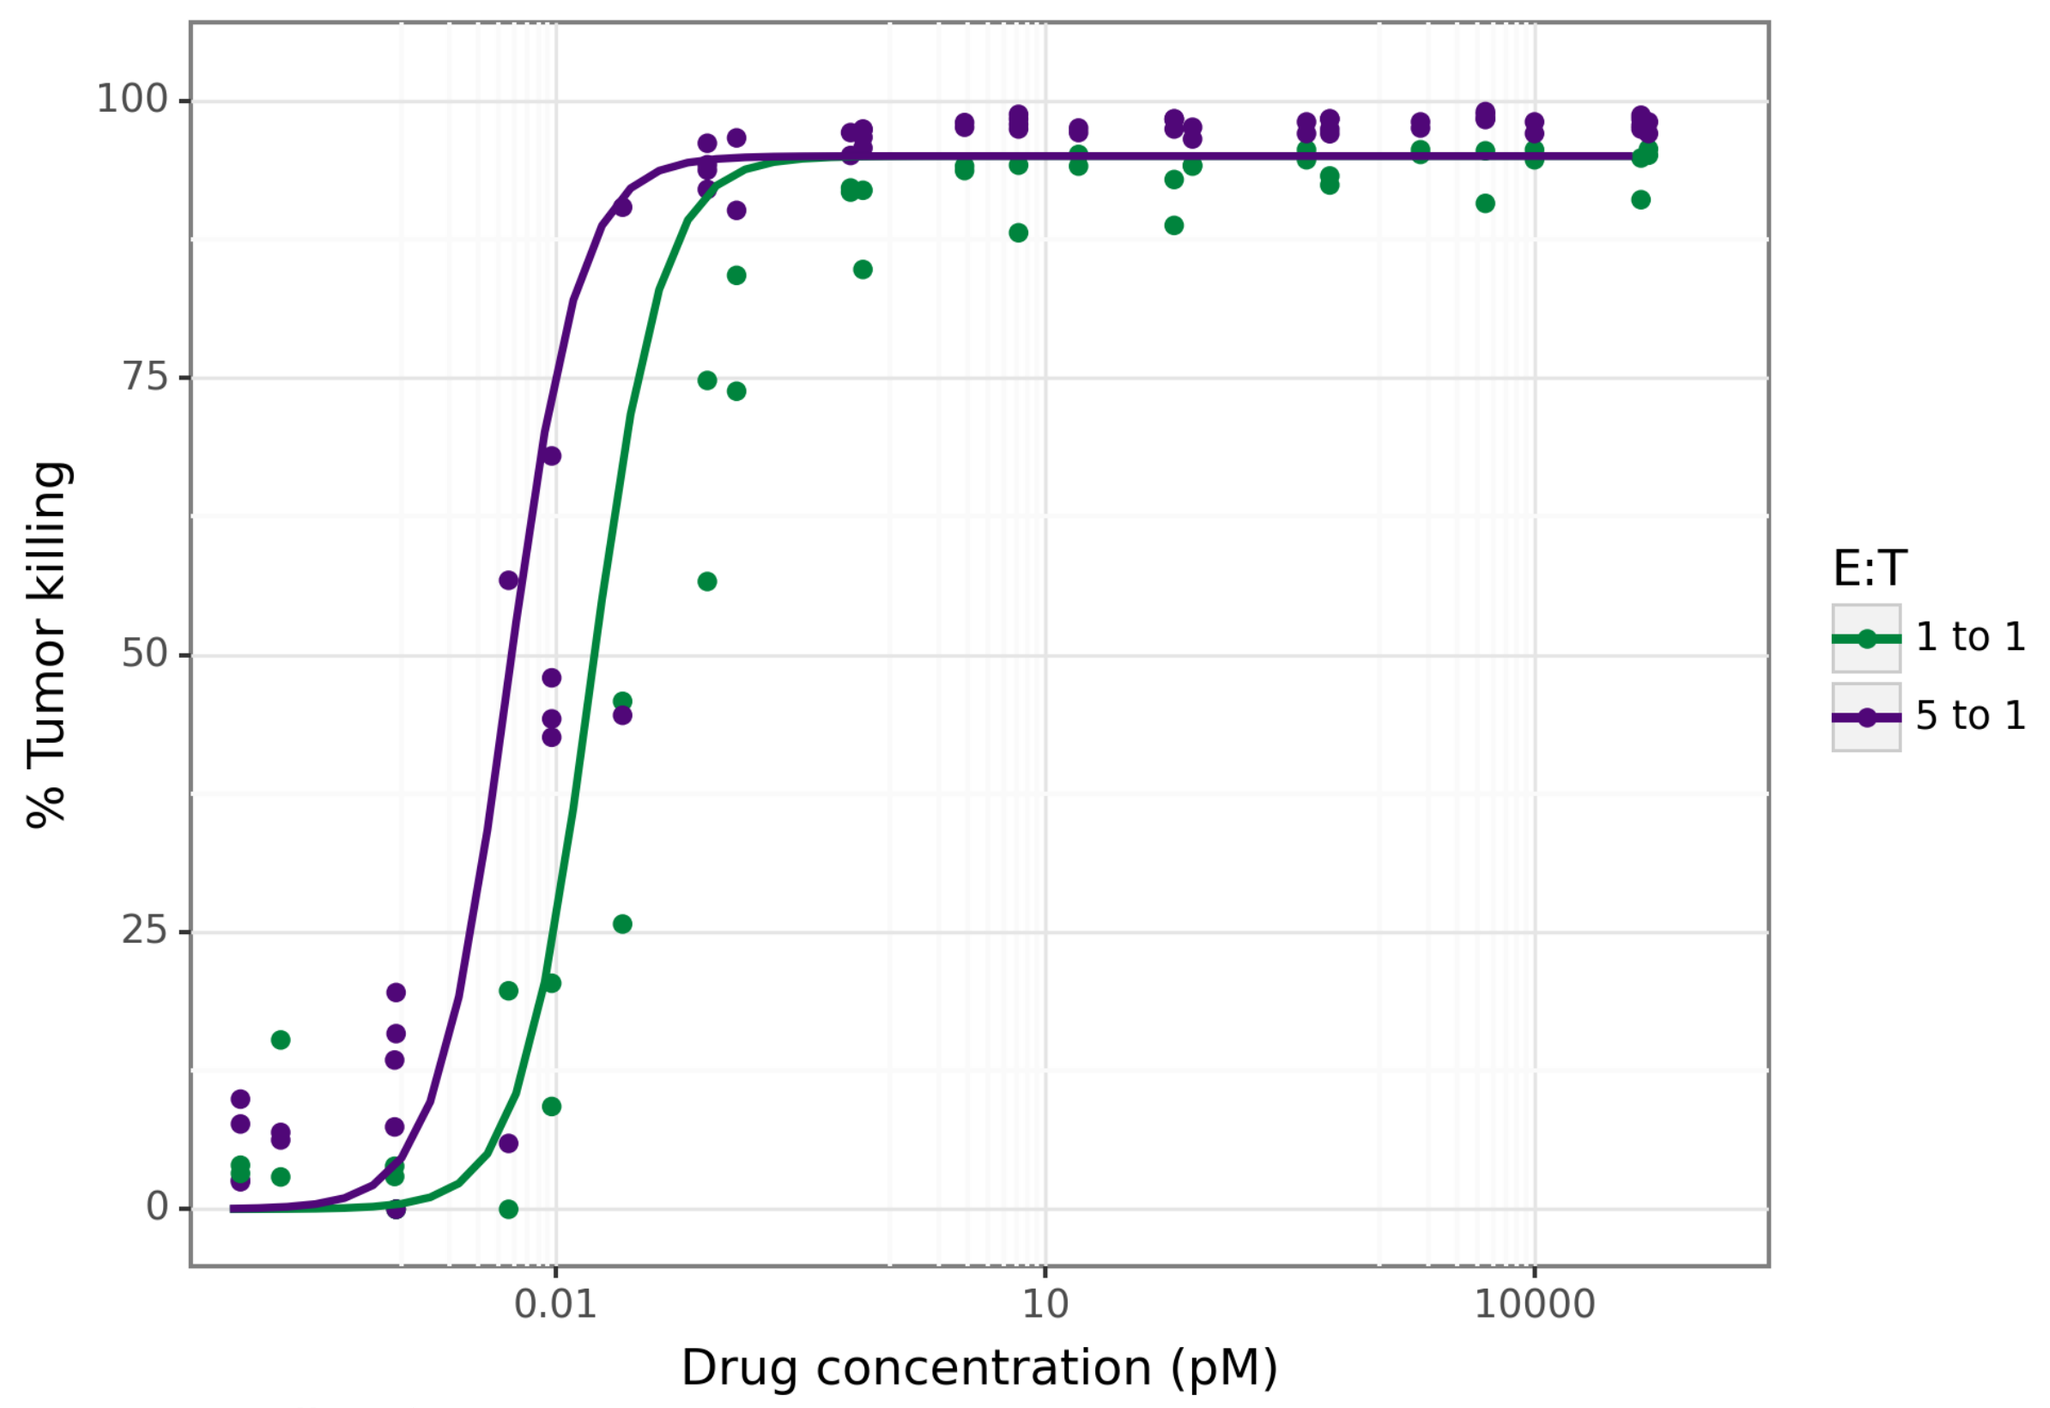
% Tumor killing
Drug concentration (pM)
E:T
1 to 1
5 to 1

