Zanidatamab zovodotin (ZW49) induces hallmarks of immunogenic cell death and is active in patient-derived xenograft models of gastric cancer

AACR Annual Meeting 2023
Abstract #2633

Stuart Barnscher, Andrea Hernández Rojas, Kevin Hamblett, Nichole Escalante, and Renee Duan
Zymeworks Inc., Vancouver, BC, Canada

Introduction

- A phase 1 clinical study of zanidatamab zovodotin (ZW49) in HER2-expressing advanced solid tumors is currently underway (NCT03821233).
- We previously presented data illustrating the internalization profile of ZW49 and anti-tumor activity in patient-derived xenograft models originating from breast carcinomas.[1]
- Here we demonstrate induction of in vitro hallmarks of immunogenic cell death (ICD) by ZW49 and illustrate its anti-tumor activity in an expanded panel of patient-derived xenograft models of HER2-expressing breast and gastric cancer.

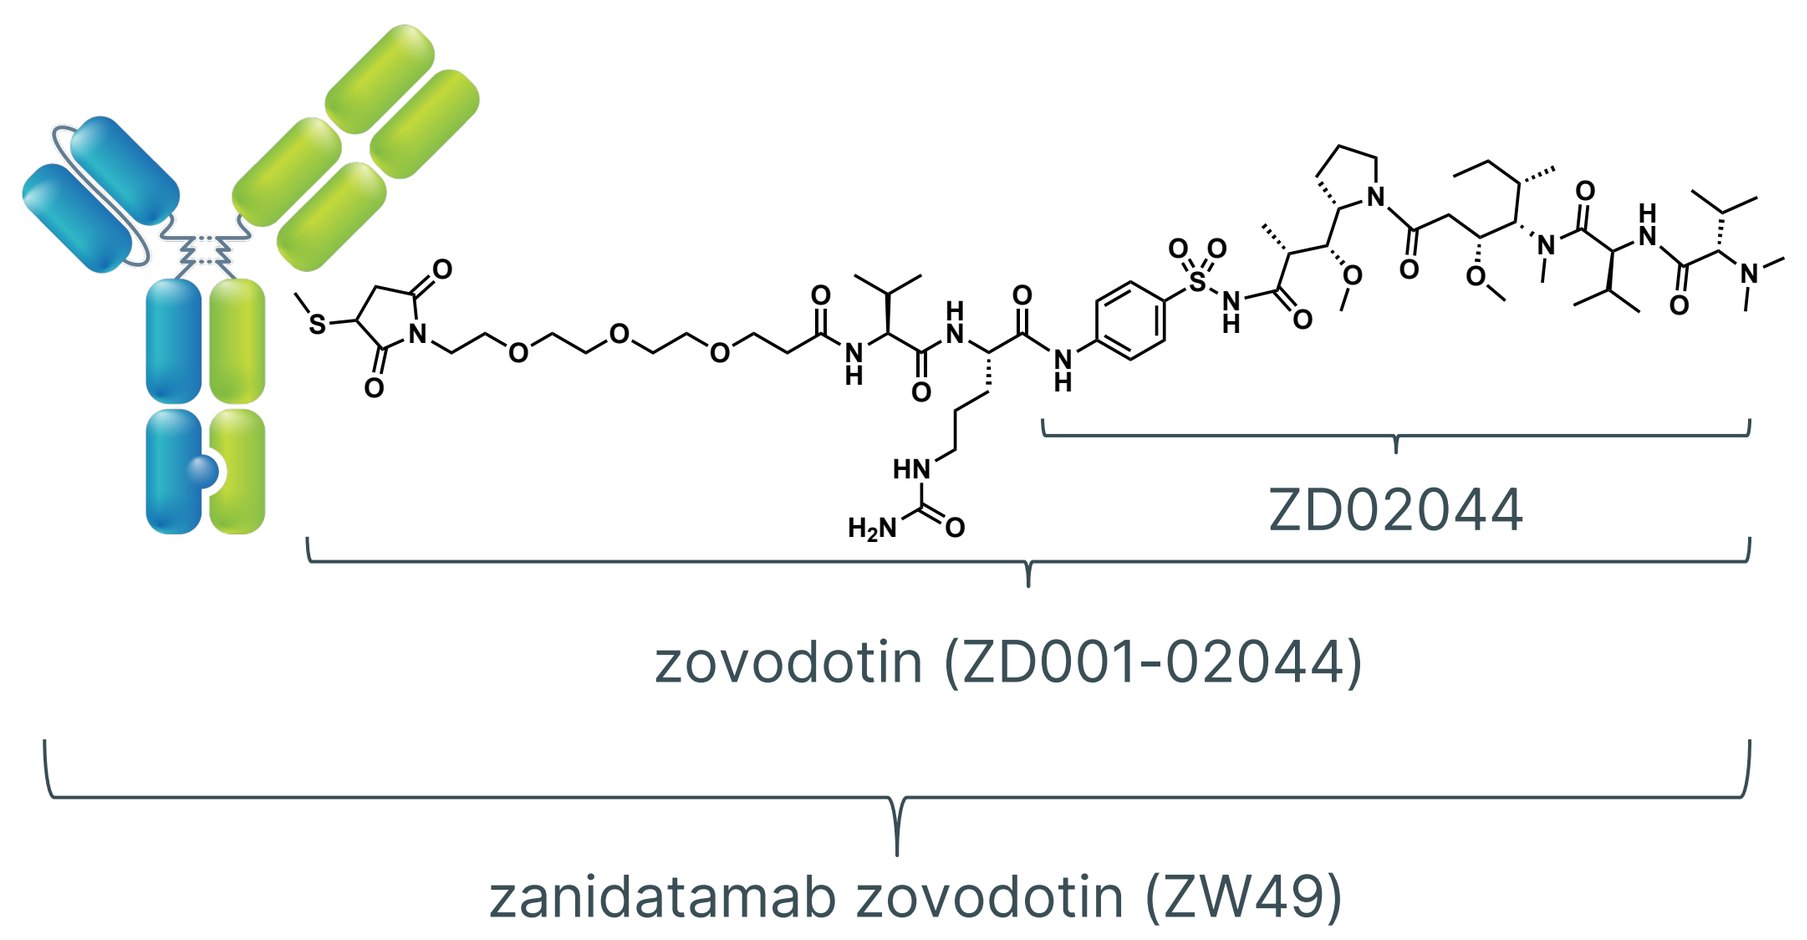
ZD02044
zovodotin (ZD001-02044)
zanidatamab zovodotin (ZW49)

Figure 1. Zanidatamab zovodotin (ZW49) consists of an anti-HER2 biparatopic IgG1 antibody (ZW25, zanidatamab) conjugated to a microtubule inhibitor auristatin payload (ZD02044) via a protease cleavable linker at a DAR of 2.

