TriTCE Co-Stim: A next generation trispecific T cell engager platform with integrated CD28 costimulation, engineered to improve T cell function and antitumor responses in hard-to-treat cancers

Abstract # C091

Lisa Newhook, Purva Bhojane, Peter Repenning, Desmond Lau, Kurt Stahl, Diana Canals Hernaez, Alec Robinson, Diego Perez Escanda, Mariana Rocha, Aditi Deshmukh, Begonia Silva Moreno, John Zhang, Kesha Patel, Marylou Vallejo, Bing Catherine Wu, Gavin Storoschuk, Polly Shao, Nichole Escalante, Chayne L. Piscitelli, Nicole Afacan, Alexandra Livernois, Gursev Anmole, Prajwal Raghunatha, Paul Moore, Thomas Spreter von Kreudenstein, Nina E. Weisser

Introduction

Low T cell infiltration and T cell anergy are challenges for the treatment of solid tumors with conventional CD3-engaging bispecific T cell engagers (TCEs)[1]. By providing balanced activation of "Signal 1" (CD3) and "Signal 2" (CD28) in a single molecule, co-stimulatory trispecific TCEs (TriTCE Co-Stim) have the potential to induce more sustainable T cell responses in the tumor and increase therapeutic responses beyond that achievable with bispecific TCE. By enhancing T cell responses, TriTCE Co-Stim have the potential to increase the depth and durability of anti-tumor responses in patients with difficult to treat solid tumors with low T cell infiltration and poor T cell function.

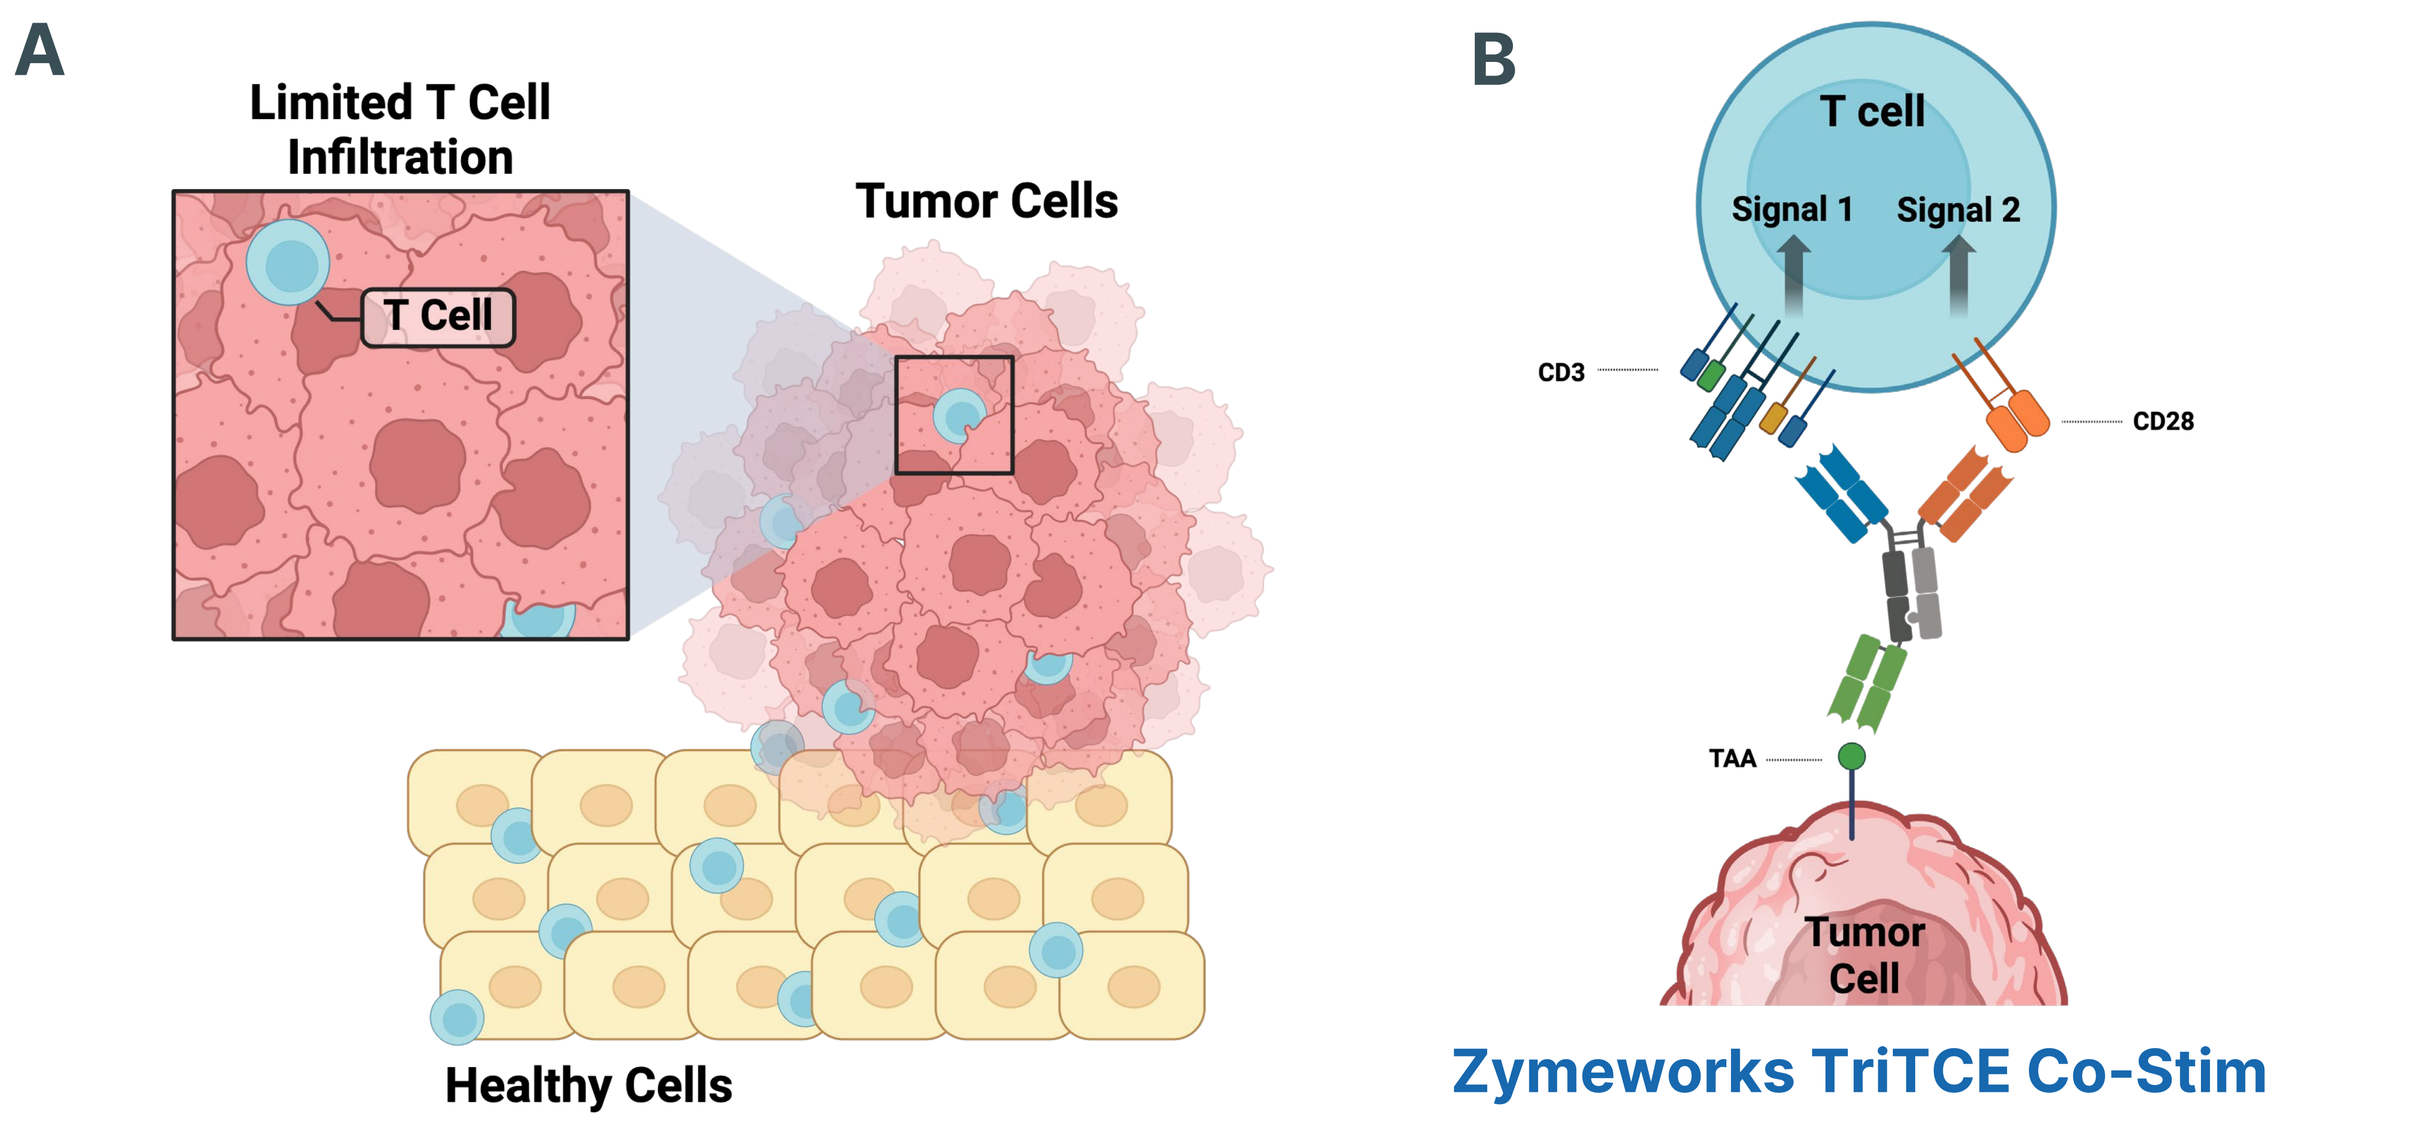
A
Limited T Cell Infiltration
T Cell
Tumor Cells
Healthy Cells
B
T cell
Signal 1
Signal 2
CD3
CD28
TAA
Tumor Cell
Zymeworks TriTCE Co-Stim

Figure 1. Proposed mechanism of action for Zymeworks' TriTCE Co-Stim. Schematic of limited T cell infiltration in solid tumors¶ (**A**). Schematic of TriTCE Co-Stim-mediated T cell activation in solid tumors¶ (**B**). TriTCE Co-Stim is designed to provide tumor-associated antigen (TAA) dependent agonism of Signal 1 (CD3) and Signal 2 (CD28) in a single molecule to increase T cell activation, fitness, and proliferation.

TriTCE Co-Stim: From Concept to Platform

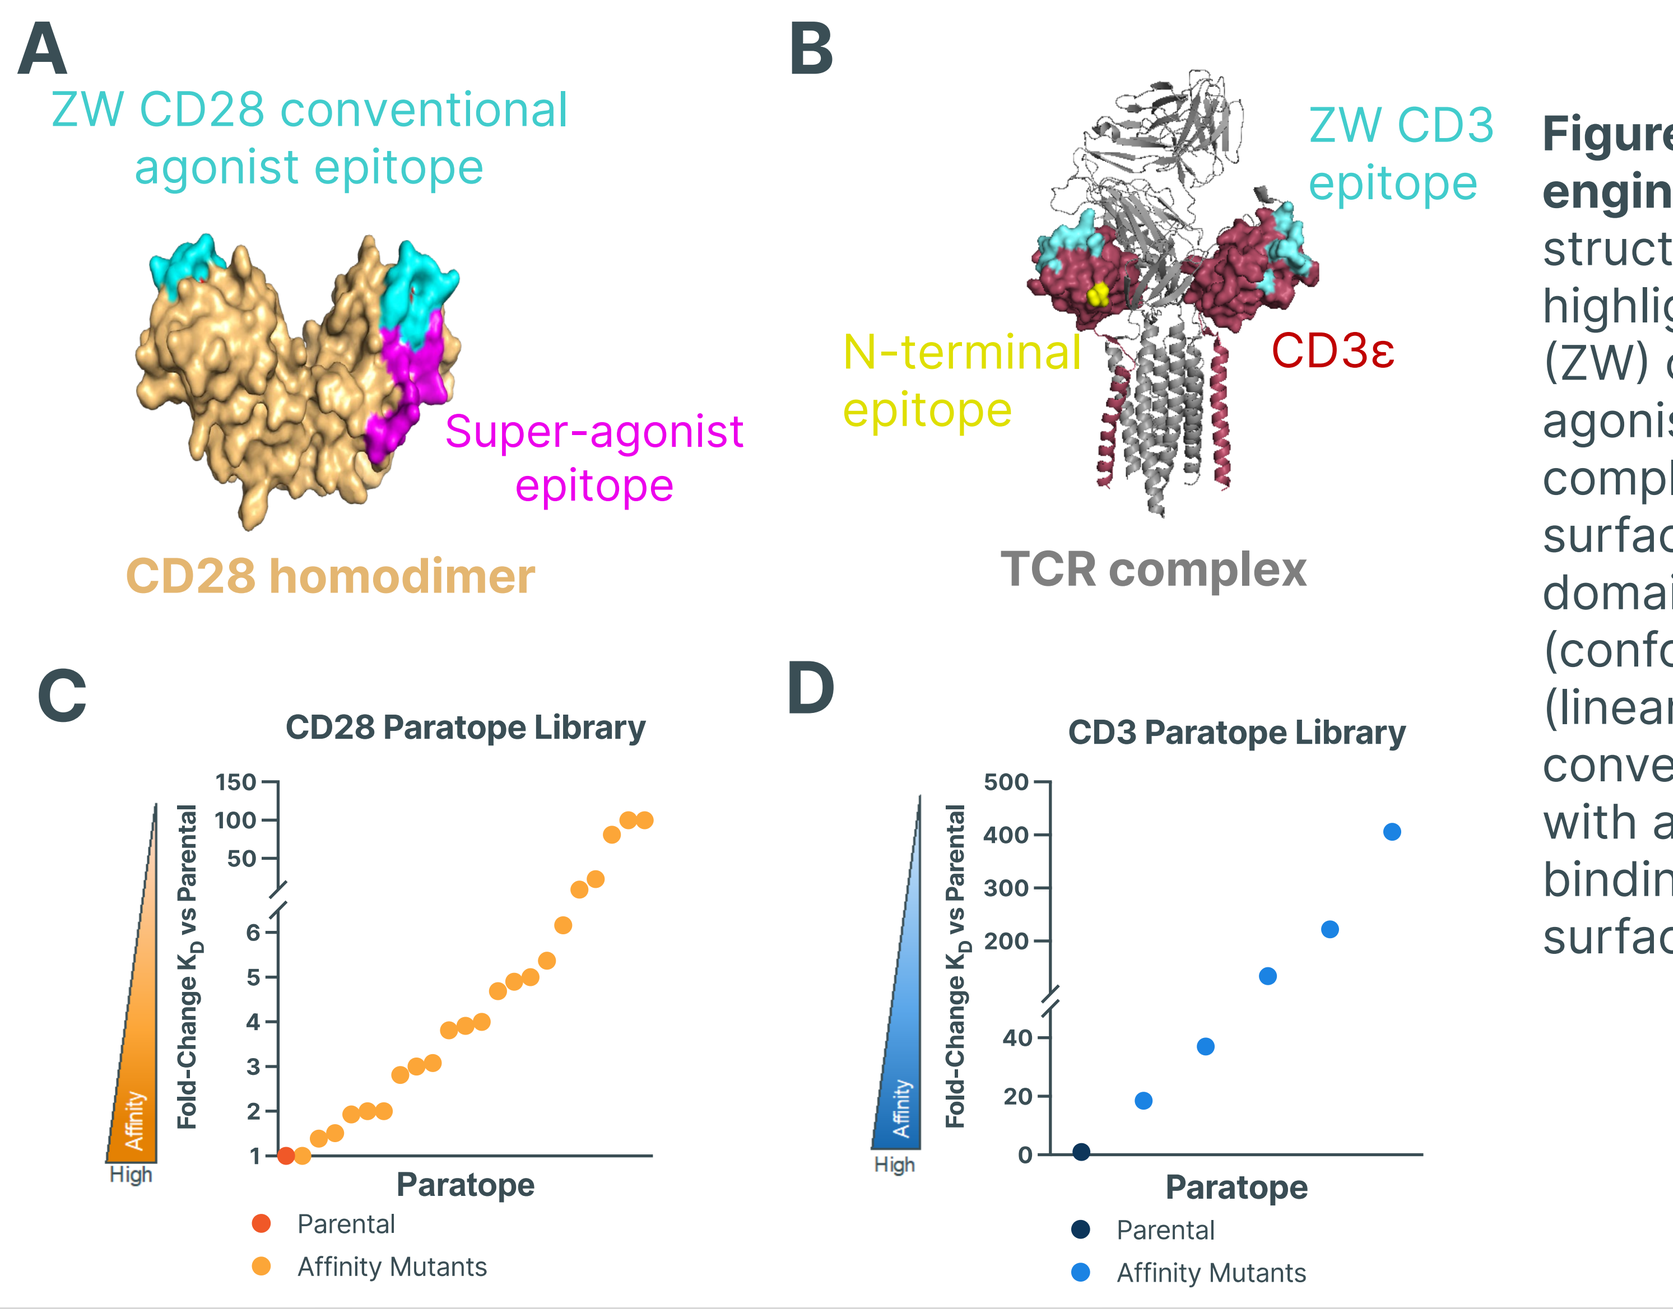
A
ZW CD28 conventional agonist epitope
Super-agonist epitope
CD28 homodimer
B
ZW CD3 epitope
N-terminal epitope
CD3ε
TCR complex
C
CD28 Paratope Library
Fold-Change K_D vs Parental
Affinity
High
Paratope
Parental
Affinity Mutants
D
CD3 Paratope Library
Fold-Change K_D vs Parental
Affinity
High
Paratope
Parental
Affinity Mutants

Figure 2. CD3 and CD28 paratope engineering. (A) CD28 homodimer structure (modelled using 1YJD) highlighting epitopes for Zymeworks' (ZW) conventional agonist vs super-agonist antibodies. **(B)** Full TCR complex (modelled using 7FJD) with surface representation of CD3ε domain, highlighting for ZW (conformational) vs the N-terminal (linear) epitopes. Libraries of conventional agonist paratope variants with a range of CD28 **(C)** and CD3 **(D)** binding affinities determined by surface plasmon resonance (SPR).

