Phase 1 Study of Zanidatamab Zovodotin (ZW49): Safety Profile and Recommended Dose (RD) in Patients with Human Epidermal Growth Factor 2 (HER2)-positive Solid Cancers

Do-Youn Oh[1]; Philippe L Bedard[2]; Keun-Wook Lee[3]; Hyo Sook Han[4]; Yoon-Koo Kang[5]; Wilson H. Miller Jr.[6]; Sun Young Rha[7]; Jwa Hoon Kim[8]; Efrat Dotan[9]; Chih-Yi Liao[10]; Anthony Tolcher[11]; Alexander Spira[12]; Erika Hamilton[13]; Christos Karapetis[14]; Lisa Boyken[15]; Charles Chen[15]; Joseph Woolery[15]; Komal Jhaveri[16]

[1]Seoul National University Hospital, Seoul, South Korea; [2]Princess Margaret Cancer Centre, Toronto, ON, Canada; [3]Seoul National University College of Medicine, Seoul National University Bundang Hospital, Seongnam, South Korea; [4]Moffitt Cancer Center, Tampa, FL, US; [5]Asan Medical Center, Seoul, South Korea; [6]Lady Davis Institute and Jewish General Hospital, McGill University, Montreal, QC, Canada; [7] Yonsei Cancer Center, Yonsei University Health System, Seoul, South Korea; [8]Korea University Anam Hospital, Seoul, South Korea; [9]Fox Chase Cancer Center, Philadelphia, PA, US; [10]University of Chicago Medicine, Chicago, IL, US; [11]NEXT Oncology, San Antonio, TX, US; [12]Virginia Cancer Specialists, PC, Fairfax, VA, US; [13]Sarah Cannon Research Institute, Tennessee Oncology, Nashville, TN, US; [14]Flinders Medical Centre and Flinders University, Adelaide, Australia; [15]Zymeworks BC Inc., Vancouver, BC, Canada; [16]Memorial Sloan Kettering Cancer Center, New York, NY, US

Abstract Number: 33234

Background

- Zanidatamab zovodotin (ZW49) is a novel antibody drug conjugate (ADC) comprised of a HER2-targeting bispecific antibody directed against 2 non-overlapping HER2 epitopes and attached to a proprietary auristatin payload with a protease-cleavable linker
- The pharmacokinetics (PK) of ZW49 is characterized by monitoring 3 PK analytes to define the relevance of each analyte to the safety and efficacy of ZW49
- We previously reported an acceptable safety profile and encouraging antitumor activity from this first-in-human, dose escalation (DE) and dose expansion (DX) study (NCT03821233)[1] in patients with HER2+ cancers.[2] Here we present the analyses of data supporting the selection of the ZW49 recommended dose (RD)

Structure and Proposed Mechanism of Action

- Immunoglobulin 1-like antibody backbone directed against extracellular domain 4 (ECD4) & ECD2 of HER2
- Antibody sequence is identical to zanidatamab (ZW25)
- Auristatin payload (ZD02044) covalently linked to the antibody via a protease cleavable valine-citrulline linker
- Antibody-induced internalization with increased toxin-mediated cytotoxicity and immunogenic cell death
- Average drug to antibody ratio (DAR) = 2

Figure 1: ZW49 Bispecific HER2 Antibody-Drug Conjugate

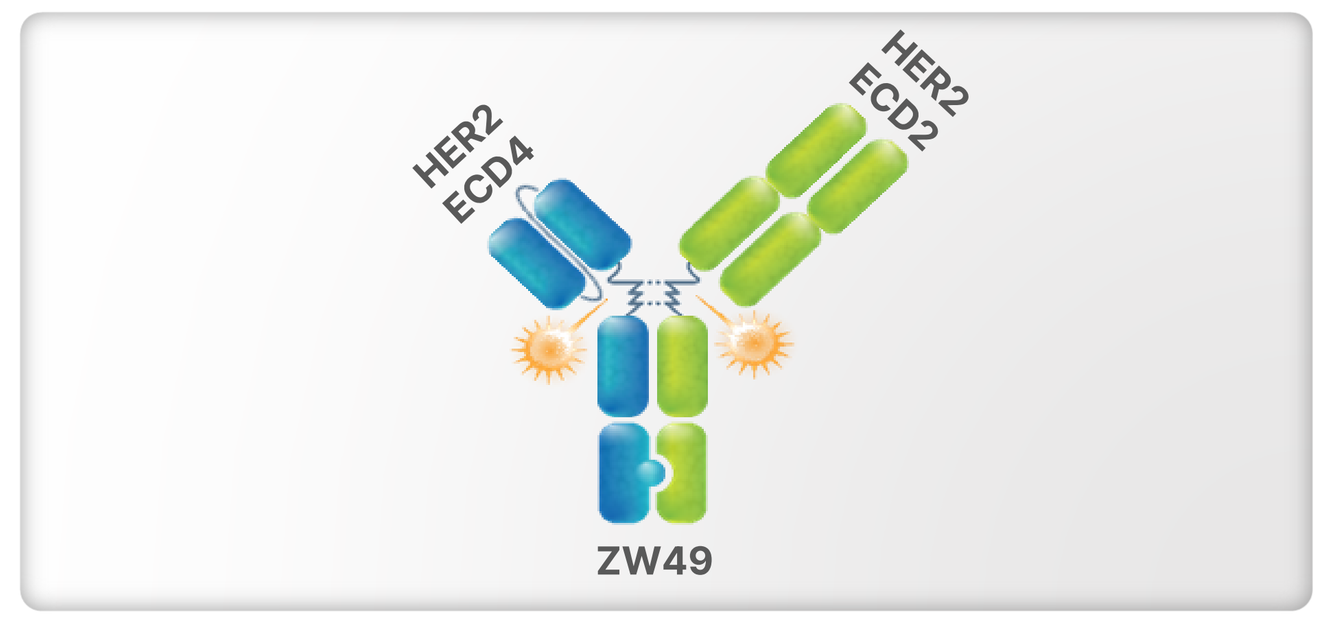

HER2 ECD4
HER2 ECD2
ZW49

Methods

- The 3+3 DE part evaluated safety and tolerability of ZW49 IV in HER2+ cancers. A safety monitoring committee (SMC) reviewed safety data and dose-limiting toxicities (DLTs) in DE to determine doses to be tested in DX. The DX part further evaluated safety, including DLTs, and antitumor activity of ZW49 IV in patients with centrally confirmed HER2+ cancers
- The DLT-evaluation period was Cycle 1 Day 1 through Day 28 (QW and Q2W regimens) or through Day 21 (Q3W regimen)
- Safety was evaluated using CTCAE V5.0 and antitumor activity was assessed using RECIST 1.1