Zanidatamab is rapidly internalized and traffics to lysosomes

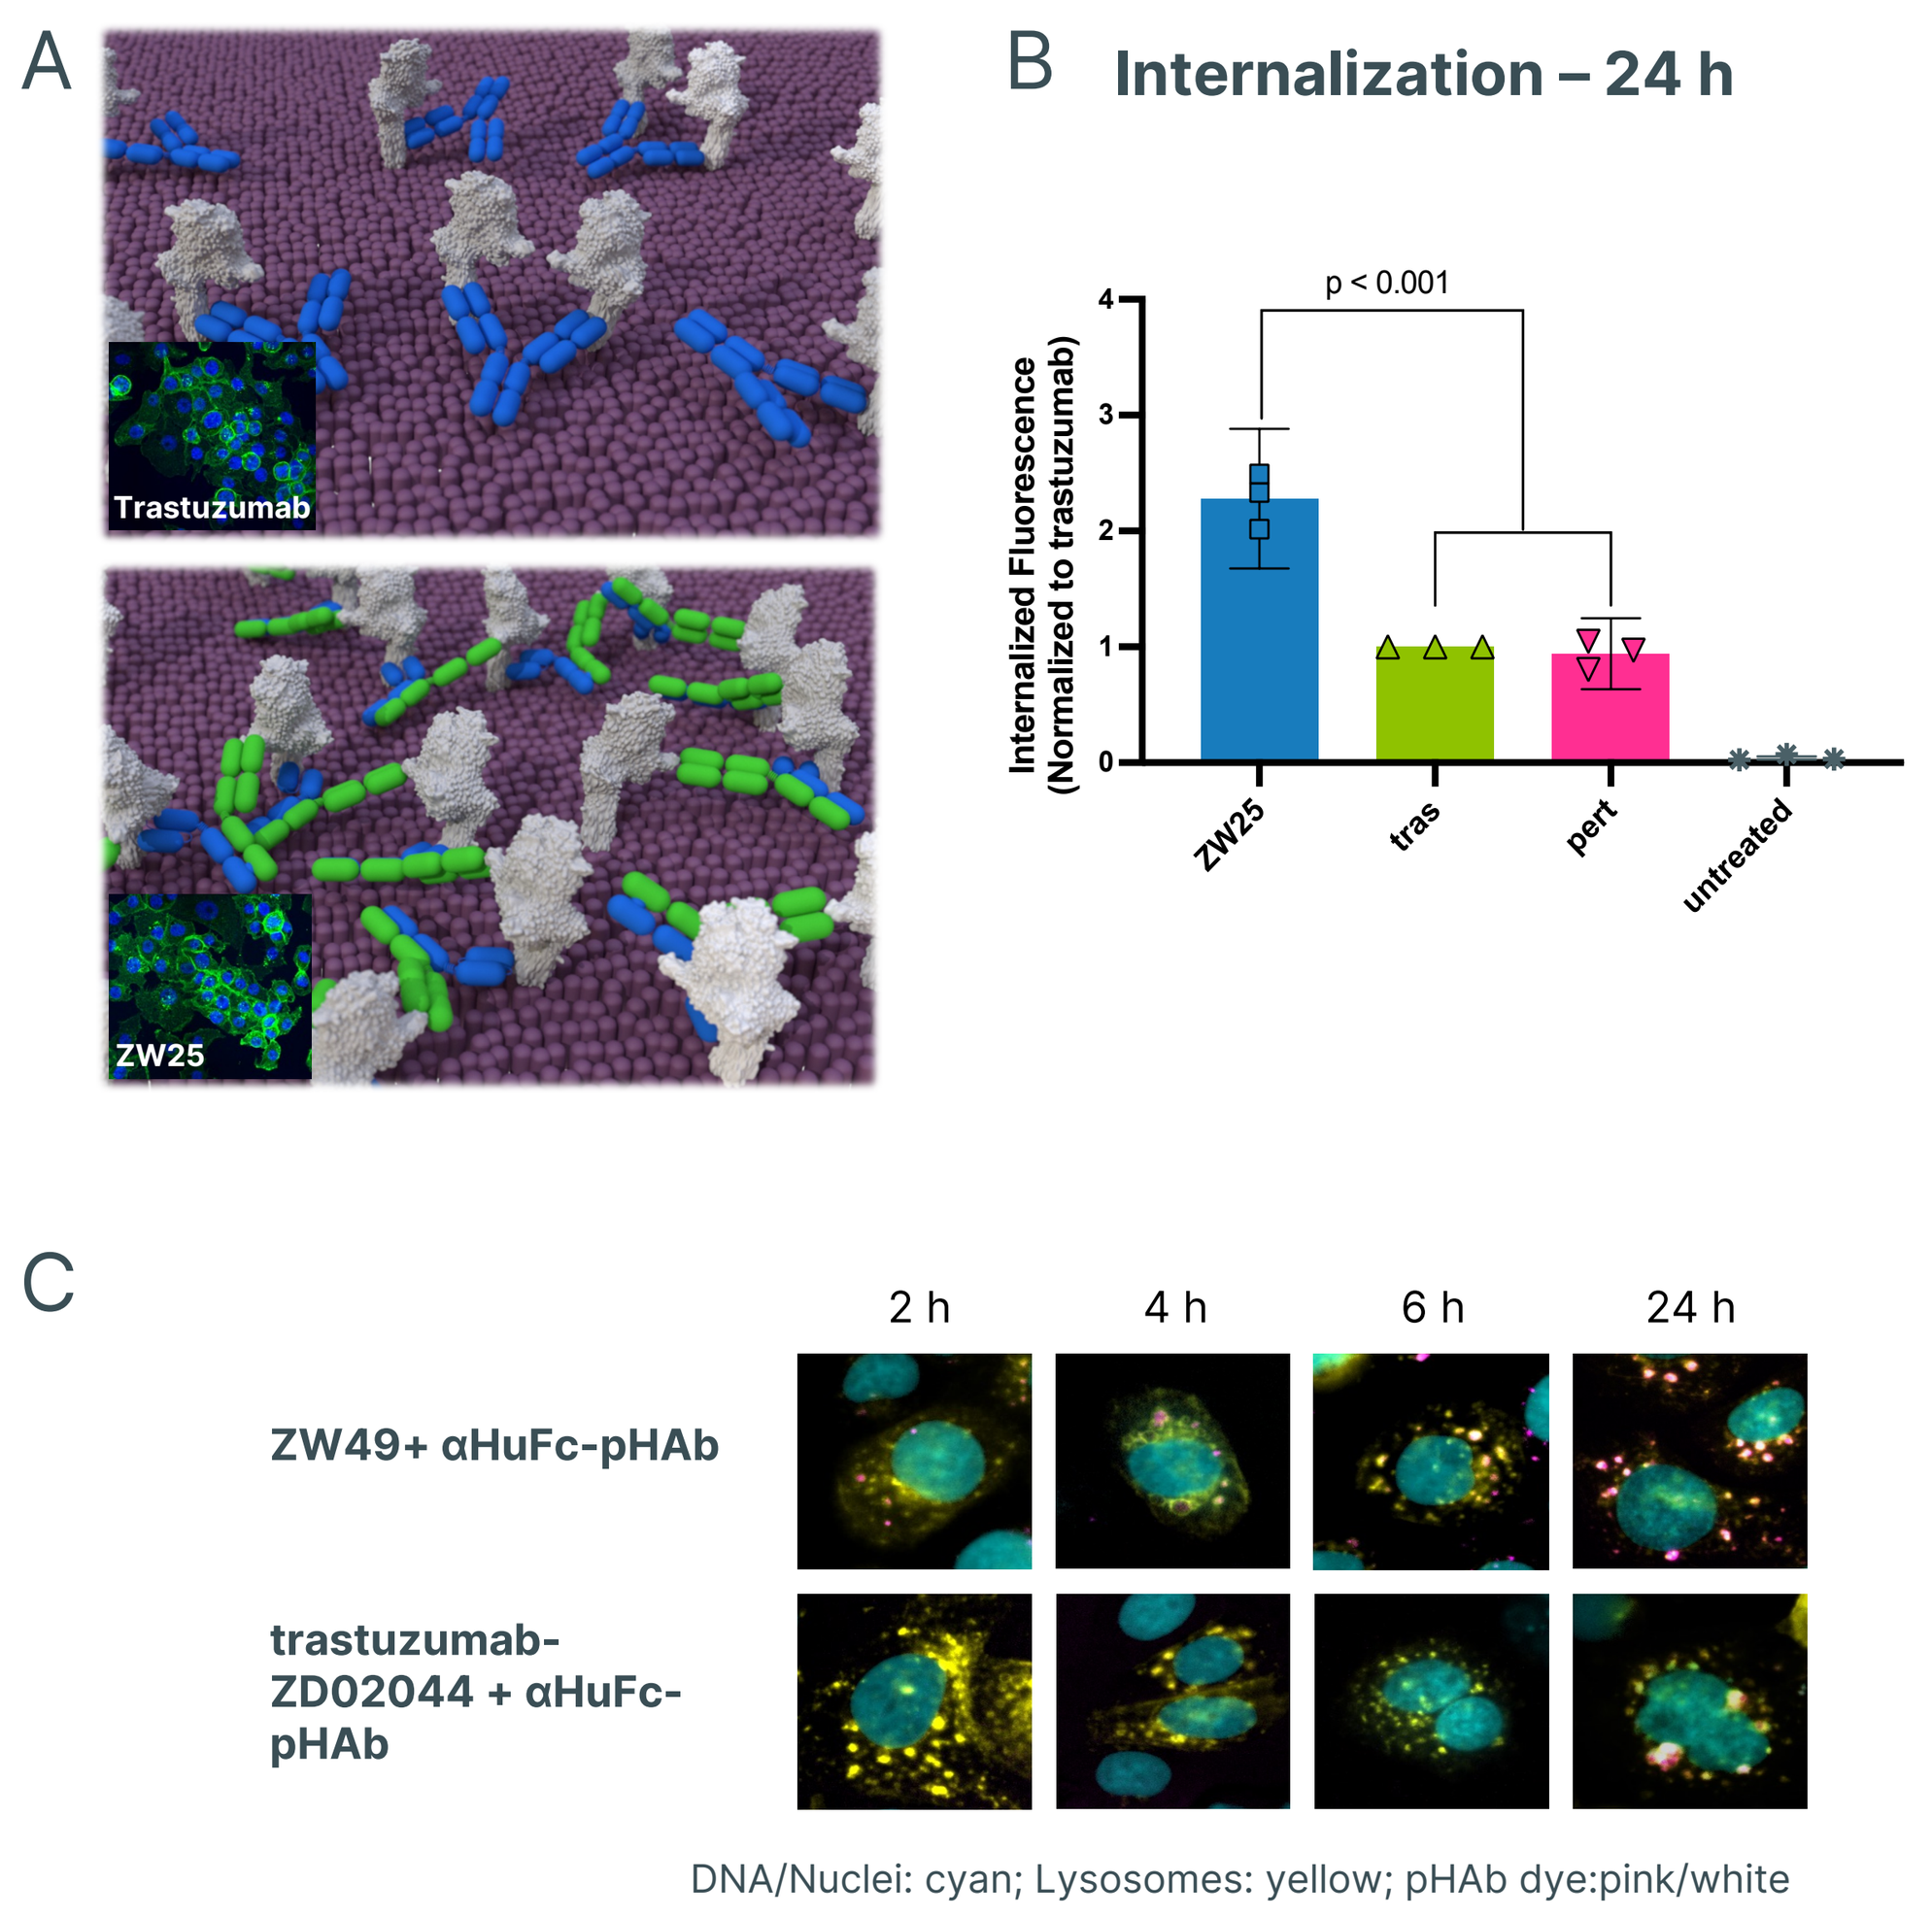
A
Trastuzumab
ZW25
B
Internalization – 24 h
p < 0.001
Internalized Fluorescence (Normalized to trastuzumab)
ZW25
tras
pert
untreated
C
2 h
4 h
6 h
24 h
ZW49+ αHuFc-pHAb
trastuzumab-ZD02044 + αHuFc-pHAb
DNA/Nuclei: cyan; Lysosomes: yellow; pHAb dye:pink/white

Figure 2. (A) Cartoon of monoparatopic antibody binding of trastuzumab (top) compared to biparatopic antibody binding of ZW25 (bottom). (B) Comparison of ZW25 internalization to trastuzumab (tras) and pertuzumab (pert) in SK-BR-3 cells measured by flow cytometry.[2] (D) Comparison of lysosomal co-localization of ZW49 to trastuzumab-ZD02044 in SK-BR-3 cells measured using a pH sensitive dye, pHAb.