TriTCE Co-Stim formats exhibit antibody-like developability with differential *in vitro* properties[2]

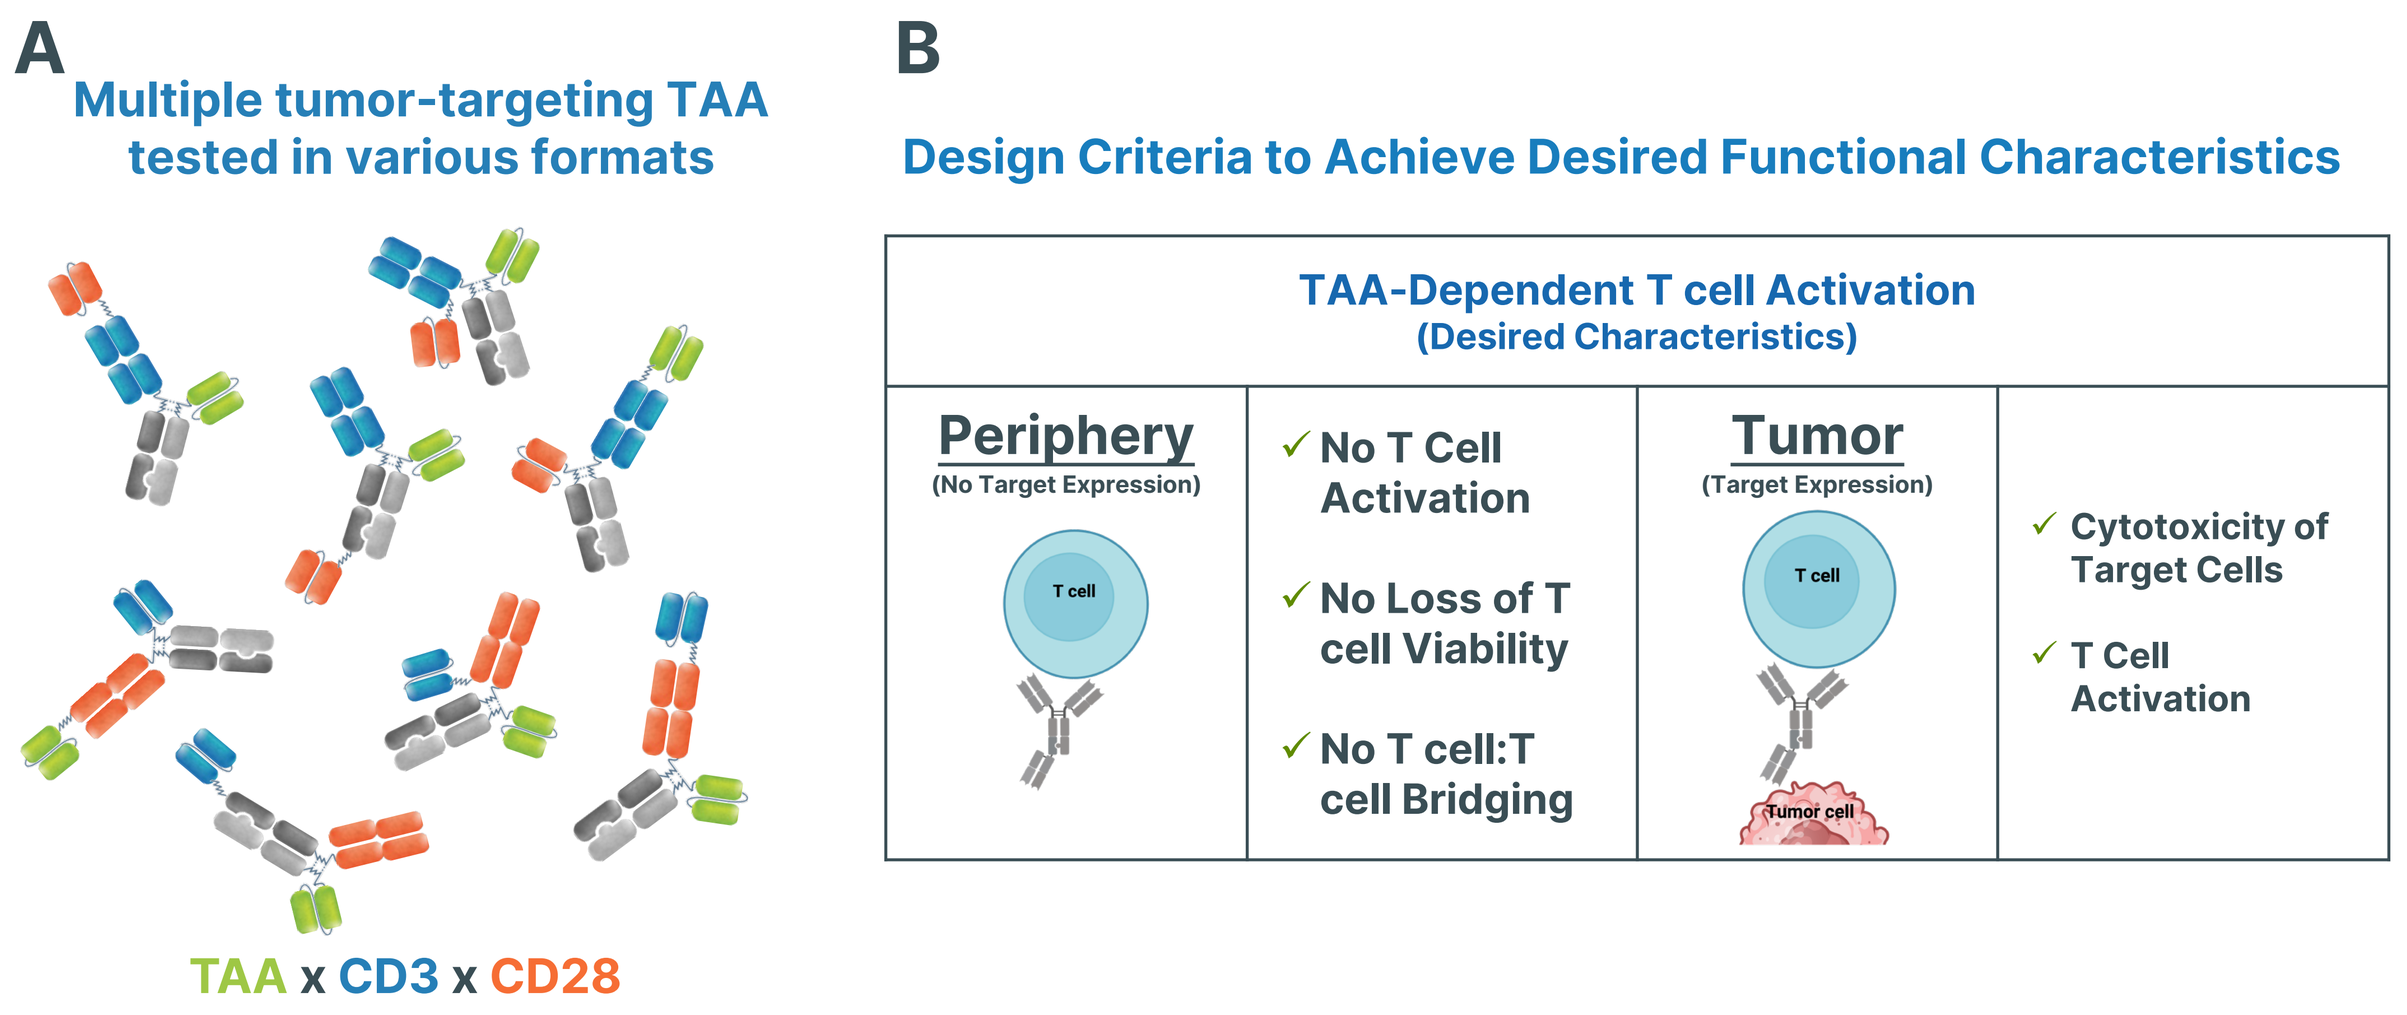
A
Multiple tumor-targeting TAA tested in various formats
TAA x CD3 x CD28
B
Design Criteria to Achieve Desired Functional Characteristics
TAA-Dependent T cell Activation
(Desired Characteristics)
Periphery
(No Target Expression)
T cell
✓ No T Cell Activation
✓ No Loss of T cell Viability
✓ No T cell:T cell Bridging
Tumor
(Target Expression)
Tumor cell
✓ Cytotoxicity of Target Cells
✓ T Cell Activation

C

Representative CLDN18.2 *in vitro* Screening to Assess Desired Functional Characteristics

Antibody Format	1	2	3	4	5	6	7	8	9	10
Target-Dependent?	✓	✓	✓	x	✓	x	x	x	x	x
Cytotoxic Potency										
Yield (mg/L)										
Stability (T_m; °C)										

Low – High

Figure 3. TriTCE Co-Stim antibodies with various paratope formats and geometries are engineered using the Azymetric™ and EFECT™ platforms to optimize the therapeutic window. Schematic representation of a subset of formats tested using multiple tumor targeting TAA. (**A**) Summary of desired target-dependent properties of TriTCE Co-Stim achieved by optimized format design¶ (**B**). TriTCE Co-Stim formats that exhibit potent cytotoxicity of target cells, target-dependency, acceptable yield, and thermal stability are selected through extensive screening *in vitro* (**C**).

TriTCE Co-Stim Lead Format Selection

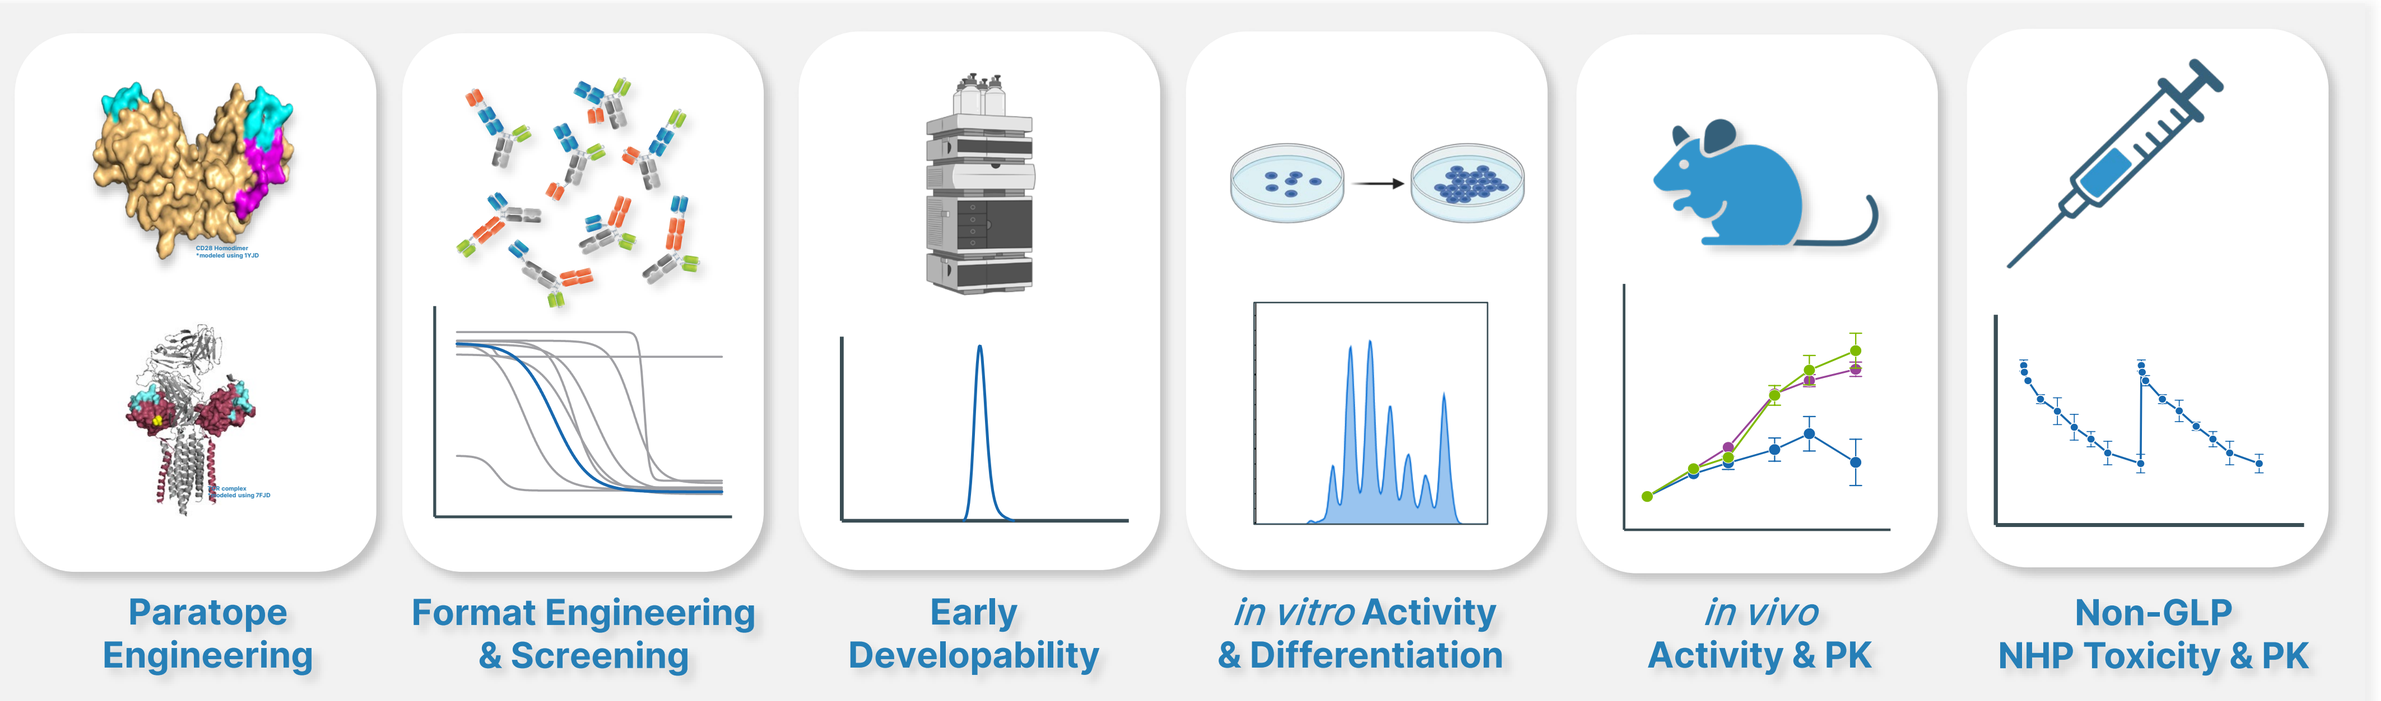
Paratope Engineering
Format Engineering & Screening
Early Developability
in vitro Activity & Differentiation
in vivo Activity & PK
Non-GLP NHP Toxicity & PK

TriTCE Co-Stim platform tested with three targets including CLDN18.2[3] and DLL3[4]

- Assessed TriTCE Co-Stim relative to first generation CD3-engaging bispecific TCEs
- Interrogated mechanism of T cell engagement relative to benchmark trispecific TCEs
- Determined safety in NHP tox for CLDN18.2 lead TriTCE Co-Stim

Figure 4. Workflow Established for the Development of TriTCE Co-Stim Platform¶.

