Super-resolution Imaging Studies of Zanidatamab: Providing Insights into its Bispecific Mode of Action

Abstract: 1032

Libin Abraham[1], Gesa Volkers[2], Liam Worrall[3], Claire Atkinson[3], Alejandro Gil-Ley[2], Joel Smith[2], Joe Schrag[4], Charles Stevens[2], Mario Sanches[2], Patrick Farber[2], Abhishek Mukhopadhyay[2], Antonios Samiotakis[2], Nina Weiser[2], Natalie Strynadka[3], Mike Gold[1], Surjit Dixit[2]

[1]Department of Immunology and Microbiology, University of British Columbia, Vancouver, BC, Canada; [2]Zymeworks Inc., 1385 West 8th Ave, Vancouver, BC, Canada; [3]Department of Biochemistry, University of British Columbia, Vancouver, BC, Canada; [4]Human Health Therapeutics Portfolio, NRC-CNRC, Montreal, Canada.

Zanidatamab: A Biparatopic Anti-HER2 Antibody

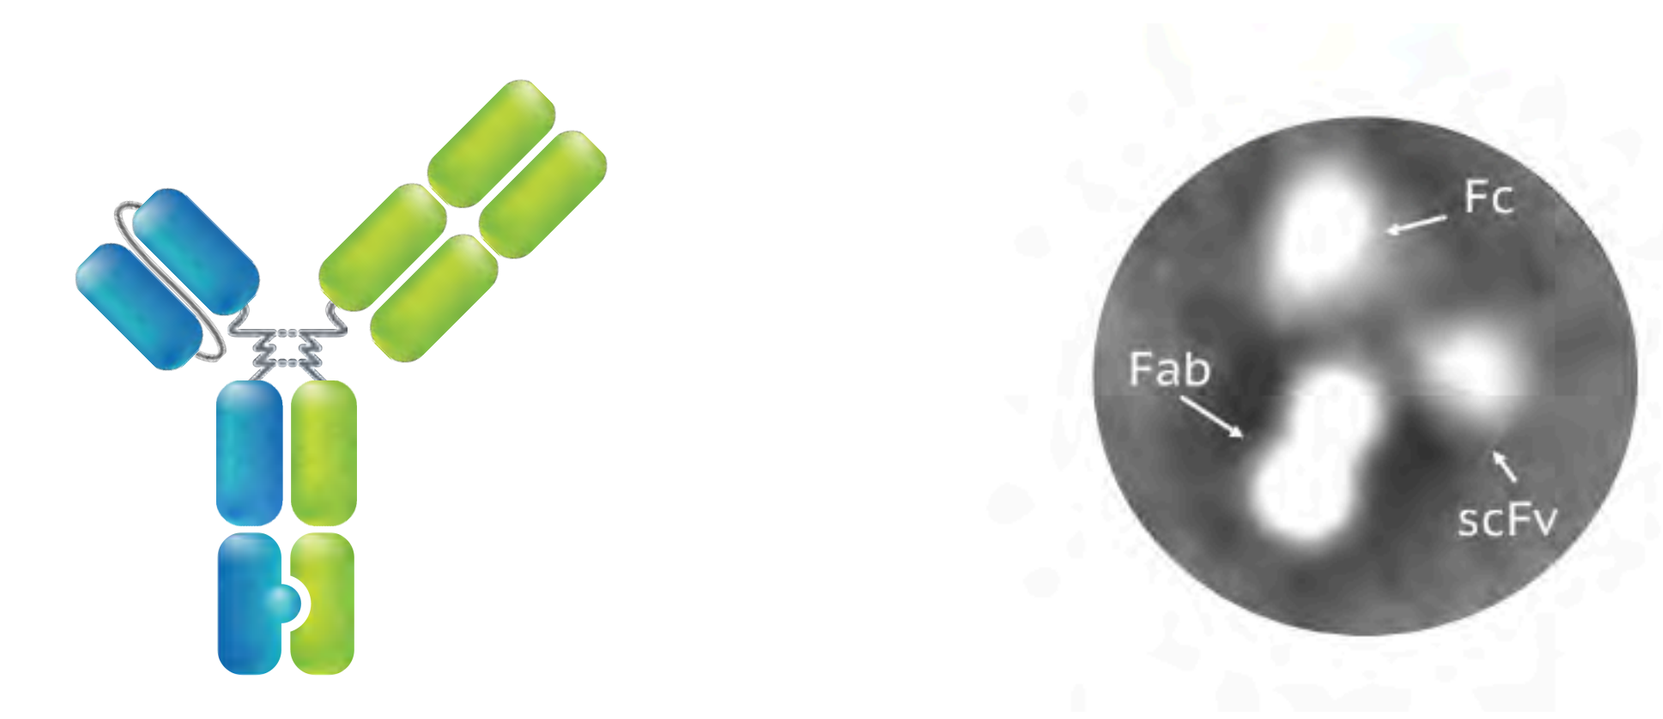
Fc
Fab
scFv

Zanidatamab (zani) simultaneously binds two distinct epitopes of the oncogenic cell surface receptor HER2. Zani is currently being evaluated in global Phase 1, Phase 2 and registration-enabling clinical trials as a potential new treatment for patients with HER2-expressing cancers, including biliary tract, gastroesophageal adenocarcinomas, breast, and other tumor types. The extracellular domain of the HER2 receptor comprises four domains with zani binding to epitopes on domain 2 and the membrane proximal domain 4. Domain 2 and domain 4 of HER2 are also respectively targeted by the antibodies pertuzumab (pert) and trastuzumab (tras), currently approved for clinical use in a subset of HER2 overexpressing cancers. The unique engineering and HER2-engagement of zani results in multiple mechanisms of action, including dual HER2 signal blockade, increased antibody binding, receptor clustering, removal of HER2 from the cell surface, and potent effector function. Preclinical data shows that zani can exhibit differentiated activity over tras or combination of tras and pert. We present results from a variety of structural and imaging studies which showcase the differentiated structural characteristics of zani:HER2 complexes.

Zanidatamab Preferentially Forms Higher Order Complex Structures with HER2 In Solution

Zani, tras, pert and tras+pert were mixed at equimolar ratio with HER2 ECD and sedimentation was characterized by Analytical Ultra Centrifugation (AUC).

