PROTECT™, A Novel Trispecific Antibody Masking Platform With Integrated Immune Modulation Displays Unique Activity and Differentiated Modes of Action

AACR Annual Meeting 2023
Abstract #2926

Anna von Rossum, Genevieve Desjardins, Nichole Escalante, Wingkie Wong, Bryant Harbourne, Janessa Li, Begonia Silva Moreno, Prajwal Raghunatha, Richard Kunze, Madeline Fung, Florian Heinkel, Harsh Pratap, Kevin Haworth, Eric Escobar-Cabrera, Brandon Clavette, Surjit Dixit, Nina Weisser and Thomas Spreter von Kreudenstein

Author Affiliations: Zymeworks Inc., Vancouver, BC, Canada

Introduction

Many T-cell engagers (TCE) have limited efficacy in the clinic due to toxicity and narrow therapeutic index.

PROTECT™ is a platform designed to tackle these challenges by combining a masking domain that, when cleaved, provides additional immunomodulatory properties.

Here we demonstrate platform proof-of-concept and data for a broadly expressed therapeutic cancer target utilizing the PROTECT™ platform.

Engineering Conditionally Active TCEs with Safer Therapeutic Index

- The PD-1 domain is engineered for improved PD-L1 binding, thus enhancing masking of the anti-CD3 paratope by the PD1:PD-L1 heterodimer.
- A protease cleavage site links the PD-L1 domain and the CD3 Fab light chain, allowing for its release in the protease rich tumor microenvironment (TME)[1].
- The remaining PD-1 domain provides steric hindrance and results in a TCE with reduced affinity for CD3.
- The PROTECT™ format drives activity and is compatible with different low affinity anti-CD3 paratopes.
- Specificity of the TCE is readily modified by changing the anti-TAA scFv.
- Azymetric™ and EFECT™ platforms were used to generate multispecific antibodies with reduced effector function.

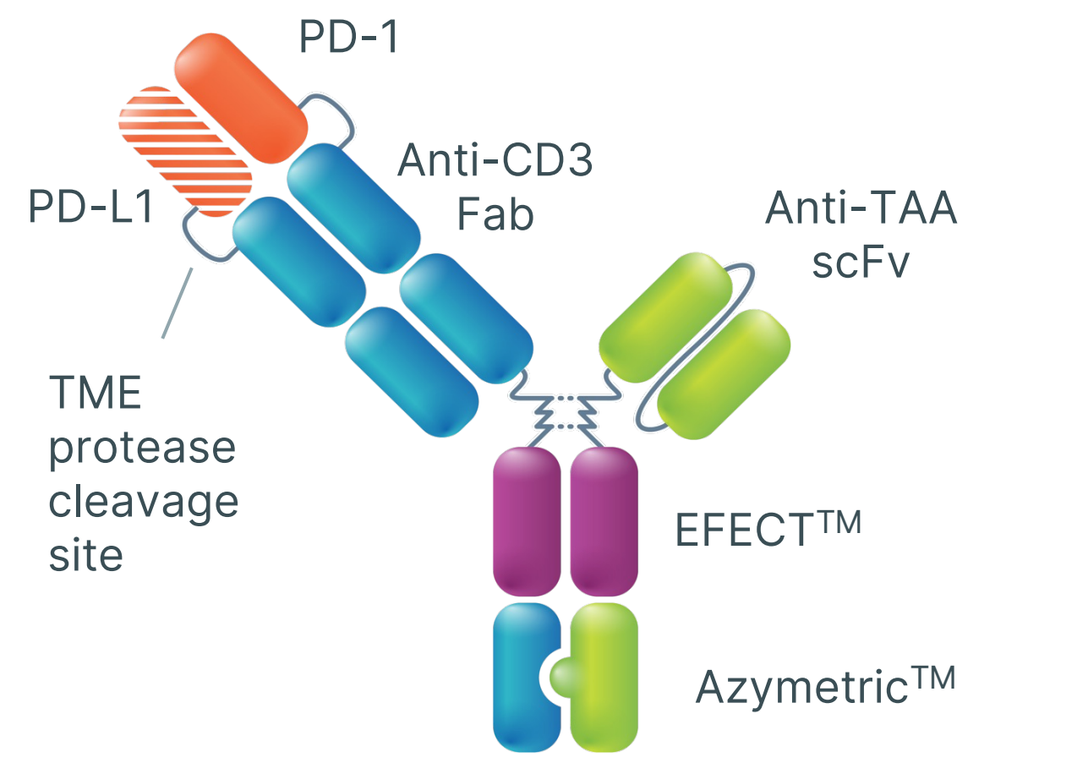
PD-1
PD-L1
Anti-CD3 Fab
Anti-TAA scFv
TME protease cleavage site
EFECT™
Azymetric™

Figure 1. PROTECT™ is a masking and immunomodulatory platform that is conditionally-active. Anti-TAA arm in scFv format is shown in green. Anti-CD3 arm is shown in blue, while the PD1:PD-L1 masking domains are in orange. The purple region represent the CH2 domain with mutations to reduce effector function.

Proposed Mechanism of Action for PROTECT™

- Anti-TAA paratope facilitates PROTECT™ therapeutics binding within the TAA+ TME.
- TME proteases cleave at the cleavage site and release the PD-L1 mask moiety.
- Activated PROTECT™ binds avidly to tumor via PD-L1 (and TAA) engagement and activates T cells via CD3.
- PD1 domain provides checkpoint inhibition (CPI) activity.

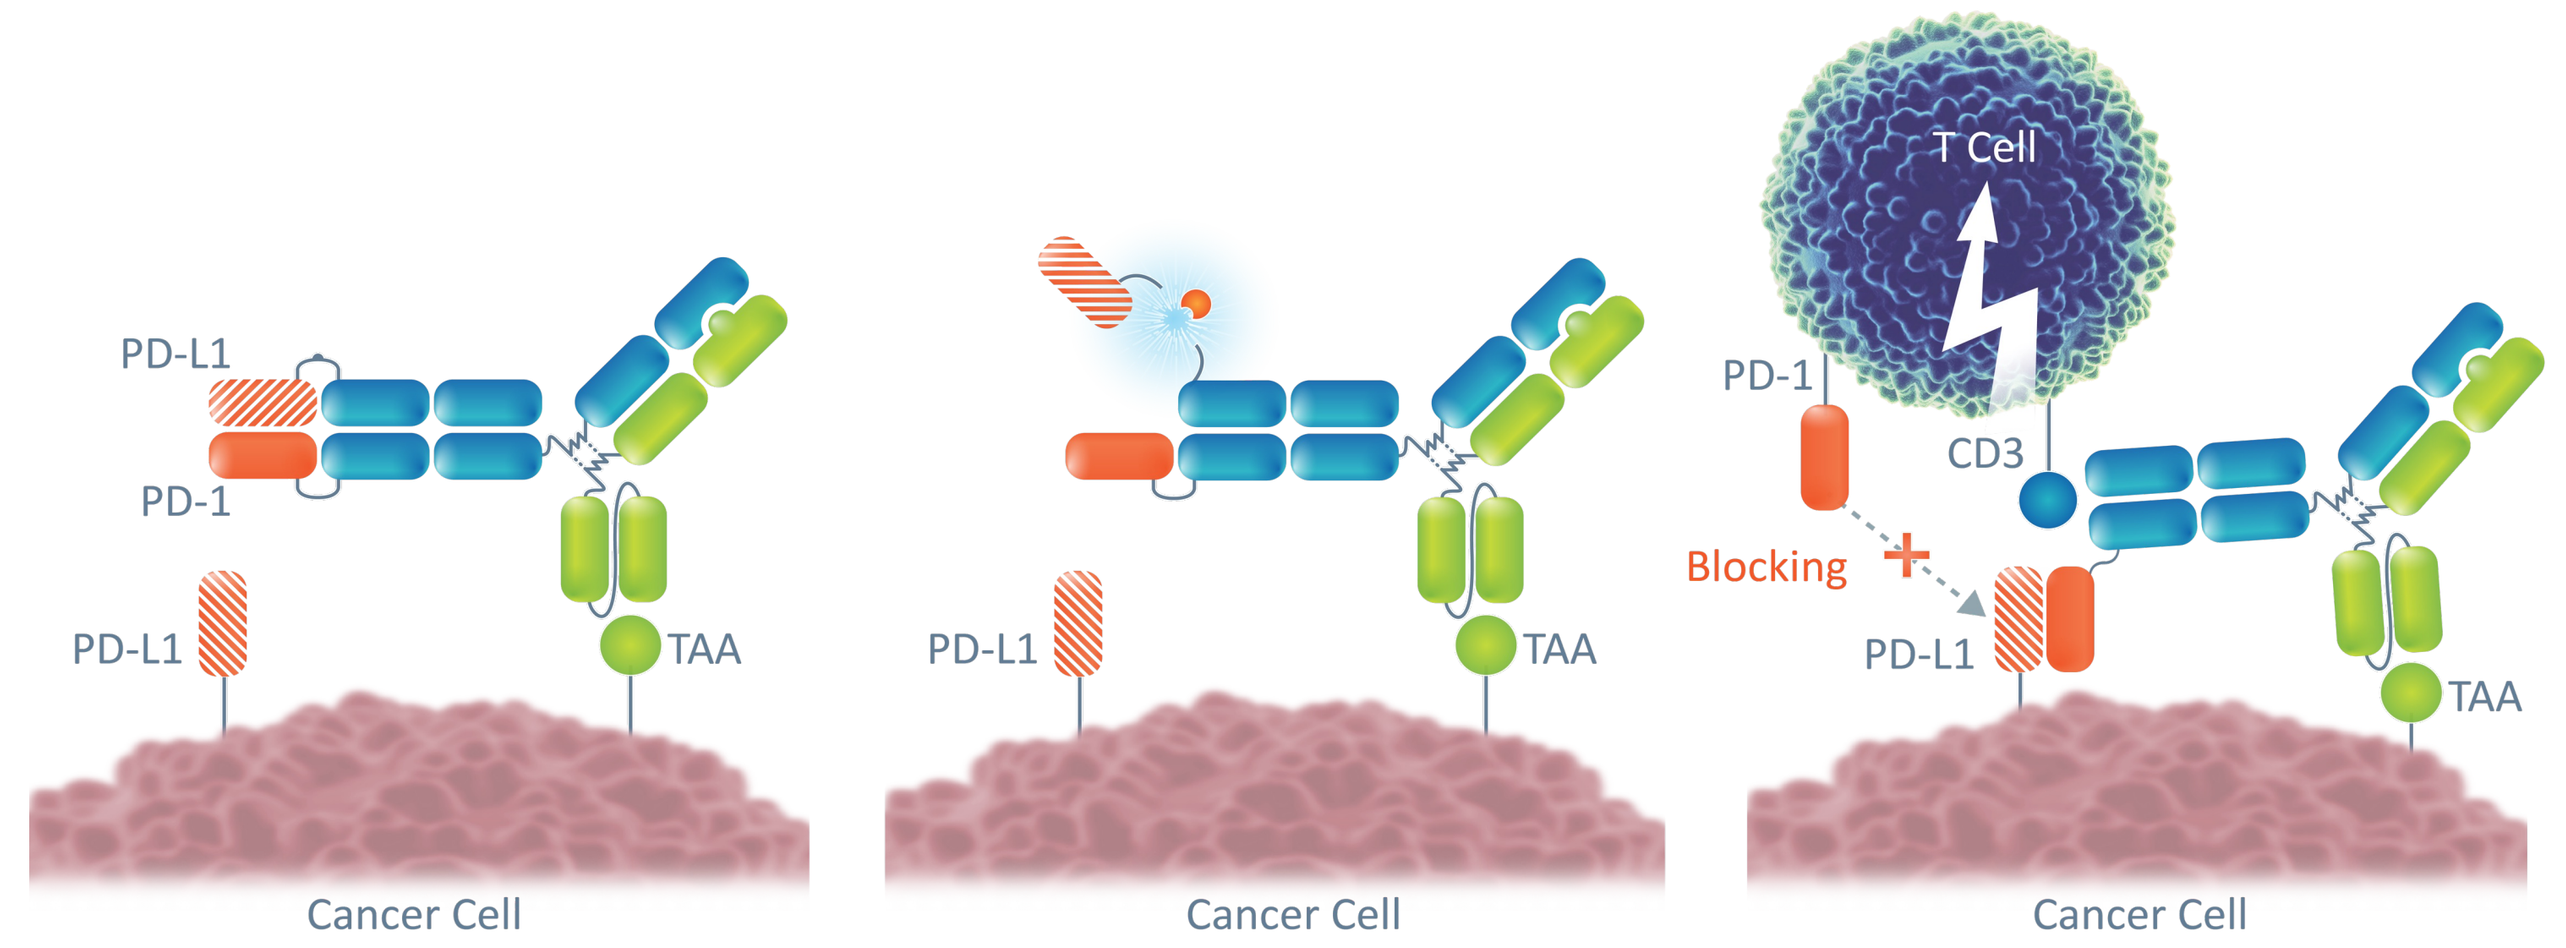
PD-L1
PD-1
PD-L1
TAA
Cancer Cell
PD-L1
TAA
Cancer Cell
T Cell
PD-1
CD3
Blocking
PD-L1
TAA
Cancer Cell