Enhanced T-cell mediated Cytotoxicity

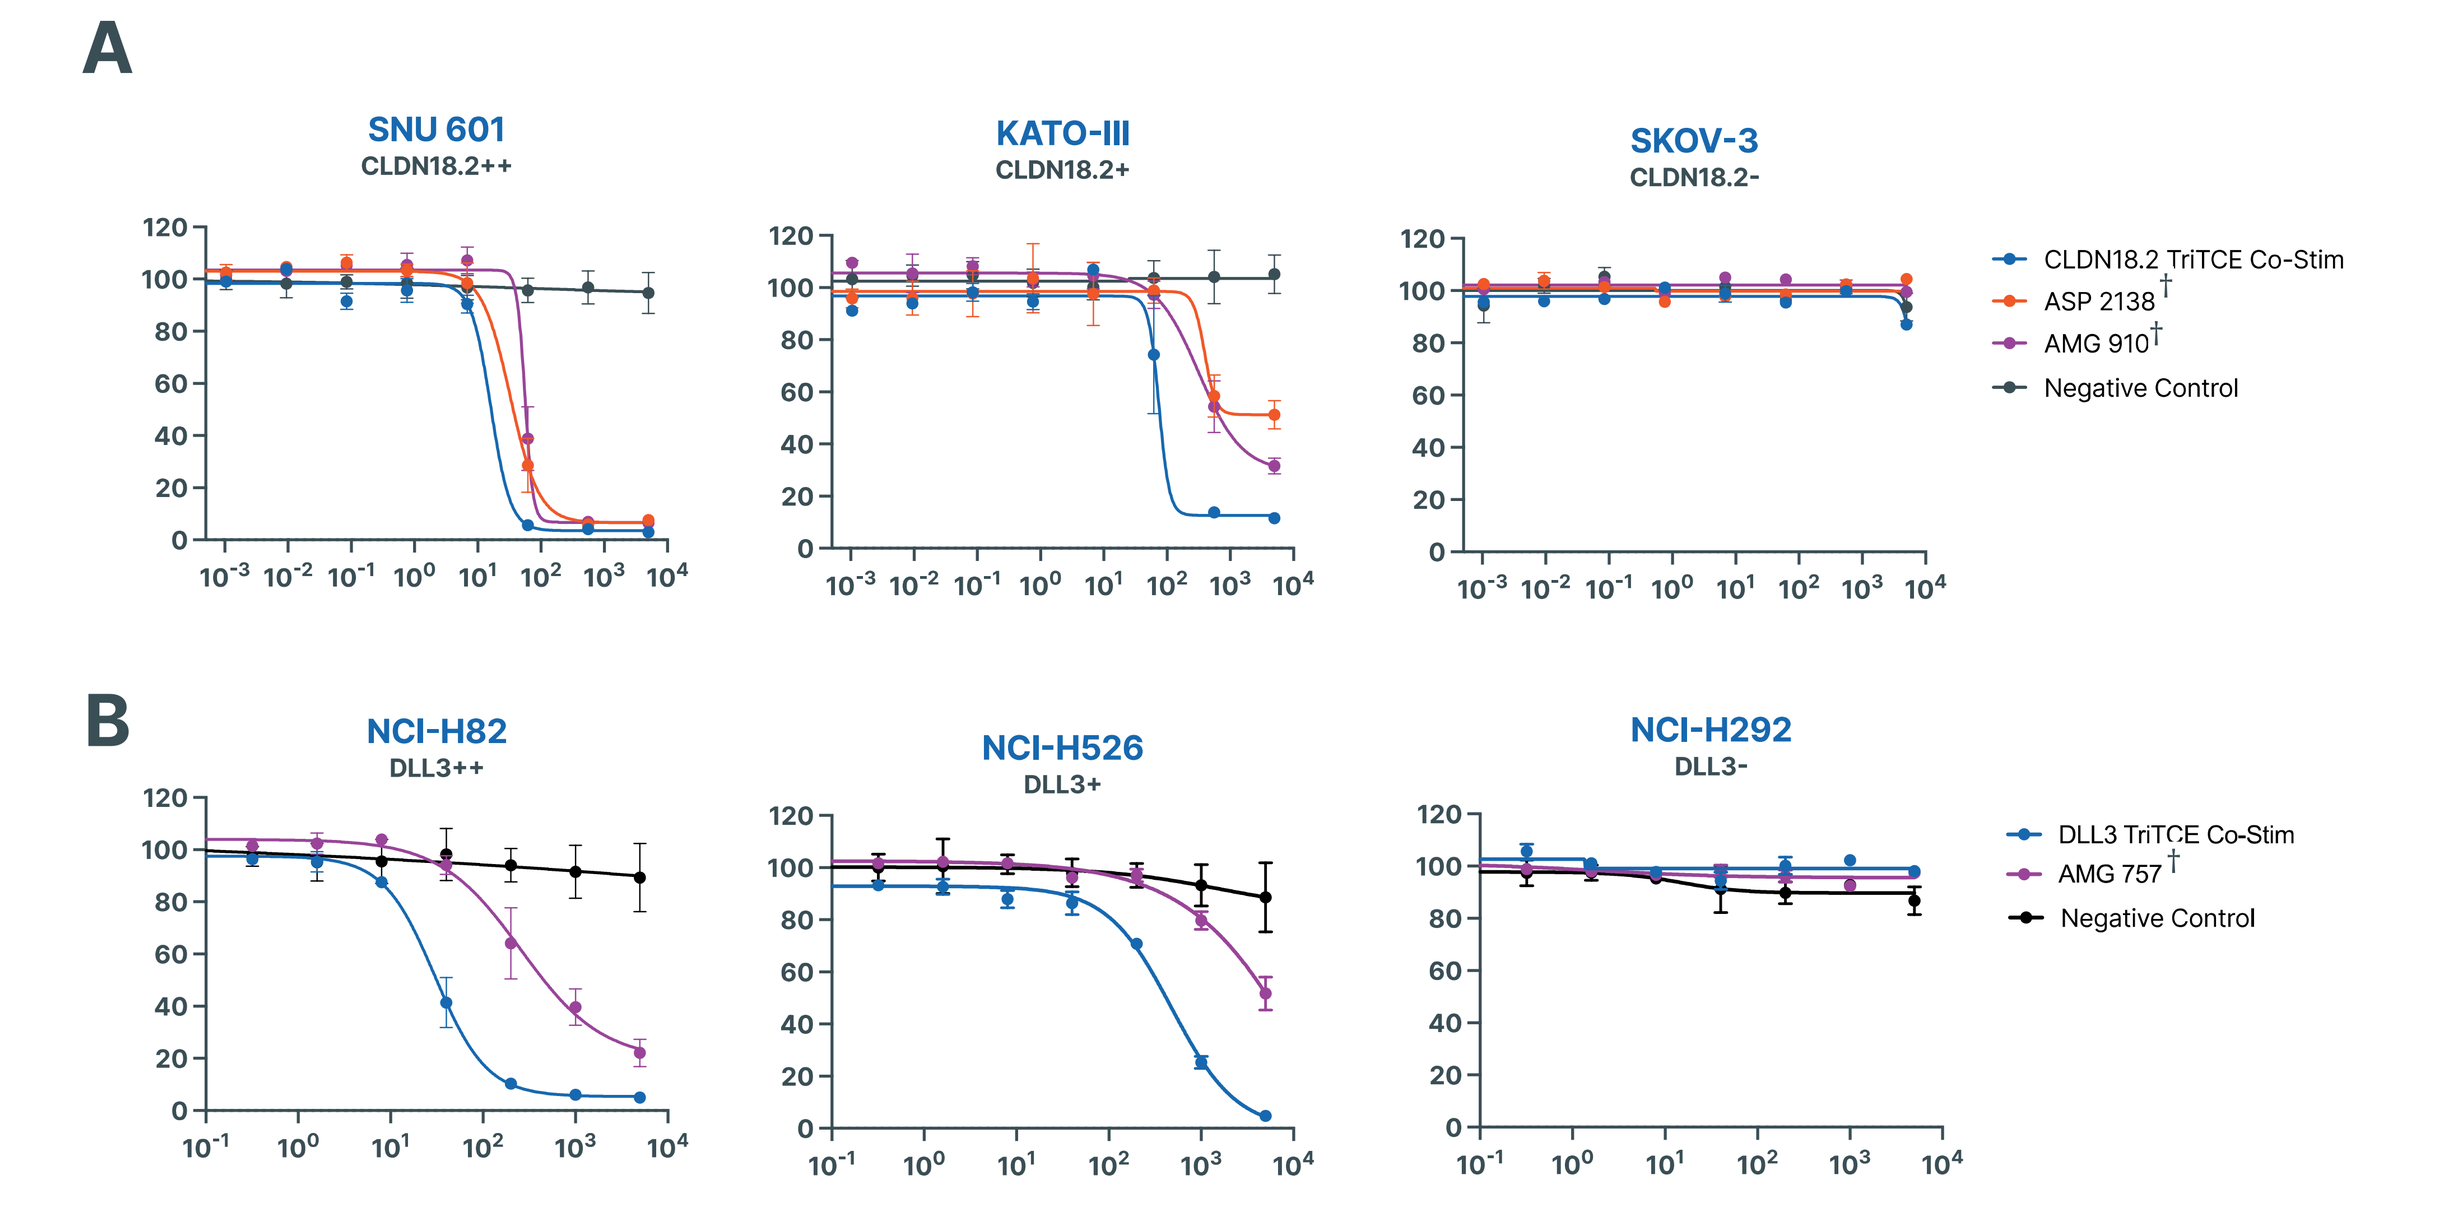
A
SNU 601
CLDN18.2++
KATO-III
CLDN18.2+
SKOV-3
CLDN18.2-
CLDN18.2 TriTCE Co-Stim
ASP 2138†
AMG 910†
Negative Control
B
NCI-H82
DLL3++
NCI-H526
DLL3+
NCI-H292
DLL3-
DLL3 TriTCE Co-Stim
AMG 757†
Negative Control

Figure 5. TriTCE Co-Stim molecules display superior cytotoxic potency in long term low E:T co-cultures. Test articles were incubated with human T cells co-cultured with CLDN18.2+ **(A)** or DLL3+ tumor cell lines for 7 days at low E:T and evaluated for cytotoxicity of target cells. TriTCE Co-Stim demonstrate superior cytotoxicity in a long-term low E:T ratio cytotoxicity assay and demonstrate no activity on TAA- cells.

Sustained Tumor Cytotoxicity with Repeated Stimulations

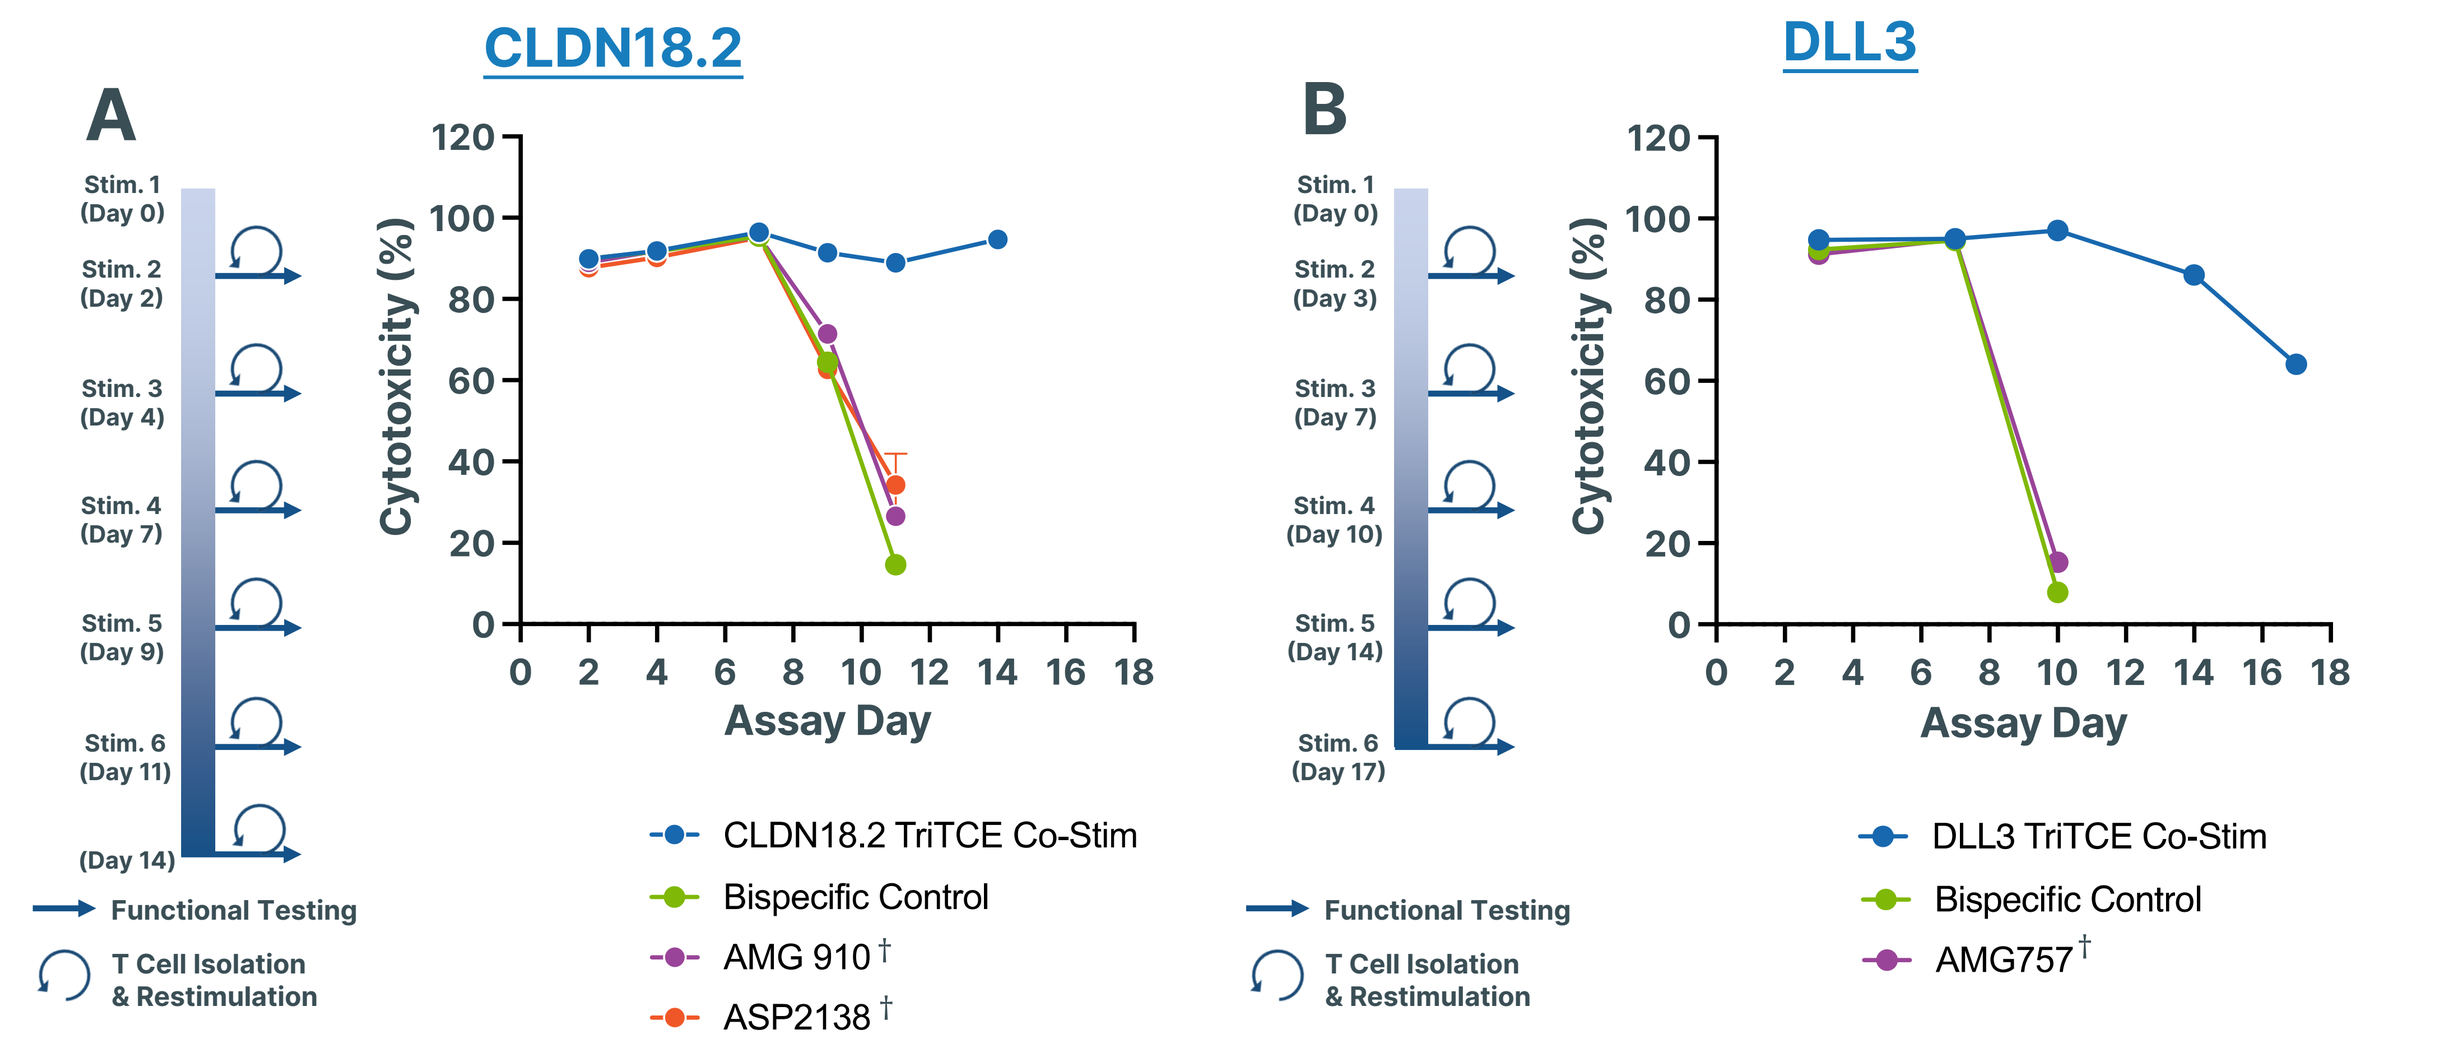
A
CLDN18.2
Stim. 1 (Day 0)
Stim. 2 (Day 2)
Stim. 3 (Day 4)
Stim. 4 (Day 7)
Stim. 5 (Day 9)
Stim. 6 (Day 11)
(Day 14)
Functional Testing
T Cell Isolation & Restimulation
Cytotoxicity (%)
Assay Day
CLDN18.2 TriTCE Co-Stim
Bispecific Control
AMG 910†
ASP2138†
B
DLL3
Stim. 1 (Day 0)
Stim. 2 (Day 3)
Stim. 3 (Day 7)
Stim. 4 (Day 10)
Stim. 5 (Day 14)
Stim. 6 (Day 17)
DLL3 TriTCE Co-Stim
Bispecific Control
AMG757†