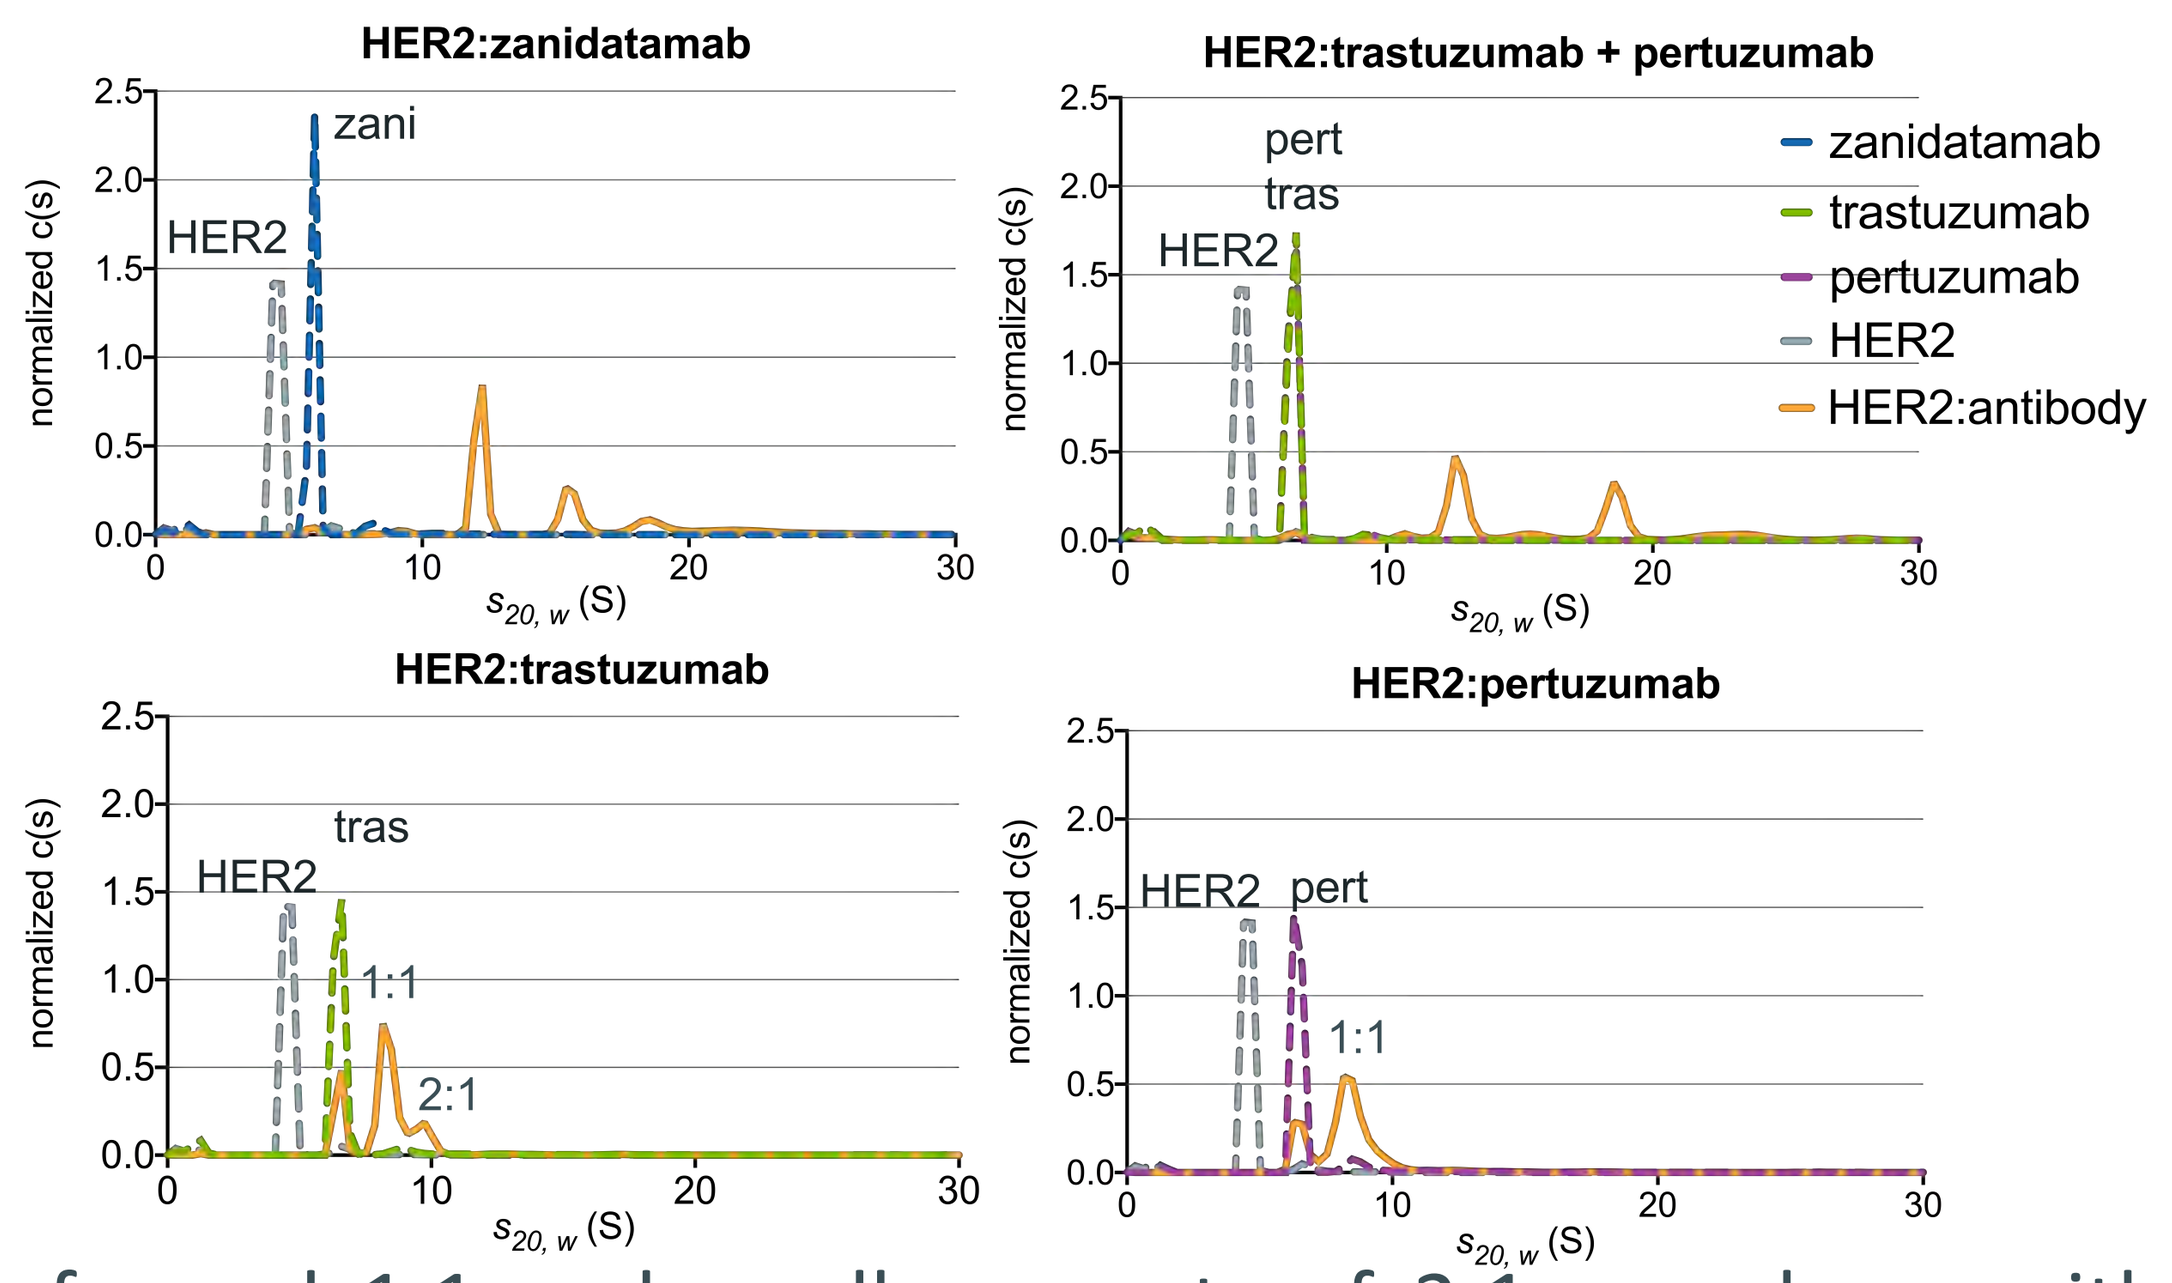
HER2:zanidatamab
HER2:trastuzumab + pertuzumab
HER2:trastuzumab
HER2:pertuzumab
normalized c(s)
$s_{20,w}$ (S)
zani
HER2
pert
tras
1:1
2:1
zanidatamab
trastuzumab
pertuzumab
HER2
HER2:antibody

Either tras or pert formed 1:1 and small amounts of 2:1 complexes with HER2. Zani or the combination of tras+pert formed predominantly higher order complexes with some differences of molecular weight distribution. Only trace amounts of 1:1 and 2:1 complexes were detected for zani or the combo of tras+pert when mixed at equimolar ratio with HER2, respectively.