E:T = Effector to Target cell ratio

Effective TPC (Trimer Per Cell) to Drive PD effect

- ZW171 can form both trimers and tetramers (due to 1:2 CD3:MSLN binding stoichiometry).
- To account for the formation of both TPC and TetPC, an **Effective TPC** was determined for ZW171 and compared to modelling results of a tool MSLNxCD3 bispecific molecule with 1:1 binding stoichiometry:

 Effective TPC = 5.4 x TetPC + TPC

TetPC = tetramer (CD3ε-ZW171-MSLN-MSLN tetramolecular complex) per cell
TPC = trimer (CD3ε-ZW171-MSLN trimolecular complex) per cell

Table 1. Effective TPC for the 5:1 and 1:1 E:T ratio

E:T Ratio	Criteria	ZW171 Concentration (pM)	Effective TPC
5:1	ET50	0.0080	3
	ET30	0.0034	1.3
	ET20	0.0020	0.75
	ET10	0.00089	0.33
1:1	ET50	0.047	5.5
	ET30	0.020	2.4
	ET20	0.012	1.4
	ET10	0.0052	0.61

Human QSP Model

Figure 3. Model-predicted ZW171 PK (left) and effective TPC (right) following single IV dose administration in human

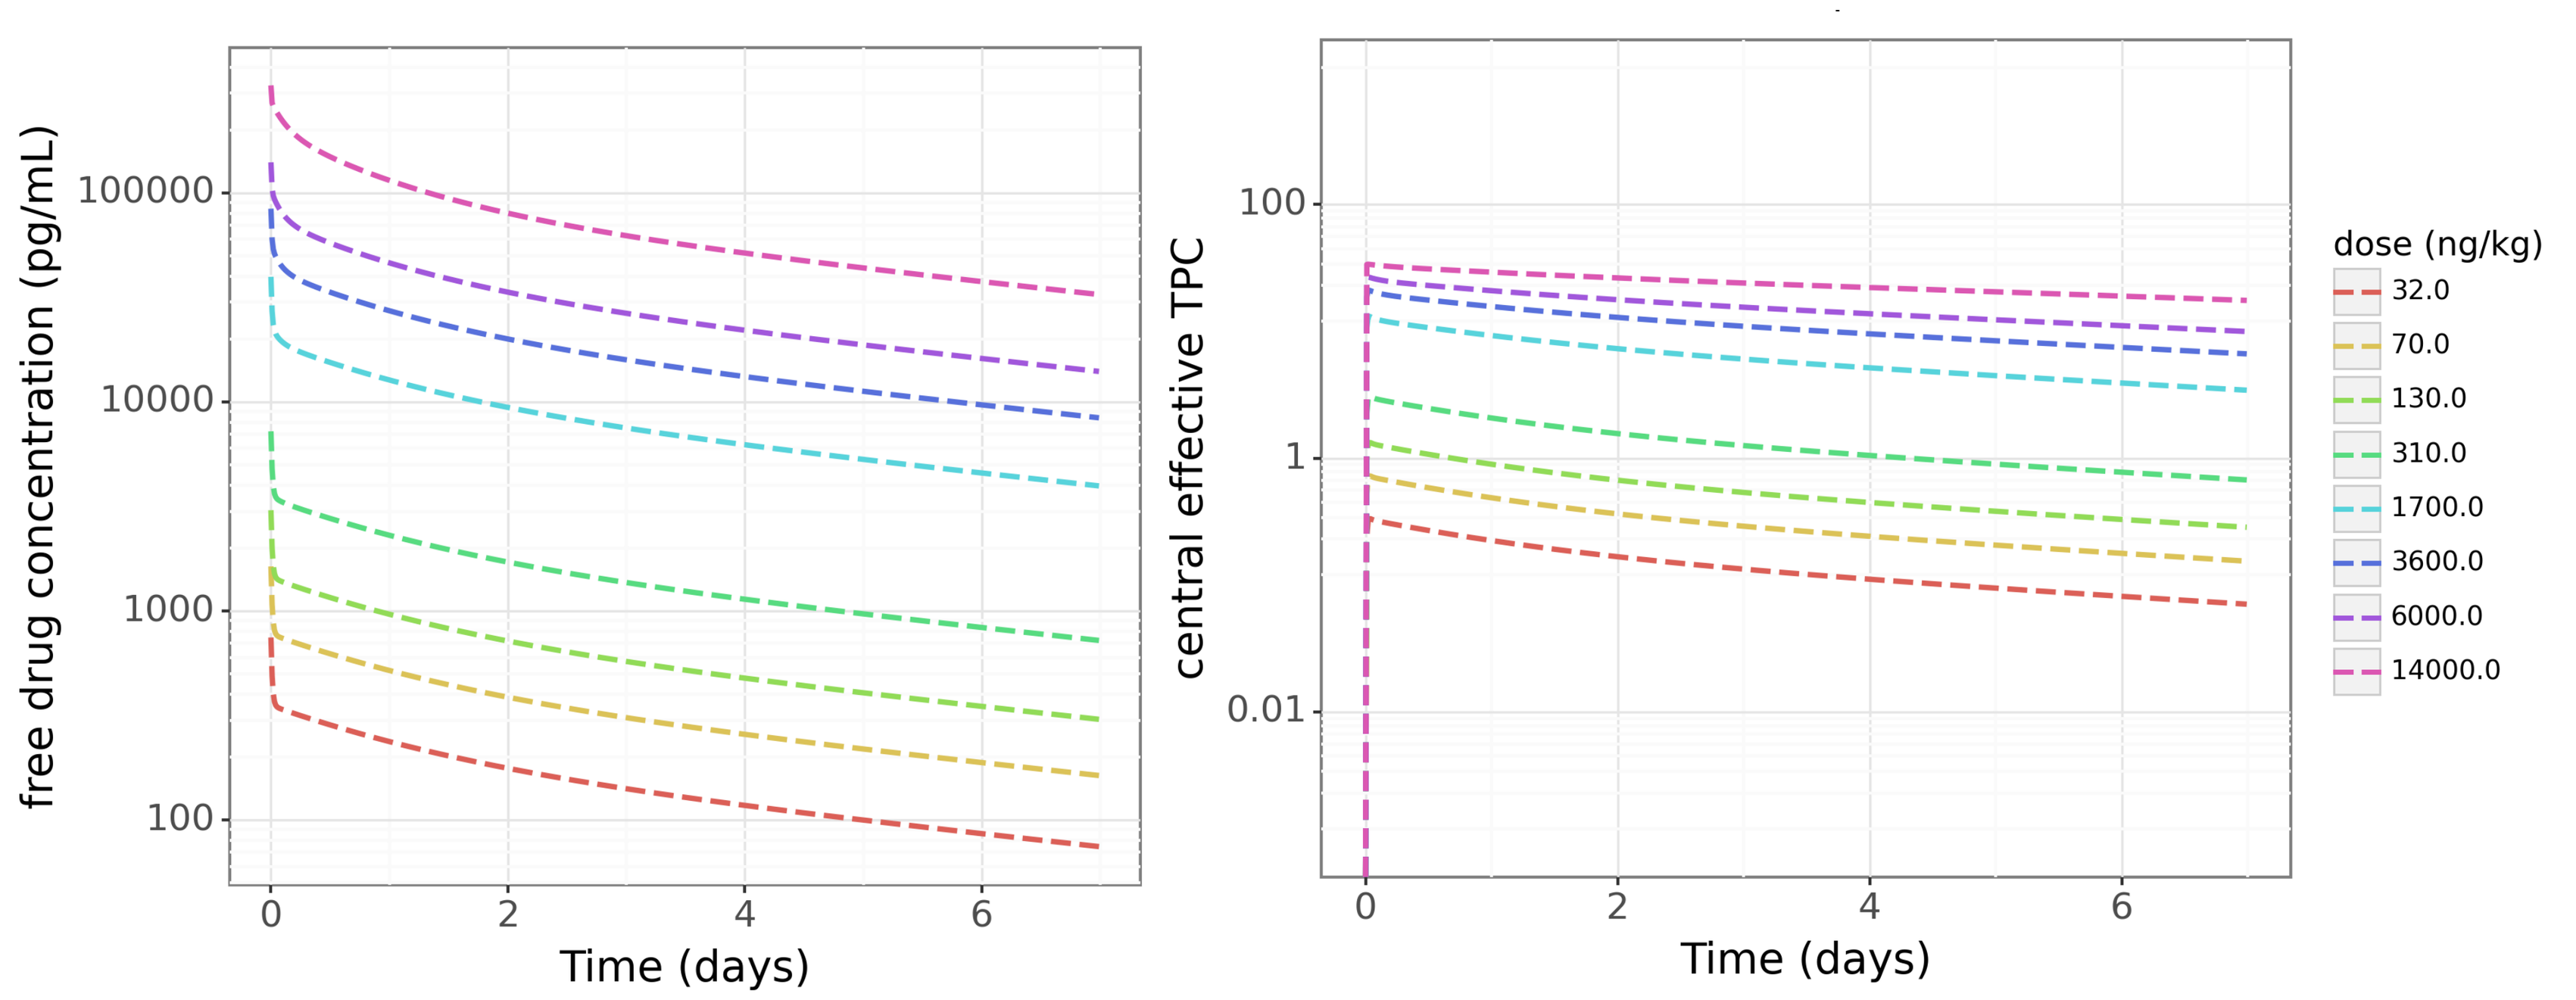
free drug concentration (pg/mL)
Time (days)
central effective TPC
Time (days)
dose (ng/kg)
32.0
70.0
130.0
310.0
1700.0
3600.0
6000.0
14000.0