Figure 6. TriTCE Co-Stim molecules display sustained T cell fitness and anti-tumor activity in a repeat challenge assay. T cells were stimulated with CLDN18.2+ tumor cell line - SNU 601 cells (5:1 E:T) and test article (1 nM), and DLL3+ tumor cell line - NCI-H82 cells (5:1 E:T) and test article (5 nM). For each subsequent round of stimulation, T cells are isolated from the T cell:tumor cell co-culture, counted, and re-stimulated with fresh tumor cell and respective test article. Schematic of T cell restimulation ¶ (**A**). Following each round of stimulation, T cell:tumor cell co-cultures were then assessed for tumor cell cytotoxicity (**B**). Insufficient T cells for continued stimulation with CLDN18.2 Bispecific, AMG 910† and ASP2138† following stimulation 5 in SNU601 cells and similar results were observed with DLL3 bispecific control and AMG 757† following stimulation 3 for NCI-H82 cells

Enhanced T-cell Proliferation and Survival

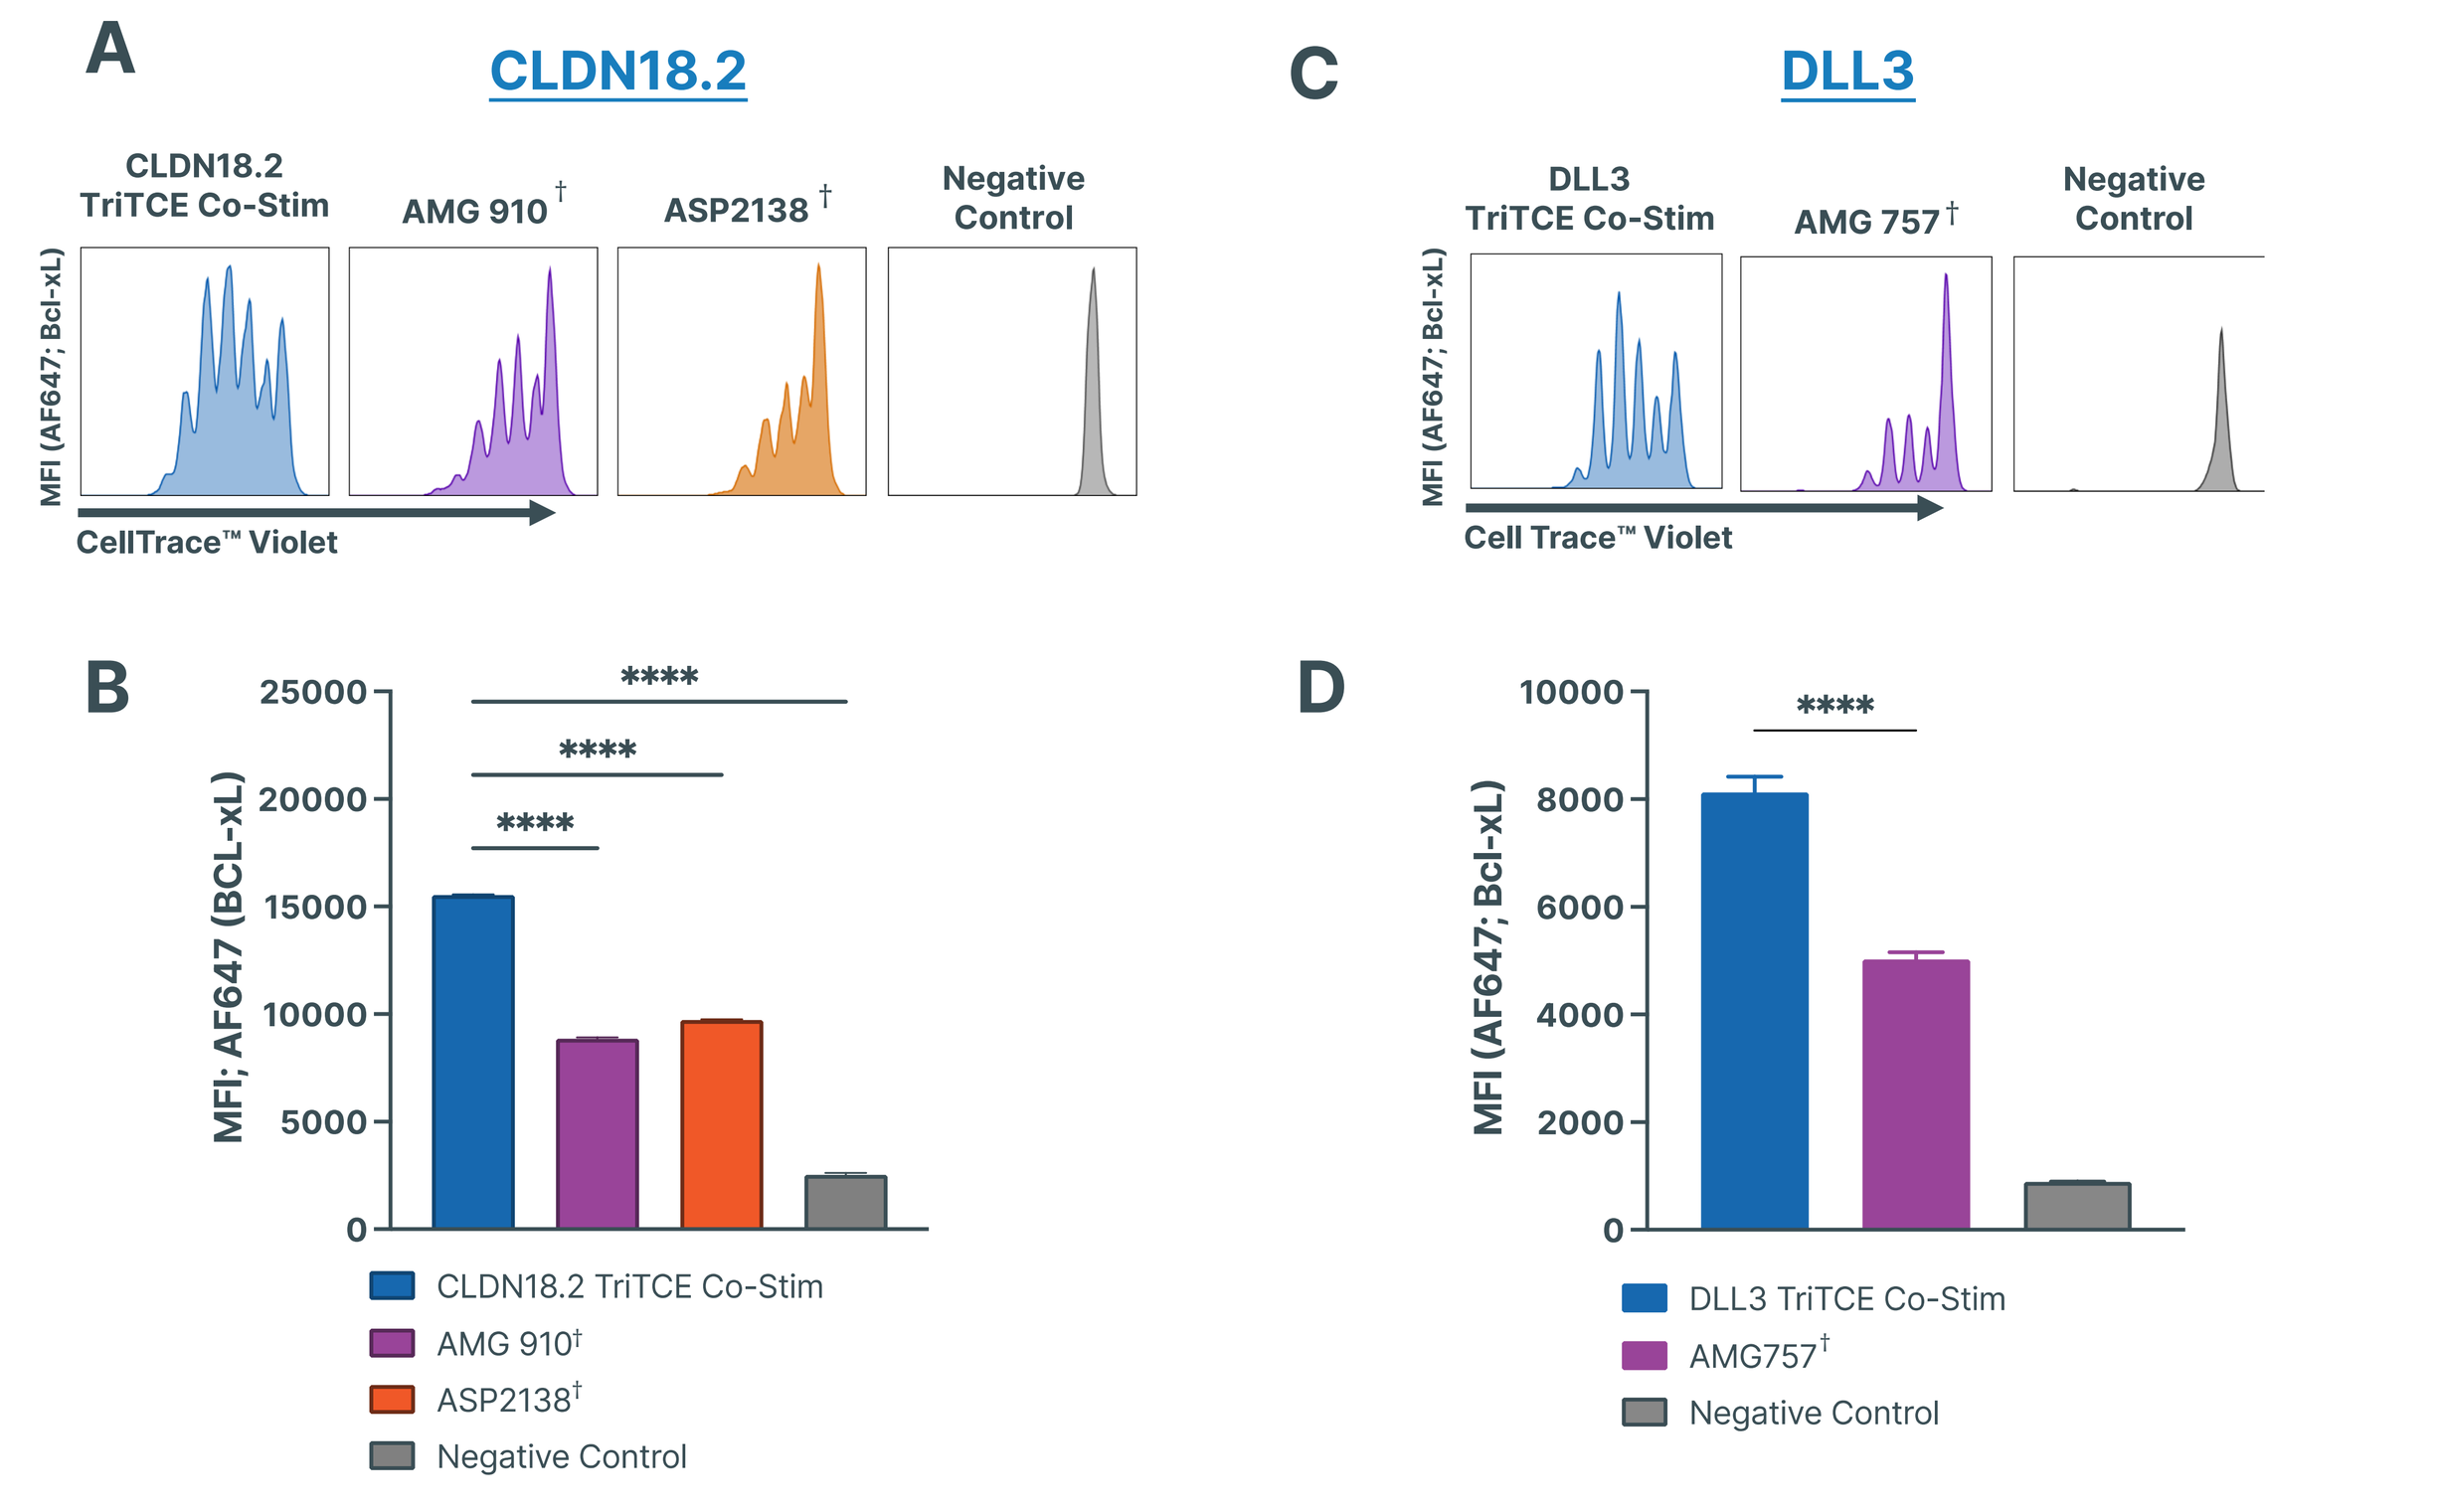
A
CLDN18.2
CLDN18.2 TriTCE Co-Stim
AMG 910†
ASP2138†
Negative Control
MFI (AF647; Bcl-xL)
CellTrace™ Violet
C
DLL3
DLL3 TriTCE Co-Stim
AMG 757†
Negative Control
Cell Trace™ Violet
B
MFI; AF647 (BCL-xL)

CLDN18.2 TriTCE Co-Stim
AMG 910†
ASP2138†
Negative Control
D
MFI (AF647; Bcl-xL)

DLL3 TriTCE Co-Stim
AMG757†
Negative Control

Figure 7. TriTCE Co-Stim molecules Increases T cell proliferation and upregulation of anti-apoptotic marker Bcl-xL. Test articles were incubated with CellTrace™ Violet-stained T cells co-cultured with tumor cells (SNU 601 cells for CLDN18.2 and NCI-H82 cells for DLL3 at 5:1 E:T) for 5 days and assessed by flow cytometry (**A, C**). Test articles were incubated with T cells co-cultured with SNU 601 cells for CLDN18.2 and NCI-H82 for DLL3 (2:1 E:T) and evaluated for Bcl-xL expression, an anti-apoptotic marker, by flow cytometry (**B, D**). Data are representative of two individual donors and are presented as mean ± SD.