Zanidatamab:HER2 Do Not Form Simple 1:1 Paired Complexes

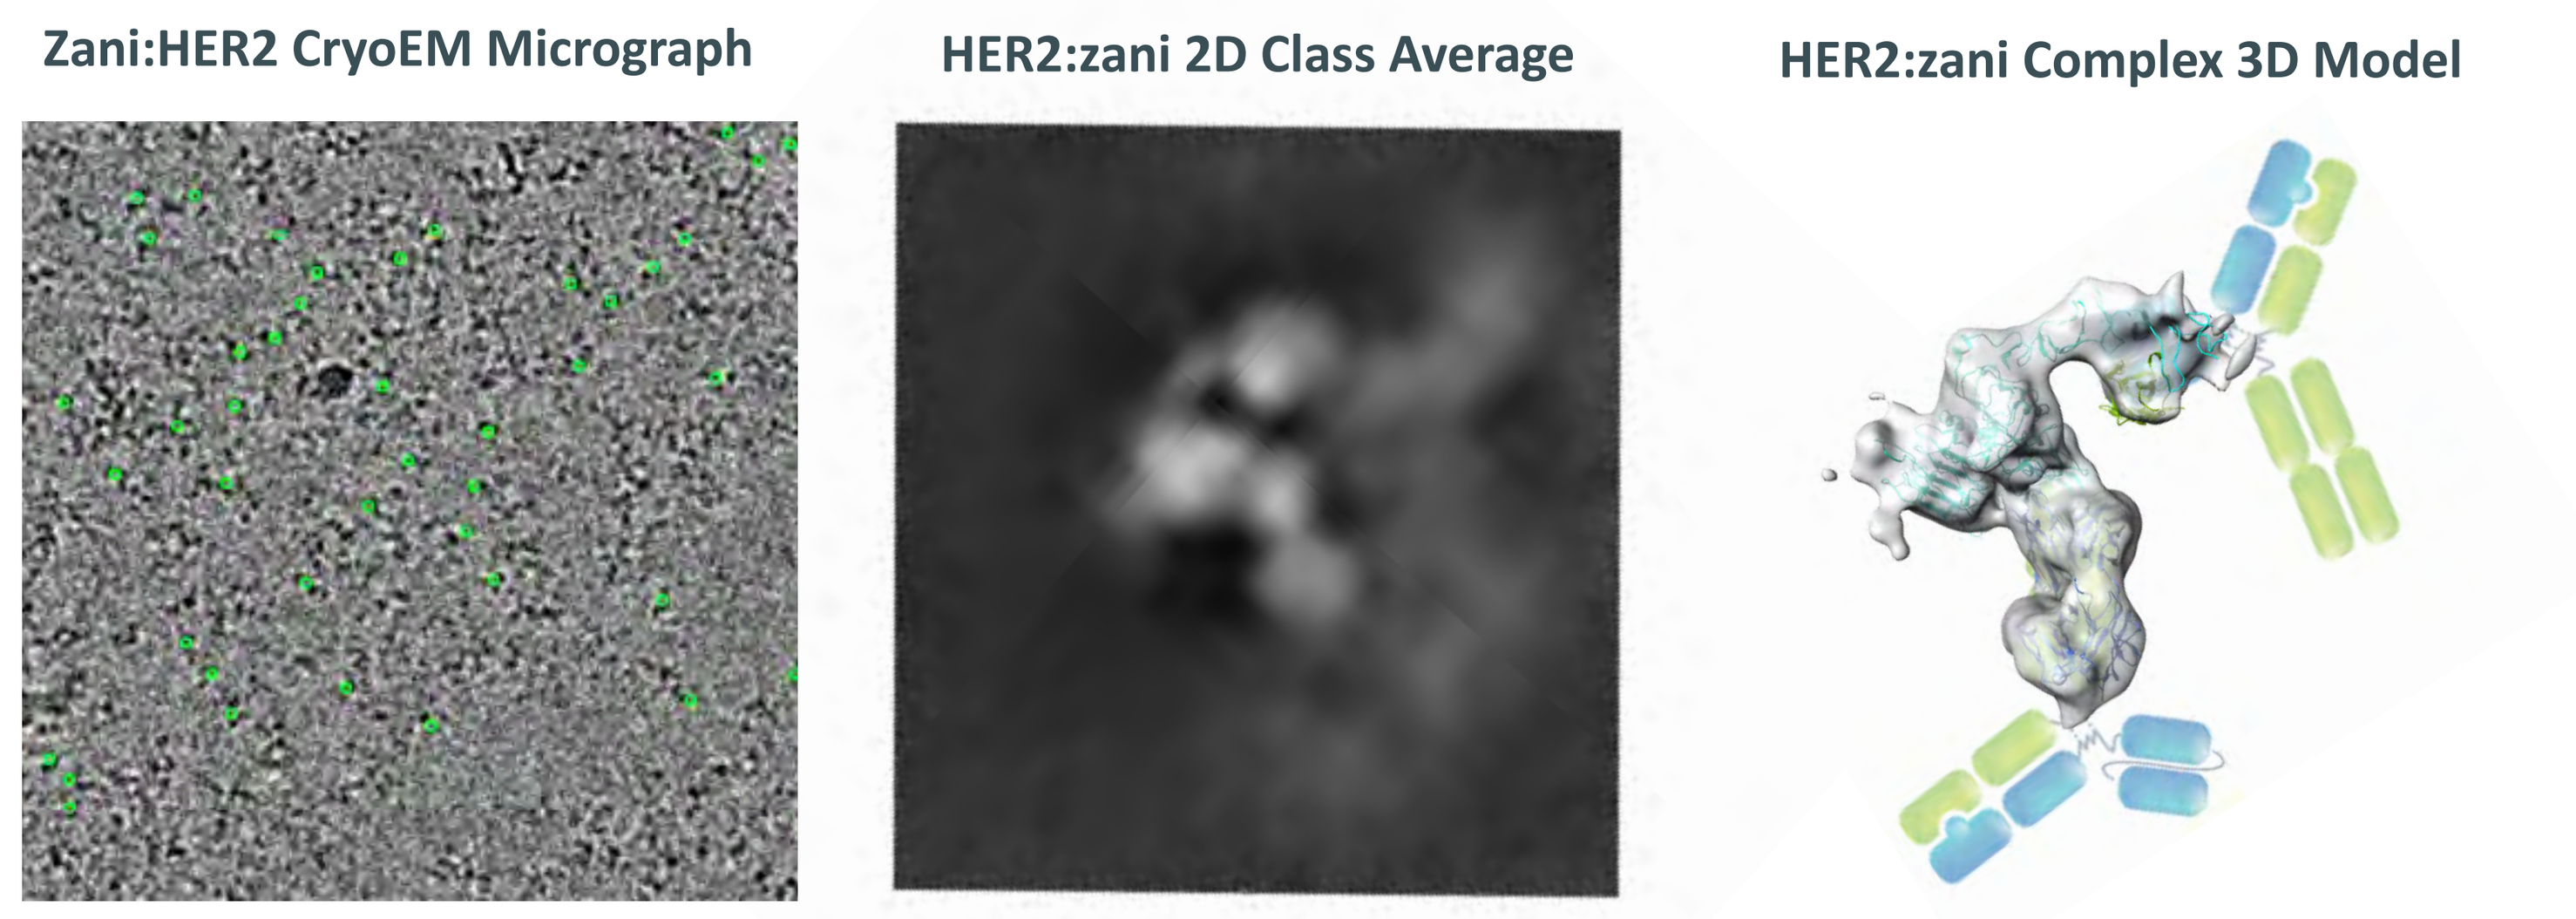
Zani:HER2 CryoEM Micrograph
HER2:zani 2D Class Average
HER2:zani Complex 3D Model