PRIMARY OBJECTIVES
- Safety and tolerability
- Maximum tolerated dose (MTD)/RD

SECONDARY OBJECTIVES
- Pharmacokinetics and immunogenicity
- Antitumor activity

Figure 2: Study design

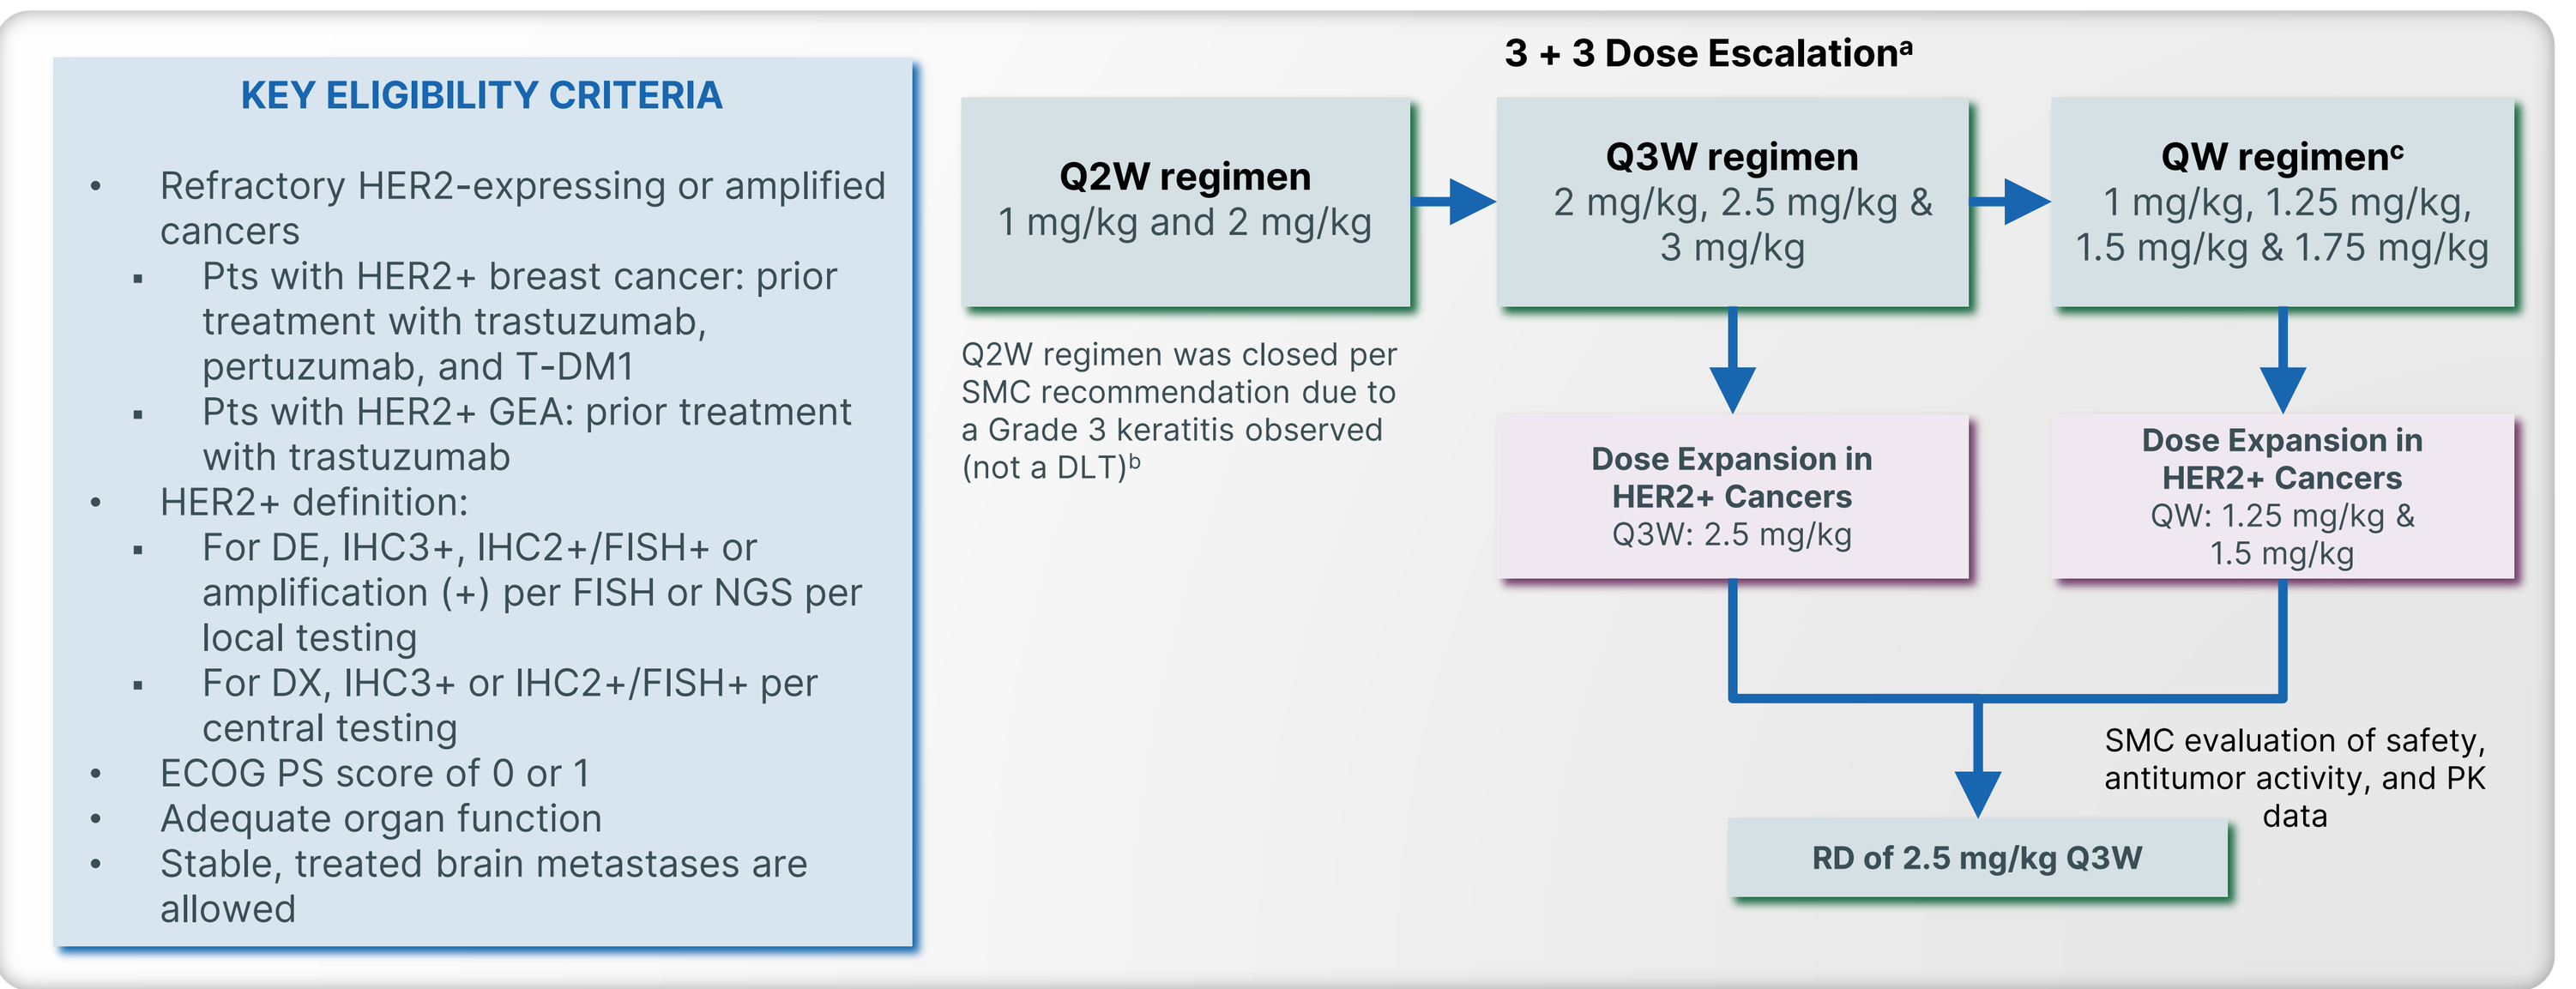

KEY ELIGIBILITY CRITERIA
Refractory HER2-expressing or amplified cancers
Pts with HER2+ breast cancer: prior treatment with trastuzumab, pertuzumab, and T-DM1
Pts with HER2+ GEA: prior treatment with trastuzumab
HER2+ definition:
For DE, IHC3+, IHC2+/FISH+ or amplification (+) per FISH or NGS per local testing
For DX, IHC3+ or IHC2+/FISH+ per central testing
ECOG PS score of 0 or 1
Adequate organ function
Stable, treated brain metastases are allowed
3 + 3 Dose Escalation[a]
Q2W regimen
1 mg/kg and 2 mg/kg
Q3W regimen
2 mg/kg, 2.5 mg/kg & 3 mg/kg
QW regimen[c]
1 mg/kg, 1.25 mg/kg, 1.5 mg/kg & 1.75 mg/kg
Q2W regimen was closed per SMC recommendation due to a Grade 3 keratitis observed (not a DLT)[b]
Dose Expansion in HER2+ Cancers
Q3W: 2.5 mg/kg
Dose Expansion in HER2+ Cancers
QW: 1.25 mg/kg & 1.5 mg/kg
SMC evaluation of safety, antitumor activity, and PK data
RD of 2.5 mg/kg Q3W

CTCAE = Common Terminology Criteria for Adverse Events; DE = dose escalation; DLT = dose-limiting toxicity; DX = dose expansion; ECOG PS = Eastern Cooperative Oncology Group performance status; FISH = fluorescence in situ hybridization; GEA = gastroesophageal adenocarcinoma; HER2 = human epidermal growth factor 2; IHC = immunohistochemistry; IRR = infusion-related reaction; NGS = next-generation sequencing; pts = patients; RD = recommended dose; QW = once every week; Q2W = once every 2 weeks; Q3W = once every 3 weeks; PK = pharmacokinetics; SMC = Safety Monitoring Committee; T-DM1 = ado-trastuzumab emtansine; ZW49 = zanidatamab zovodotin. a One additional pt was eligible to enroll in a DE cohort to account for potential non-DLT evaluable pts per SMC recommendation. b Ocular prophylaxis was subsequently implemented for other dose levels. c In QW regimen, 1.5 mg/kg QW initially selected to be tested in expansion, but closed due to the frequency of DLTs and dose modifications observed. SMC recommended to test the 1.25 mg/kg QW dose level in DX to further characterize the safety profile of the QW regimen.
Note: All pts received prophylaxis for IRRs prior to ZW49 administration.