Enhanced Anti-tumor Activity in Humanized Xenograft Models

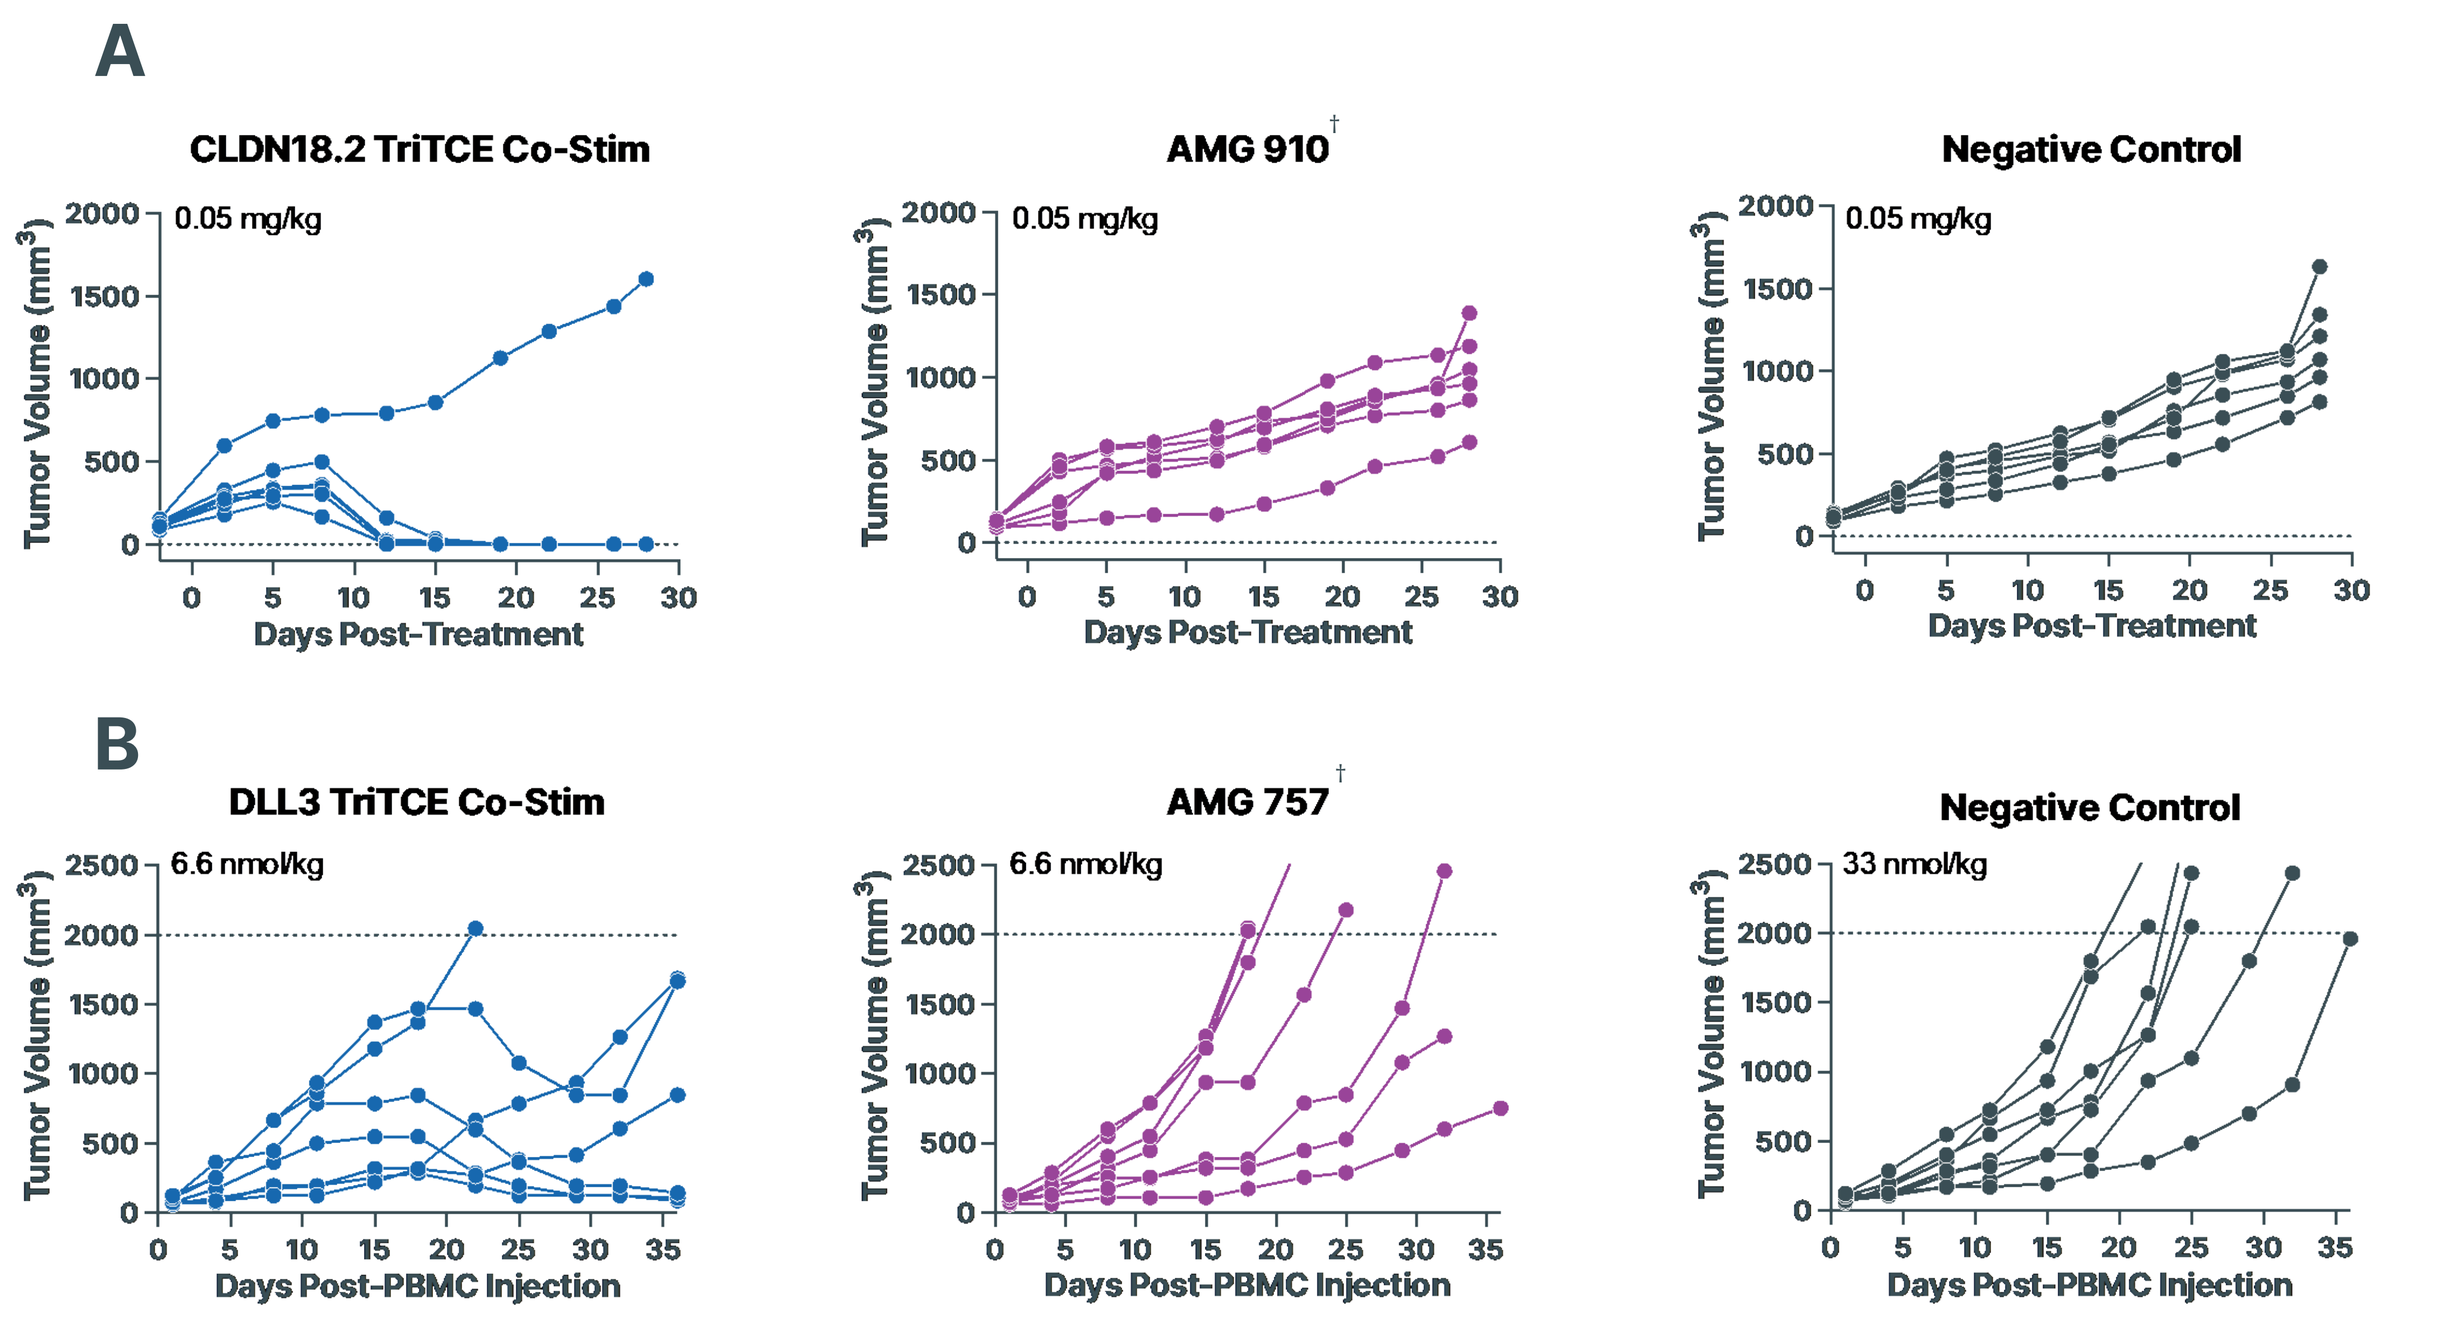
A
CLDN18.2 TriTCE Co-Stim
AMG 910†
Negative Control
0.05 mg/kg
Tumor Volume (mm³)
Days Post-Treatment
B
DLL3 TriTCE Co-Stim
AMG 757†
Negative Control
6.6 nmol/kg
33 nmol/kg
Days Post-PBMC Injection

Figure 8. *in vivo* efficacy following treatment with TriTCE Co-Stim molecules. NCG mice (n=6) were injected SC with SNU 620 target cells, engrafted with human PBMCs, and treated intravenously with CLDN18.2 TriTCE Co-stim q1wx4 and full tumor regression is observed in 5/6 mice **(A)**. PBMC-engrafted SHP-77 (SC) xenograft mouse model used to evaluate DLL3 TriTCE Co-Stim *in vivo* efficacy. Tumor volume over time of mice treated intravenously with DLL3 TriTCE Co-Stim, AMG 757 and irrelevant mAb. Full or partial tumor regression is observed in 4/7 mice treated with DLL3 TriTCE Co-Stim when IV treated q1wx4. **(B)**.