Complexes of zani with HER2 appeared as strings of molecular assemblies on the Cryo-electron micrographs. A model of HER2 (cyan) bound to zani was determined to approximately 8 Å resolution from a dataset of about 25,000 particles. Only the Fab (blue ribbons) bound to domain 2 of HER2 and scFv (green ribbon) bound to domain 4 of HER2 are resolved. The relative orientation of the Fab and scFv does not sterically allow for a 1:1 pairing of HER2 and zani. Instead, 2 copies of zani are expected to bind HER2 as shown in the example 2D class average and 3D model, with the rest of the structure appearing as a diffuse halo. The unresolved domains of two bound zani molecules are presented in the 3D structure as cartoons.

Computational Modeling Helps Understand Characteristics Of Zani:HER2 Complexation

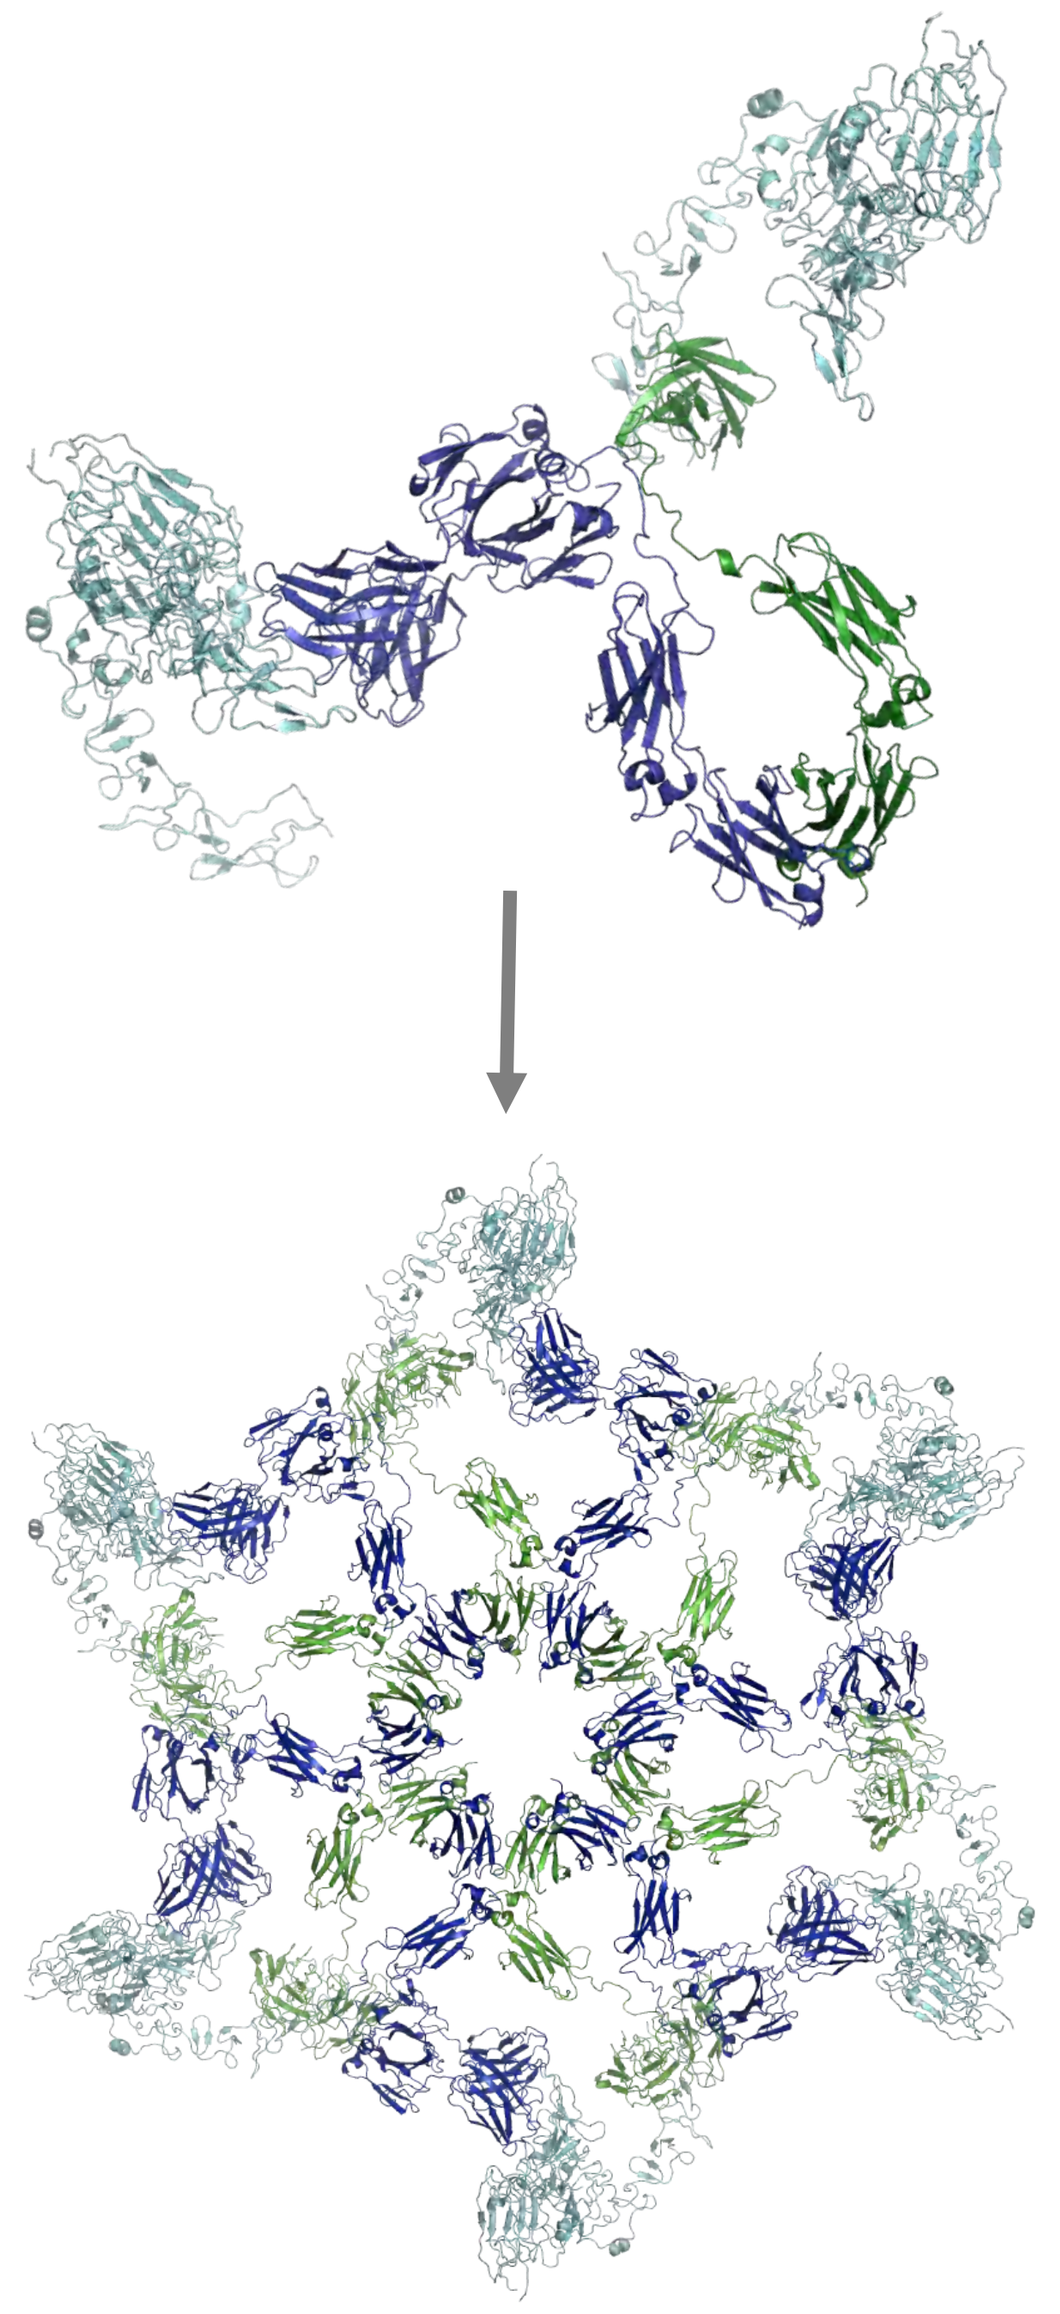

Hexamerization of IgG Fc promotes C1q engagement and CDC activity. In light of the CDC activity observed for zani (See Poster #1005), we evaluated if a HER2:zani complex could form a six-fold cyclic structure. Applying symmetry operations to a model of HER2 bound to two copies of zani (top left) based on the structure of HER2 + tras/pert complexes (PDB ID: 1N8Z, 1S78, 6OGE) resulted in a six-fold ring structure (bottom left) with HER2 naturally aligned in a plane, akin to presentation on the cell membrane (below). The Fc is aligned in a configuration suitable for interaction with C1q (PDB ID: 6FCZ). Symmetry limitations did not allow an equivalent model of HER2 with the combination of tras and pert.