ZW49 drives higher intracellular payload accumulation compared to a trastuzumab ADC

A

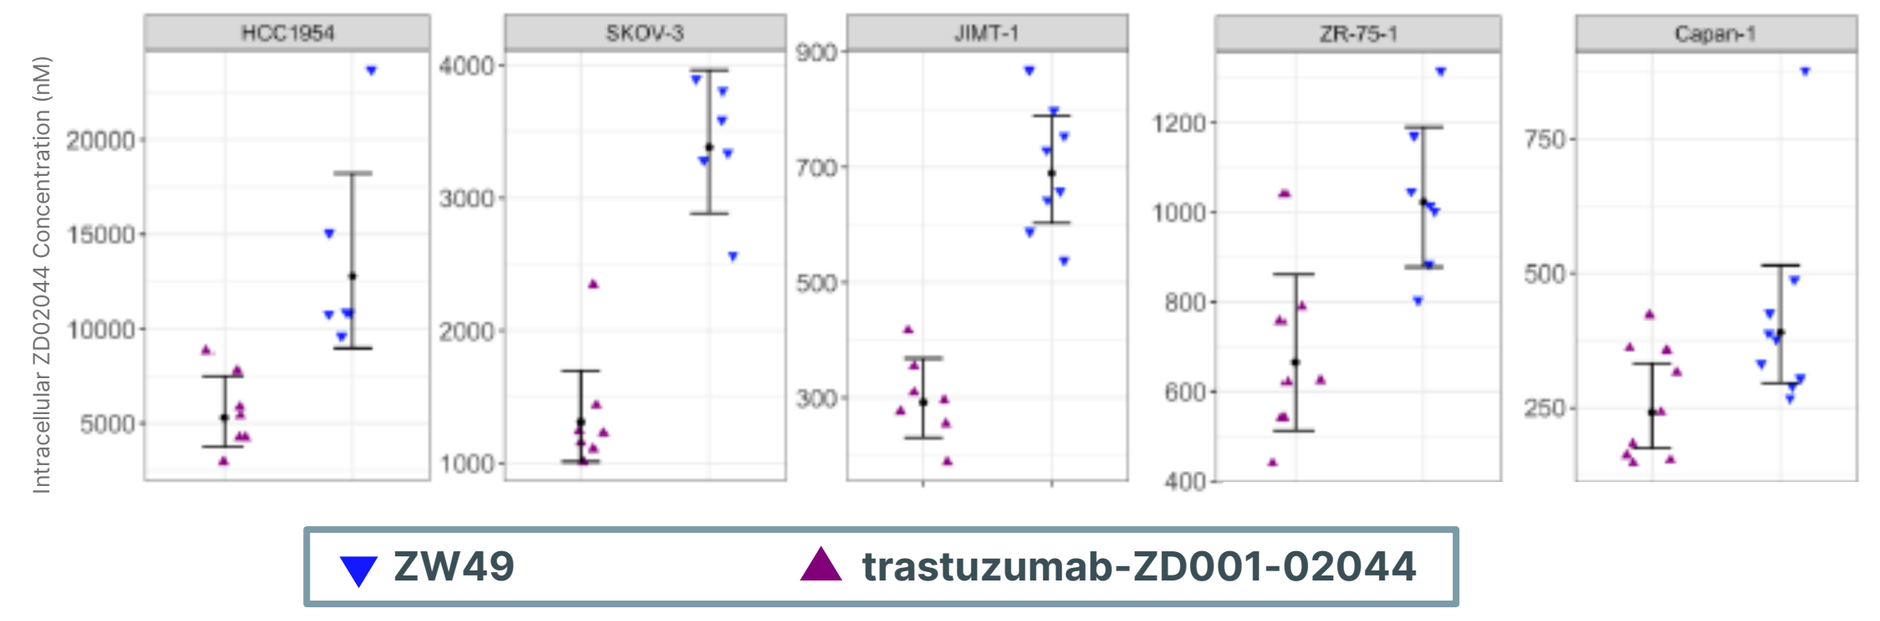
HCC1954
SKOV-3
JIMT-1
ZR-75-1
Capan-1
Intracellular ZD02044 Concentration (nM)
ZW49
trastuzumab-ZD001-02044

B

Cell line	HER2 receptors/cell	Fold difference in ZD02044 concentration ZW49/trastuzumab-ZD001-02044	P-value
HCC1954	4,930,000	**2.41**	0.003
SKOV-3	1,310,000	**2.58**	<0.0001
JIMT-1	336,000	**2.36**	<0.0001
ZR-75-1	352,000	**1.54**	0.0116
Capan-1	167,000	**1.62**	0.0184

Figure 3. (A) Intracellular payload (ZD02044) concentration was determined by LC-MS/MS after tumor cells were treated for 24 hours. (B) Table of fold difference in ZD02044 concentration for ZW49 compared to trastuzumab-ZD001-02044.

ZW49 has potent cell killing activity against HER2-expressing breast cancer cells in vitro

Cell type	Cell line	HER2 receptors/cell	EC50 (nM) ZW49	EC50 (nM) ZW25	EC50 (nM) ZD02044
Breast Cancer	HCC1954	6,000,000	**0.04 ± 0.01**	No activity	4.70 ± 0.93
	SK-BR-3	3,660,000	**0.04 ± 0.02**	0.31	10.01 ±1.24
	JIMT-1	526,000	**0.50 ± 0.09**	No activity	44.35 ± 4.71
	ZR-75-1	378,00	**0.58 ± 0.46**	No activity	19.65 ± 3.43
	MDA-MB-175	287,000	**0.62 ± 0.10**	1.96 ± 0.38	13.88 ± 1.22
	T-47D	206,000	**No activity**	No activity	61.46 ± 5.78

Figure 4. Table of cytotoxicity for ZW49, ZW25 (unconjugated antibody), and ZD02044 (free payload) in breast cancer cell lines representing a range of HER2 expression.

ZW49 is active in xenograft models of breast cancer with a range of HER2 expression