TriTCE Co-Stim Facilitates Desirable T-cell Engagement

Conditional binding of CD28 requiring CD3 co-engagement

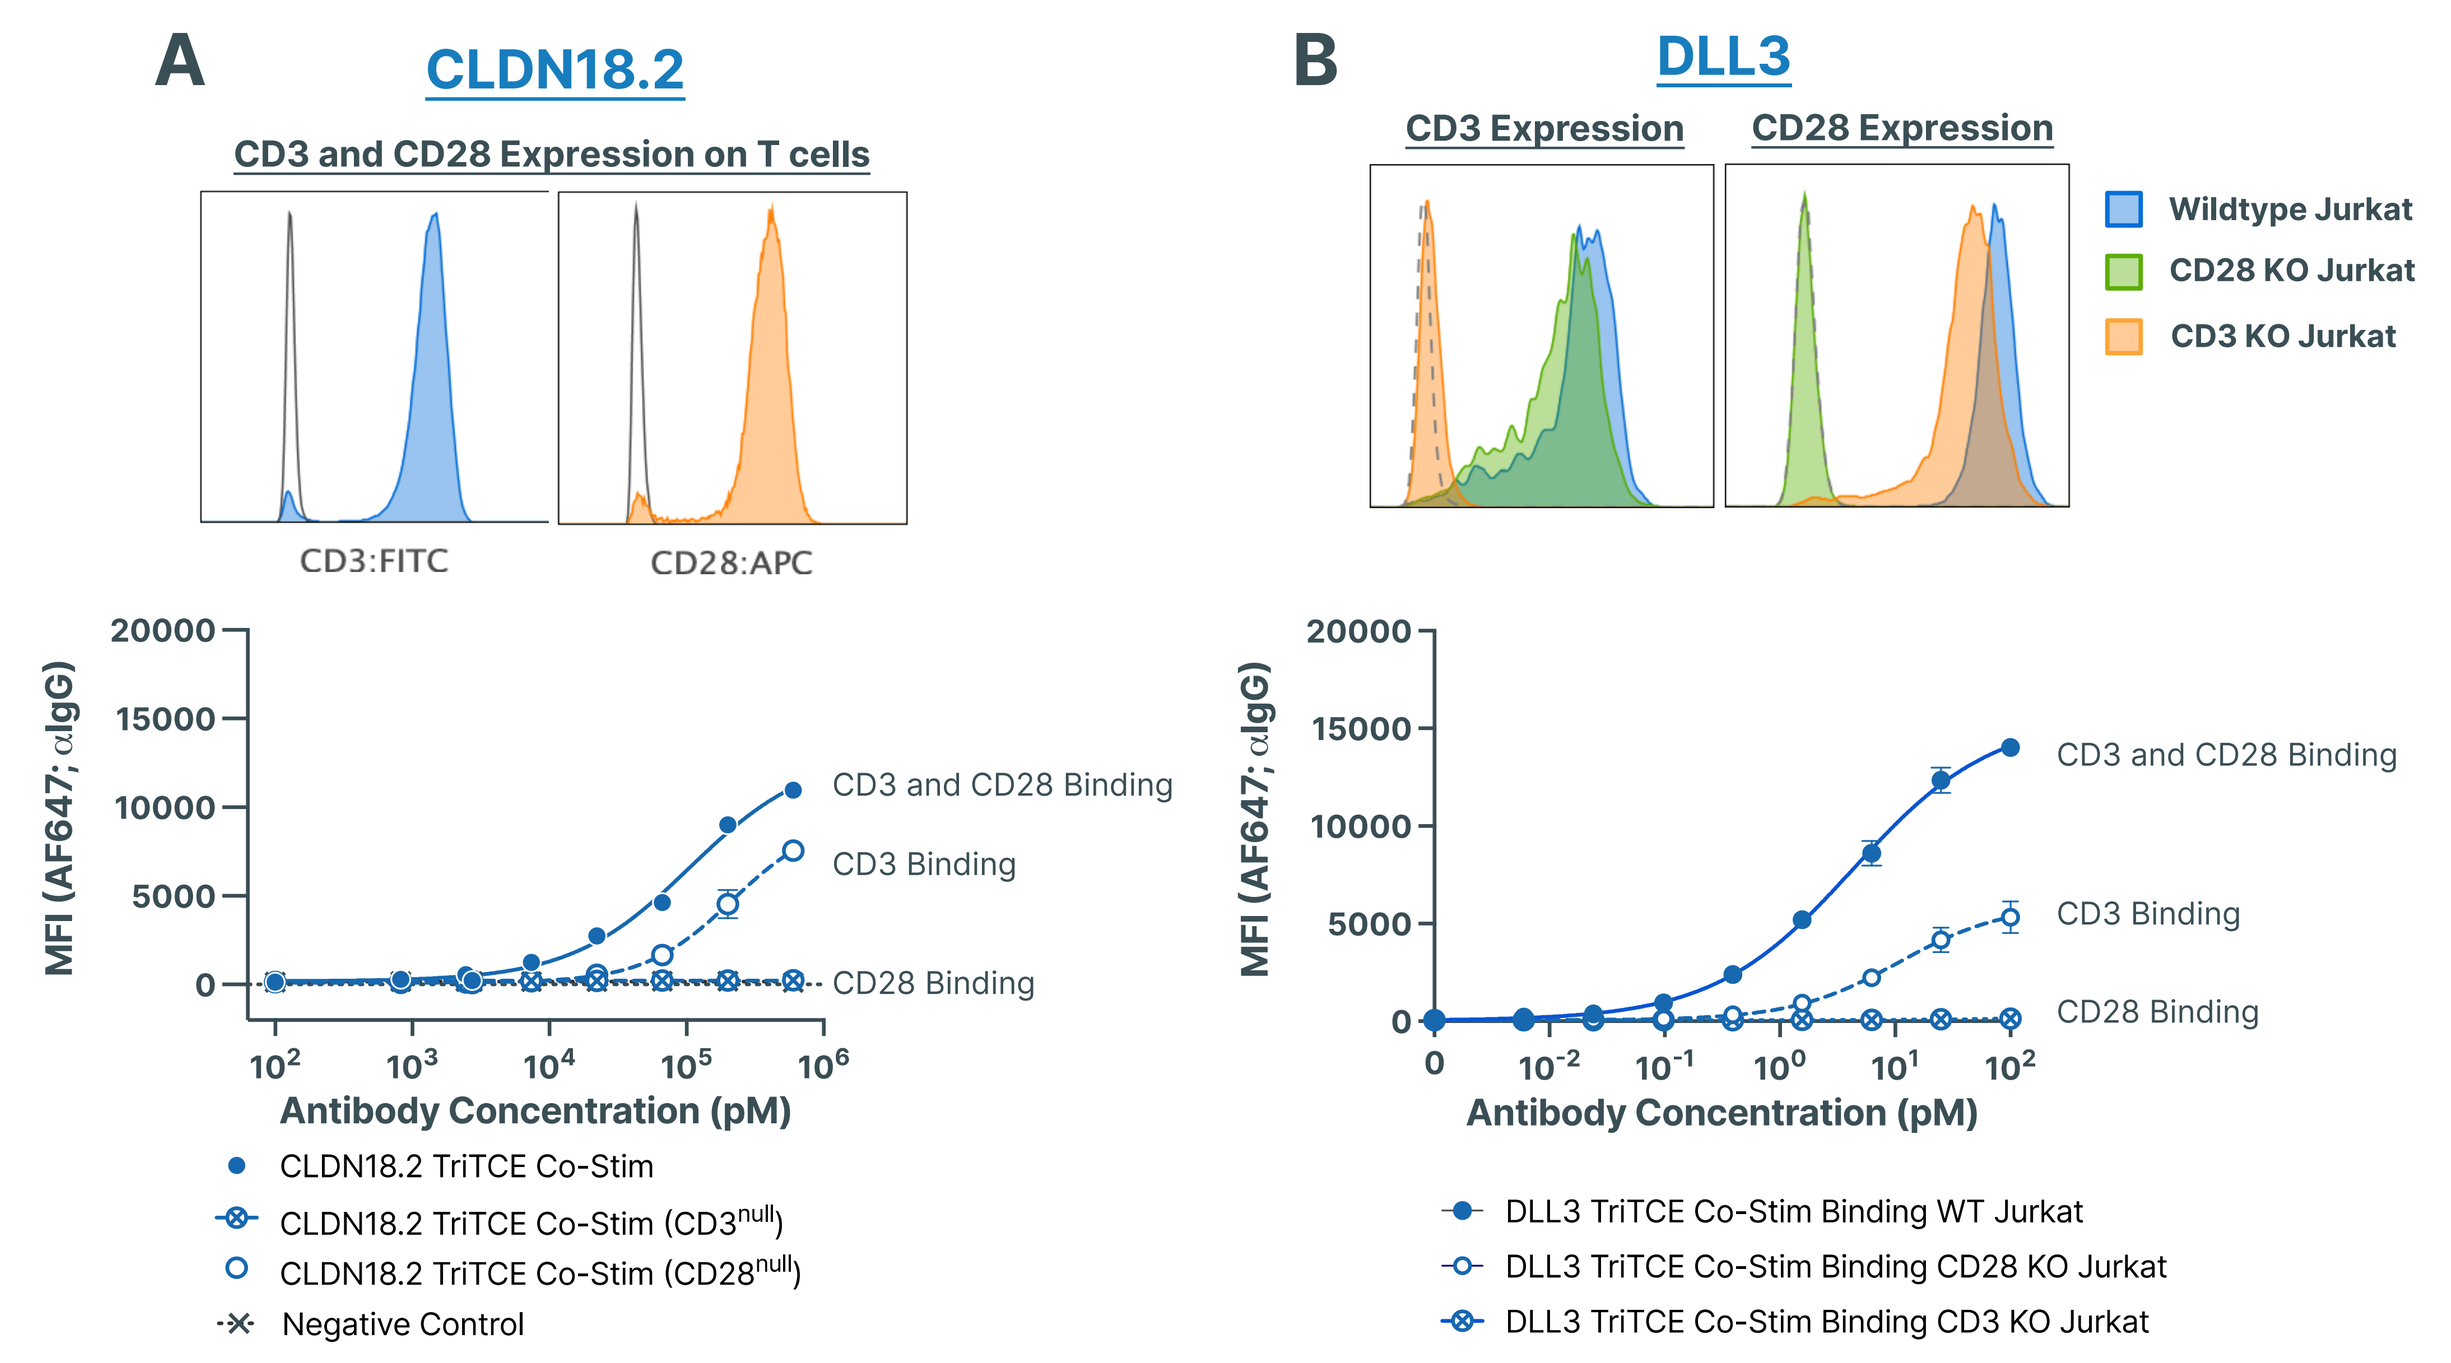
A
CLDN18.2
CD3 and CD28 Expression on T cells
CD3:FITC
CD28:APC
MFI (AF647; αIgG)
Antibody Concentration (pM)
CD3 and CD28 Binding
CD3 Binding
CD28 Binding
CLDN18.2 TriTCE Co-Stim
CLDN18.2 TriTCE Co-Stim (CD3null)
CLDN18.2 TriTCE Co-Stim (CD28null)
Negative Control
B
DLL3
CD3 Expression
CD28 Expression
Wildtype Jurkat
CD28 KO Jurkat
CD3 KO Jurkat
DLL3 TriTCE Co-Stim Binding WT Jurkat
DLL3 TriTCE Co-Stim Binding CD28 KO Jurkat
DLL3 TriTCE Co-Stim Binding CD3 KO Jurkat

No cross-linking of T cells via CD3 and CD28 *trans* binding

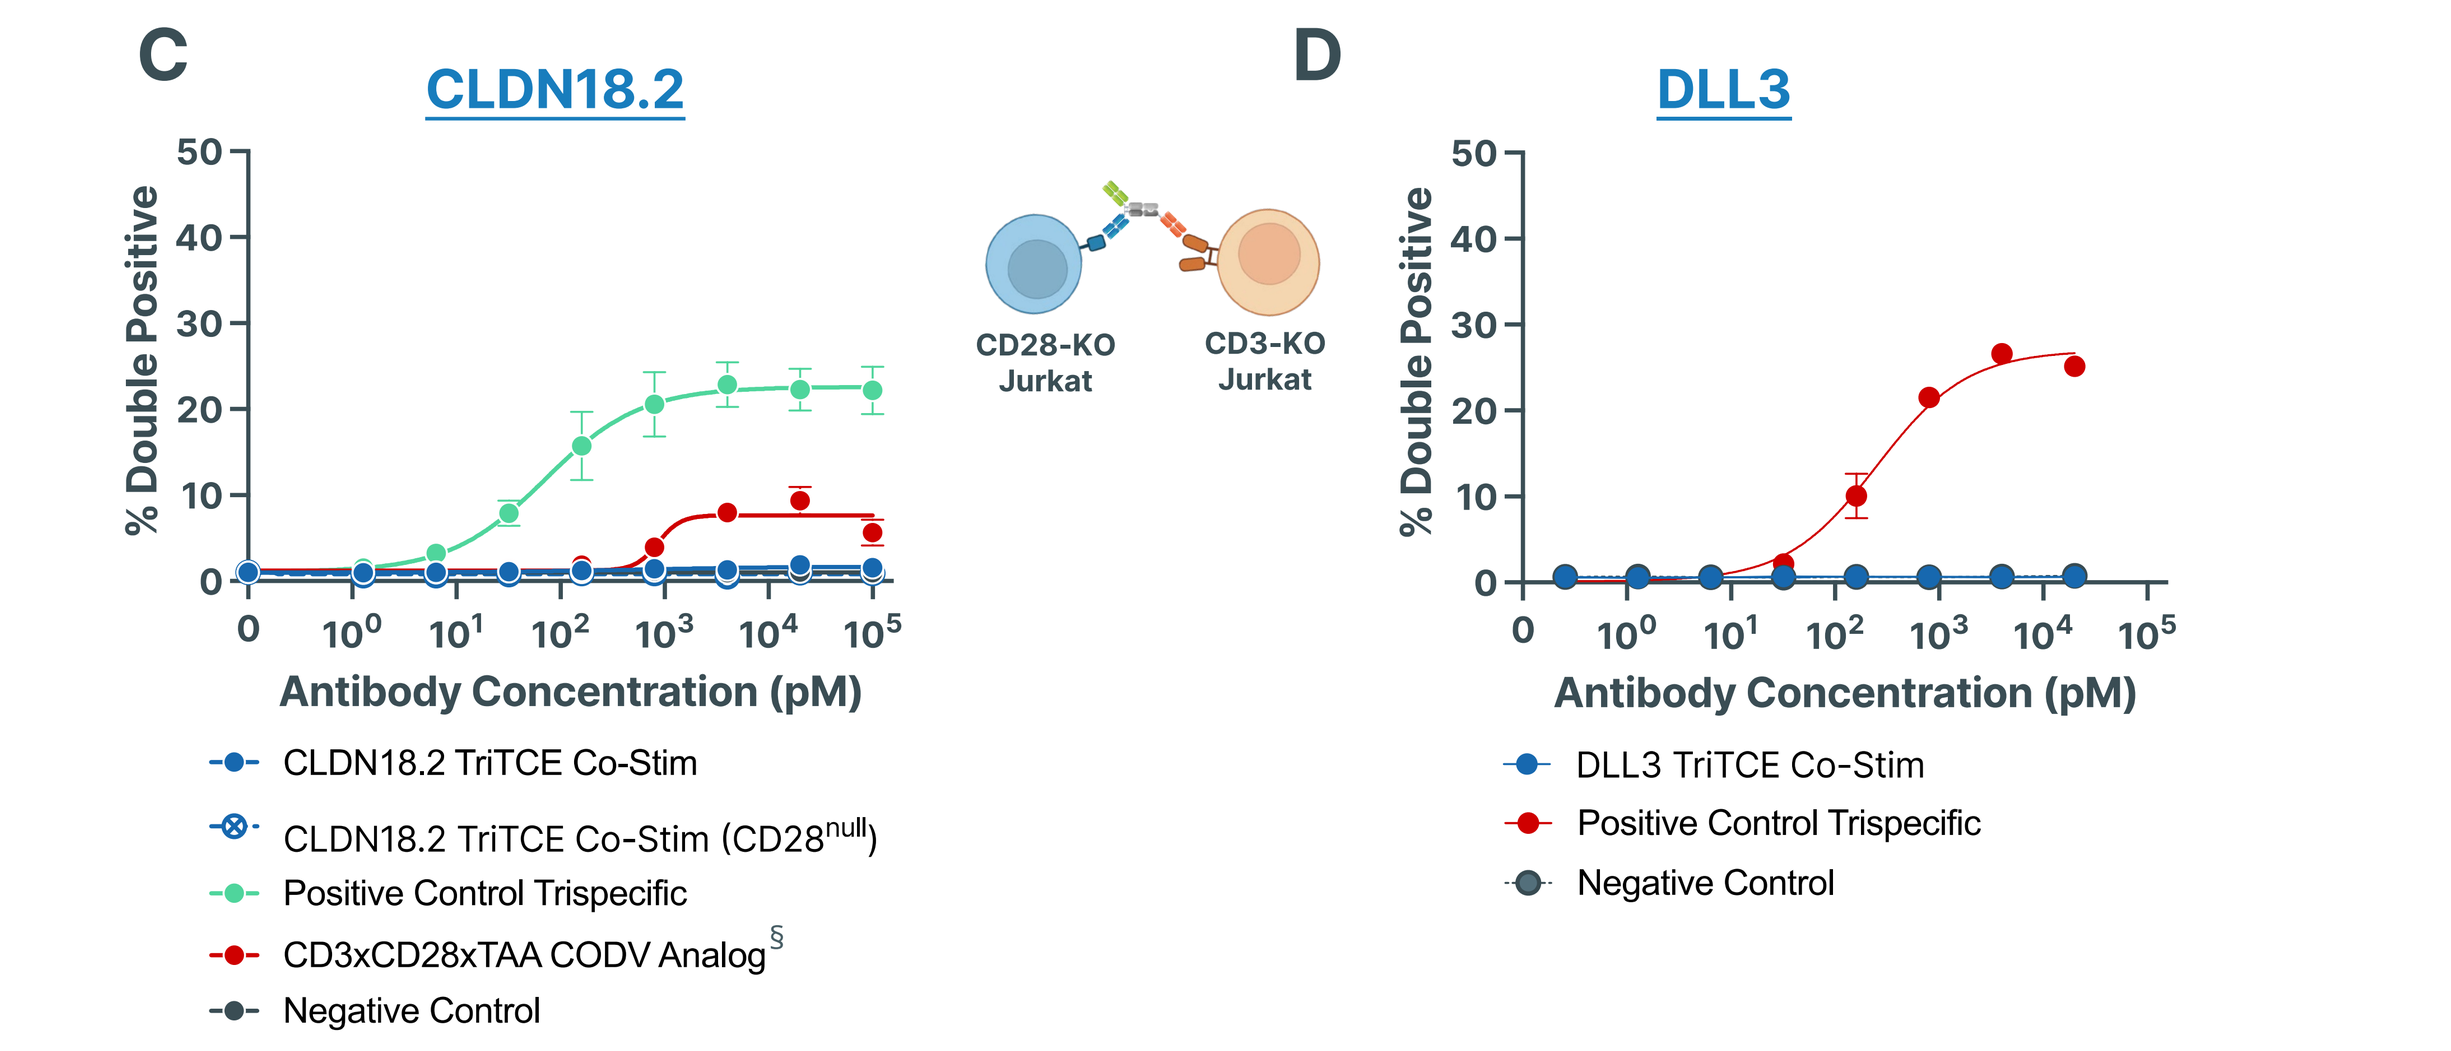
C
CLDN18.2
% Double Positive
Antibody Concentration (pM)
CD28-KO Jurkat
CD3-KO Jurkat
CLDN18.2 TriTCE Co-Stim
CLDN18.2 TriTCE Co-Stim (CD28null)
Positive Control Trispecific
CD3xCD28xTAA CODV Analog§
Negative Control
D
DLL3
DLL3 TriTCE Co-Stim
Positive Control Trispecific
Negative Control