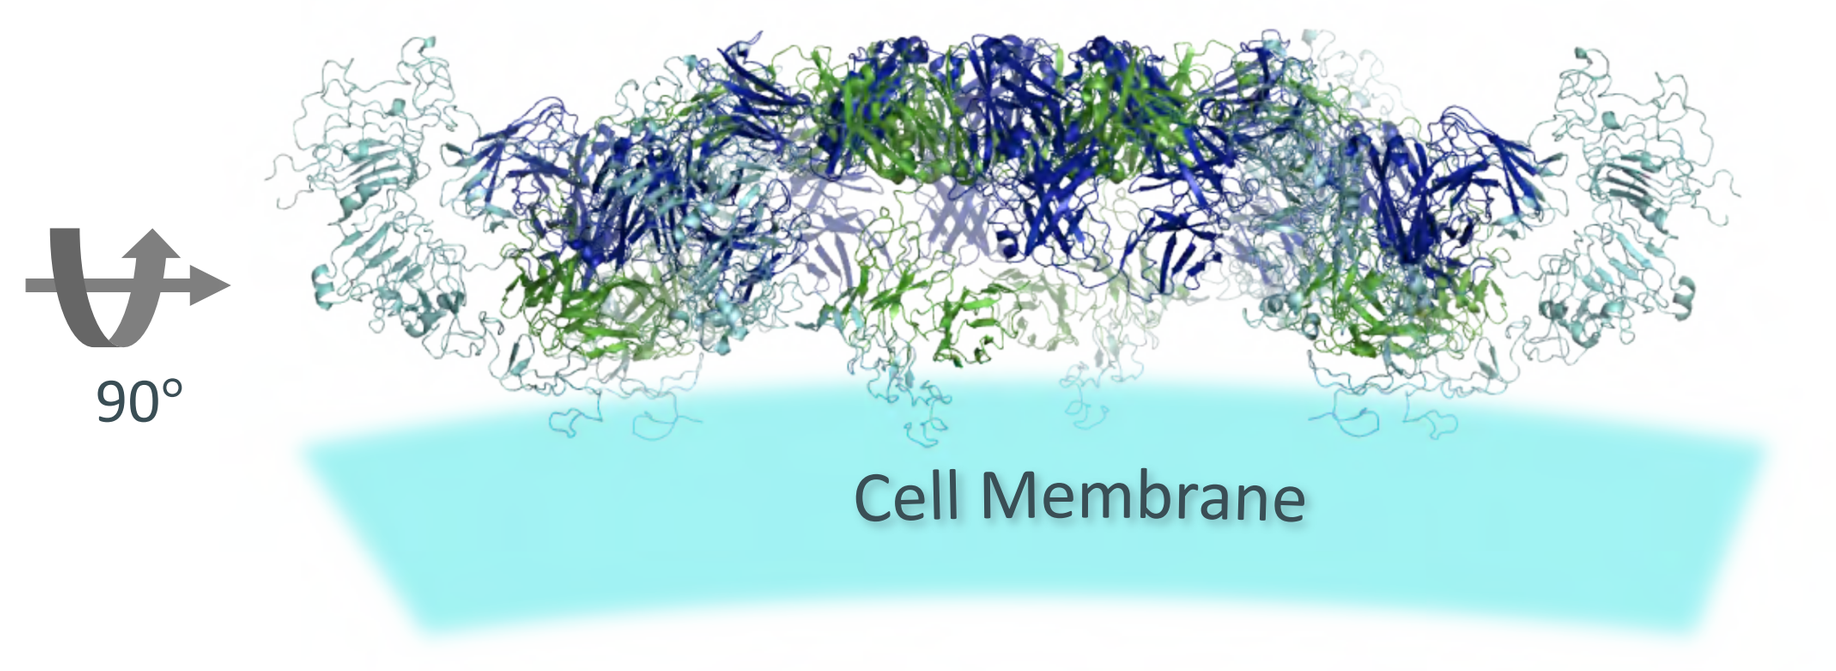
90°
Cell Membrane

Zanidatamab Induces Distinctly Large Clusters Of HER2 On Target Cell Surface

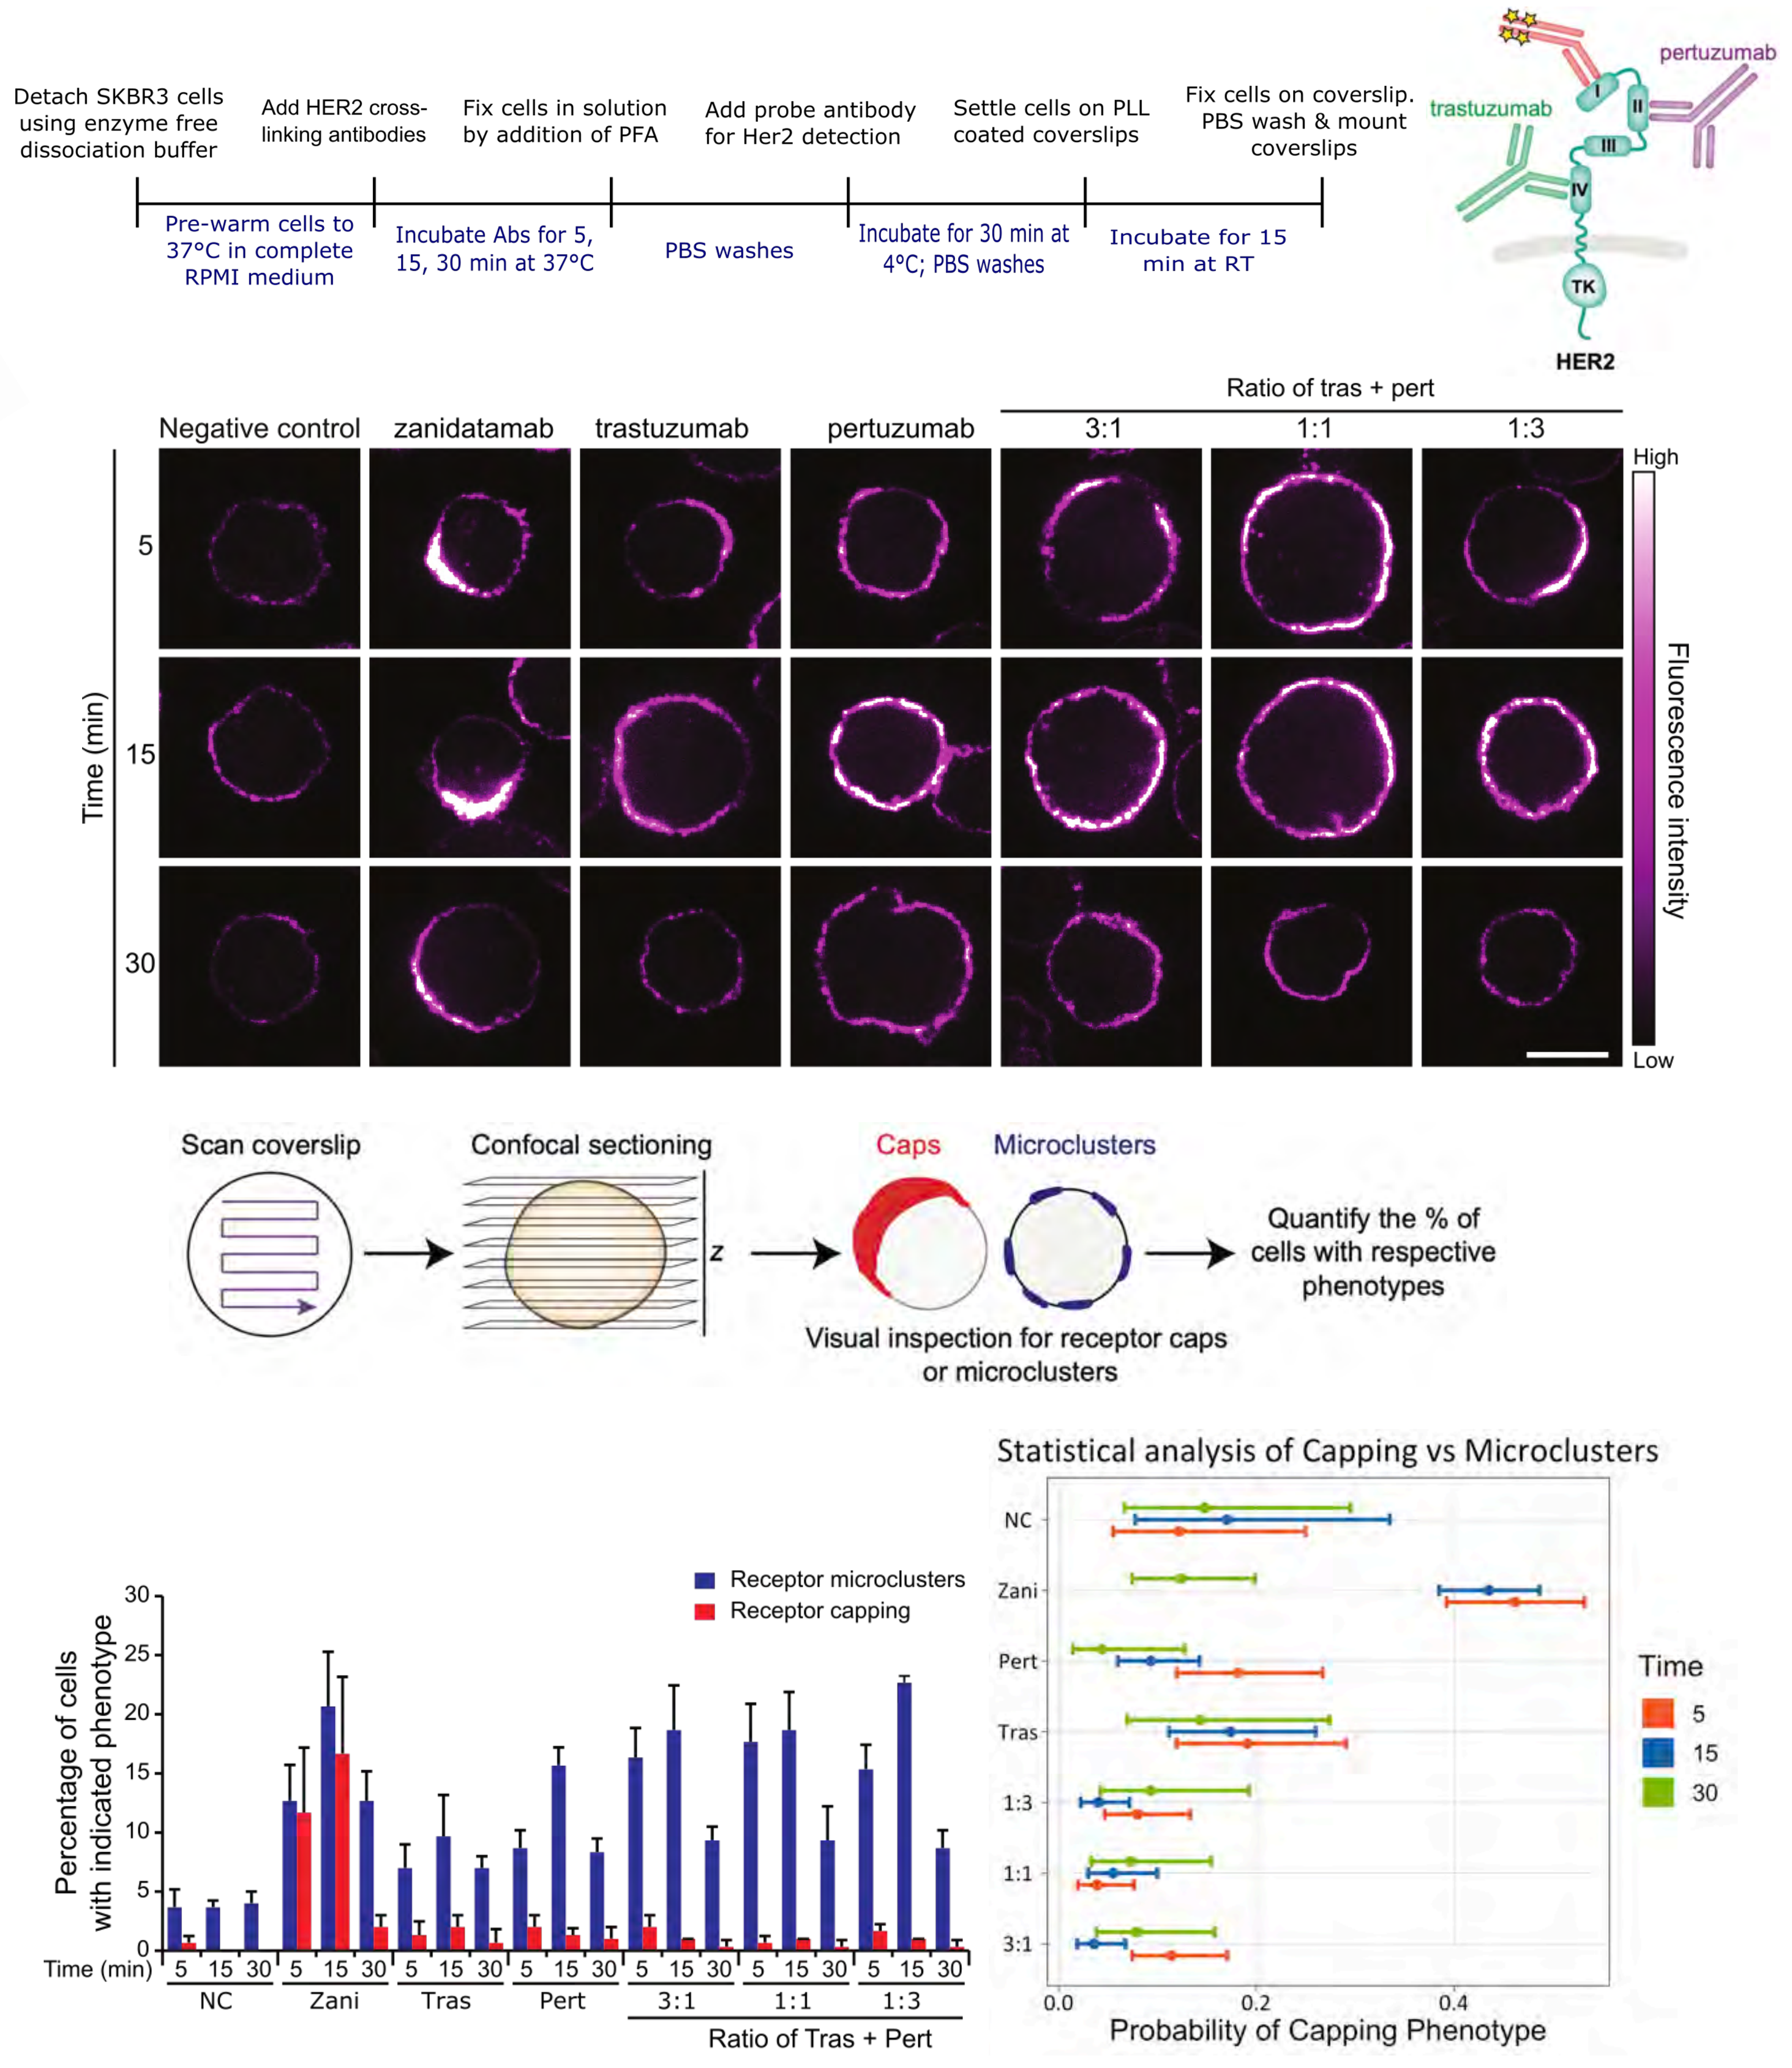
Detach SKBR3 cells using enzyme free dissociation buffer
Add HER2 cross-linking antibodies
Fix cells in solution by addition of PFA
Add probe antibody for Her2 detection
Settle cells on PLL coated coverslips
Fix cells on coverslip. PBS wash & mount coverslips
Pre-warm cells to 37°C in complete RPMI medium
Incubate Abs for 5, 15, 30 min at 37°C
PBS washes
Incubate for 30 min at 4°C; PBS washes
Incubate for 15 min at RT
pertuzumab
trastuzumab
HER2
TK
Ratio of tras + pert
Negative control
zanidatamab
trastuzumab
pertuzumab
3:1
1:1
1:3
Time (min)
High
Low
Fluorescence intensity
Scan coverslip
Confocal sectioning
Caps
Microclusters
Quantify the % of cells with respective phenotypes
Visual inspection for receptor caps or microclusters
Receptor microclusters
Receptor capping
Percentage of cells with indicated phenotype
Time (min)
NC
Zani
Tras
Pert
Ratio of Tras + Pert
Statistical analysis of Capping vs Microclusters
Probability of Capping Phenotype
Time