PROTECT™ Platform: Proof of Concept Studies Demonstrate Enhanced Functionality and Anti-Tumor Activity

Masked HER2xCD3 PROTECT™ mediates reduced *in vitro* cytotoxicity while cleaved HER2xCD3 PROTECT™ recovers cytotoxicity and mediates checkpoint inhibition

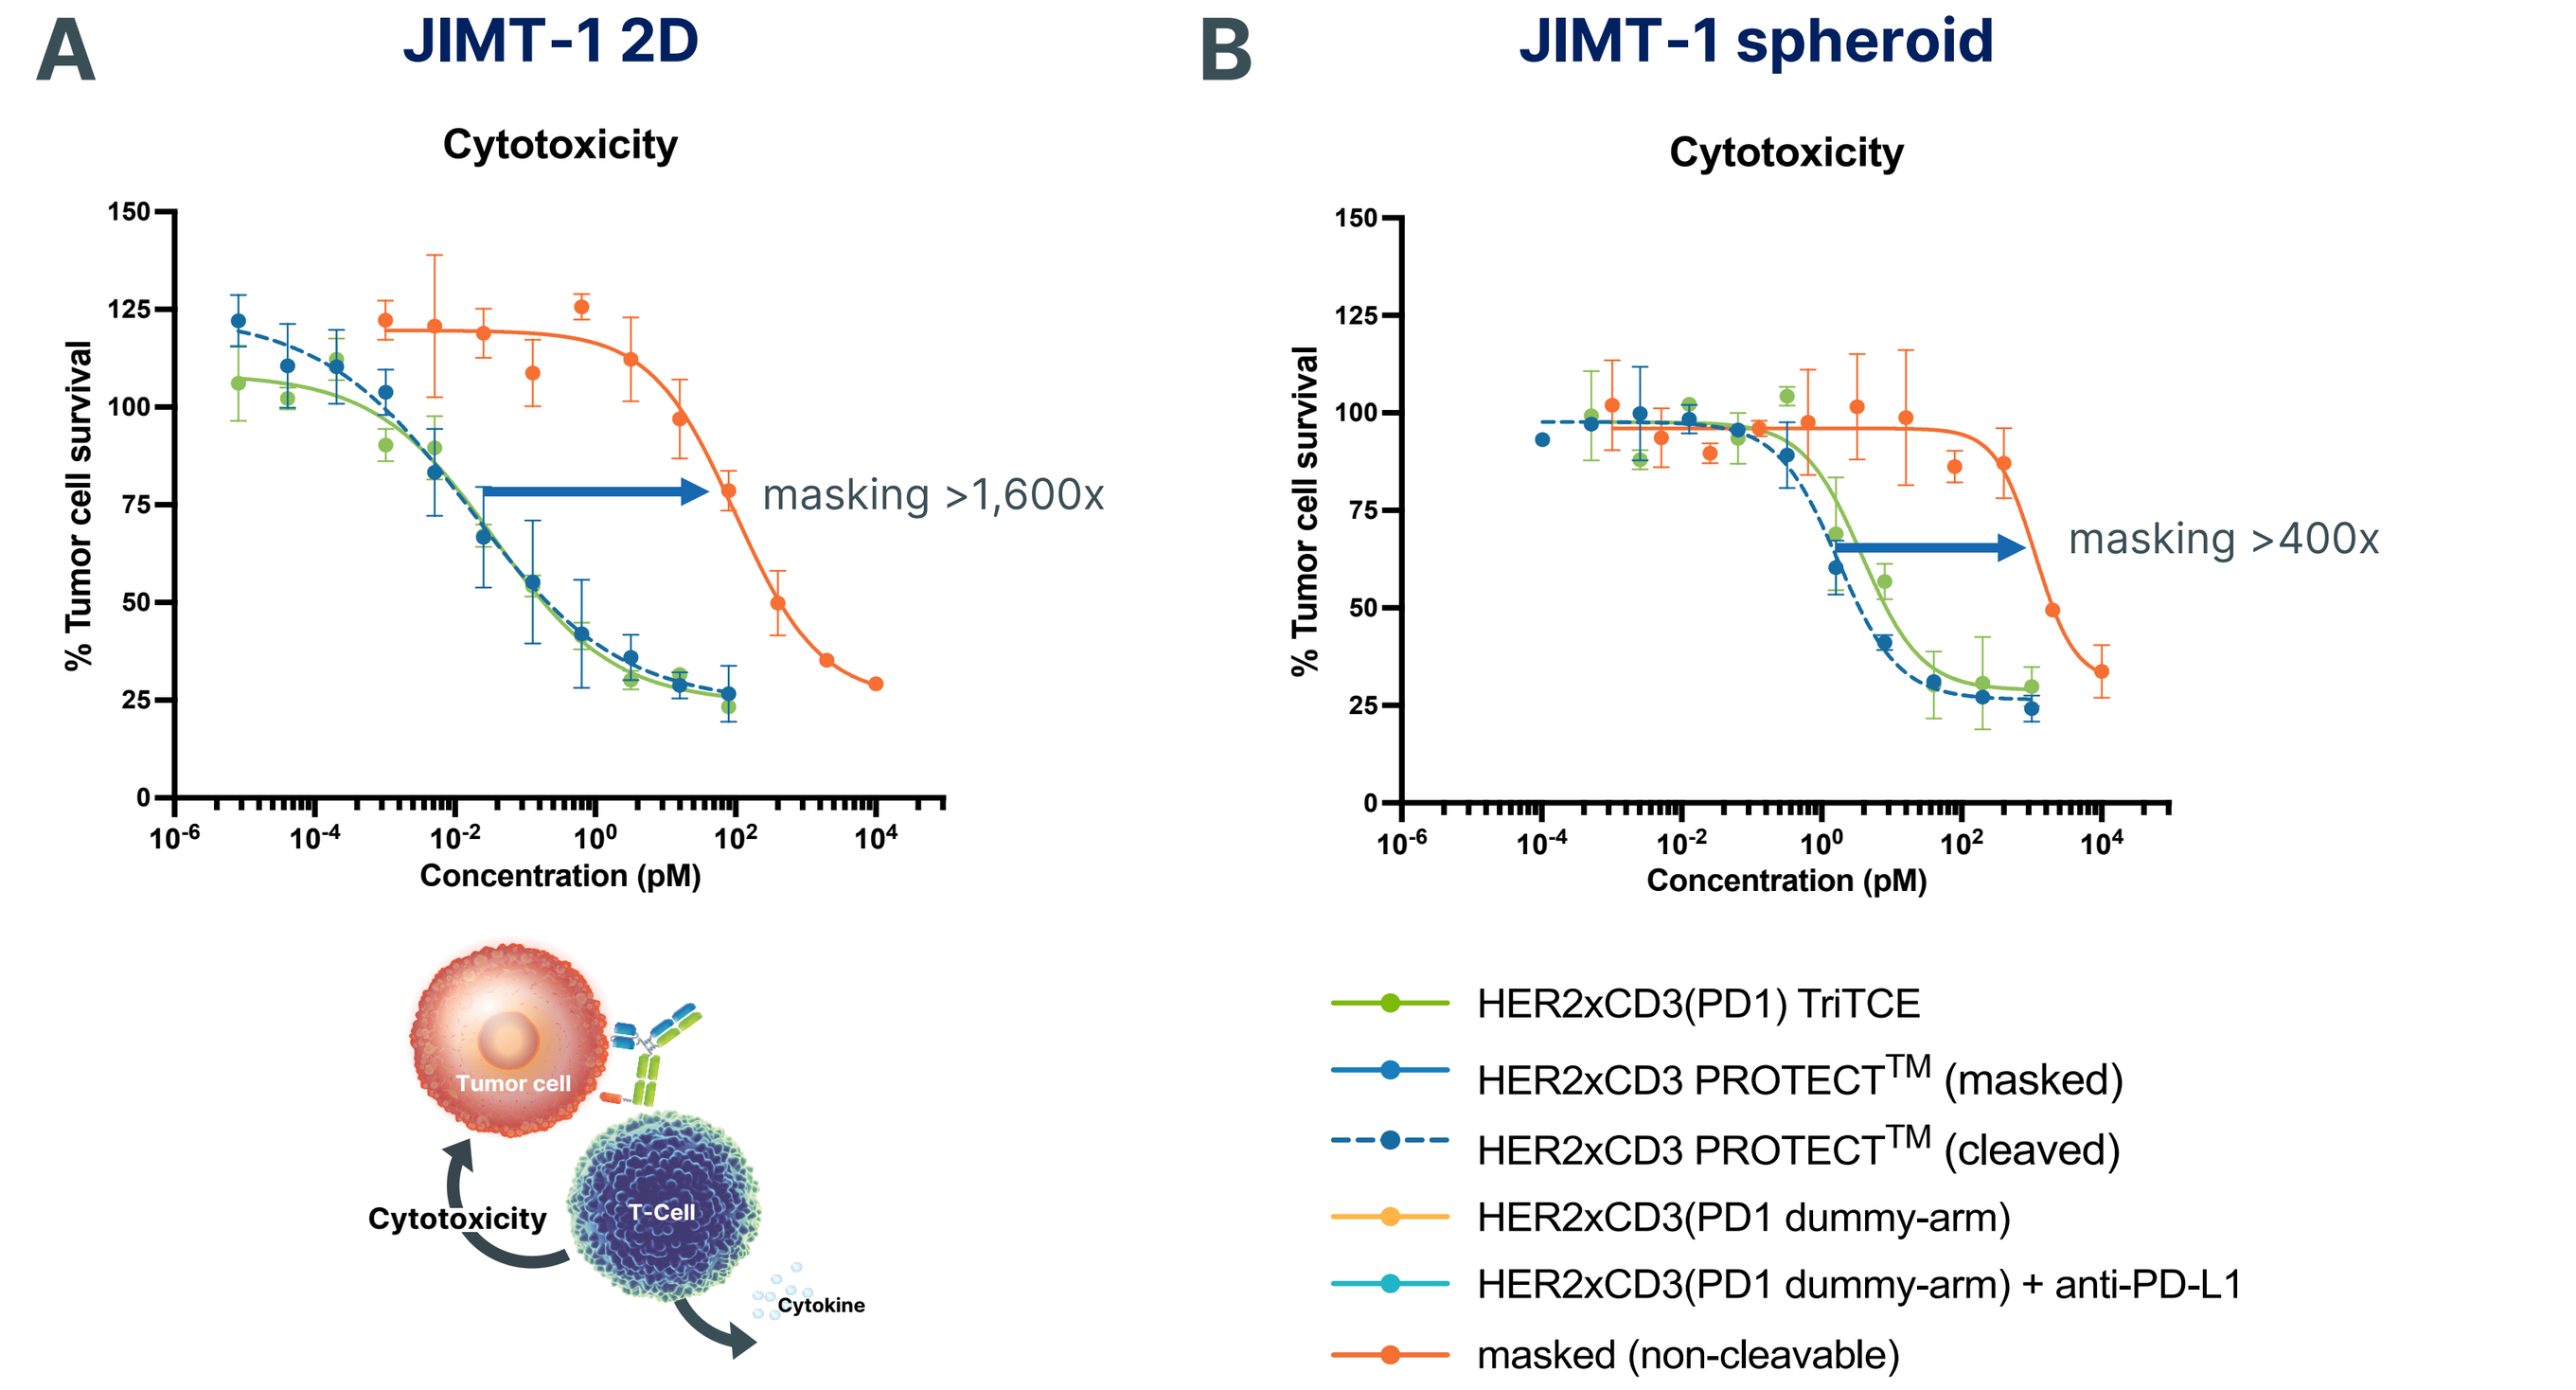
A
JIMT-1 2D
Cytotoxicity
% Tumor cell survival
Concentration (pM)
masking >1,600x
Tumor cell
T-Cell
Cytotoxicity
Cytokine
B
JIMT-1 spheroid
Cytotoxicity
masking >400x
HER2xCD3(PD1) TriTCE
HER2xCD3 PROTECT™ (masked)
HER2xCD3 PROTECT™ (cleaved)
HER2xCD3(PD1 dummy-arm)
HER2xCD3(PD1 dummy-arm) + anti-PD-L1
masked (non-cleavable)