No reduction of T-cell viability

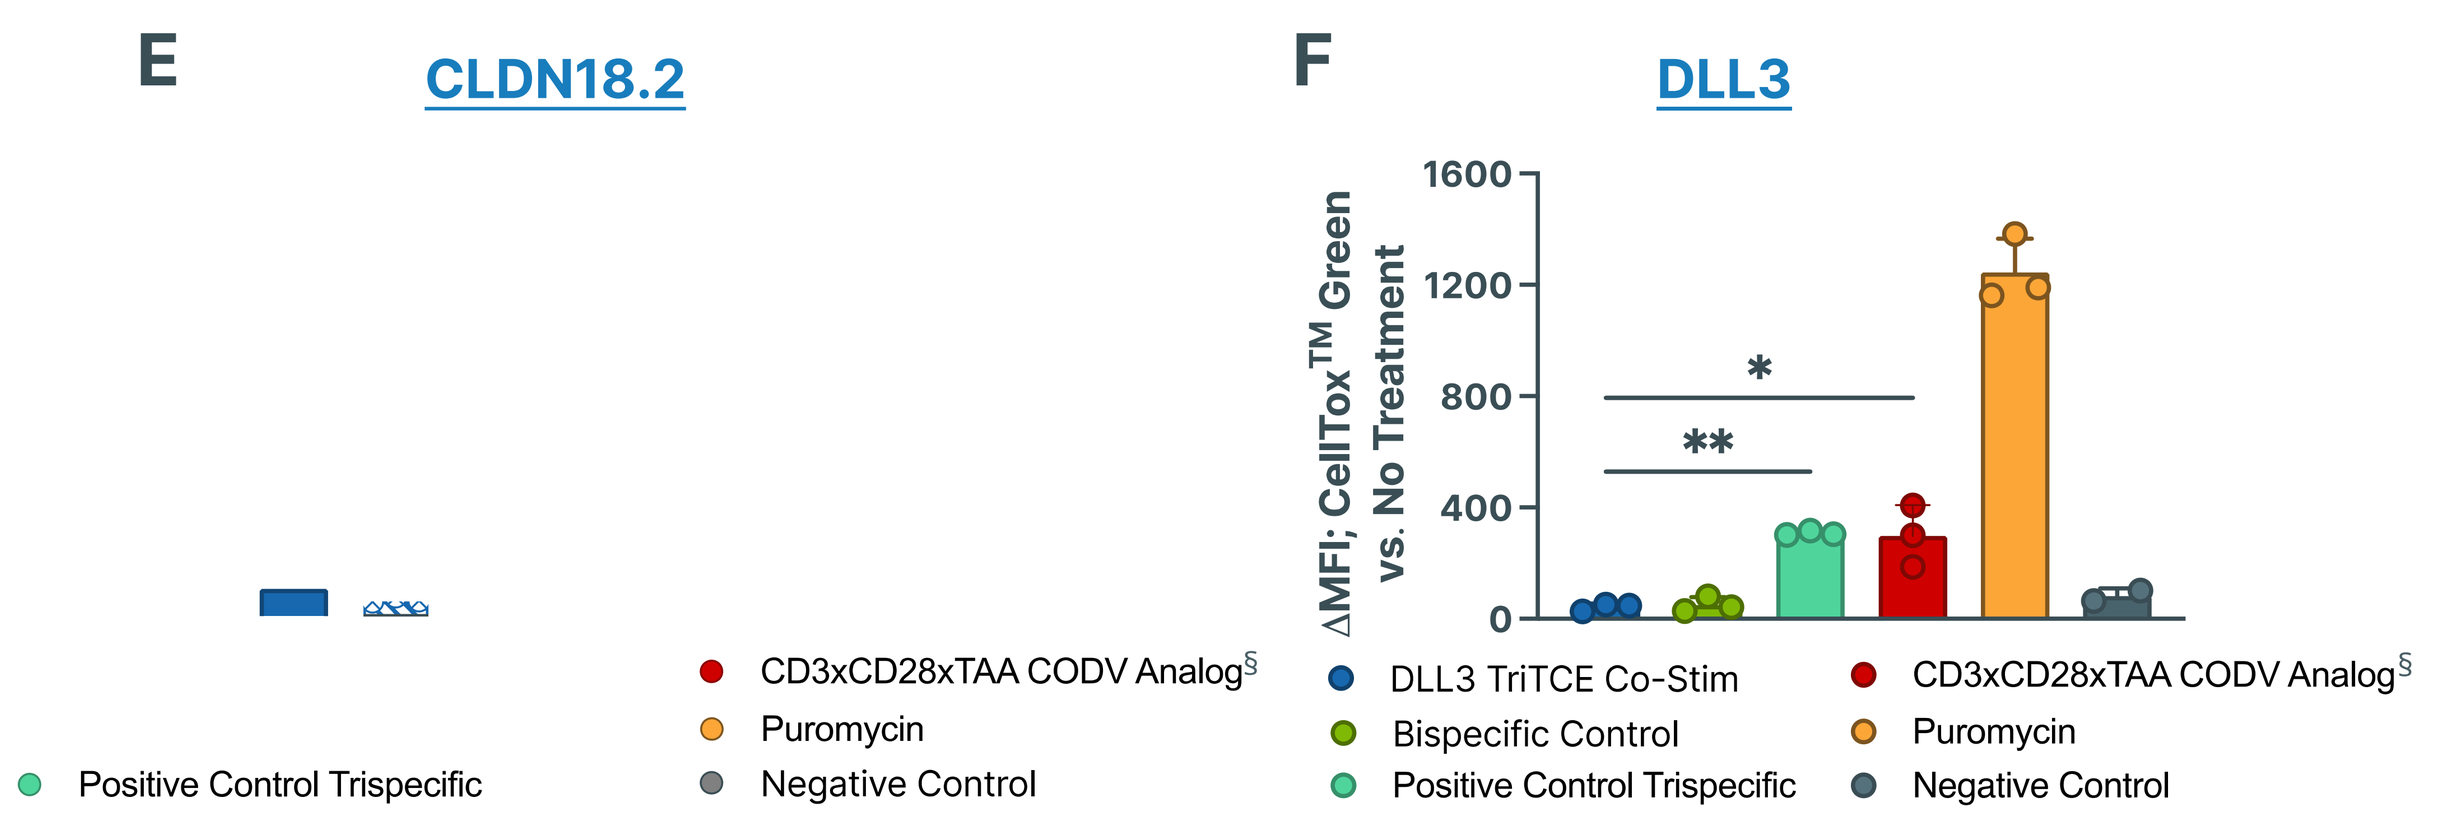
E
CLDN18.2
Positive Control Trispecific
CD3xCD28xTAA CODV Analog§
Puromycin
Negative Control
F
DLL3
ΔMFI; CellTox™ Green vs. No Treatment
DLL3 TriTCE Co-Stim
Bispecific Control
Positive Control Trispecific
CD3xCD28xTAA CODV Analog§
Puromycin
Negative Control

Figure 9. TriTCE Co-Stim displays desirable T-cell engagement and does not cross link T cells in *trans*. TriTCE Co-Stim molecules need CD3 engagement on T-cells in order to bind CD28, therefore CD28 engagement is conditional. No T-cell binding is observed with the CLDN18.2 TriTCE Co-Stim CD3 null **(A)**. DLL3 TriTCE Co-Stim does not bind CD3 KO Jurkat T cells **(B)**. Cell bridging by immune cell engaging antibodies can induce fratricide and thus reduce therapeutic efficacy[4]. The ability of TriTCE Co-Stim molecules to cross link T cells was tested by incubating CD3 KO and CD28 Jurkat T cells fluorescently labelled with dye in the presence of TriTCE. No T-cell bridging is observed with both CLDN18.2 and DLL3 TriTCE Co-Stim molecules **(C,D)**. To assess effects of TriTCE Co-Stim molecules on T-cell viability, test article and T cells were incubated with CellTox™ green and signal was measured 48 hours post incubation. CLDN18.2 and DLL3 TriTCE Co-Stim show significantly less reduction in T-cell viability relative to positive control TriTCE and the CD3xCD28xTAA CODV Analog **(E,F)** . * $p<0.05$, ** $p<0.01$, **** $p<0.0001$.

Lack of IL-2 production in solid phase CRS Assay

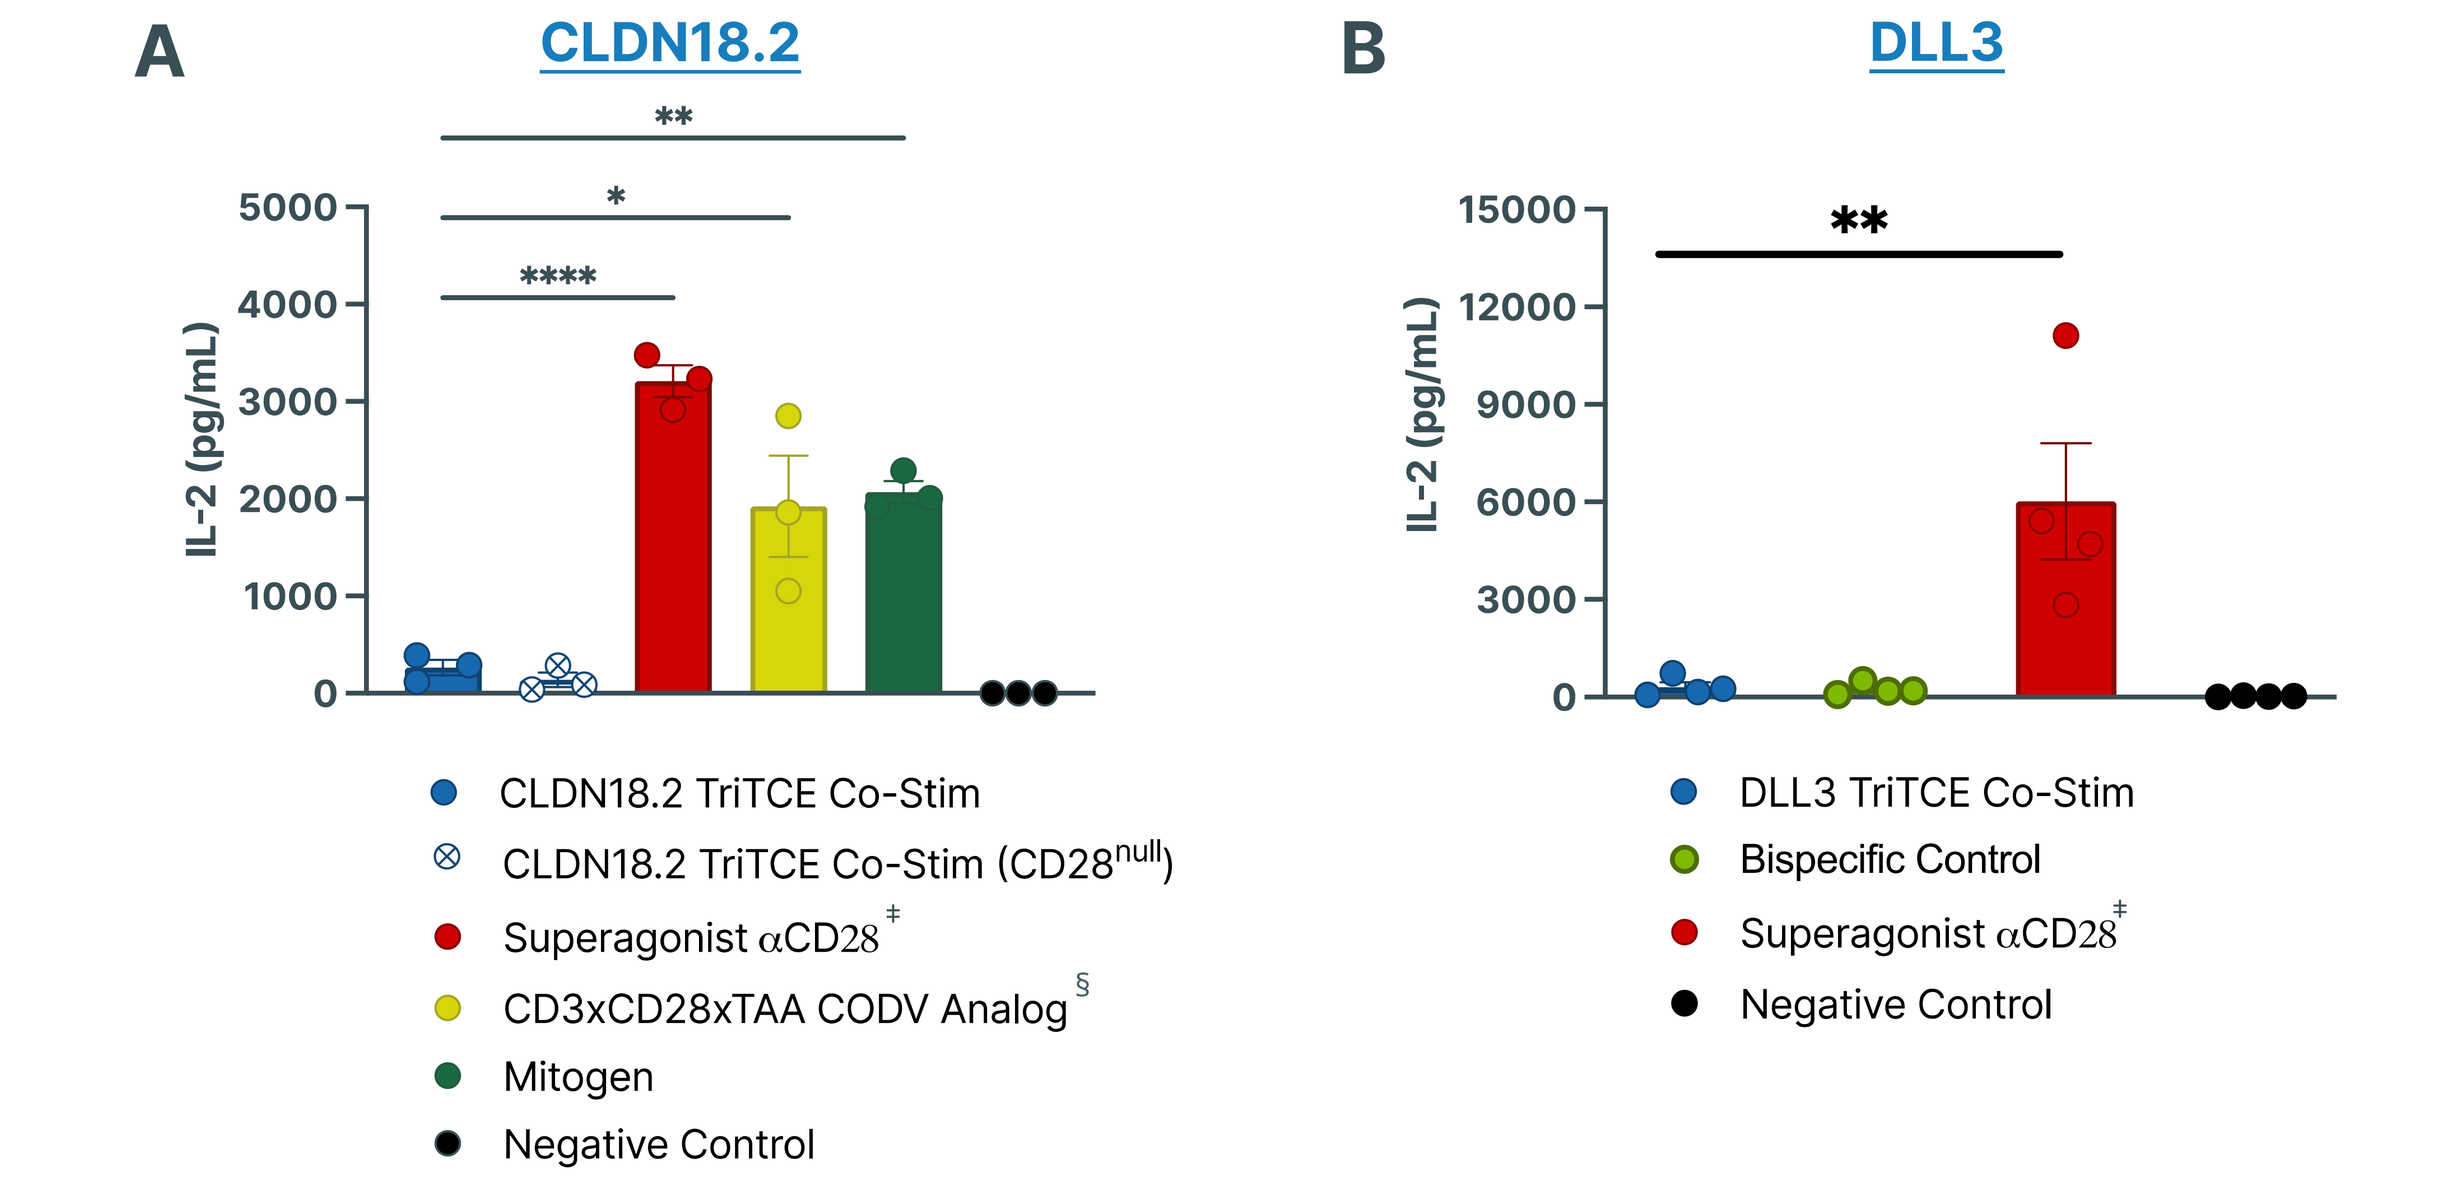
A
CLDN18.2
IL-2 (pg/mL)
CLDN18.2 TriTCE Co-Stim
CLDN18.2 TriTCE Co-Stim (CD28null)
Superagonist αCD28‡
CD3xCD28xTAA CODV Analog§
Mitogen
Negative Control
B
DLL3
DLL3 TriTCE Co-Stim
Bispecific Control
Superagonist αCD28‡
Negative Control

Figure 10. Predictive *in vitro* model for cytokine release syndrome (CRS). IL-2 production in a solid phase CRS assay is correlated with severity of CRS by TGN1412, an anti-CD28 superagonist antibody[5,6]. To test IL-2 production, immobilized test articles (1 μg/well) were incubated with PBMCs for 48 hours and cytokine levels were measured from supernatants via MSD. CLDN18.2 TriTCE Co-Stim **(A)** and DLL3 TriTCE Co-Stim **(B)** did not exhibit T-cell cytokine release. Superagonist αCD28 is TGN1412 replica produced in-house. Mitogen is Staphylococcal enterotoxin B. Data presented are mean ± SEM of at least three individual PBMC donors. * $p<0.05$, ** $p<0.01$, **** $p<0.0001$.