In a 3D spinning confocal microscopy imaging study with breast cancer cell line SKBR3 that over-expresses HER2, fraction of cells treated with zani showed strikingly large HER2 'caps' that are polarized to one side of the cell membrane. While all α-HER2 antibodies tested induced receptor aggregation and microcluster formation, such receptor caps were largely absent in cells treated with tras, pert or both in combination at various ratios. We observed a time dependence for the capping effect in zani cross-linked samples, the percentage of cells with HER2 caps increased until 15 min and gradually resolved by 30 min. The imaging agent is a fluorophore conjugated single arm antibody that binds ECD1 of HER2. A multinomial logistic regression model fit to the confocal microscopy phenotype data suggests that the probability of observing a capping phenotype over a microcluster, at t=5min and t=15min with zani is higher than with any other treatment tested in the experiment.

Zanidatamab Induced Clusters Are Larger Than Those Formed By Trastuzumab (T), Pertuzumab (P) Or T+P

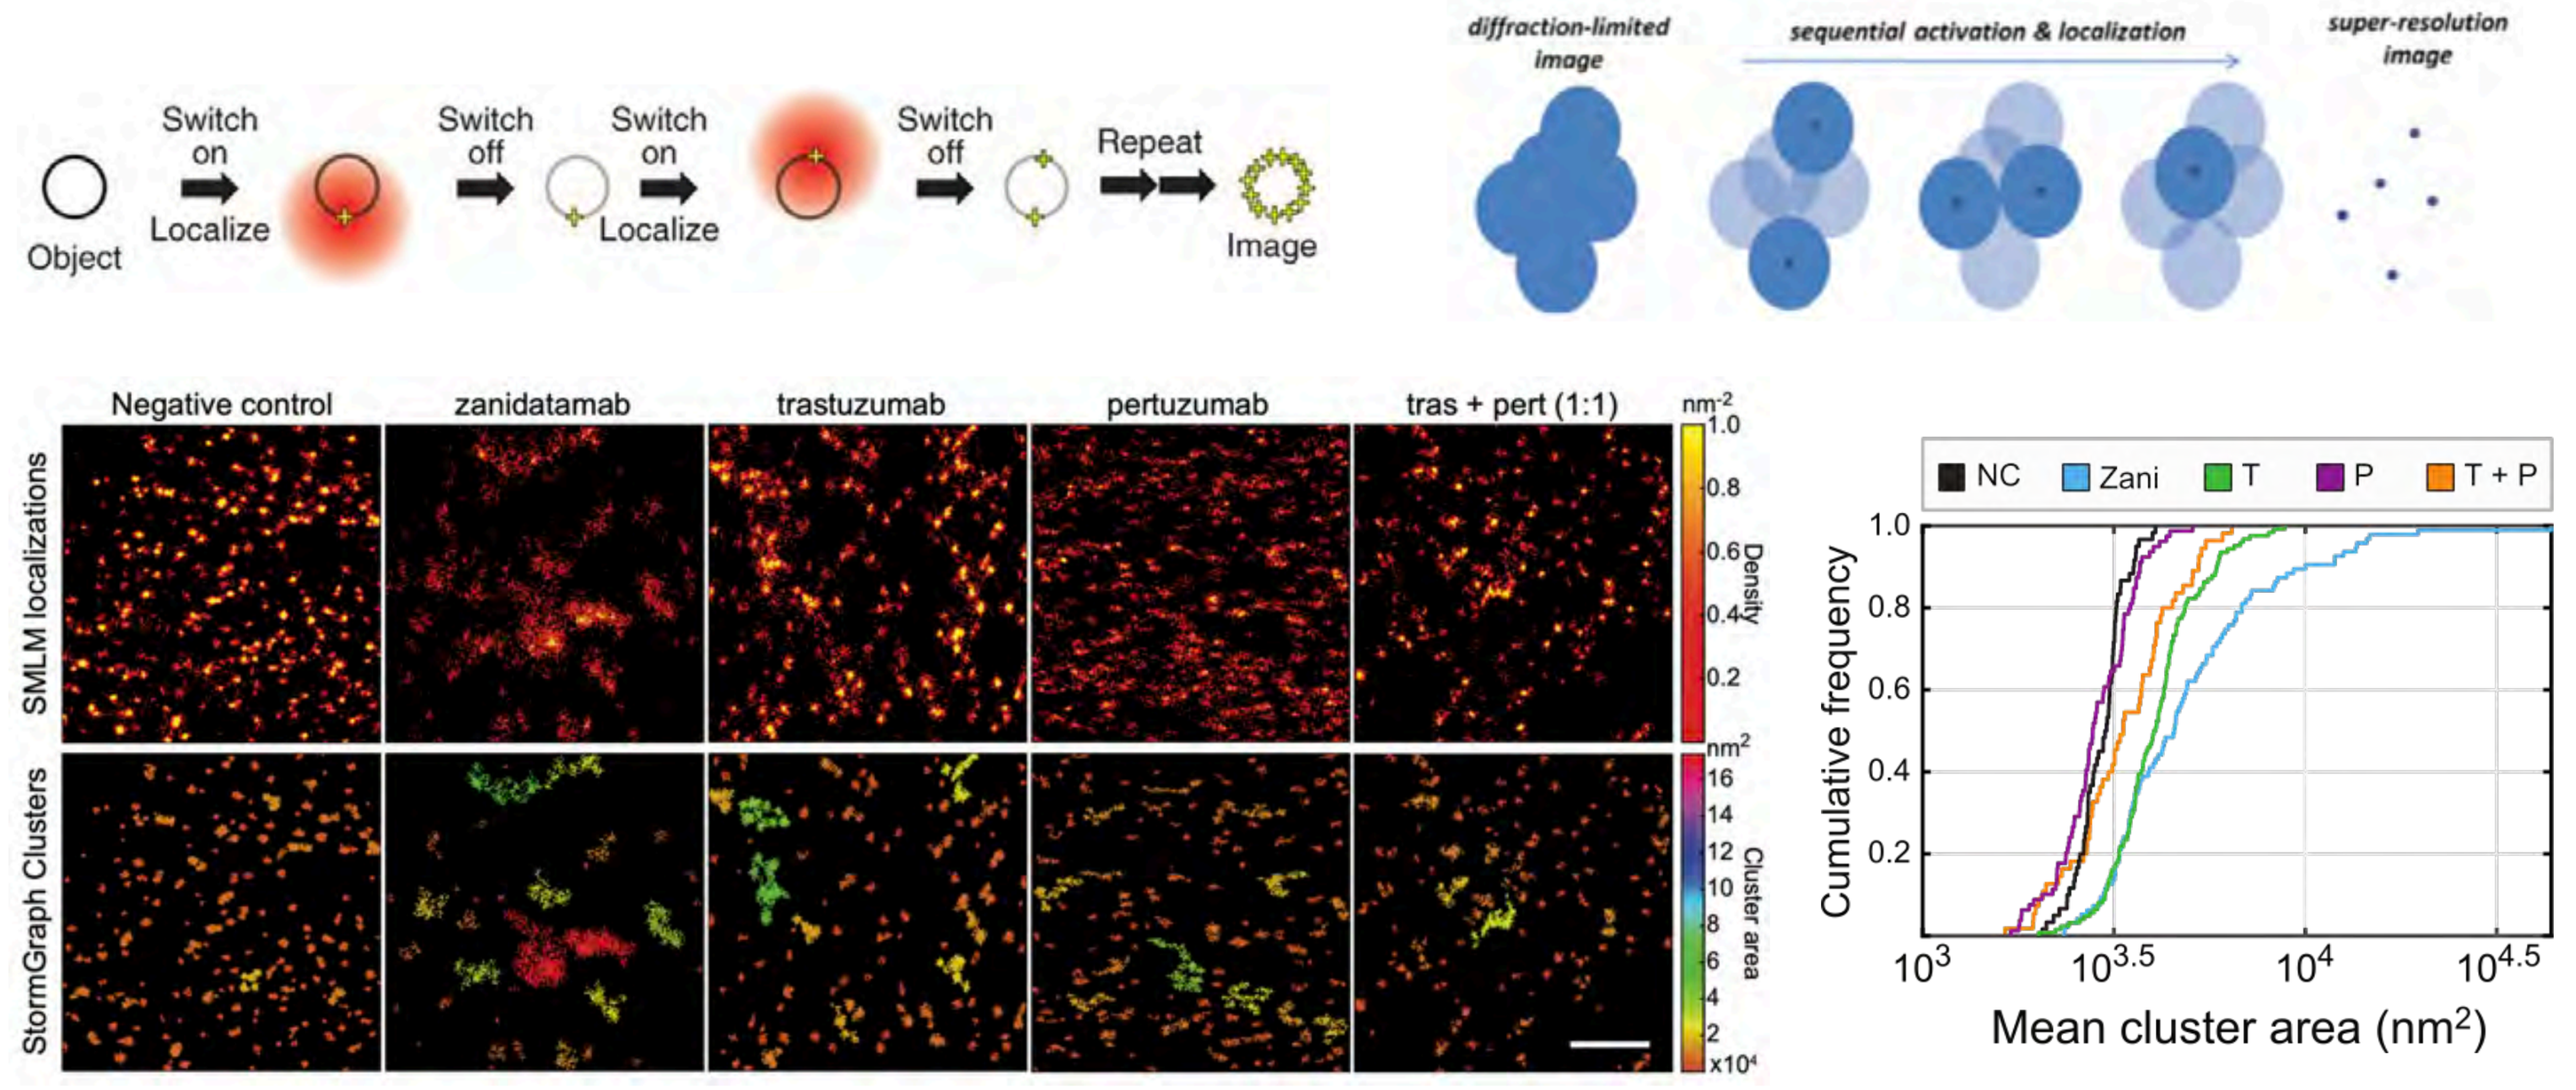
Object
Switch on
Localize
Switch off
Repeat
Image
diffraction-limited image
sequential activation & localization
super-resolution image
Negative control
zanidatamab
trastuzumab
pertuzumab
tras + pert (1:1)
SMLM localizations
StormGraph Clusters
Density
Cluster area
NC
Zani
T
P
T + P
Cumulative frequency
Mean cluster area (nm²)