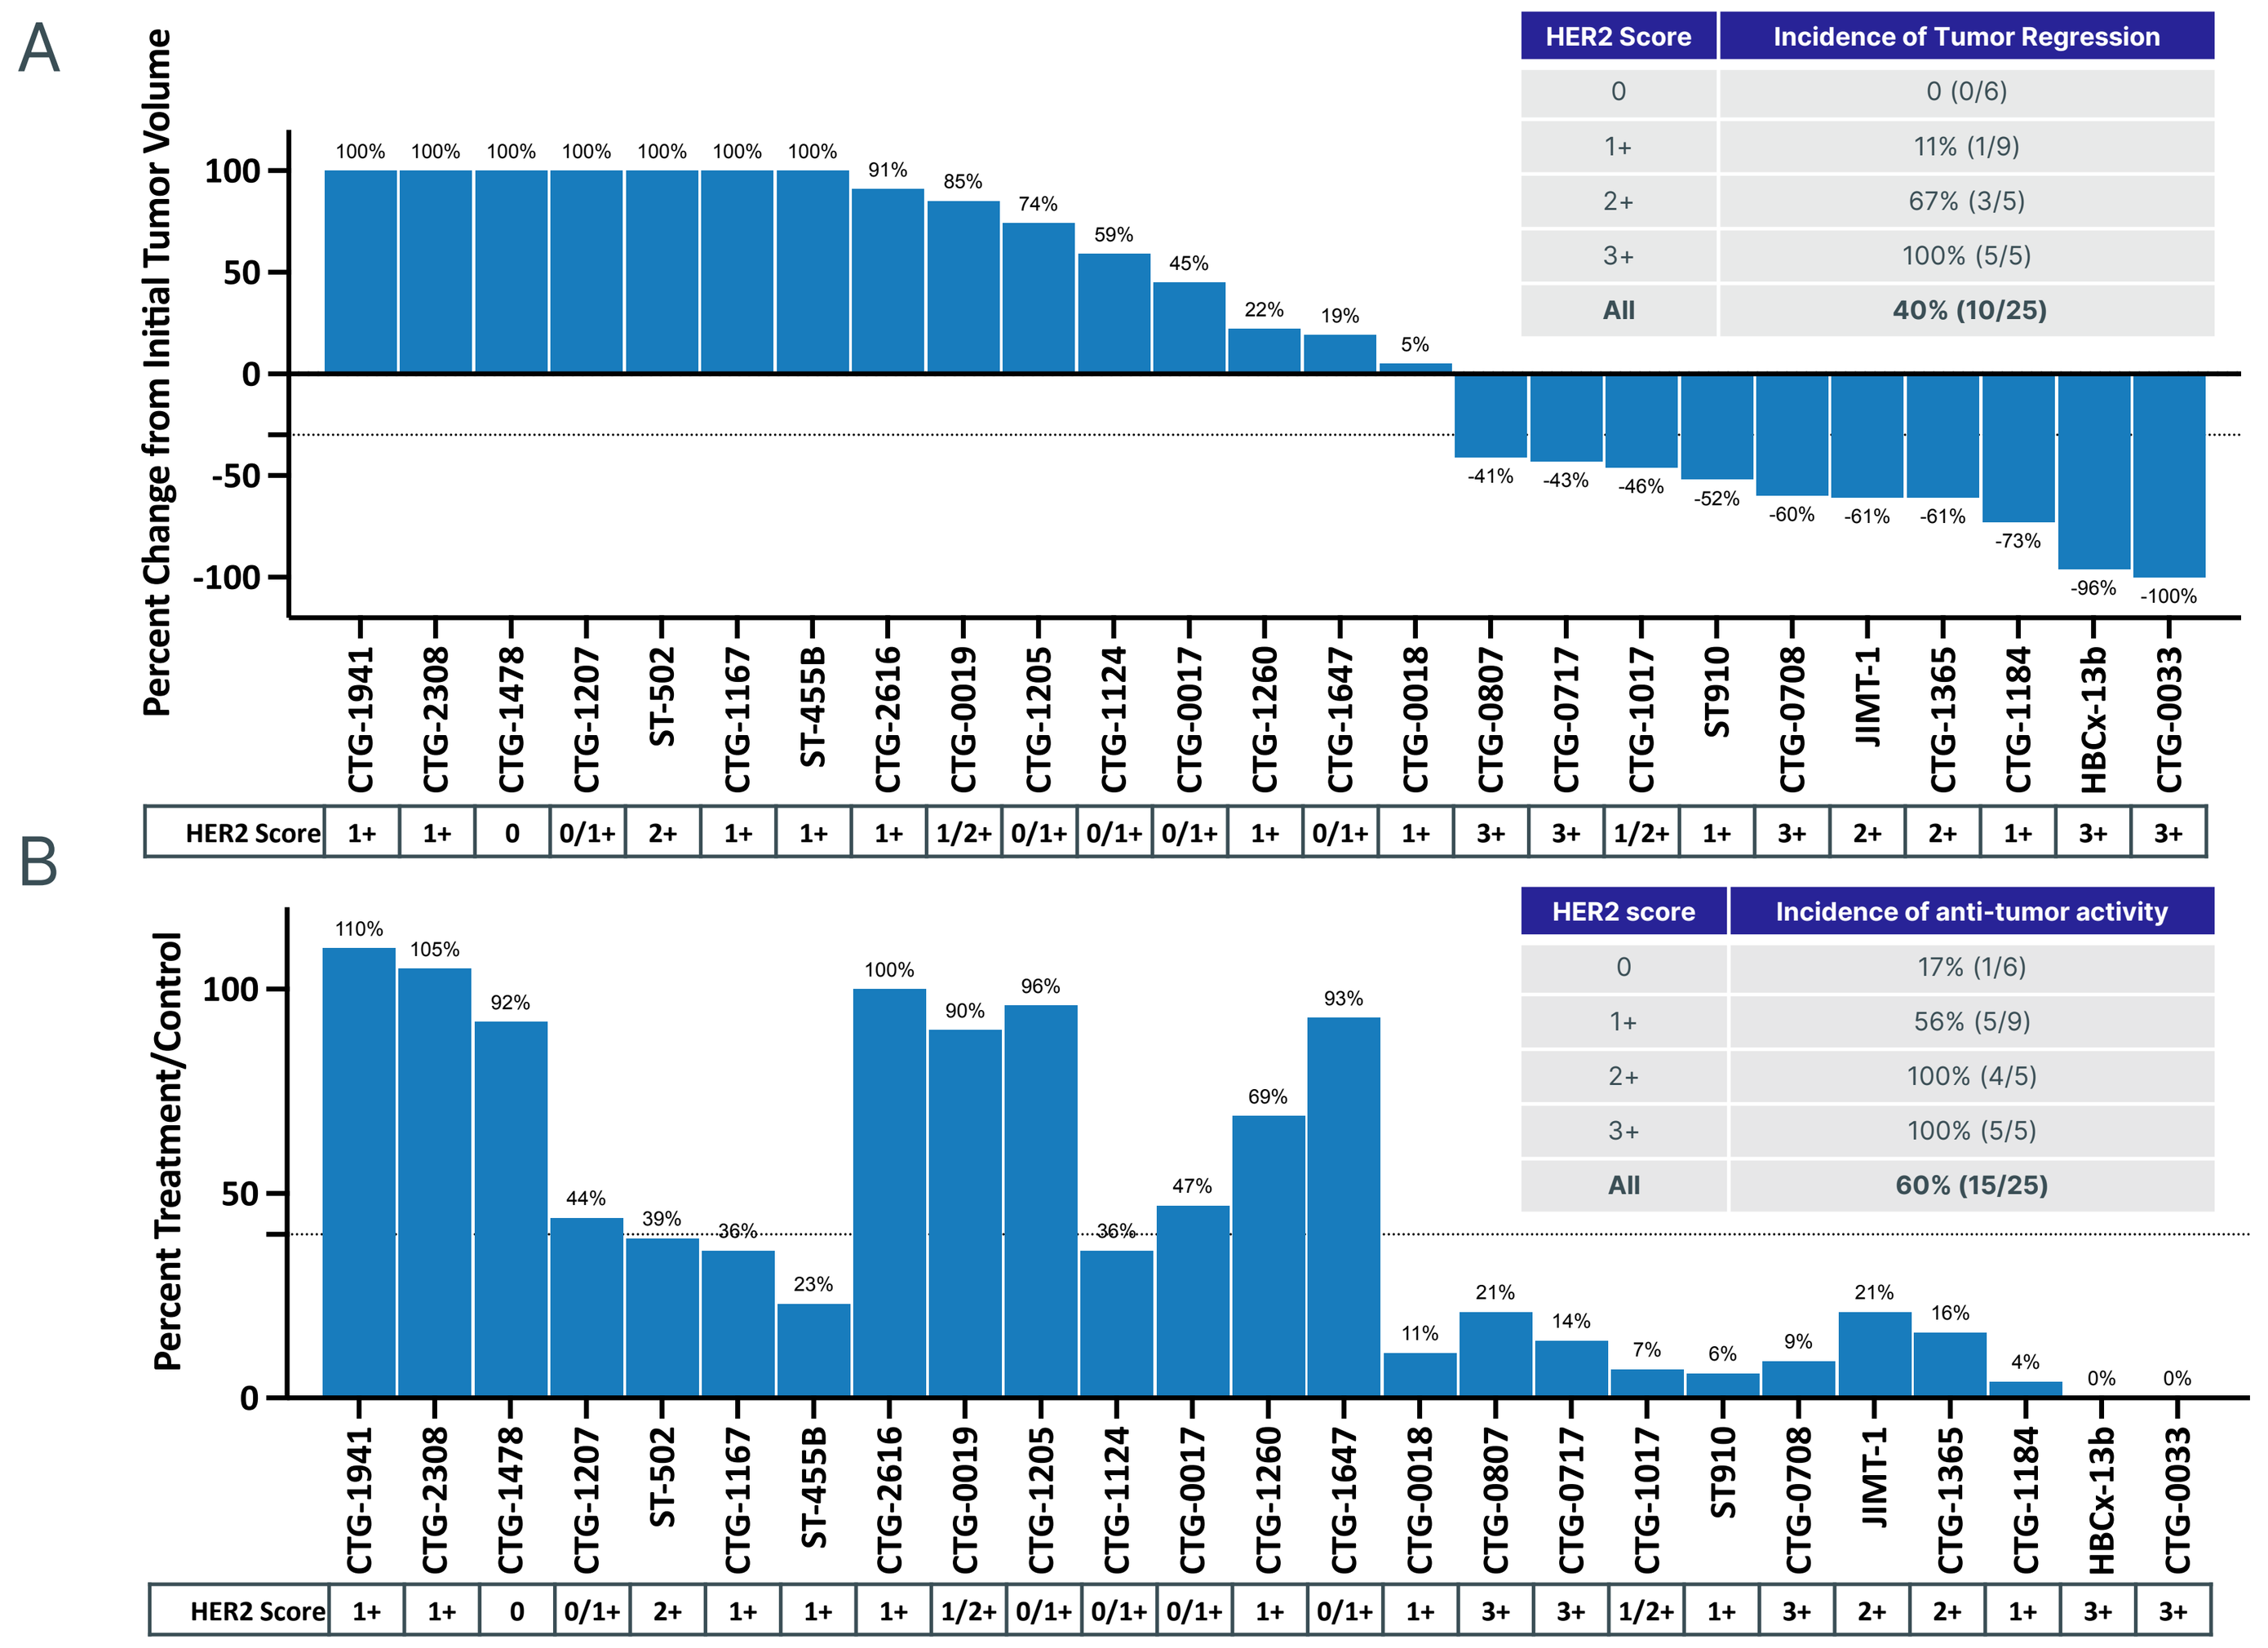
A
Percent Change from Initial Tumor Volume
B
Percent Treatment/Control
HER2 Score

HER2 Score	Incidence of Tumor Regression
0	0 (0/6)
1+	11% (1/9)
2+	67% (3/5)
3+	100% (5/5)
All	**40% (10/25)**

HER2 score	Incidence of anti-tumor activity
0	17% (1/6)
1+	56% (5/9)
2+	100% (4/5)
3+	100% (5/5)
All	**60% (15/25)**

Figure 5. Tumor regression and anti-tumor activity summary for 25 breast cancer xenograft models treated with ≤ 6 mg/kg of ZW49. For CTG models 2308, 1478, 1207, 2616, 0019, 1205, 1124, 0017, 1260, 1647, and 1365 the vehicle cohorts were n=3 and the ZW49 cohorts were n=1. For all other models, n ≧ 6 per cohort. (A) Tumor regression was determined by calculating the percent change in best response from the initial tumor volume. Tumor regression was defined as a percent change from initial tumor volume < 0. (B) Anti-tumor activity was determined by calculating the percent treatment over control at the time of best response. Anti-tumor response was defined as a percent T/C < 40%. HER2 Score was determined by HercepTest™.