Cleaved HER2xCD3 PROTECT™ displays checkpoint inhibition on PD-L1-expressing tumor cells

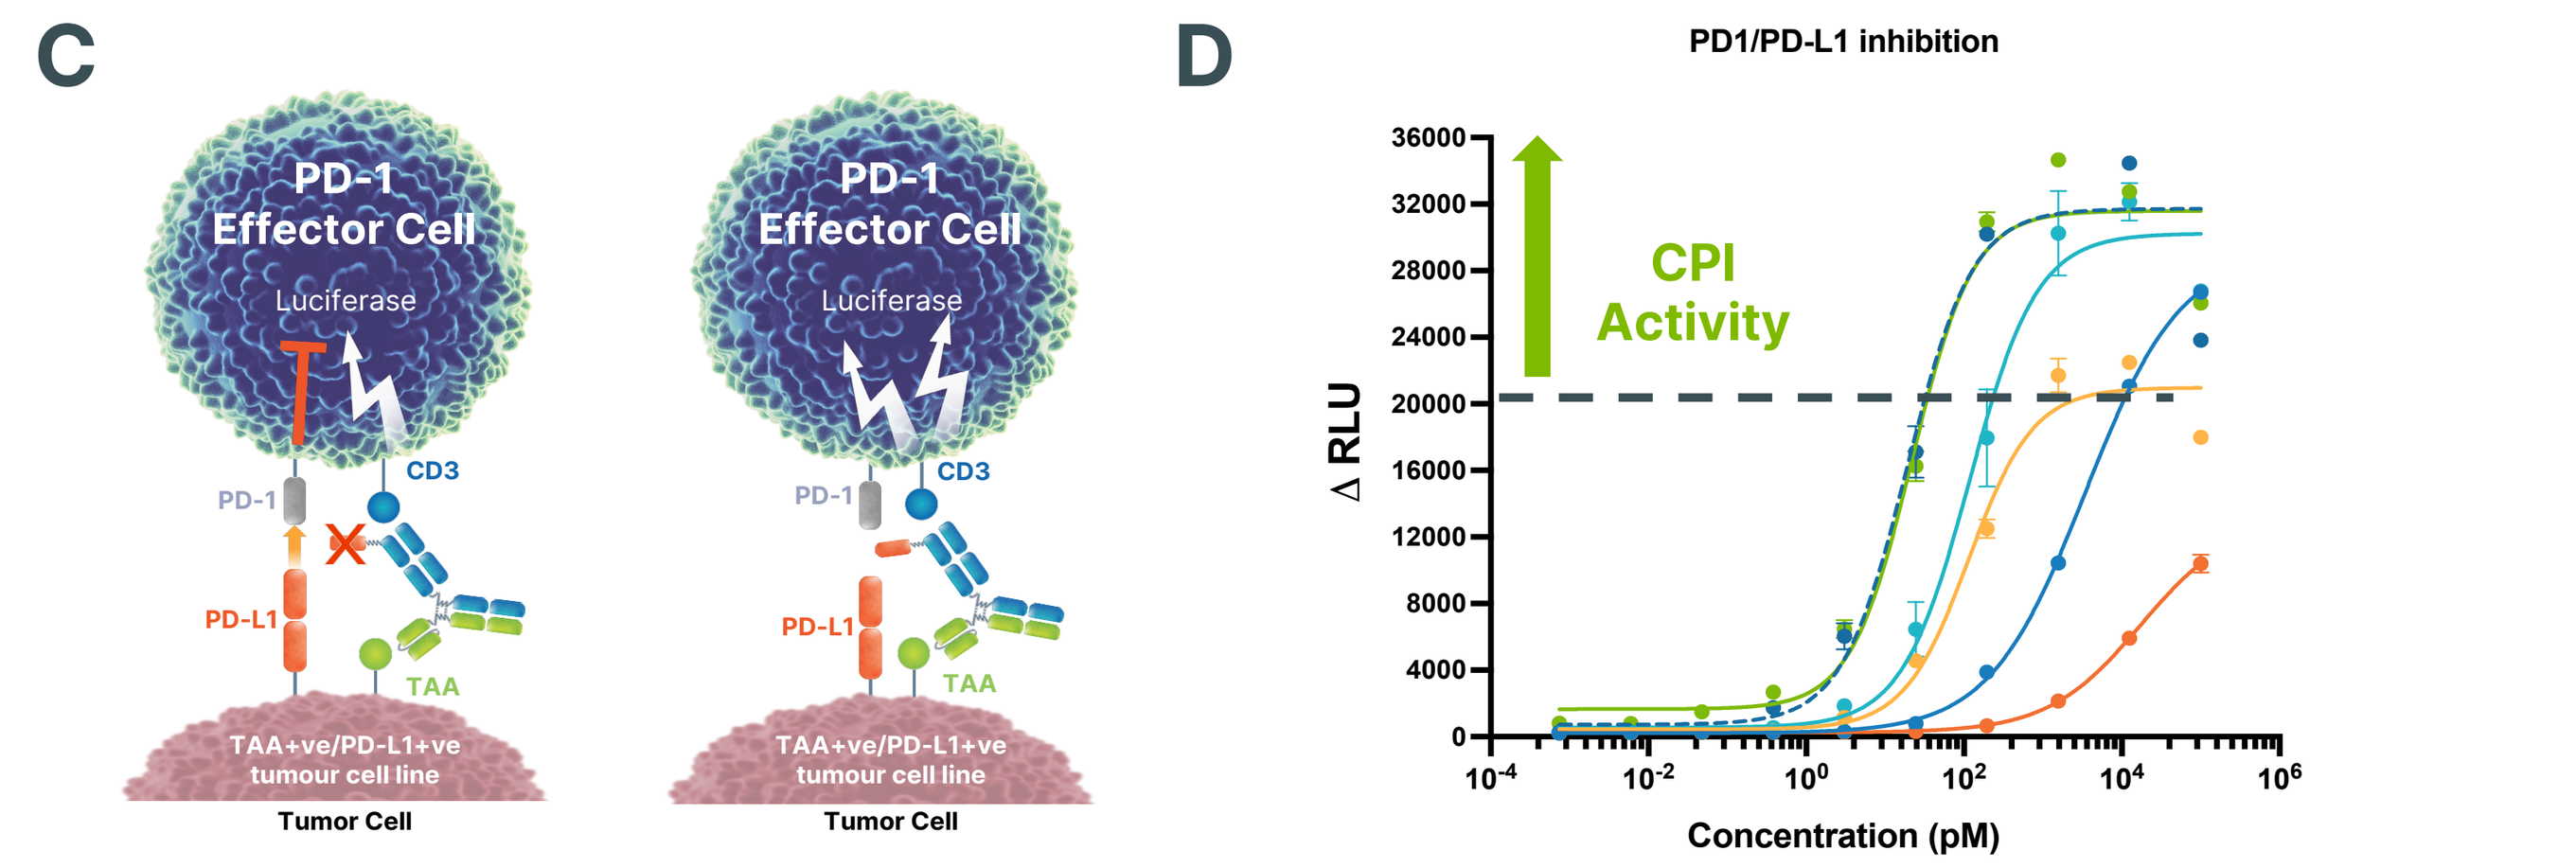
C
PD-1 Effector Cell
Luciferase
PD-1
CD3
PD-L1
TAA
TAA+ve/PD-L1+ve tumour cell line
Tumor Cell
D
PD1/PD-L1 inhibition
CPI Activity
Δ RLU
Concentration (pM)

PROTECT™ induces complete and durable individual tumor responses *in vivo* with an improved therapeutic index