Results

Analyses of data for the dose determining population (DE+DX) as of the data extract date of 10 Jul 2023 are presented

Patient disposition:

- Dose determining population included 67 patients treated with ZW49 at select cohorts in the 1.25 mg/kg (n =18) and 1.5 mg/kg (n = 18) QW; 2.5 mg/kg Q3W (n = 31) dosing regimens
- The most common (≥ 10%) cancer types were gastric (36%), breast (15%), and colorectal and biliary (both 10%) cancers
- Of the 67 patients, 4 (6%) continue on ZW49 treatment; 63 (94%) patients have discontinued ZW49 treatment: 51 (81%) due to disease progression, 8 (13%) due to adverse event (AE), and 4 (6%) due to withdrawal of consent
- The median duration of ZW49 exposure was 2.3 (range, 0.03 – 21.09) months

Results

Table 1: Demographics and Baseline Characteristics

Parameter	1.25 mg/kg QW (n = 18)	1.5 mg/kg QW (n= 18)	2.5 mg/kg Q3W (n = 31)	Total (n = 67)
Median age (range), years	63.5 (35, 83)	62.5 (41, 76)	59.0 (32, 75)	60.0 (32, 83)
Sex, n (%)				
Male	11 (61)	7 (39)	17 (55)	35 (52)
Female	7 (39)	11 (61)	14 (45)	32 (48)
Race, n (%)				
Asian	10 (56)	12 (67)	13 (42)	35 (52)
White	5 (28)	3 (17)	16 (52)	24 (36)
Black or African American	3 (17)	0	1 (3)	4 (6)
Other[a]	0	3 (17)	1 (3)	4 (6)
ECOG PS, n (%)				
0	8 (44)	8 (44)	11 (35)	27 (40)
1	10 (56)	10 (56)	20 (65)	40 (60)
HER2 Status[b], n (%)				
IHC 3+	12 (67)	12 (67)	22 (71)	46 (69)
IHC 2+/FISH +	3 (17)	4 (22)	9 (29)	16 (24)
ERBB2 Gene amp	3 (17)	2 (11)	0	5 (7)
Prior HER2 therapies, n(%)	12 (67)	11 (61)	22 (71)	45 (67)
Trastuzumab	12 (67)	11 (61)	22 (71)	45 (67)
Pertuzumab	2 (11)	4 (22)	6 (19)	12 (18)
T-DM1	1 (6)	3 (17)	8 (26)	12 (18)
T-DXd	2 (11)	1 (6)	3 (10)	6 (9)
ZW25	1 (6)	2 (11)	3 (10)	6 (9)
Lapatinib	1 (6)	1 (6)	2 (6)	4 (6)
Tucatinib	0	0	2 (6)	2 (3)
Neratinib	0	0	1 (3)	1 (1)
Margetuximab	0	0	1 (3)	1 (1)
Prior systemic cancer therapy in metastatic setting, median (range)	3 (1, 9)	2 (1, 5)	3 (1, 12)	3 (1, 12)

Amp = amplification; ECOG PS = Eastern Cooperative Oncology Group performance status; FISH = fluorescence *in situ* hybridization; HER2 = human epidermal growth factor 2; IHC = immunohistochemistry; QW = once every week; Q3W = once every 3 weeks; T-DM1 = ado-trastuzumab emtansine; T-DXd = trastuzumab deruxtecan; ZW25 = zanidatamab. a Other included: Not Reported (n=2); Unknown (n=1); and Other (n=1). b Missing HER2 status for 2 patients (1 each at 1.25 mg/kg QW and 2.5 mg/kg Q3W, respectively) as of the data extract date were subsequently entered into the database and the updated data are reflected on this table.

Safety:

- The 2.5 mg/kg Q3W and 1.5 mg/kg QW regimens were initially selected by the SMC to be tested in DX
- The 1.5 mg/kg QW regimen was subsequently closed due to the frequency of DLTs and dose modifications observed, and the SMC recommended to test the 1.25 mg/kg QW dose level in DX to further characterize the safety profile of the QW regimen

Summary:

- Of the 67 patients treated with ZW49 in the dose determining population (DE + DX), 60 (90%) experienced at least 1 treatment-related adverse event (TRAE; considered by the investigator as related to ZW49 treatment); the most common (≥ 20%) TRAEs were keratitis/punctate keratitis (49%), alopecia (25%), and diarrhoea (24%) with the majority of TRAEs Grade 1 or 2 in severity
- Grade ≥ 3 TRAEs occurred in 14 (21%) patients; of which all events were Grade 3 in severity except in 2 patients who experienced Grade 4 IRR and Grade 4 neutrophil count decreased, respectively
- In DE, no DLTs occurred for the 1.25 mg/kg QW, 1.5 mg/kg QW, or 2.5 mg/kg Q3W regimens, and an MTD was not reached. In DX, 7 DLTs occurred, including 4 DLTs in 1.5 mg/kg QW (1 Grade 3 and 2 Grade 2 keratitis/punctate keratitis events, and 1 Grade 3 diarrhea), 2 DLTs in 1.25 mg/kg QW (Grade 3 blurred vision and Grade 3 IRR), and 1 DLT in 2.5 mg/kg Q3W (Grade 2 keratitis) regimens

 Notes:
 - Grade 2 keratitis was considered a DLT if it delayed Cycle 2 by more than 2 weeks and had not resolved to ≤ Grade 1 or baseline
 - Grade 3 fatigue reported as a DLT for 1 patient (1.5 mg/kg QW) as of the data extract date was subsequently withdrawn and therefore, not included as a DLT
- Eight patients discontinued (D/C) due to TRAEs, including 5 patients D/C in 2.5 mg/kg Q3W (3 Grade 2 and 1 Grade 3 keratitis; and 1 serious TRAE of Grade 4 IRR); 1 patient D/C in 1.25 mg/kg QW (Grade 2 ophthalmic herpes zoster); and 2 patients D/C in 1.5 mg/kg QW (2 Grade 2 keratitis)
- TRAEs leading to dose reduction of ZW49 occurred in 20 (30%) patients; of which 14 (21%) patients had an ophthalmic event (12 [17%] patients had keratitis/punctate keratitis, 2 [3%] patients each had dry eye and vision blurred) and 2 (3%) patients had an IRR. No other dose reductions due to TRAE were reported for more than 1 patient.
- Across the 3 dosing regimens, treatment-related keratitis/punctate keratitis were observed in 33 (49%) patients. Of the 33 patients, 11 patients experienced maximum Grade 1 and 22 patients experienced maximum Grade ≥ 2 keratitis/punctate keratitis. Grade ≥ 2 keratitis/punctate keratitis events improved to Grade 1 or completely resolved with dose reductions, dose delays, and more frequent treatment with steroid and lubricating eye drops
 - Ocular prophylaxis with prednisolone, tetrahydrozoline or naphazoline (or equivalent eye drops), and application of cooling eye masks during the infusion is required for all patients
- No non-infectious pulmonary toxicity or treatment-related deaths occurred