ZW49 demonstrates strong activation of multiple ICD hallmarks when compared to T-MMAE and T-DXd

A

ADC	Antibody	Drug-linker (payload)	DAR	Payload class	Mechanism of action
ZW49	zanidatamab (ZW25)	zovodotin (ZD02044)	2	auristatin	microtubule disruption
T-MMAE	trastuzumab	vedotin (MMAE)	4	auristatin	microtubule disruption
T-DXd	trastuzumab	deruxtecan (DXd)	8	camptothecin	topoisomerase-1 inhibition

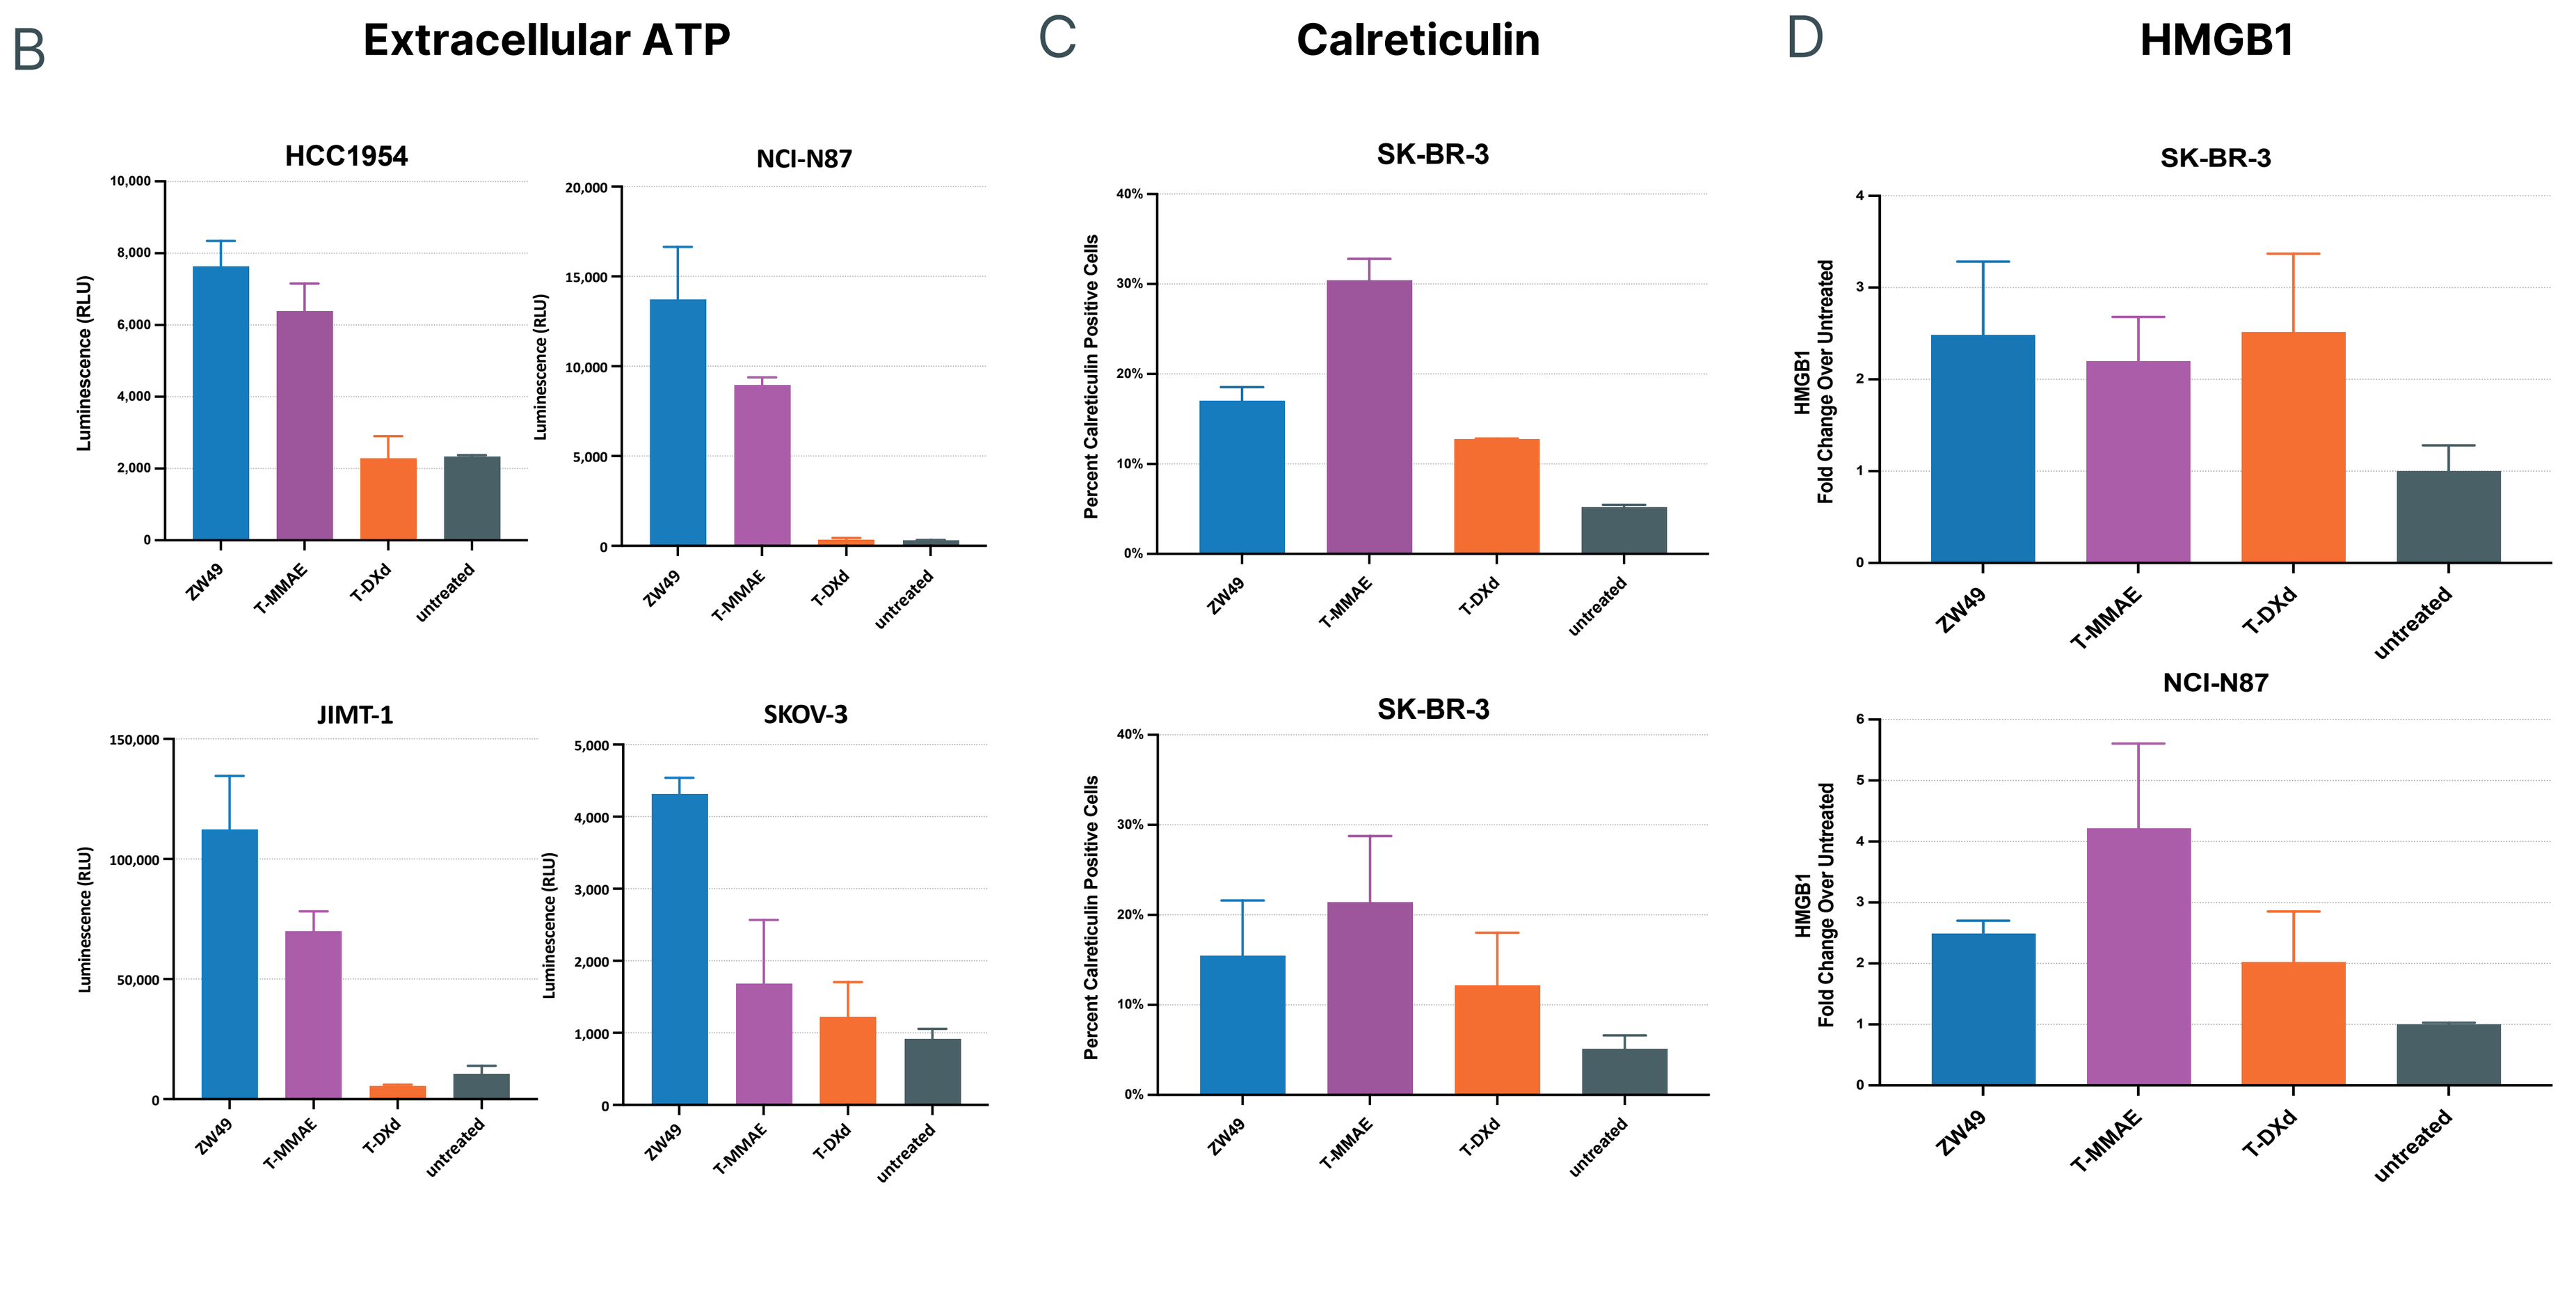
B
Extracellular ATP
C
Calreticulin
D
HMGB1
HCC1954
NCI-N87
JIMT-1
SKOV-3
SK-BR-3
NCI-N87
Luminescence (RLU)
Percent Calreticulin Positive Cells
HMGB1 Fold Change Over Untreated
ZW49
T-MMAE
T-DXd
untreated