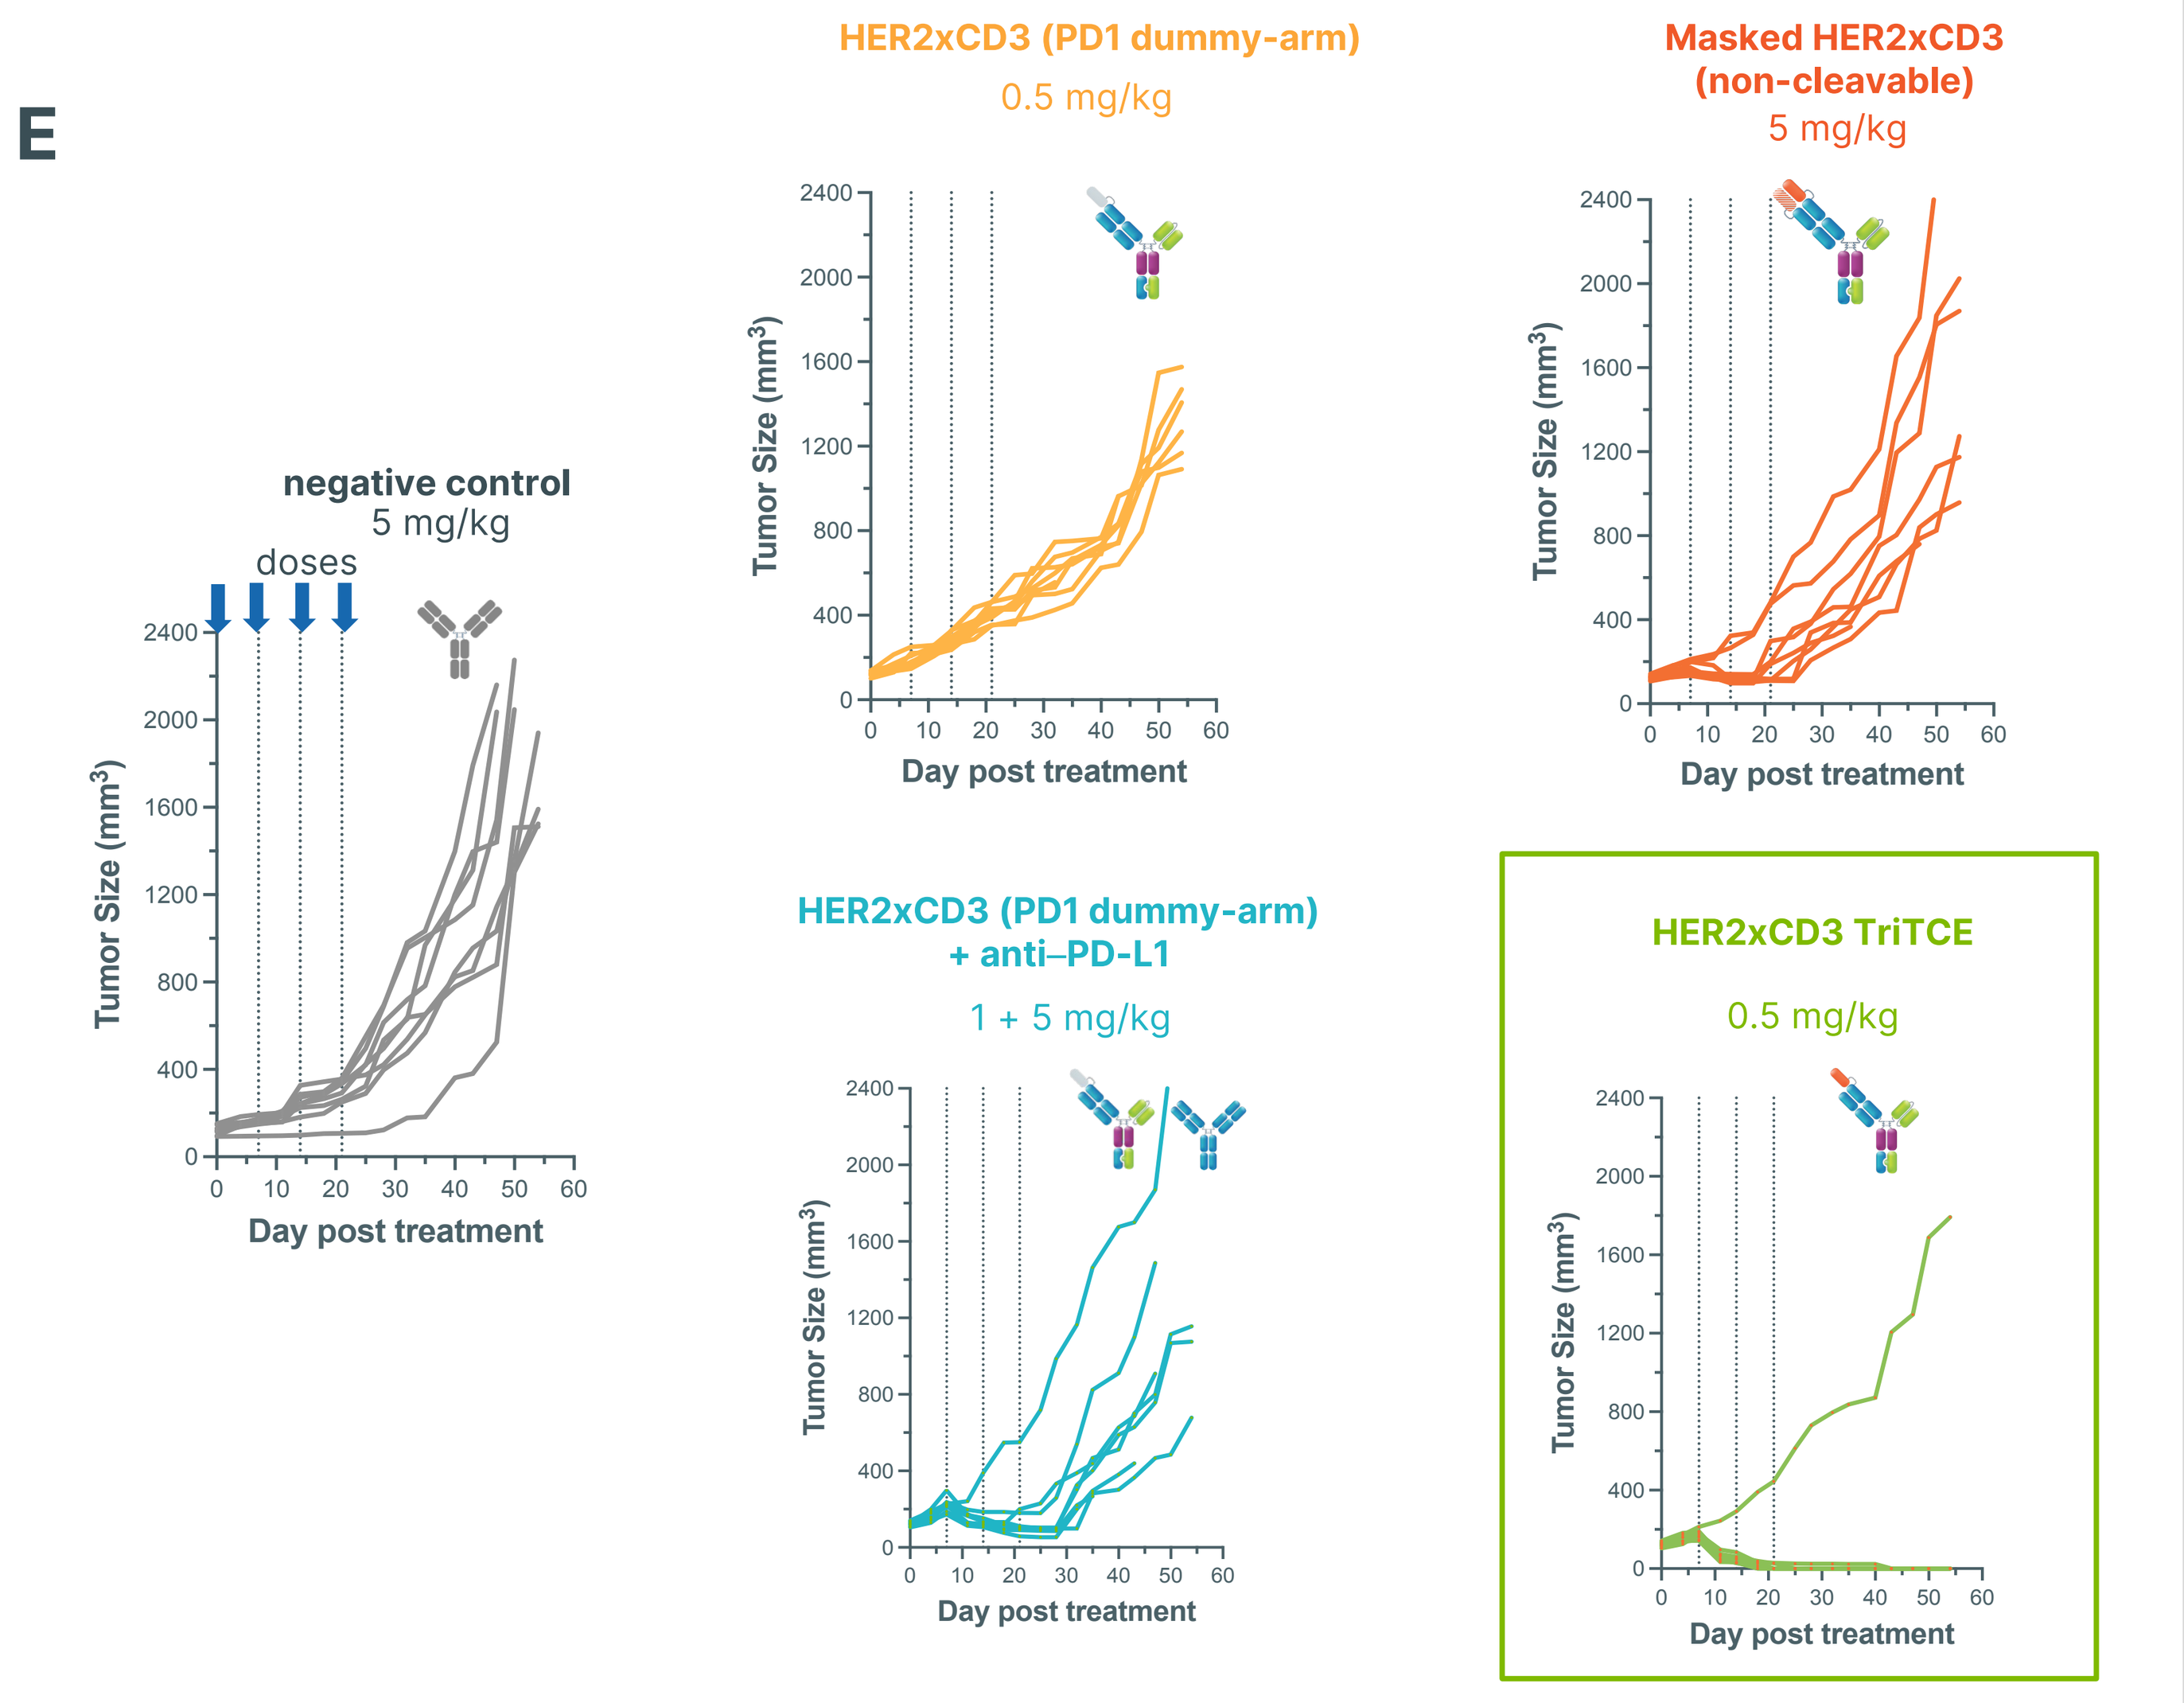
E
negative control
5 mg/kg
doses
HER2xCD3 (PD1 dummy-arm)
0.5 mg/kg
Masked HER2xCD3 (non-cleavable)
5 mg/kg
HER2xCD3 (PD1 dummy-arm) + anti–PD-L1
1 + 5 mg/kg
HER2xCD3 TriTCE
0.5 mg/kg
Tumor Size (mm³)
Day post treatment

Figure 2. HER2xCD3 PROTECT™ mediates increased anti-tumor cytotoxicity, induces CPI upon mask release *in vitro* and results in complete and durable tumor responses *in vivo*. T-cell mediated cytotoxicity was evaluated in standard T cell cytotoxicity assay using PBMCs effectors at 10:1 ratio with 2D JIMT-1 tumor cells **(A)** or 3D JIMT-1 spheroids **(B)** as targets. Masking window is calculated compared to non-cleavable mask control. Checkpoint inhibition was evaluated using a PD-1/PD-L1 reporter-gene assay (RGA) **(C)** with JIMT-1 cells as targets and PD-1-expressing Jurkat cells as effectors **(D)**. Unmasked PROTECT™ provides checkpoint inhibition at lower concentrations than the bispecific combo with anti-PD-L1. The non-cleavable and masked PROTECT™ do not provide significant checkpoint inhibition. **(E)** NOG mice were inoculated with JIMT-1 tumor cells that were grown to 100-120 mm³ before they were randomized and engrafted with PBMCs. Mice were dosed weekly for 4 weeks, except for combo where anti-PD-L1 was dosed biweekly. Tumor growth and body weight were monitored twice weekly for 60 days. The TriTCE provides complete and durable response compared to the bispecific HER2xCD3(PD1 dummy), the bispecific HER2xCD3(PD1 dummy) + anti-PD-L1 combo and the masked non-cleavable variants.