Table 2: ZW49-related AEs in ≥ 20% of Patients, ≥ Grade 3 TRAE in 3 or more Patients, TR SAEs in ≥ 1 Patient, and TR AESIs

	1.25 mg/kg QW (n=18)		1.5 mg/kg QW (n=18)		2.5 mg/kg Q3W (n=31)		Total (n=67)	
	Any Grade	Gr ≥ 3	Any Grade	Gr ≥ 3	Any Grade	Gr ≥ 3	Any Grade	Gr ≥ 3
Any TRAE, n(%)	14 (78)	4 (22)	18 (100)	5 (28)	28 (90)	5 (16)	60 (90)	14 (21)
TRAE in ≥ 20% patients or Grade ≥ 3 TRAE in 3 or more pts								
Keratitis/punctate keratitis	7 (39)	0	12 (67)	1 (6)	14 (45)	1 (3)[a]	33 (49)	2 (3)
Alopecia	2 (11)	0	10 (56)	0	5 (16)	0	17 (25)	0
Diarrhoea	1 (6)	0	6 (33)	2 (11)	9 (29)	0	16 (24)	2 (3)
IRR	5 (28)	1 (6)	3 (17)	1 (6)	5 (16)	1 (3)	13 (19)	3 (4)
TR SAEs ≥ 1 pt								
Any AE	1 (6)	0	1 (6)	1 (6)	2 (6)	2 (6)	4 (6)	3 (4)
IRR	0	0	1 (6)	1 (6)	1 (3)	1 (3)	2 (3)	2 (3)
ECG QT Prolonged	0	0	0	0	1 (3)	1 (3)	1 (1)	1 (1)
Herpes zoster	1 (6)	0	0	0	0	0	1 (1)	0
TR AESIs								
Gr ≥ 2 ophthalmic events	5 (28)[b]	1 (6)	8 (44)[b]	1 (6)	12 (39)[b]	1 (3)	25 (37)[b]	3 (4)
Keratitis	3 (17)	0	6 (33)	1 (6)	10 (32)	1 (3)	19 (28)	2 (3)
Vision blurred	2 (11)	1 (6)	0	0	1 (3)	0	3 (4)	1 (1)
Punctate keratitis	0	0	2 (11)	0	1 (3)	0	3 (4)	0
Dry eye	0	0	1 (6)	0	2 (6)	0	3 (4)	0
Visual impairment	0	0	0	0	1 (3)	0	1 (1)	0
Cardiac events[c,d]	1 (6)	0	1 (6)	0	1 (3)	0	3 (4)	0
IRR	5 (28)	1 (6)	3 (17)	1 (6)	5 (16)	1 (3)	13 (19)	3 (4)
Non-infectious pulmonary toxicity	0	0	0	0	0	0	0	0

AE = adverse event; AESI = adverse event of special interest; ECG = electrocardiogram; ECHO = echocardiogram; Gr = Grade; IRR = infusion-related reaction; LVEF = left ventricular ejection fraction; MedDRA = Medical Dictionary for Regulatory Activities; MUGA = multigated acquisition scan; QT = QT interval; QW = once every week; Q3W = once every 3 weeks; TEAE = treatment-emergent adverse event; TRAE = treatment-related adverse event; SAE = serious adverse event; SMQ = standardized MedDRA query. a This event started as a Grade 2 keratitis (> 14 days) and the Grade 2 event was assessed as a DLT. b Includes only Grade ≥ 2 ophthalmic events. c Potential cardiac events are defined as either ECHO or MUGA results for LVEF decrease ≥ 10 percentage points from pretreatment baseline LVEF, and/or Grade ≥ 2 TEAEs meeting the broad cardiac failure SMQ or the narrow myocardial infarction SMQ. d All reported events were Grade 2 ejection fraction decreased.

PK Summary:

- Pharmacokinetics of ZW49 is characterized by monitoring three analytes:
 - Antibody drug conjugate (ADC), which quantifies antibody with at least one payload conjugated
 - Total antibody (TAb), including fully conjugated, partially deconjugated, and fully deconjugated antibodies
 - Unconjugated ZD02044 (toxin)
- Preliminary PK parameters of each analyte for patients at 1.25 mg/kg QW, 1.5 mg/kg QW, and 2.5 mg/kg Q3W extensive PK sampling schedule were derived from non-compartmental analysis (NCA) using serum or plasma concentration versus sampling time profile (Figure 3)
- Following first IV infusion of ZW49, concentrations declined with a mean terminal half-life ($t_{1/2}$) of approximately 2.13~2.85 days and 1.91~2.43 days for ADC and TAb, respectively (Table 3)
- Based on PK of ADC and TAb, $AUC_{(0-\infty)}$ appeared dose-proportional among the three dose regimens. The maximal toxin concentration ranged from 0.354 to 0.693 ng/mL

Table 3: Geometric Mean (CV%) PK Parameters Following First Dose of ZW49 (Extensive PK Sampling)

PK Analyte	Dosing Regimen	$t_{1/2}$ (d)	C_{max} (µg/mL)[a]	C_{trough}[b] (µg/mL)	$AUC_{0-\infty}$ (d* µg /mL)	V_z (mL/kg)	C_L (mL/d/kg)
ADC	1.25 mg/kg QW (N=7)	2.13 (43)	40.9 (31)	1.98 (111)	109 (40)	31.2 (48)	11.1 (40)
	1.5 mg/kg QW (N=9)	2.44 (47)	48.4 (27)	2.71 (517)	146 (59)	32.5 (22)	10.3 (59)
	2.5 mg/kg Q3W (N=13)	2.85 (24)	64.5 (20)	0.233 (155)	213 (33)	47.0 (24)	11.7 (33)
TAb	1.25 mg/kg QW (N=7)	1.91 (51)	31.5 (37)	1.02 (225)	81.4 (44)	36.4 (54)	14.7 (44)
	1.5 mg/kg QW (N=9)	2.14 (37)	39.3 (23)	3.49 (82)	113 (70)	38.0 (27)	13.3 (70)
	2.5 mg/kg Q3W (N=13)	2.43 (22)	60.8 (20)	0.162 (156)	215 (36)	39.9 (25)	11.6 (36)
Toxin	1.25 mg/kg QW (N=1)	N/C	0.354	0.109	N/C	N/C	N/C
	1.5 mg/kg QW (N=2)	N/C	0.168 (24)	0.142 (31)	N/C	N/C	N/C
	2.5 mg/kg Q3W (N=13)	N/C	0.693 (81)	0.069 (33)	N/C	N/C	N/C