Figure 6. (A) Table of ADCs tested. (B) Extracellular ATP was measured by CellTiter-Glo™ after collecting supernatants from tumor cells treated for 24 hours at 20 nM. (C) Cell surface calreticulin was measured by flow cytometry in tumor cells treated for 72 hours (top) or 48 hours (bottom) at 20 nM. (D) Extracellular high mobility group box 1 (HMGB1) was measured by ELISA after collecting supernatants from tumor cells treated for 48 hours at 100 nM.

ZW49 induces hallmarks of immunogenic cell death (ICD) in HER2-expressing tumor cells

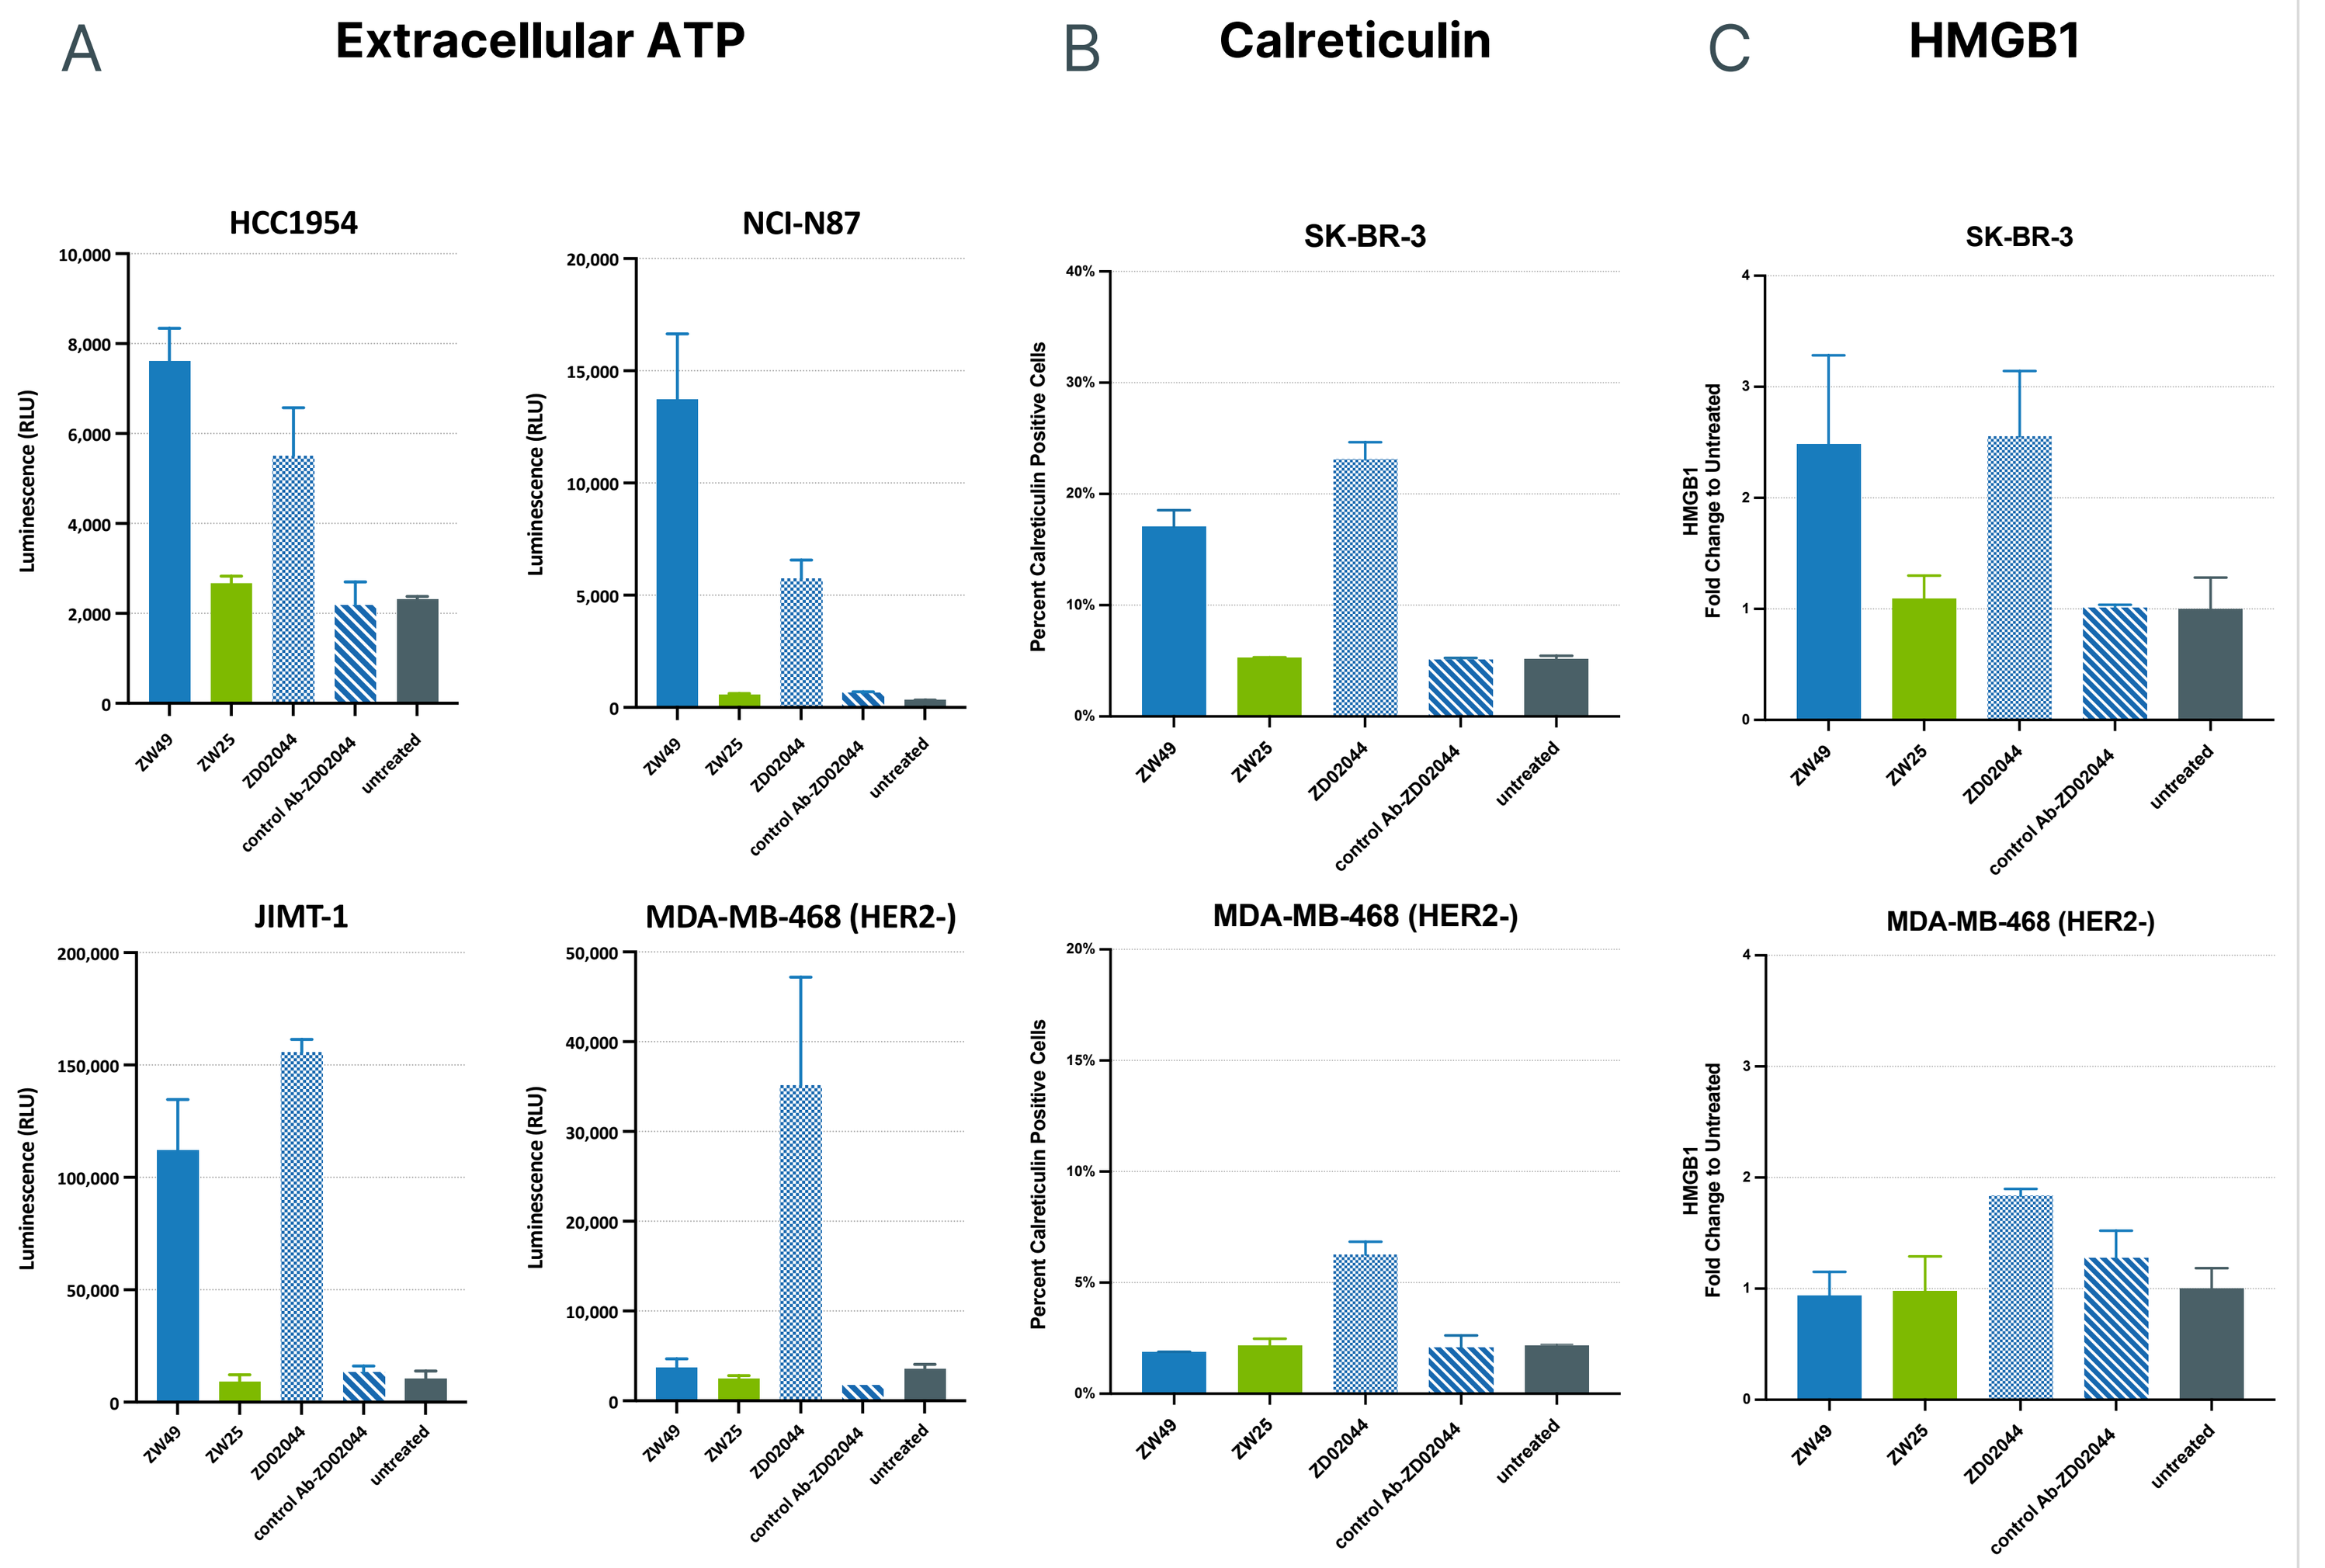
A
Extracellular ATP
B
Calreticulin
C
HMGB1
HCC1954
NCI-N87
SK-BR-3
JIMT-1
MDA-MB-468 (HER2-)
Luminescence (RLU)
Percent Calreticulin Positive Cells
HMGB1 Fold Change to Untreated
ZW49
ZW25
ZD02044
control Ab-ZD02044
untreated