We used single molecule localization microscopy (SMLM) with a real-time drift-stabilized direct Stochastic Optical Reconstruction Microscope (dSTORM) to resolve the nature of HER2 nanocluster on the cell surface. Single molecule localizations showed that HER2 mainly exists as small nanoclusters in cells treated with NC antibody. In zani treated cells HER2 molecules coalesce to form large spatially-separated clusters. This is confirmed by clustering and mean area analysis of images from multiple regions of interest. Though cells stimulated with tras, pert and tras+pert displayed varying degrees of HER2 organization, large nanoclusters observed with zani were absent in cells treated with these agents.

Conclusion

- The biparatopic design of zanidatamab results in a unique networked complex geometry upon binding to HER2.
- Structural modeling of zanidatamab with HER2 supports its ability to form complexes with distinct higher order geometry on target cell surface, potentially promoting novel function such as CDC (Complement Dependent Cytotoxicity) that is not observed with either tras, pert or their combination (See Abstract number 1005).
- High resolution imaging of HER2 organization on cells supports the structural modeling studies, confirming the presence of unique large cluster formations not seen with tras, pert or their combination.
- The observed differences in the geometry, size and persistence of these coalesced cell surface receptor clusters likely contribute to the differentiated activity of zanidatamab.

This study was sponsored by Zymeworks.

AACR Annual Meeting 2021