ADC = antibody drug conjugate; $AUC_{0-\infty}$ = area under the curve the serum concentration versus time curve from time zero to infinity; C_L = serum clearance; C_{max} = maximum serum concentration; C_{trough} = trough concentration; CXDX = Cycle X Day X; N/C = not calculated due to R2 value for Kel less than 0.80; PK = pharmacokinetics; QW = once a week; Q3W = once every 3 weeks; $t_{1/2}$= terminal half-life; TAb = total antibody; V_z= terminal elimination phase; ZW49 = zanidatamab zovodotin.
a µg/mL for ADC and TAb; ng/mL for Toxin. b C_{trough} is summarize based on predose of C1D8 for QW regimen and predose of C2D1 for Q3W regimen.
Note: All PK parameters are reported in geometric means (CV%) except for $t_{1/2}$ (arithmetic mean with %CV).

Figure 3: Concentration vs. Time Profile of ADC/TAb (Left) and Toxin (Right) after First Dose of ZW49 (Extensive + Sparse PK Sampling)

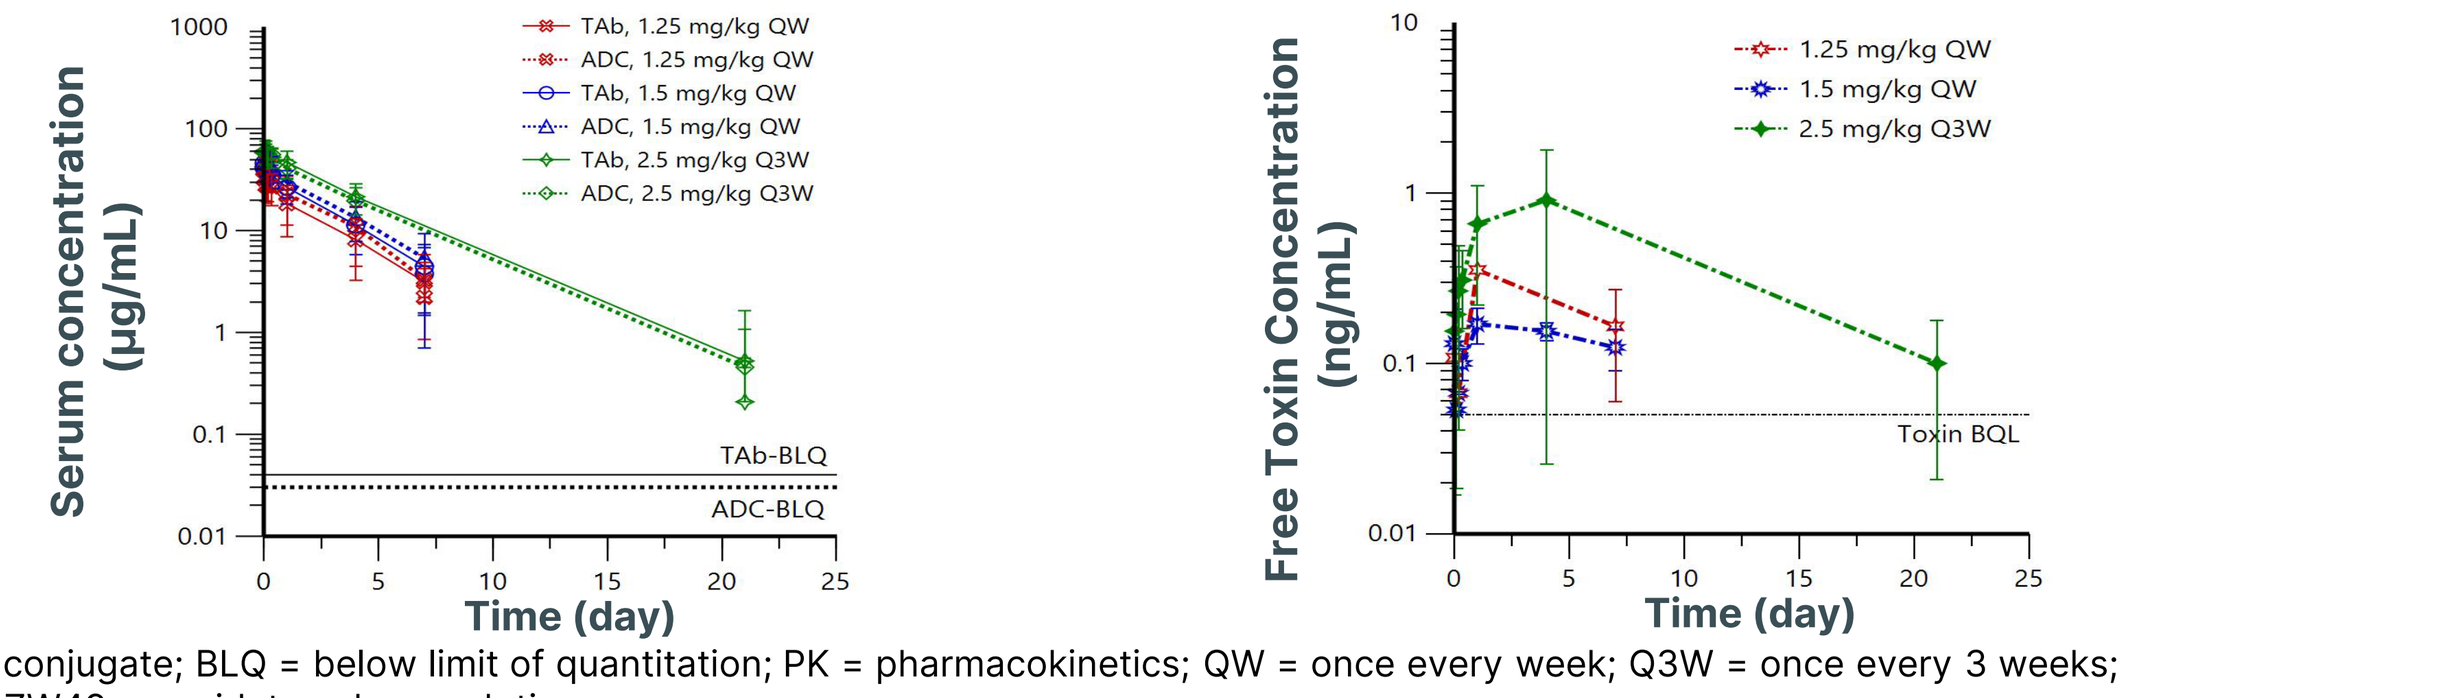

Serum concentration (µg/mL)
Time (day)
TAb, 1.25 mg/kg QW
ADC, 1.25 mg/kg QW
TAb, 1.5 mg/kg QW
ADC, 1.5 mg/kg QW
TAb, 2.5 mg/kg Q3W
ADC, 2.5 mg/kg Q3W
TAb-BLQ
ADC-BLQ
Free Toxin Concentration (ng/mL)
Time (day)
1.25 mg/kg QW
1.5 mg/kg QW
2.5 mg/kg Q3W
Toxin BQL

ADC = antibody drug conjugate; BLQ = below limit of quantitation; PK = pharmacokinetics; QW = once every week; Q3W = once every 3 weeks; TAb = total antibody; ZW49 = zanidatamab zovodotin.

Anti-tumor activity:

Efficacy data for the 64 HER2+ response-evaluable patients treated with ZW49 in the dose determining population (DE + DX) are presented in Table 4. The median duration of response in the 2.5 mg/kg Q3W regimen was 6.8 (range, 1.4 – 19.8*) months with 1 response ongoing

Table 4: Efficacy for Response-evaluable Patients with HER2+ Cancers per Central Results[a,b]:

	1.25 mg/kg QW (N = 17)	1.5 mg/kg QW (N = 17)	2.5 mg/kg Q3W (N = 30)	Total (N = 64)
cORR, %(95% CI)	12 (1.5, 36.4)	18 (3.8, 43.4)	30 (14.7, 49.4)	22 (12.5, 34.0)
PR, n (%)	2 (12)	3 (18)[c]	9 (30)	14 (22)
SD, n (%)	7 (41)	10 (59)	13 (43)	30 (47)
PD, n (%)	8 (47)	4 (24)	7 (23)	19 (30)
NE, n (%)	0	0	1 (3)	1 (2)
DCR[d], % (95% CI)	53 (27.8. 77.0)	77 (50.1, 93.2)	73 (54.1, 87.7)	69 (55.9, 79.8)
CBR[e], % (95% CI)	29 (10.3, 56.0)	35 (14.2, 61.7)	37 (19.9, 56.1)	34 (22.9, 47.3)

CBR = clinical benefit rate; cORR = confirmed objective response rate; CR = complete response; DCR = disease control rate; GEA = gastroesophageal adenocarcinoma; NE = not evaluable; PR = partial response; RECIST = Response Evaluation Criteria in Solid Tumours; SD = stable disease; PD = progressive disease.
a Response-evaluable includes all patients with measurable disease who had at least one post-baseline disease assessment (per RECIST 1.1) or discontinued the study due to death or clinical progression. b HER2-positive was defined as IHC3+ or IHC 2+/FISH+. c GEA = 2 pts and salivary gland carcinoma = 1 pt. d DCR = CR, PR, or SD.
e CBR = SD ≥ 24 weeks or best overall response of CR or PR.

Figure 4: Treatment Duration: Patients with HER2+ Cancers treated with ZW49 at 1.25 mg/kg QW and 2.5 mg/kg Q3W Dosing Regimens