PROTECT™ Platform: Plug and Play Feature Enables Application to Additional Tumor Targets

Masked TAAxCD3 PROTECT™ mediates reduced *in vitro* cytotoxicity while cleaved TAAxCD3 PROTECT™ recovers cytotoxicity

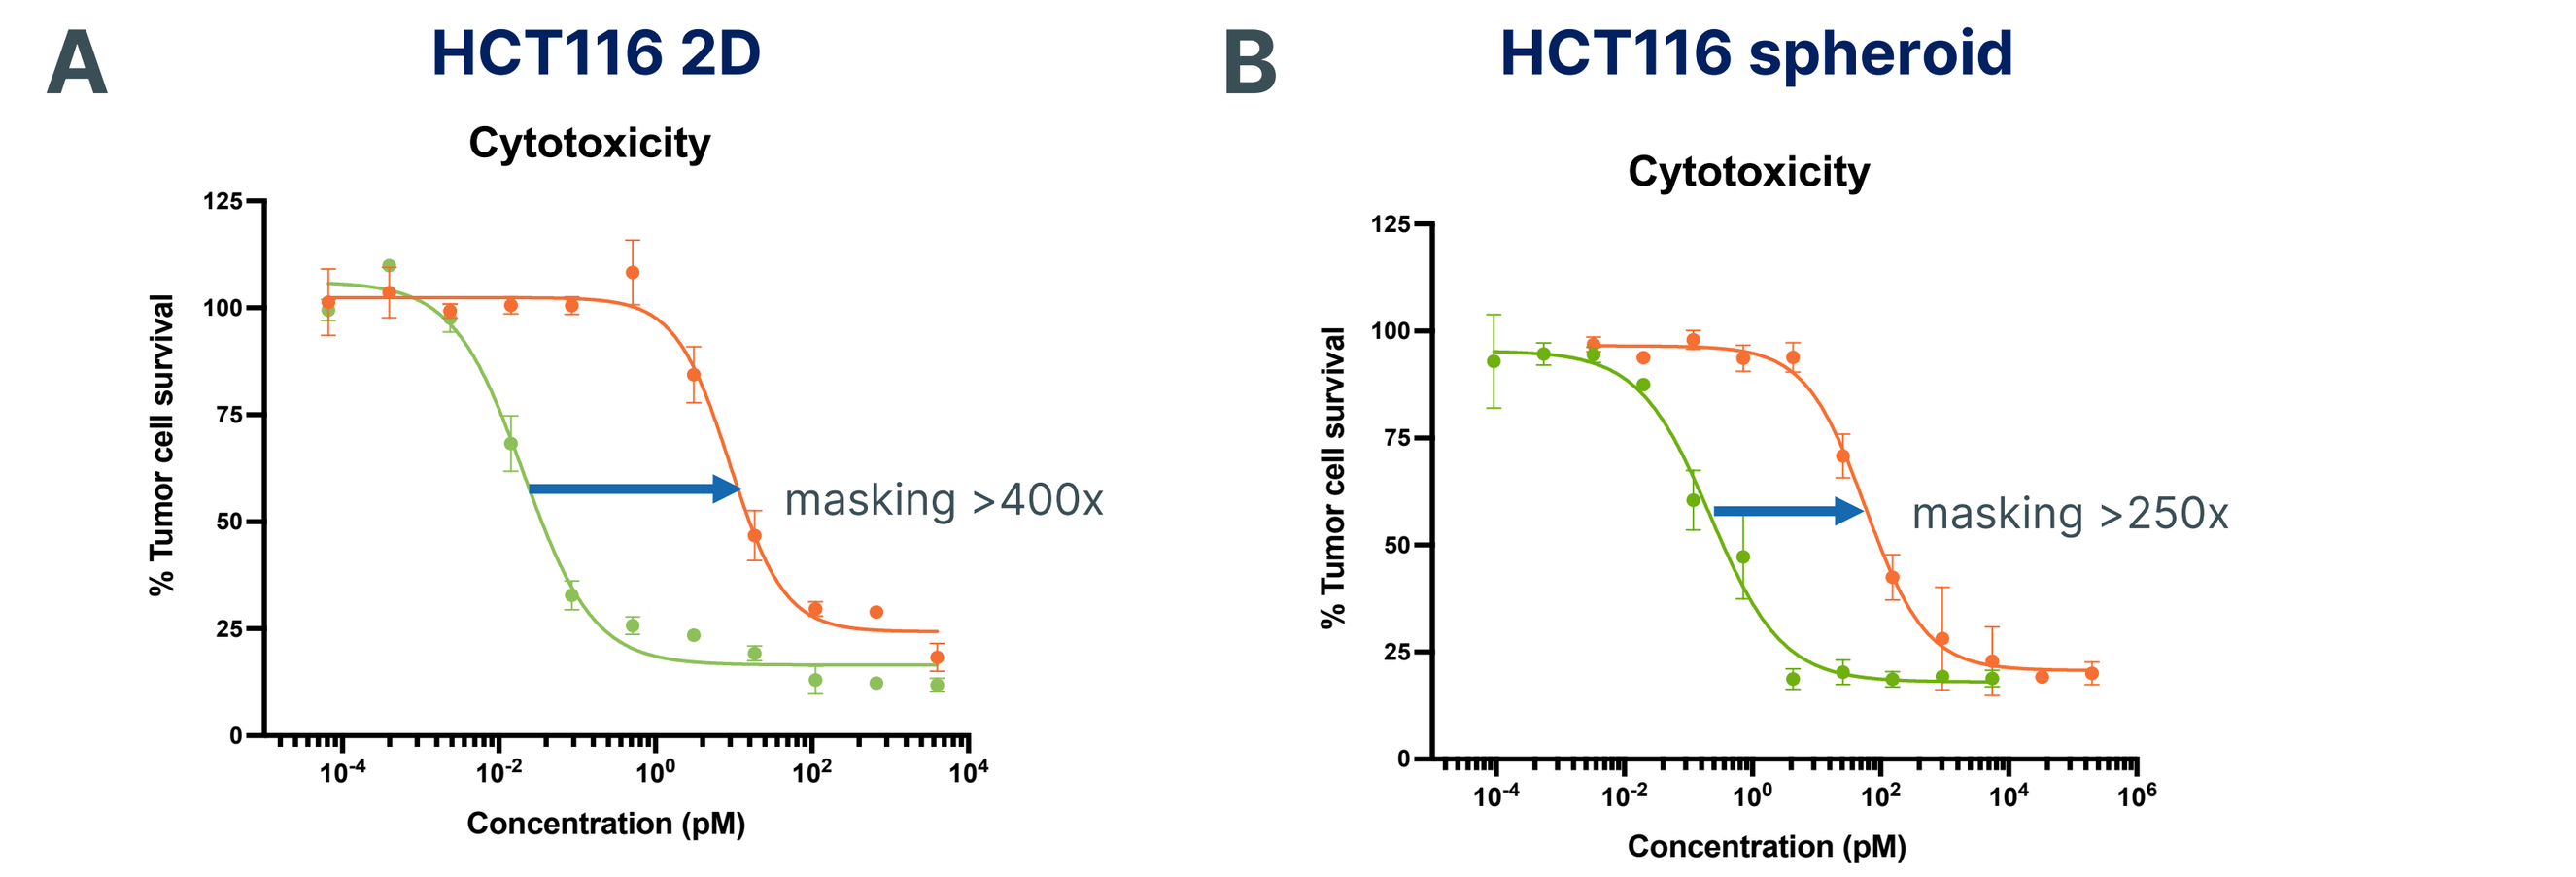
A
HCT116 2D
Cytotoxicity
% Tumor cell survival
Concentration (pM)
masking >400x
B
HCT116 spheroid
Cytotoxicity
masking >250x

Cleaved TAAxCD3 PROTECT™ increases binding selectivity to exhausted T cells

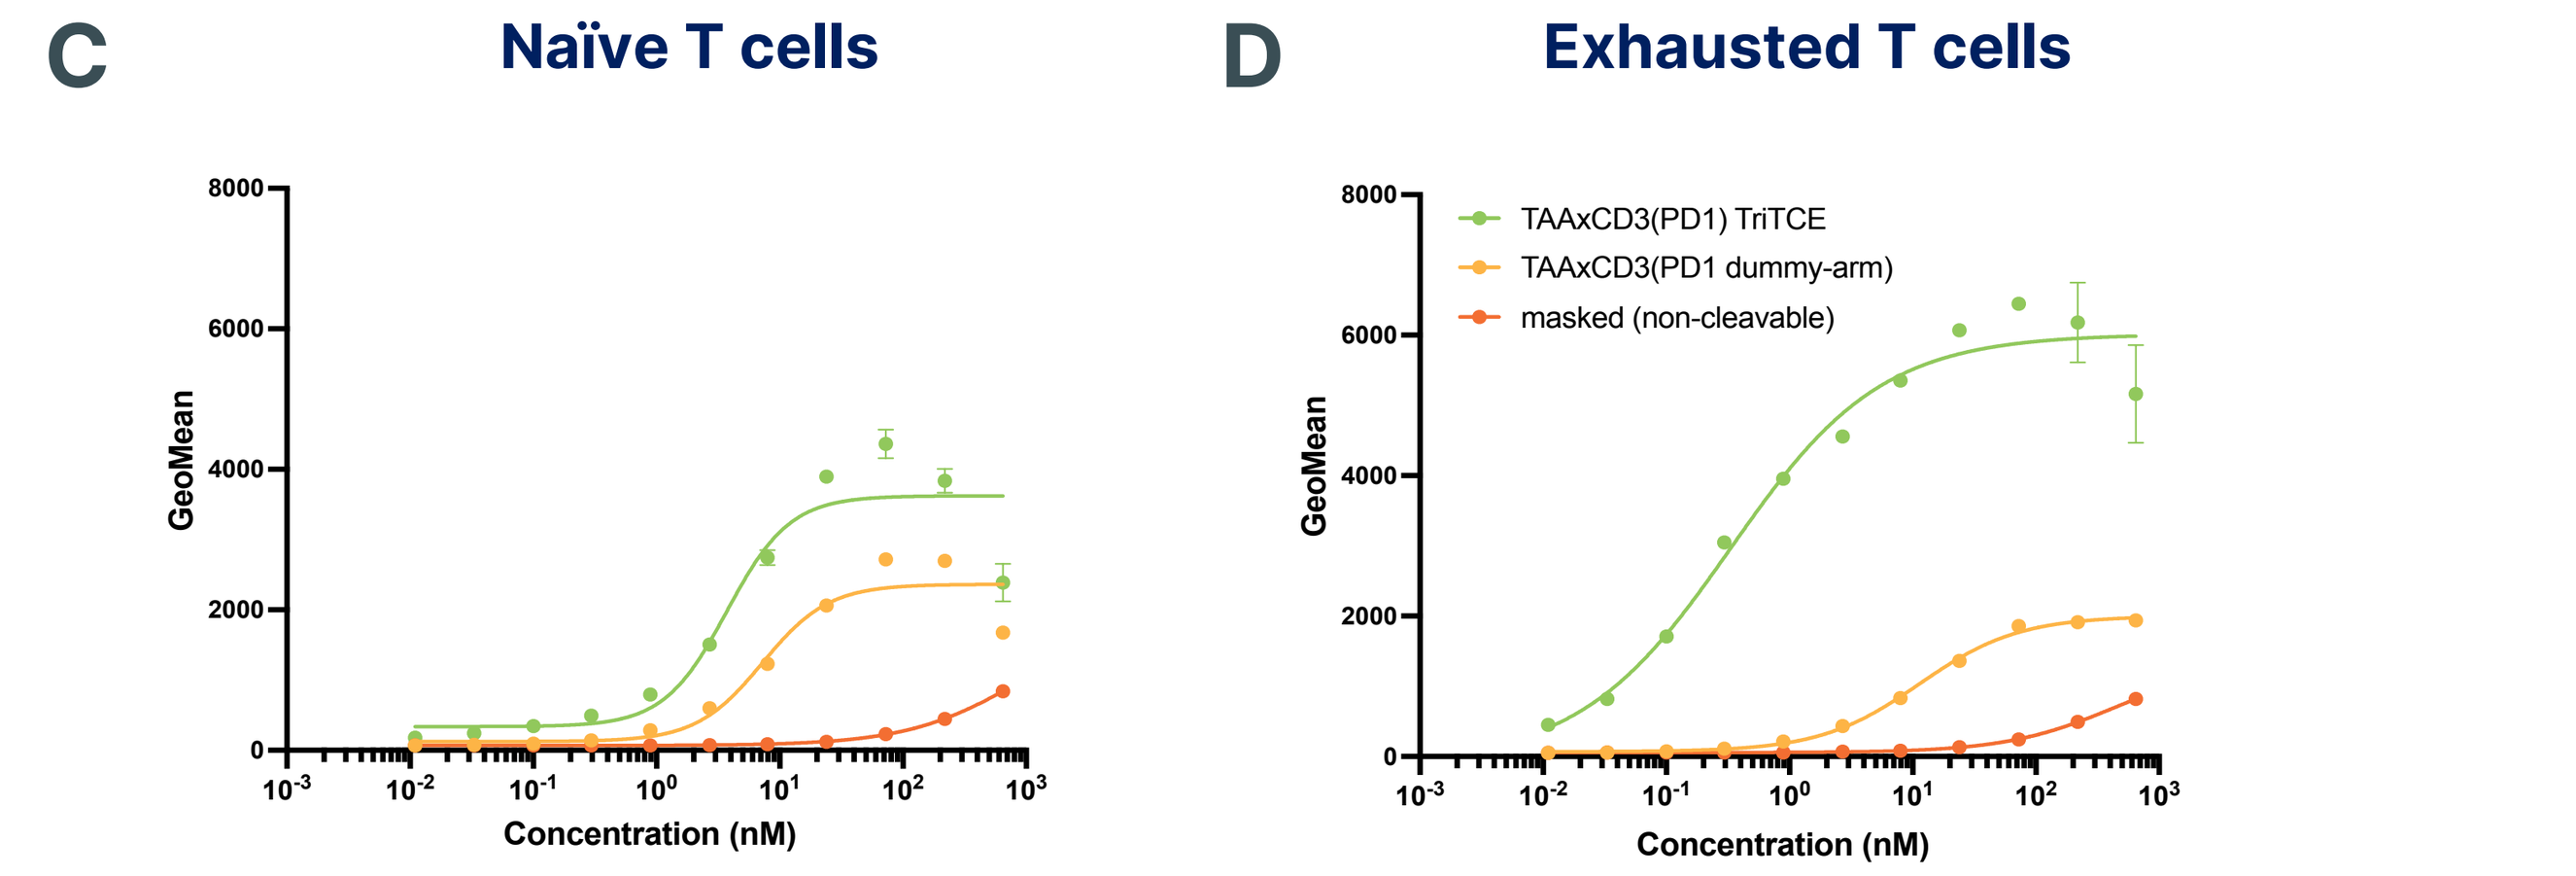
C
Naïve T cells
D
Exhausted T cells
TAAxCD3(PD1) TriTCE
TAAxCD3(PD1 dummy-arm)
masked (non-cleavable)
GeoMean
Concentration (nM)