CLDN18.2 TriTCE Co-Stim Displays Favourable Safety Profile

Cynomolgus monkeys (n=3) were administered a repeat dose of 3mg/kg of a cynomolgus surrogate CLDN18.2 TriTCE Co-Stim* on day 0 and day 8.

- Toxicology findings were mild and associated with the known mechanism of action of TCEs[3]
- No histopathological changes observed in the stomach, where CLDN18.2 is expressed[7]
- Other histopathological changes were secondary to decreased food consumption and body weight loss

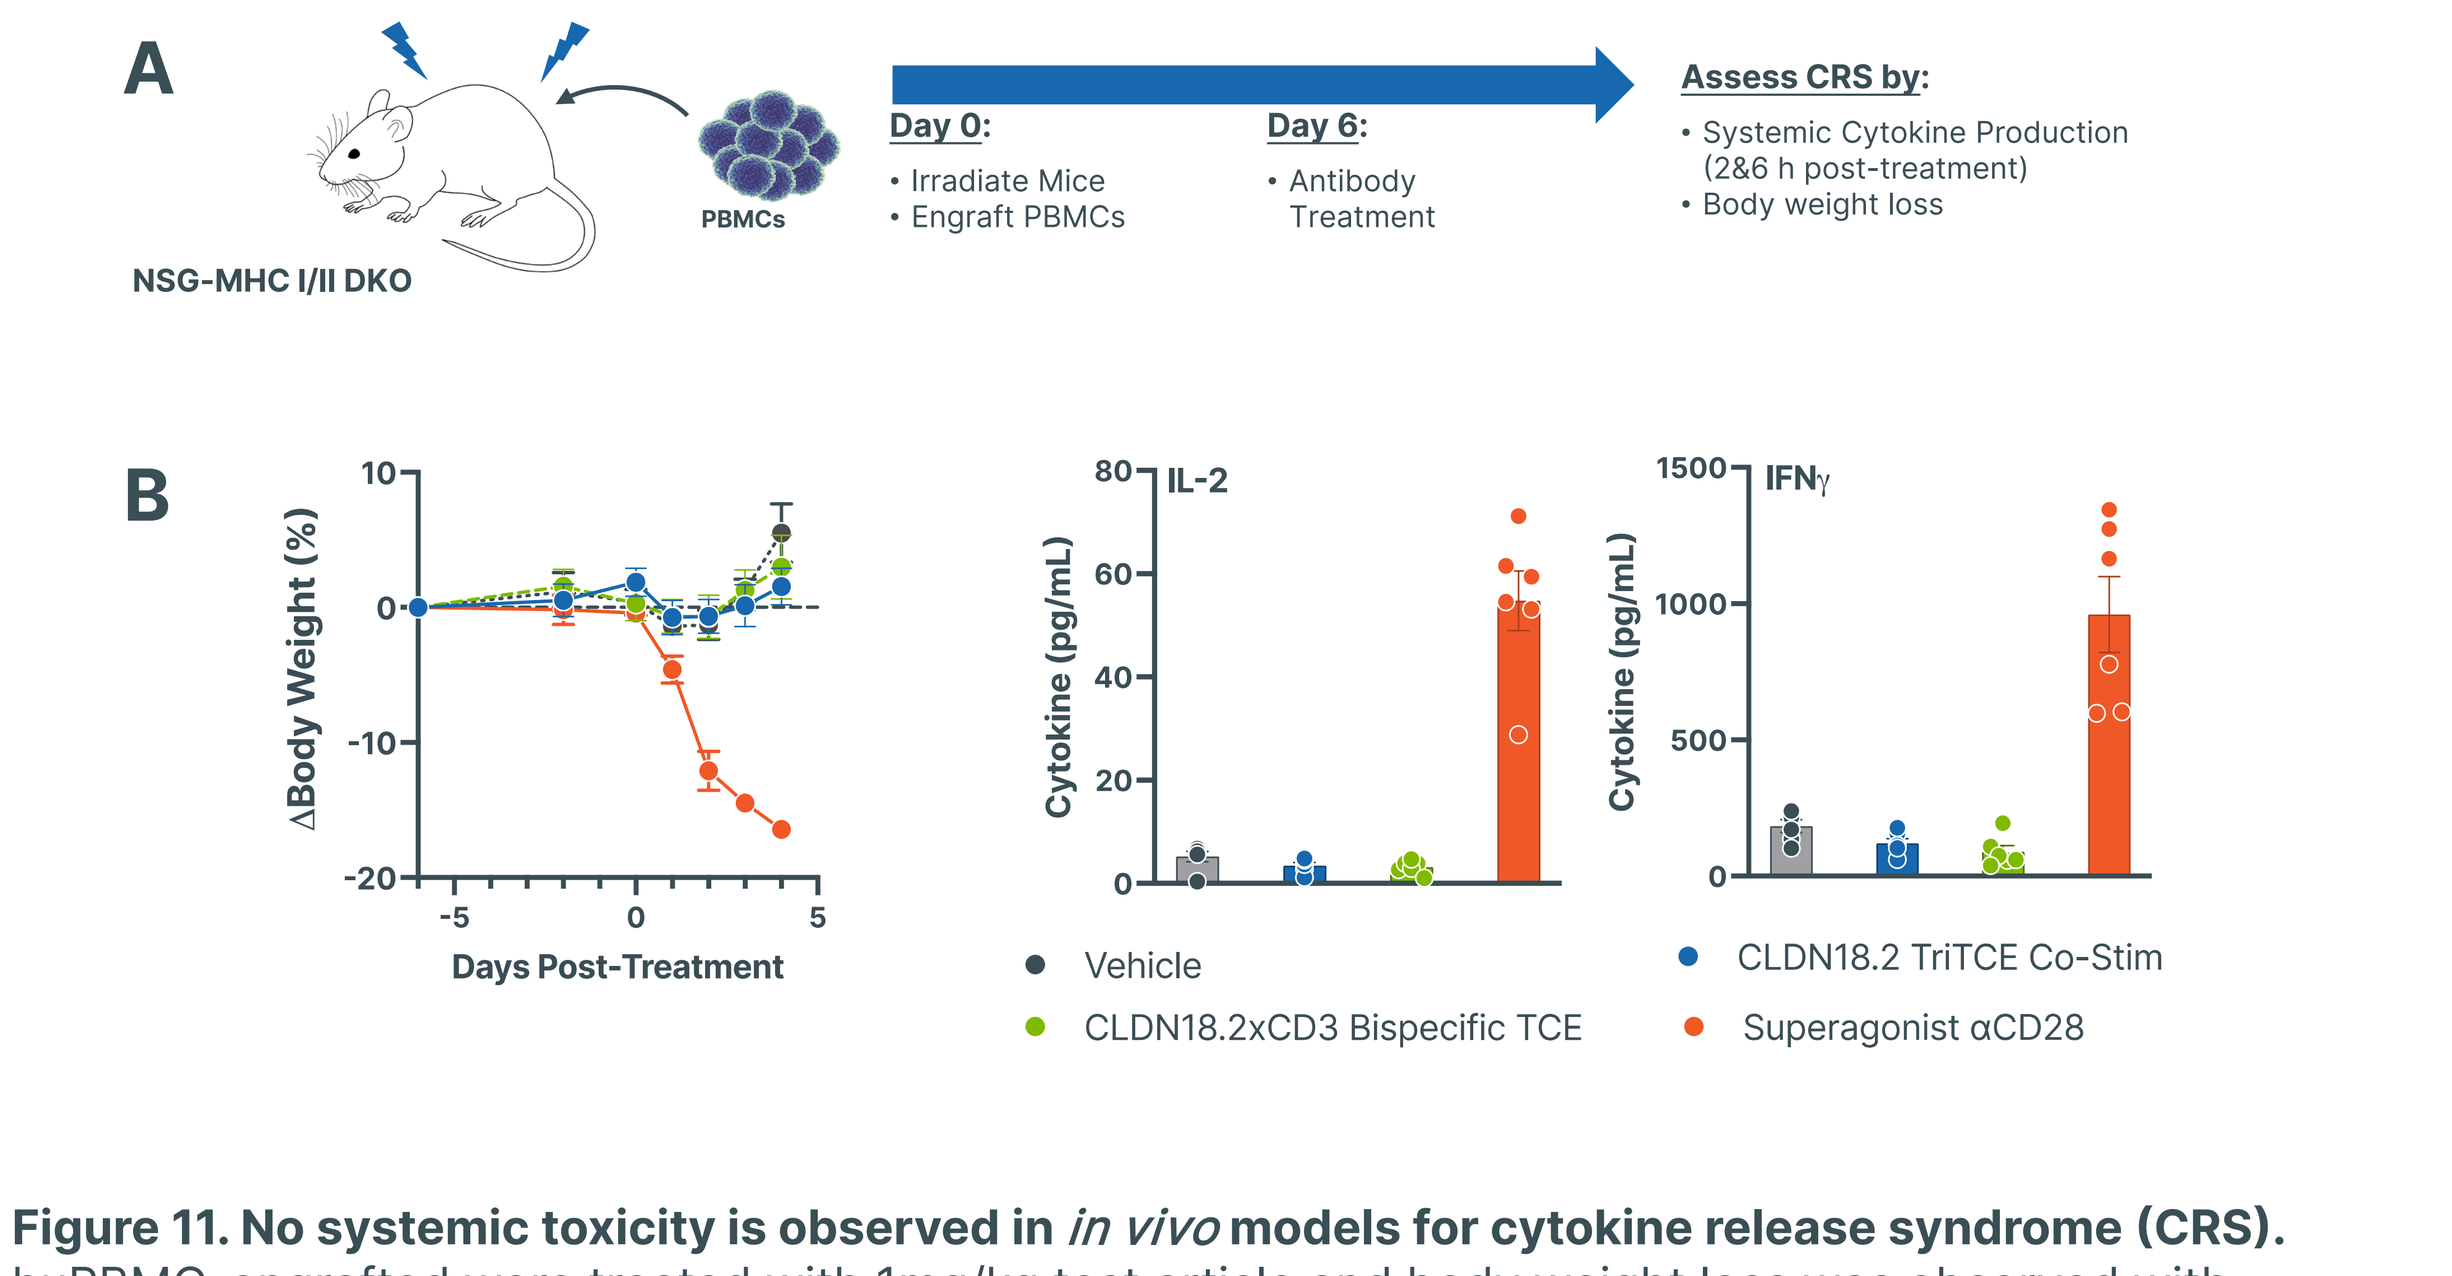
A
NSG-MHC I/II DKO
PBMCs
Day 0:
• Irradiate Mice
• Engraft PBMCs
Day 6:
• Antibody Treatment
Assess CRS by:
• Systemic Cytokine Production (2&6 h post-treatment)
• Body weight loss
B
ΔBody Weight (%)
Days Post-Treatment
IL-2
IFNγ
Cytokine (pg/mL)
Vehicle
CLDN18.2xCD3 Bispecific TCE
CLDN18.2 TriTCE Co-Stim
Superagonist αCD28

Figure 11. No systemic toxicity is observed in *in vivo* models for cytokine release syndrome (CRS). huPBMC-engrafted were treated with 1mg/kg test article and body weight loss was observed with superagonist αCD28 (ANC28.1/5D10) **(A)** and IL-2 production is correlated with severity of cytokine release syndrome by TGN1412[5,6]. High levels of systemic cytokine production was also observed with superagonist αCD28 6 h post-treatment when compared to CLDN18.2 TriTCE or Bispecific control **(B)**. Superagonist αCD28 used for *in vivo* assessment is ANC28.1/5D10 (mIgG1). CLDN18.2 TriTCE Co-Stim is cross-reactive with mouse CLDN18.2 (data not shown).

Conclusions

TriTCE Co-Stim antibodies with various paratope formats and geometries are engineered using the Azymetric™ and EFECT™ platforms. The evaluation of multiple formats, geometries, and paratope affinities allowed for optimization of selectivity and activity to promote a widened therapeutic index with enhanced anti-tumor activity. Zymeworks Co-Stim Abs using 2 different TAA's **CLDN18.2 and DLL3** show:

- **Greater *in vitro* cytotoxicity at low E:T ratios and improved T cell proliferation and survival** compared to bispecific TCEs
- **Sustained cytotoxicity, T cell viability and proliferation in serial challenge assays**
- **Obligate cis T cell binding to CD3 and CD28, and conditional CD28 binding requiring co-engagement of CD3**
- **Improved *in vivo* tumor regression** relative to clinical benchmark bispecific
- **No systemic cytokine release** in an *in vivo* CRS model study
- **CLDN18.2 TriTCE surrogate well tolerated in NHP[3]**

PEGS Boston
2024

References:
1. Arvedson T., et al. 2022. Targeting Solid Tumors with Bispecific T Cell Engager Immune Therapy (Vol. 6, pp.17-34).
2. Newhook L., et al. 2023. TRITCE Co-Stim: A Novel Trispecific T Cell Engager Platform, with Integrated CD28 Costimulation, Engineered to Widen the Therapeutic Window for Treatment of Poorly Infiltrated Tumors. [Poster Presentation] SITC. San Diego, CA
3. Newhook et al., 2024. TriTCE Co-Stim: A next generation trispecific T-cell engager platform with integrated CD28 costimulation, engineered to improve responses in the treatment of solid tumors. (Poster Presentation) AACR. San Diego, CA.
4. Repenning et al., 2024. DLL3 TriTCE Co-Stim: A next generation Trispecific T cell engager with integrated CD28 co-stimulation for the treatment of DLL3-expressing cancers. (Poster Presentation) AACR. San Diego, CA.
5. Eastwood D., et al. 2013. Severity of the TGN1412 Trial Disaster Cytokine Storm Correlated with IL-2 Release. *Br J Clin Pharmacol.* (Vol. 76, No. 2, pp.299-315).
6. Newhook L., et al. 2023. TriTCE Co-Stim, Next Generation Co-Stimulatory Trispecific T cell Engagers for the Treatment of Solid Tumors. [Poster Presentation] AACR. Orlando, FL
7. Türeci O., et al. 2011. Claudin-18 gene structure, regulation, and expression is evolutionary conserved in mammals. Gene. (Vol. 481, No 2, pp 83-92).

† Following molecules were produced in house: AMG 910 (CLDN18.2/CD3 BiTE), ASP2138 (CLDN18.2/CD3 2+1 bsAb) and AMG 757 (DLL3/CD3 BiTE)
‡ TGN1412 replica produced in-house
§ CD3xCD28xTAA CODV Analog is a CD3xCD28xMSLN trispecific with the same format as the Sanofi Trispecific containing a CD3xCD28 CODV-Fab; produced in-house.
∗ Surrogate CLDN18.2 TriTCE Co-Stim exhibited ~10 fold increased cytotoxic potency vs. lead TriTCE Co-Stim and ~15-fold reduced cytotoxic potency vs. AMG 910 in cynomolgus T-cell dependent cytotoxicity asssays *in vitro*
¶ Image Created with BioRender.com