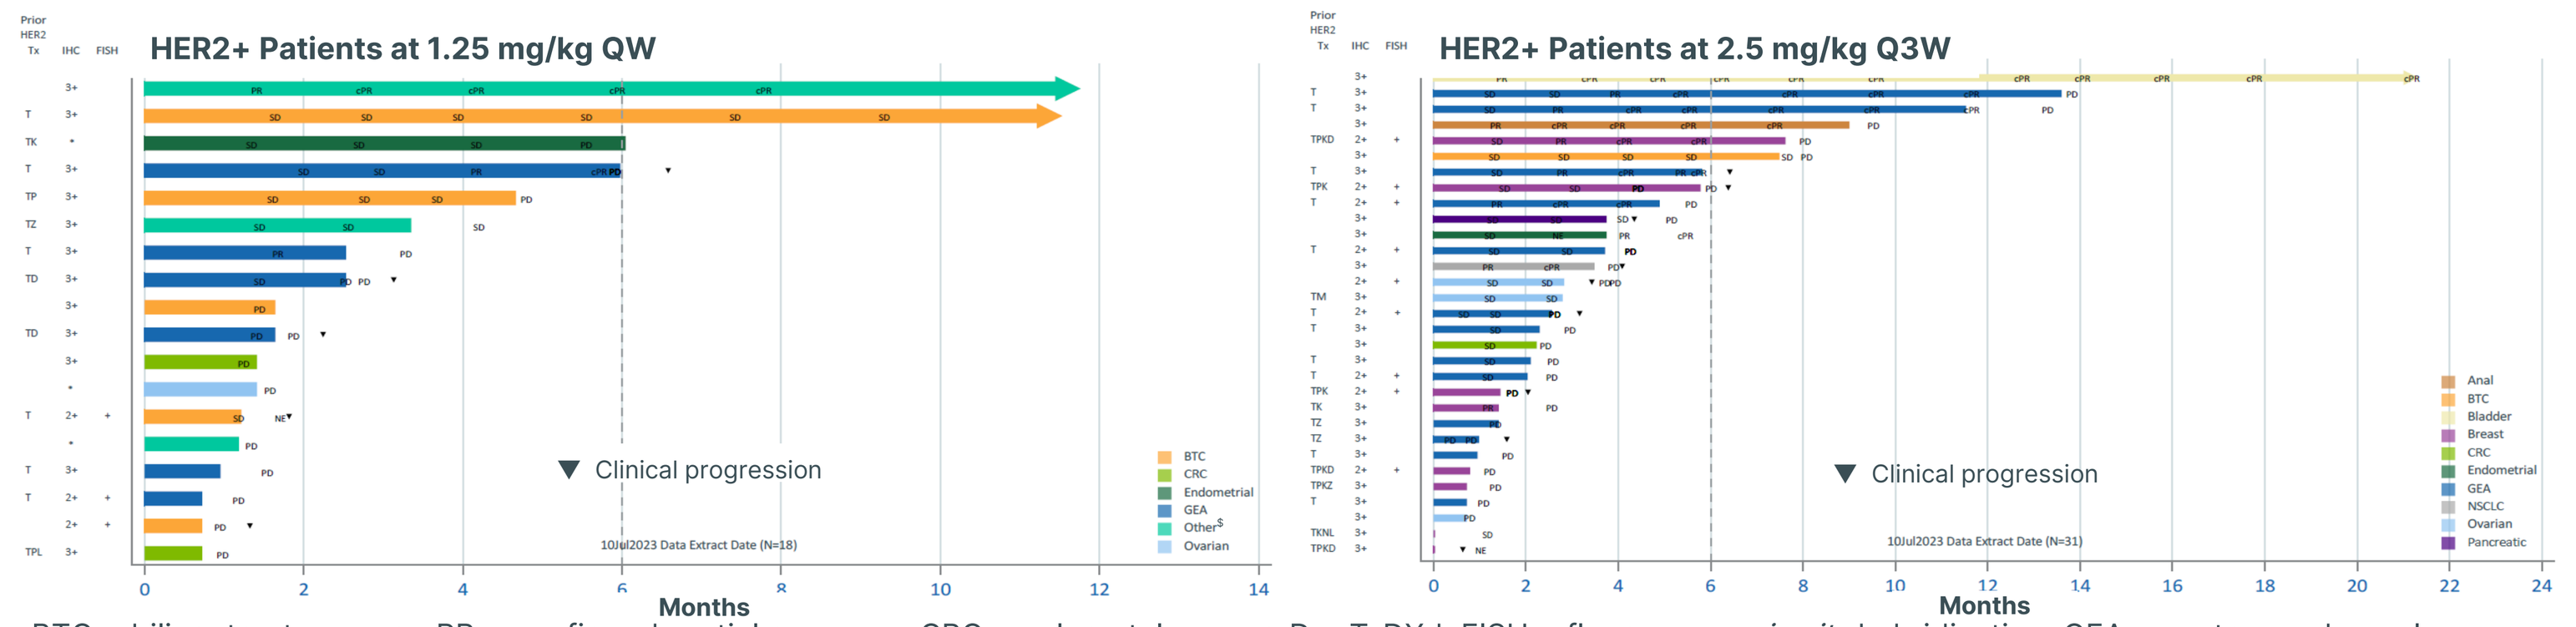

HER2+ Patients at 1.25 mg/kg QW
Prior HER2 Tx
IHC
FISH
Clinical progression
10Jul2023 Data Extract Date (N=18)
BTC
CRC
Endometrial
GEA
Other$
Ovarian
Months
HER2+ Patients at 2.5 mg/kg Q3W
Clinical progression
10Jul2023 Data Extract Date (N=31)
Anal
BTC
Bladder
Breast
CRC
Endometrial
GEA
NSCLC
Ovarian
Pancreatic
Months

BTC = biliary tract cancer; cPR = confirmed partial response; CRC = colorectal cancer; D = T-DXd; FISH = fluorescence *in situ* hybridization; GEA = gastroesophageal adenocarcinoma; IHC = immunohistochemistry; K = T-DM1; L = lapatinib; M = margetuximab; N = neratinib; NE = not evaluable; NSCLC = non-small cell lung cancer; P = pertuzumab; PD = progressive disease; PR = partial response; pts = patients; QW = once every week; Q3W = once every 3 weeks; SD = stable disease; T = trastuzumab; Tx = therapy; Z = zanidatamab; ZW49 = zanidatamab zovodotin.
* = ERBB2 gene amplification status. $ Other included bladder cancer = 2 pts and fallopian tube carcinoma = 1 pt.

Conclusions

- Safety profile observed was comparable between the 1.25 mg/kg QW and 2.5 mg/kg Q3W regimens
- The PK of ADC and TAb was comparable and appeared to be linear among the three dose regimens examined
- Based on a comprehensive review of the safety and preliminary antitumor activity data, ZW49 2.5 mg/kg Q3W IV was identified as the RD
- ZW49 2.5 mg/kg Q3W IV has an acceptable safety profile where the majority of patients experienced AEs of maximum Grade 1 or 2 in severity
 - Keratitis events were low grade, manageable and reversible
 - No interstitial lung disease or pneumonitis
 - No treatment-related deaths
- ZW49 2.5 mg/kg Q3W IV showed encouraging antitumor activity in heavily pretreated patients with advanced HER2+ cancers
 - 30% cORR with a response duration range of 1.4 – 19.8* months
- These data support further investigation of ZW49 2.5 mg/kg Q3W IV as a potential novel treatment option for advanced HER2+ cancers

References

1. ClinicalTrials.gov. https://clinicaltrials.gov/study/NCT03821233?term=zw49&checkSpell=false&rank=1 Accessed 16 Aug 2023.
2. Jhaveri, et al. Presented at: European Society for Medical Oncology; 2022. Oral Presentation.

Acknowledgements

We sincerely thank all patients and their families. We thank all the investigators, clinical trial researchers, personnel, and staff who contributed to the trial.
ZWI-ZW49-101 study is sponsored by Zymeworks BC Inc.

Copies of this e-Poster obtained through QR codes are for personal use only and may not be reproduced without written permission of the authors

Presented at the AACR-NCI-EORTC Conference October 11 – 15, 2023