Cleaved TAAxCD3 PROTECT™ displays checkpoint inhibition on PD-L1 expressing tumor cells

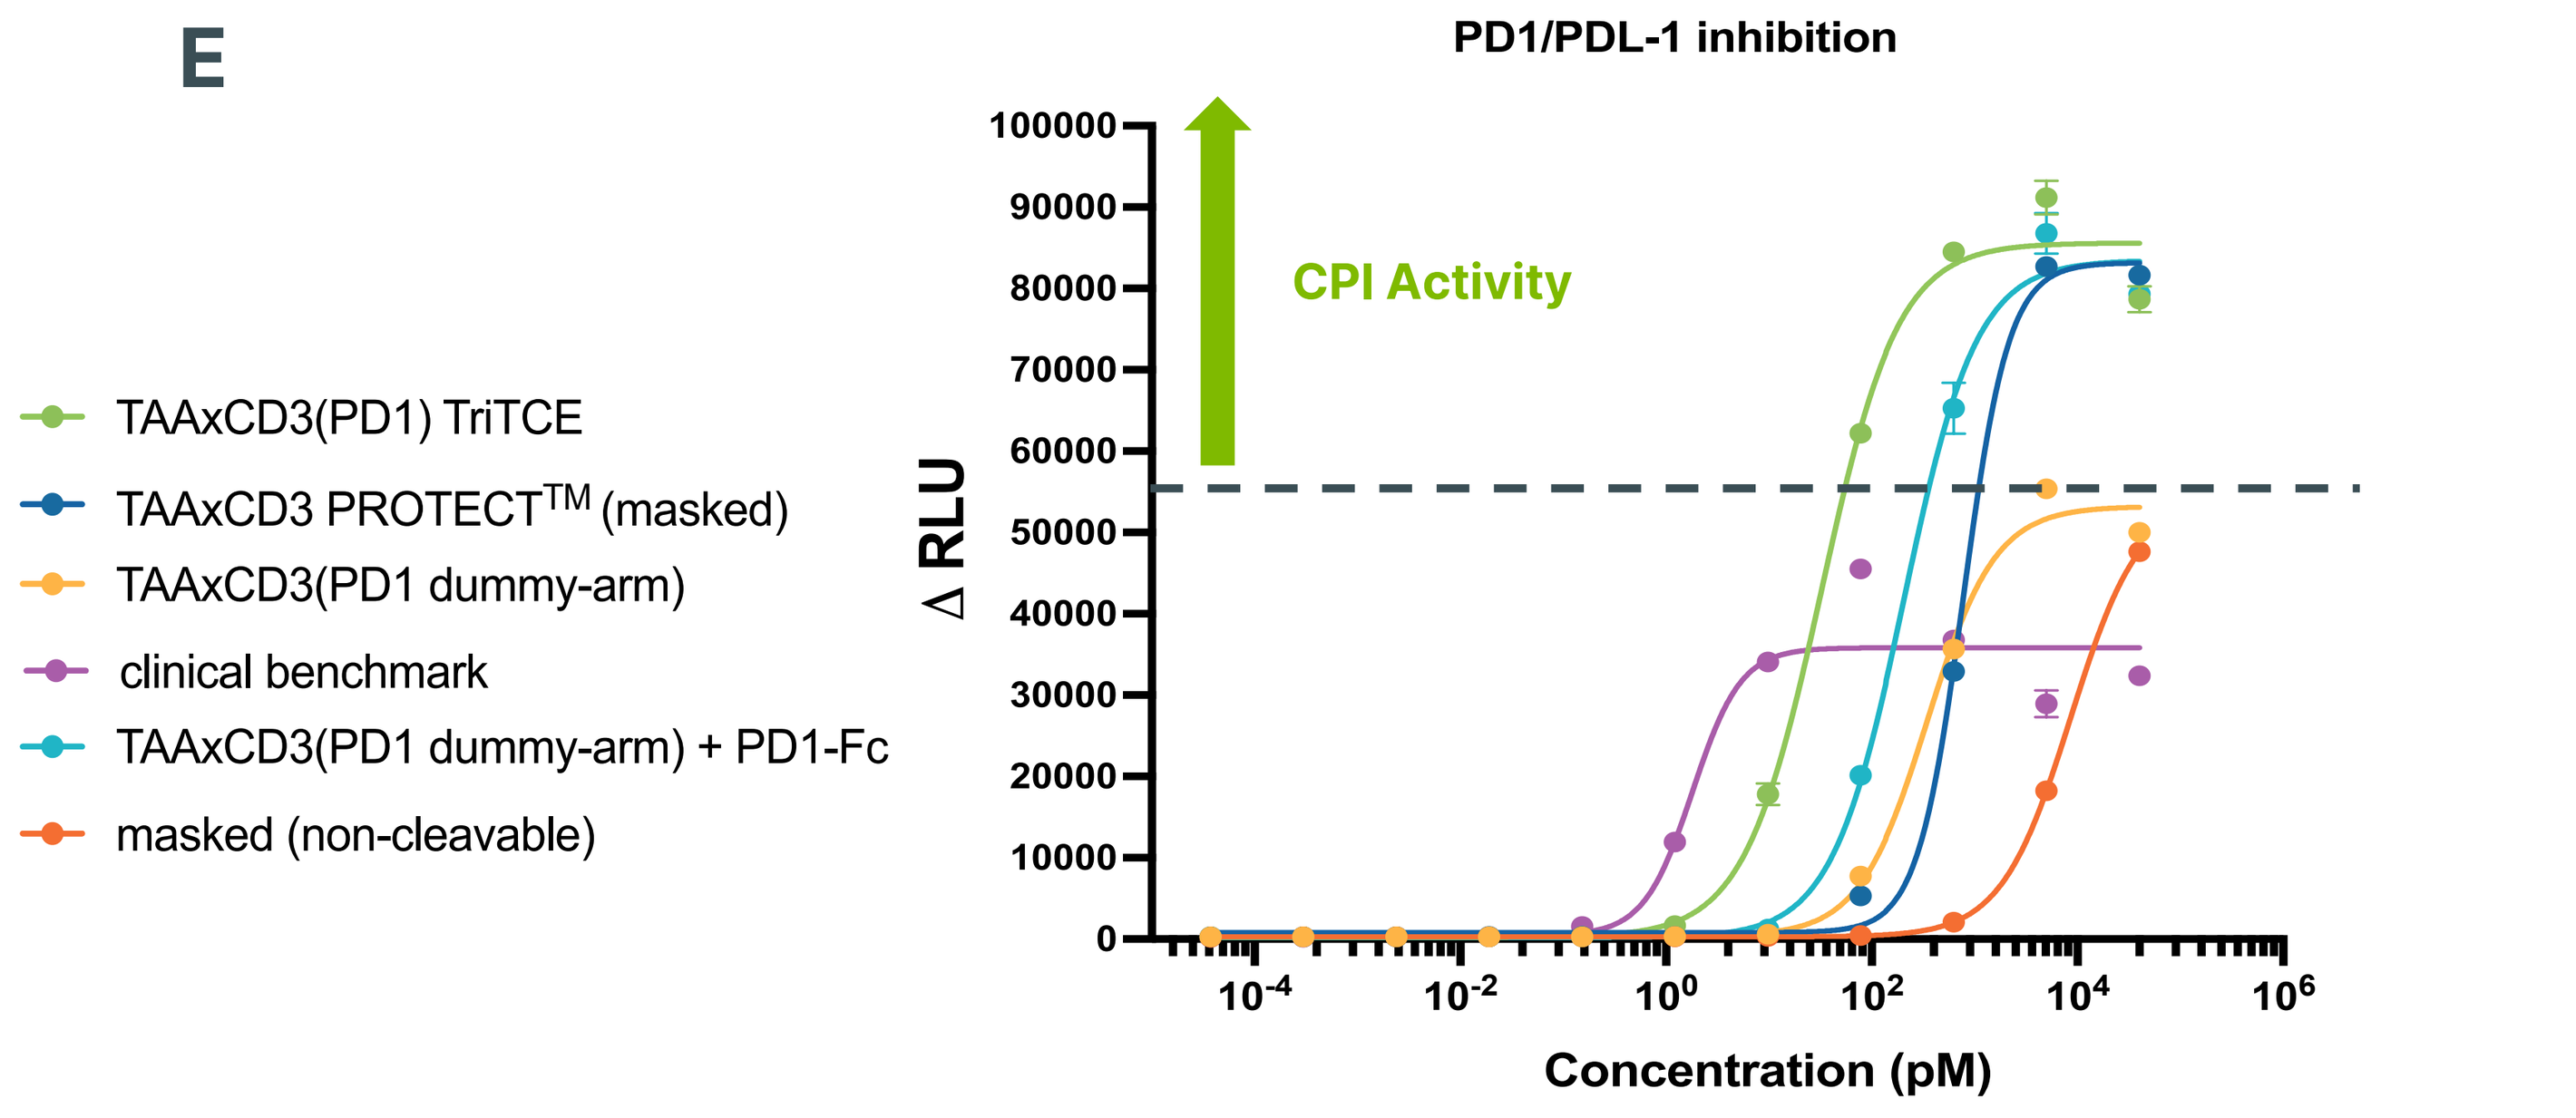
E
PD1/PDL-1 inhibition
TAAxCD3(PD1) TriTCE
TAAxCD3 PROTECT™ (masked)
TAAxCD3(PD1 dummy-arm)
clinical benchmark
TAAxCD3(PD1 dummy-arm) + PD1-Fc
masked (non-cleavable)
CPI Activity
Δ RLU
Concentration (pM)

Figure 3. TAAxCD3 PROTECT™ mediates increased anti-tumor cytotoxicity, greater on-cell binding to exhausted T-cells and CPI upon mask release. T-cell mediated cytotoxicity was evaluated in standard T cell cytotoxicity assay using PBMCs effectors at 10:1 ratio with HCT116 tumor cells **(A)** or 3D HCT116 spheroids **(B)** as targets. Masking window is calculated compared to non-cleavable mask control. Exhausted T cells were produced by subjecting T cells to repeated rounds of re-stimulation using CD3/CD28 beads. Expression of PD1/TIM3/LAG3 and PD-L1 were confirmed as well as inability to secrete cytokines in response to further activation (data not shown). On-cell binding was measured by flow cytometry. TAAxCD3(PD1) TriTCE bound preferentially to exhausted vs naïve T-cells. Masked non-cleavable variants showed little binding and TAAxCD3 with PD1 dummy showed similar binding for both naïve **(C)** and exhausted T cells **(D)**. Checkpoint inhibition was evaluated using a PD-1/PD-L1 RGA **(E)** with HCT116 cells as targets and PD-1-expressing Jurkat cells as effectors. The CPI assay demonstrate PROTECT™ can block PD-1 and PD-L1 interaction while stimulating CD3 in a TAA dependent manner. TAAxCD3(PD1) TriTCE provides checkpoint inhibition at lower concentrations than the bispecific combo with anti-PD-L1. The non-cleavable and masked PROTECT™ do not provide significant checkpoint inhibition.