Conclusion

- A MABEL approach is commonly used to select the starting dose for bispecific CD3 T-cell engagers in first-in-human (FIH) studies, though it is often considered overly conservative.
- The fit-for-purpose QSP model for ZW171 was established to integrate physiological information (e.g., T cells, MSLN+ cells, CD3/MSLN expression, receptor dynamics), *in vitro* cytotoxicity data, and cynomolgus monkey PK data.
- Unlike traditional MABEL, which relies solely on *in vitro* data, the QSP-based MABEL approach characterizes the complex interplay between ZW171 and its target antigens.
- The QSP-based MABEL approach was used to determine the starting dose for the ZW171 Phase 1 clinical study.

References

1. Dudal, Sherri et al. Application of a MABEL Approach for a T-Cell-Bispecific Monoclonal Antibody: CEA TCB *J Immunother.* 2016 Sep;39(7):279-89.
2. Schaller, Teilo H et al. First in human dose calculation of a single-chain bispecific antibody targeting glioma using the MABEL approach. *J Immunother Cancer.* 2020 Apr;8(1):e000213.
3. Afacan N. et al. (2023, April). ZW171, a T Cell-Engaging, Bispecific Antibody for the Treatment of Mesothelin-Expressing Solid Tumors [Poster presentation]. American Association for Cancer Research Annual Meeting, Orlando, FL.
4. Betts A. et al. Linear pharmacokinetic parameters for monoclonal antibodies are similar within a species and across different pharmacological targets: A comparison between human, cynomolgus monkey and hFcRn Tg32 transgenic mouse using a population-modeling approach. *MAbs.* 2018 Jul;10(5):751-764.
5. Betts A. et al. Mechanistic Quantitative Pharmacology Strategies for the Early Clinical Development of Bispecific Antibodies in Oncology. *Clin Pharmacol Ther.* 2020 Sep;108(3):528-541.

Acknowledgements
We would like to thank Ritesh Korat and Siran Cao for the contribution to this project.
This study was sponsored by Zymeworks Inc.

zymeworks