Figure 7. (A) Extracellular ATP was measured by CellTiter-Glo™ after collecting supernatants from tumor cells treated for 24 hours at 20 nM. (B) Cell surface calreticulin was measured by flow cytometry in tumor cells treated for 72 hours. ZW49, ZW25, and control Ab-02044 were tested at 20 nM. ZD02044 was tested at 100 nM. (C) Extracellular high mobility group box 1 (HMGB1) was measured by ELISA after collecting supernatants from tumor cells treated for 48 hours at 100 nM.

ZW49 is active in PDX models of gastric cancer representing a range of HER2 expression

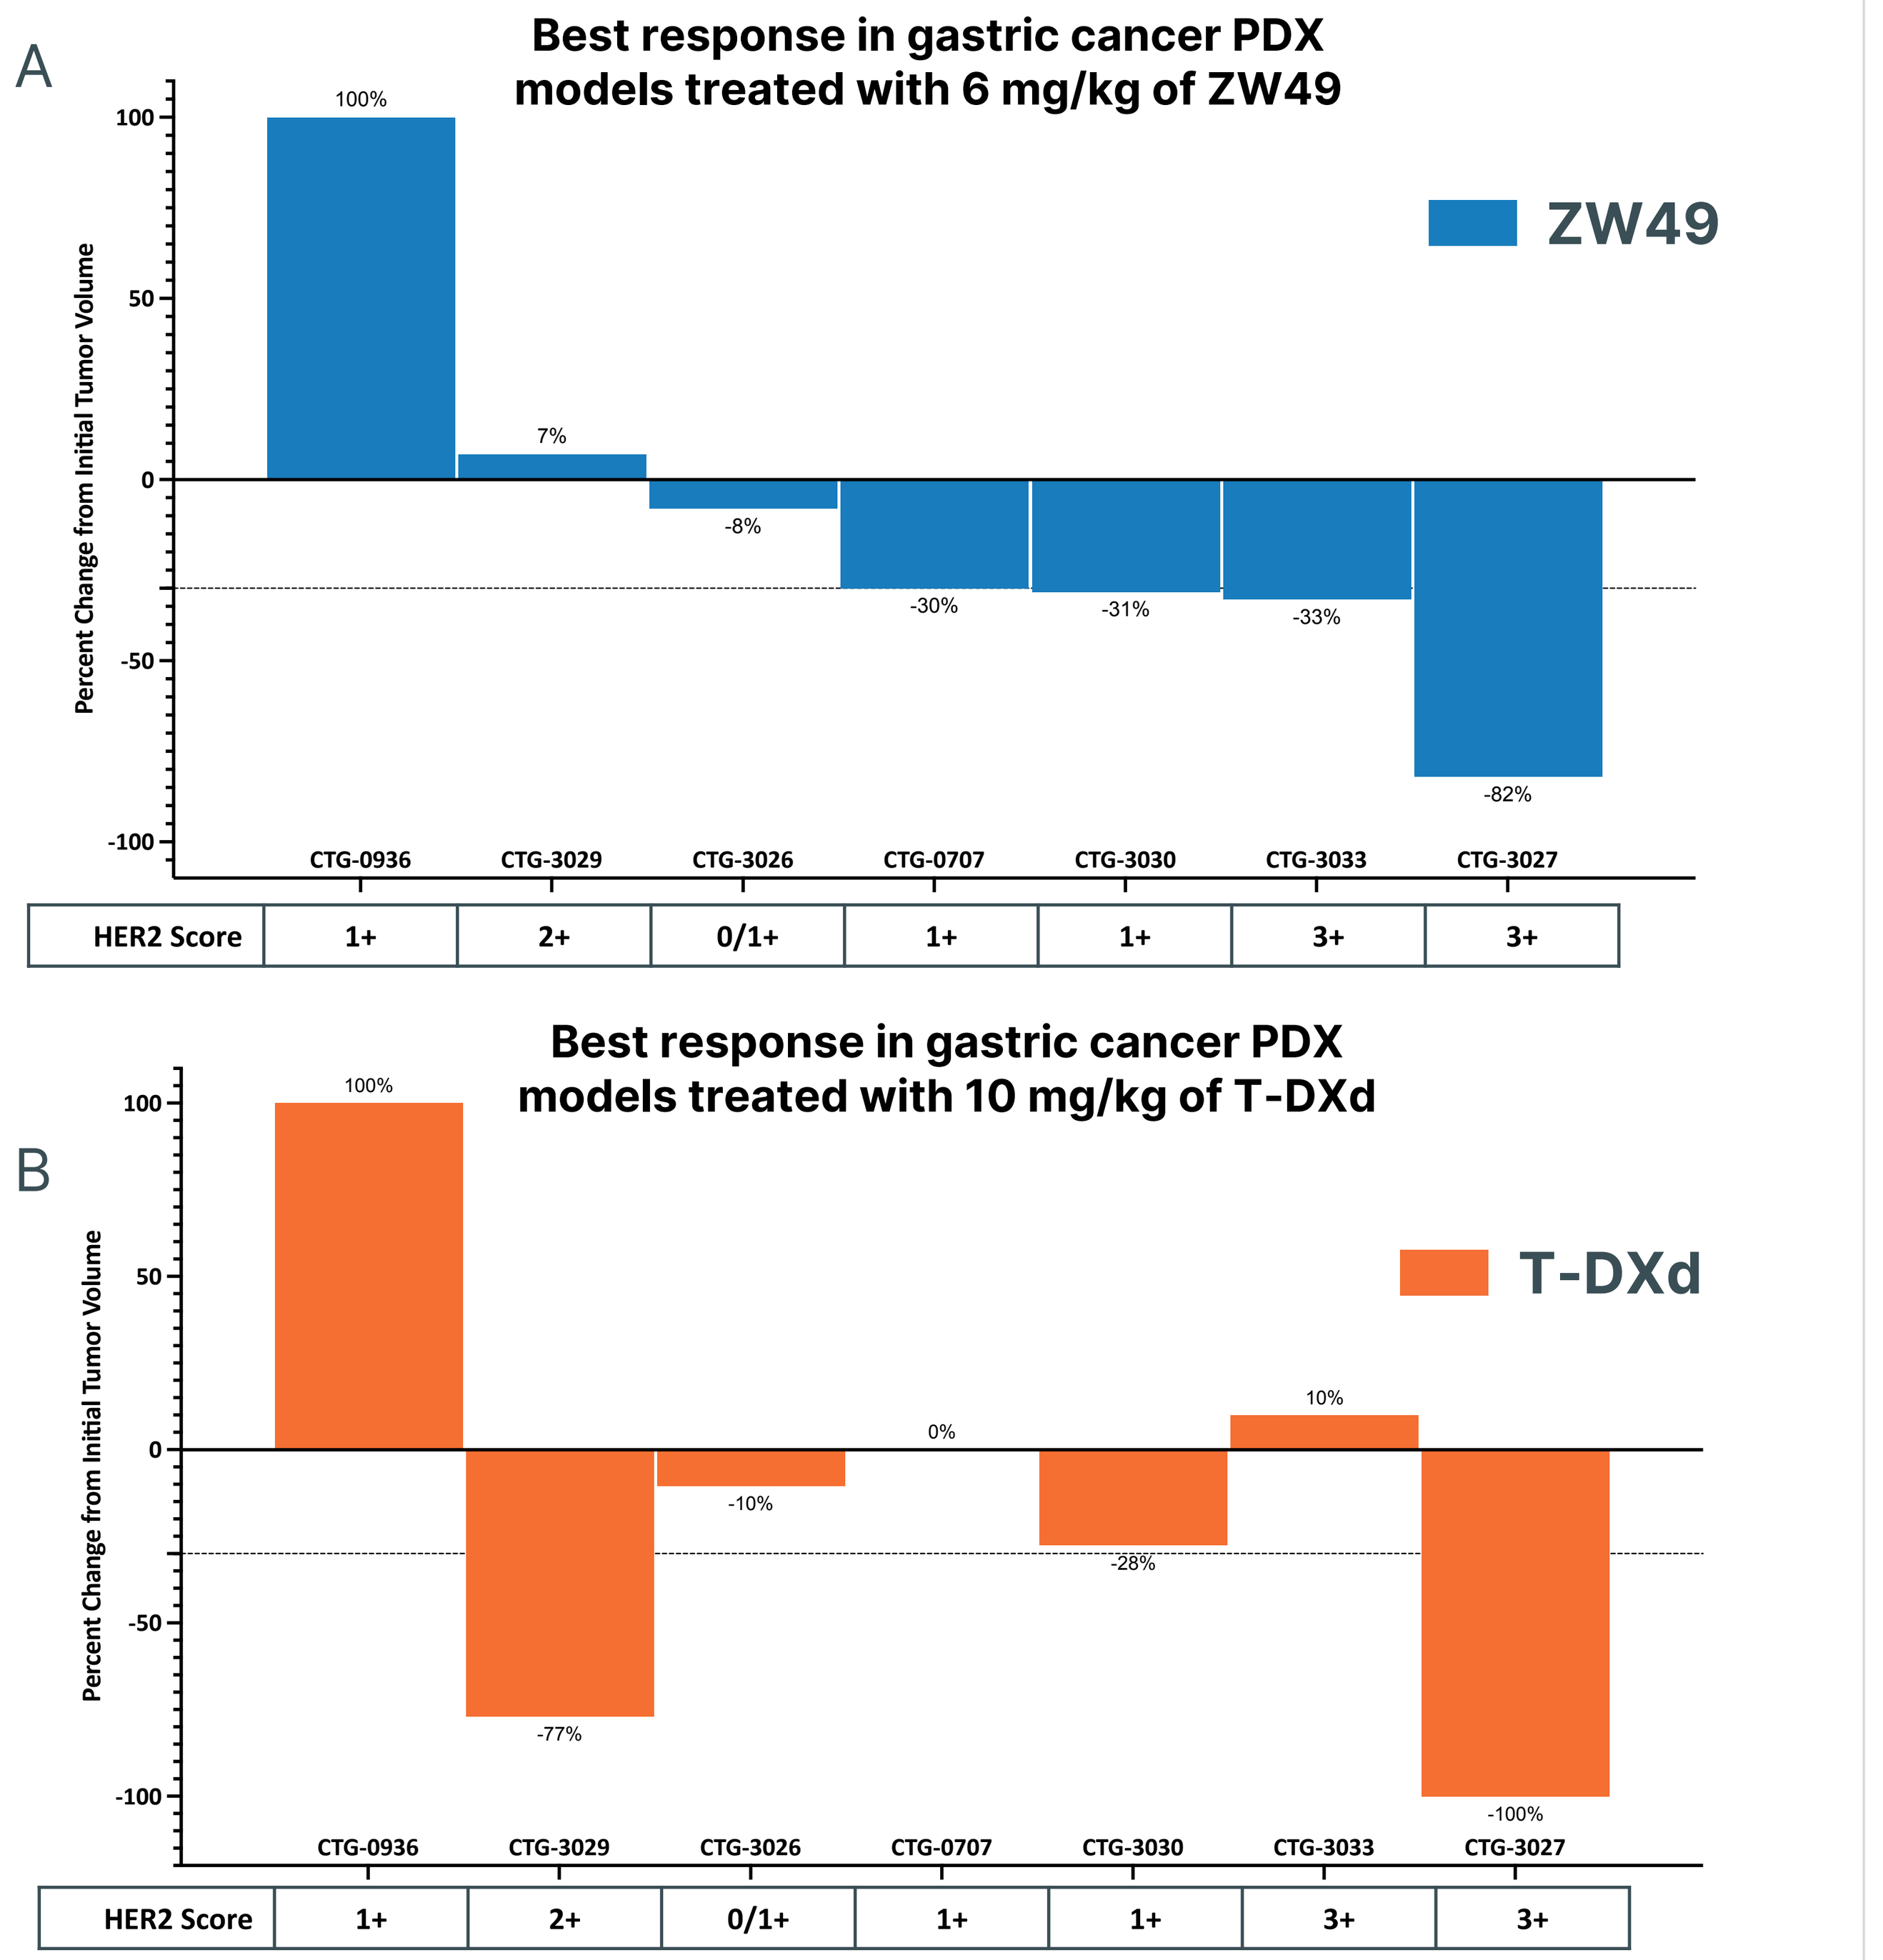
A
Best response in gastric cancer PDX models treated with 6 mg/kg of ZW49
ZW49
Percent Change from Initial Tumor Volume
100%
7%
-8%
-30%
-31%
-33%
-82%
CTG-0936
CTG-3029
CTG-3026
CTG-0707
CTG-3030
CTG-3033
CTG-3027
HER2 Score 1+ 2+ 0/1+ 1+ 1+ 3+ 3+
B
Best response in gastric cancer PDX models treated with 10 mg/kg of T-DXd
T-DXd
100%
-77%
-10%
0%
-28%
10%
-100%
CTG-0936
CTG-3029
CTG-3026
CTG-0707
CTG-3030
CTG-3033
CTG-3027
HER2 Score 1+ 2+ 0/1+ 1+ 1+ 3+ 3+

Figure 8. Tumor regression summary for seven gastric cancer patient derived xenograft models in athymic nude mice treated with either ZW49 or T-DXd. Vehicle group, n=3; treatment groups, n=1. Tumor regression was determined by calculating the percent change in best response from initial tumor volume. HER2 Score was determined by HercepTest™. (A) Tumor regression summary for models treated with a single dose of 6 mg/kg of ZW49. (B) Tumor regression summary for models treated with a single dose of 10 mg/kg of T-DXd.