PROTECT™ Potentiates T cell Activation by Engaging Dendritic Cells

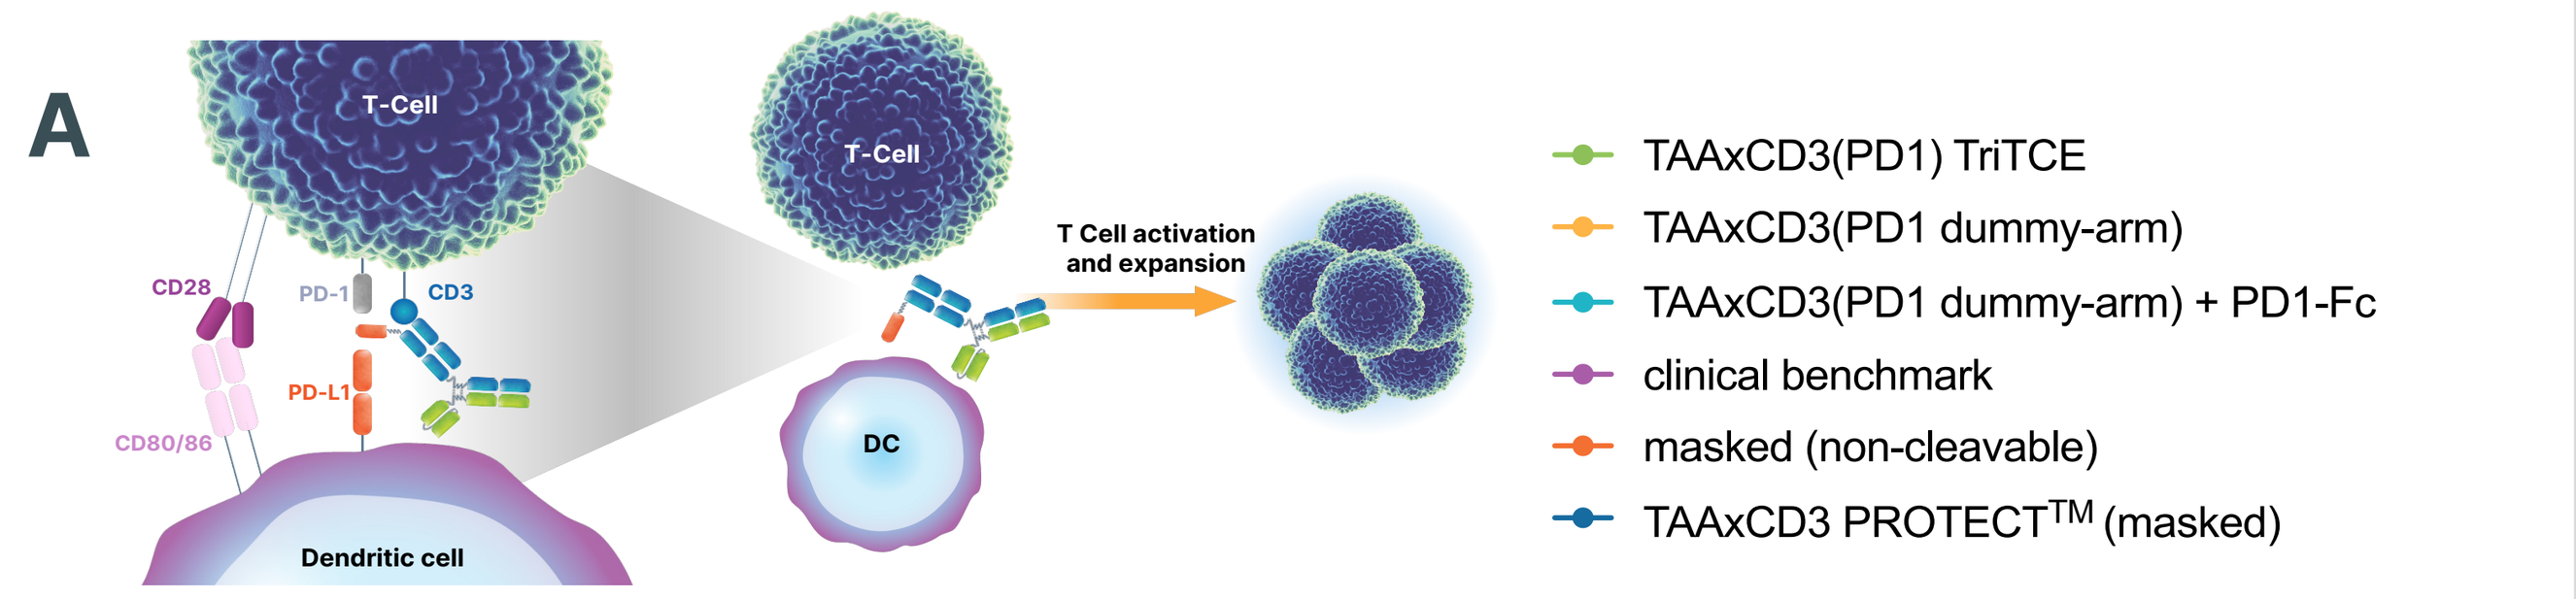
A
T-Cell
CD28
PD-1
CD3
PD-L1
CD80/86
Dendritic cell
T-Cell
DC
T Cell activation and expansion
TAAxCD3(PD1) TriTCE
TAAxCD3(PD1 dummy-arm)
TAAxCD3(PD1 dummy-arm) + PD1-Fc
clinical benchmark
masked (non-cleavable)
TAAxCD3 PROTECT™ (masked)

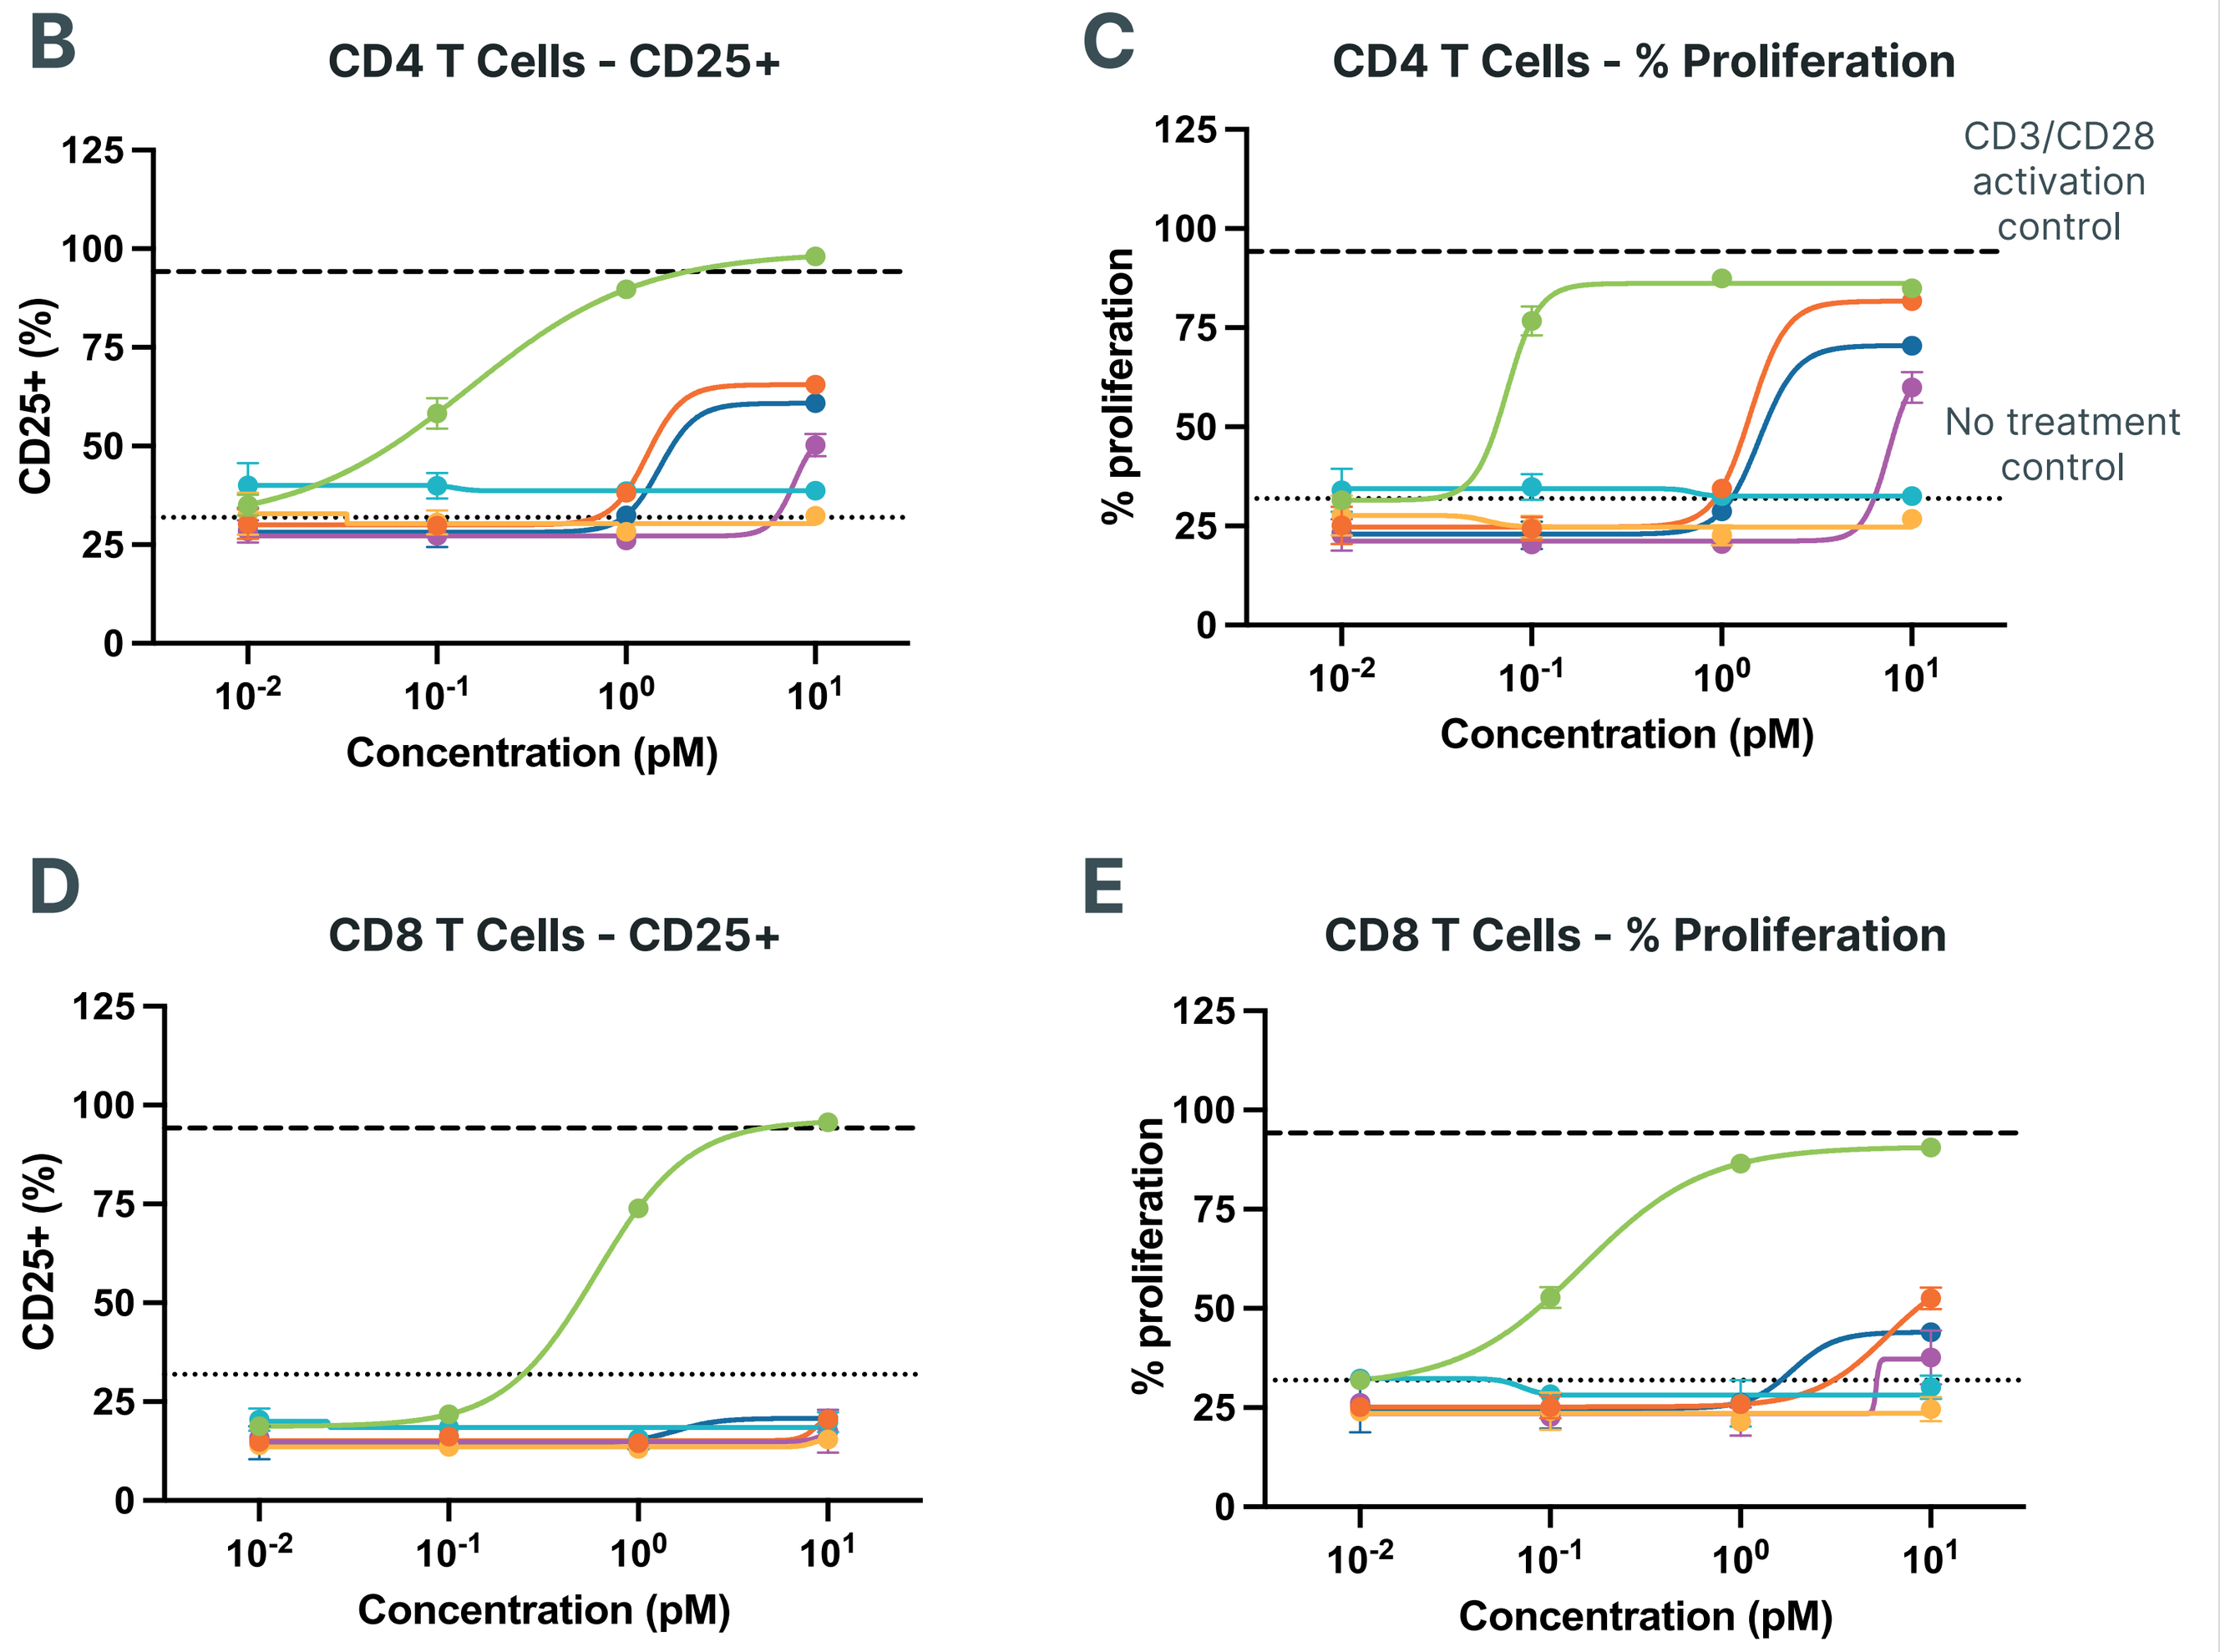
B
CD4 T Cells - CD25+
CD25+ (%)
Concentration (pM)
C
CD4 T Cells - % Proliferation
% proliferation
CD3/CD28 activation control
No treatment control
D
CD8 T Cells - CD25+
E
CD8 T Cells - % Proliferation

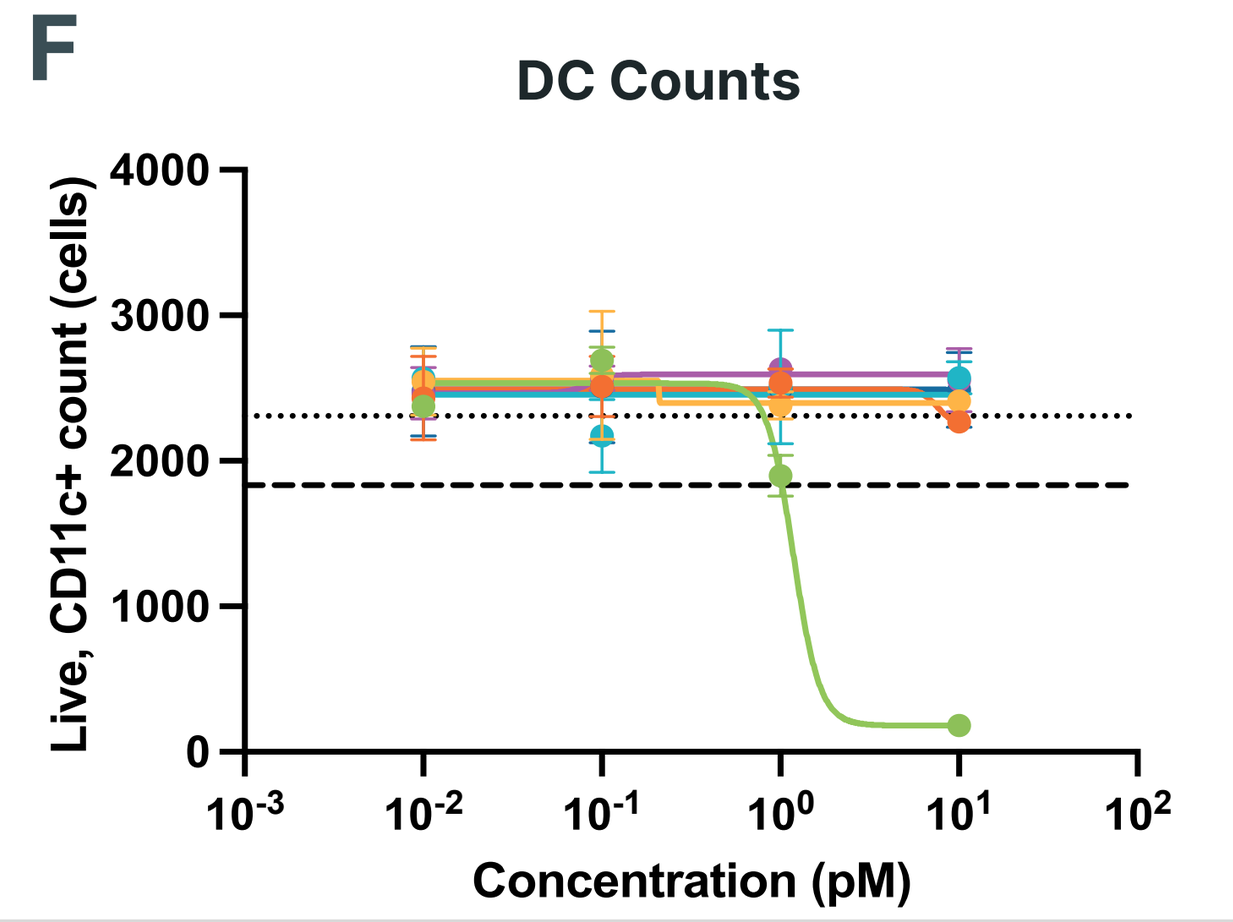
F
DC Counts
Live, CD11c+ count (cells)
Concentration (pM)

Figure 4. Bridging between DCs and T cells by the TAAxCD3(PD1) TriTCE potentiates T cell activation and proliferation. (A) DCs in the TME express high levels of PD-L1. TriTCE can potentiate T cell activation by bridging DCs and T cells via PD-L1 and CD3, respectively. **(B-E)** Mature DC and T cells were co-cultured for 5 days with PROTECT™ test articles. Treatment with the TriTCE mediated increased proliferation and CD25 expression in CD4 **(B-C)** and CD8 positive T cells **(D-E)** compared to masked or non-cleavable TAAxCD3 PROTECT™. The TAAxCD3 bispecific (PD1-dummy arm) in combination with a PD1-Fc fusion protein had no effect. **(F)** At high concentrations the TriTCE is able to deplete PD-L1-expressing DCs, but not the masked variants.

Conclusions

- PROTECT™ engineering platform combines a tumor-specific masking/unmasking approach with a unique trispecific format to enhance the therapeutic index of T cell engagers.
- Platform incorporates several engineering solutions to improve tolerability: anti-CD3 masking, low affinity CD3 binding, enhanced binding to tumor cells, and selectivity for exhausted T cells over naïve T cells.
- Cleaved trispecific displays unique MOAs to increase efficacy including increased avidity, checkpoint inhibition, and ability to potentiate T cell activation via PD-L1 on APCs.
- Zymeworks is currently applying the PROTECT™ platform to additional cancer targets.

References

1. Hart, PC. The tumor proteolytic Landscape: A challenging frontier in cancer diagnosis and therapy. Int J Mol Sci, 2021; 22(5):2514

Acknowledgements

We would like to thank Dr. Paul Moore for scientific review and all former employees who contributed to this project over the years.