Conclusions

- Enhanced binding and internalization of ZW49 leads to higher intracellular payload concentrations in HER2-expressing cells.
- ZW49 is a strong inducer of immunogenic cell death hallmarks (extracellular ATP, calreticulin, and HMGB1) when compared to trastuzumab-based ADCs with either DXd or MMAE payloads.
- Anti-tumor activity was observed in breast cancer and gastric cancer PDXs representing a range of HER2 expression.
- The strong anti-tumor activity and the ability to induce ICD markers support ZW49 as a promising ADC for the treatment of HER2-expressing cancers and warrants further investigation and potential combination with checkpoint inhibitors.

Acknowledgements

We would like to thank to Dr. Michael Press and Ivonne Villalobos at USC Medical Center Pathology Lab for HER2 testing.

References

[1]Hamblett, K.J., Hammond, P.W., Barnscher, S.D., et al. ZW49, a HER2-targeted biparatopic antibody-drug conjugate for the treatment of HER2-expressing cancers [abstract]. In: Proceedings of the American Association for Cancer Research Annual Meeting 2018; 2018 Apr 14-18; Chicago, IL. Philadelphia (PA): AACR; Cancer Res 2018;78(13 Suppl):Abstract nr 3914.
[2]Weisser, N.E., Sanches, M., Escobar-Cabrera, E., et al. An anti-HER2 biparatopic antibody that induces unique HER2 clustering and complement-dependent cytotoxicity. Nat Commun 14, 1394 (2023).